Poster P3082

Health Care Resource Utilization Decreases Among Patients With Constipation-Predominant Irritable Bowel Syndrome Following Initiation of Tenapanor: A Real-World Analysis

Kyle Staller,[1] Alan Fossa,[2] Megan O'Byrne,[2] Luisa L. Scott[3]

[1]Massachusetts General Hospital, Boston, MA, USA; [2]Arbor Research Collaborative for Health, Ann Arbor, MI, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Background

- Irritable bowel syndrome with constipation (IBS-C) is a chronic disorder of gut-brain interaction[1] associated with high health care resource utilization (HCRU):
 - A retrospective United States health care claims cohort analysis from over 100,000 individuals with IBS found that patients with IBS-C have higher annual all-cause and IBS-specific spending compared with patients with other subtypes of IBS.[2]
 - The most common costs were attributable to emergency department visits, hospitalizations, and radiological testing.[2]
- Reduction in gastrointestinal (GI) HCRU is likely an indicator of treatment efficacy and improved GI symptoms.
- This study explored GI-HCRU outcomes before and after tenapanor initiation and treatment discontinuation by patient characteristics that may affect response to treatment.

Methods

Patients

- Electronic health record (EHR) data were obtained from a national gastroenterology practice group that operates in 7 states with 150 locations.
- Eligible patients had a diagnosis of IBS-C, initiated tenapanor with one year of follow-up before and after initiation, had at least 1 GI encounter in the year prior to tenapanor initiation, and had complete patient characteristics data.

Key Definitions

- Pre-tenapanor period: 1 year prior to tenapanor initiation (including the initiation date).
- Post-tenapanor period: 1 year after tenapanor initiation.
- Treatment discontinuation: 30 consecutive days off tenapanor.
- Event dates (survival curves): date of the first encounter a patient was indicated as off tenapanor.

Statistical Analyses

- Descriptive statistics were generated for total HCRU and within-patient change in HCRU before versus after tenapanor initiation.
- A paired Wilcoxon signed-rank test compared pre- versus post-tenapanor HCRU.
- A Kaplan-Meier approach was used to construct survival curves and calculate cumulative incidence of treatment discontinuation at 1 year.
- All analyses were stratified by IBS-C treatment use before tenapanor initiation, body mass index (BMI), and Charlson Comorbidity Index (CCI) scores.

Results

Patients

- The study included 470 tenapanor users (**Table 1** and **Table 2**).

Table 1. Characteristics of Eligible Patients

Variable	Patients (N=470)
Mean (SD) age at treatment initiation, y	56.6 (16.6)
Female	404 (86.0)
Race/ethnicity	
Asian	5 (1.1)
Black or African American	38 (8.1)
Declined/unspecified/other	143 (30.4)
Hispanic	25 (5.3)
Multiple Races	50 (10.6)
White	209 (44.5)
BMI, kg/m²	
<30	349 (74.3)
≥30	121 (25.7)
On Medicare[a]	206 (44.1)
On Medicaid[b]	2 (0.4)
CCI, mean (SD)	0.8 (1.6)
Psychiatric comorbidities present[c]	92 (19.6)
Somatic comorbidities present[d]	54 (11.5)
≥1 prior IBS-C medications	330 (70.2)

Units are n (%) unless indicated otherwise.
[a]206 patients were on Medicare, 261 were not; Medicare status data were missing for 3 patients. [b]2 patients were on Medicaid, 465 were not; Medicaid status data were missing for 3 patients. [c]Psychiatric comorbidities include anxiety, depression, bipolar disorder, and post-traumatic stress disorder. [d]Somatic comorbidities include chronic pain, fibromyalgia, chronic fatigue syndrome, chronic back pain, migraines, chronic pelvic pain, and temporomandibular joint disorder.
BMI, body mass index; CCI, Charlson Comorbidity Index; IBS-C, irritable bowel syndrome with constipation; y, year

GI-HCRU

- Total GI-HCRU decreased after tenapanor initiation compared with the year prior, driven mainly by a reduction in labs and regular visits (**Figure 1**).
- Median (interquartile range [IQR]) within-patient HCRU was 4.5 (2.0, 9.0) encounters per year before tenapanor treatment and 3.0 (1.0, 6.0) encounters per year after (paired Wilcoxon signed-rank test $P<0.001$).

Figure 1: Total HCRU Encounters Before and After Tenapanor Treatment by Labs, Regular Visits, Procedures, and Imaging

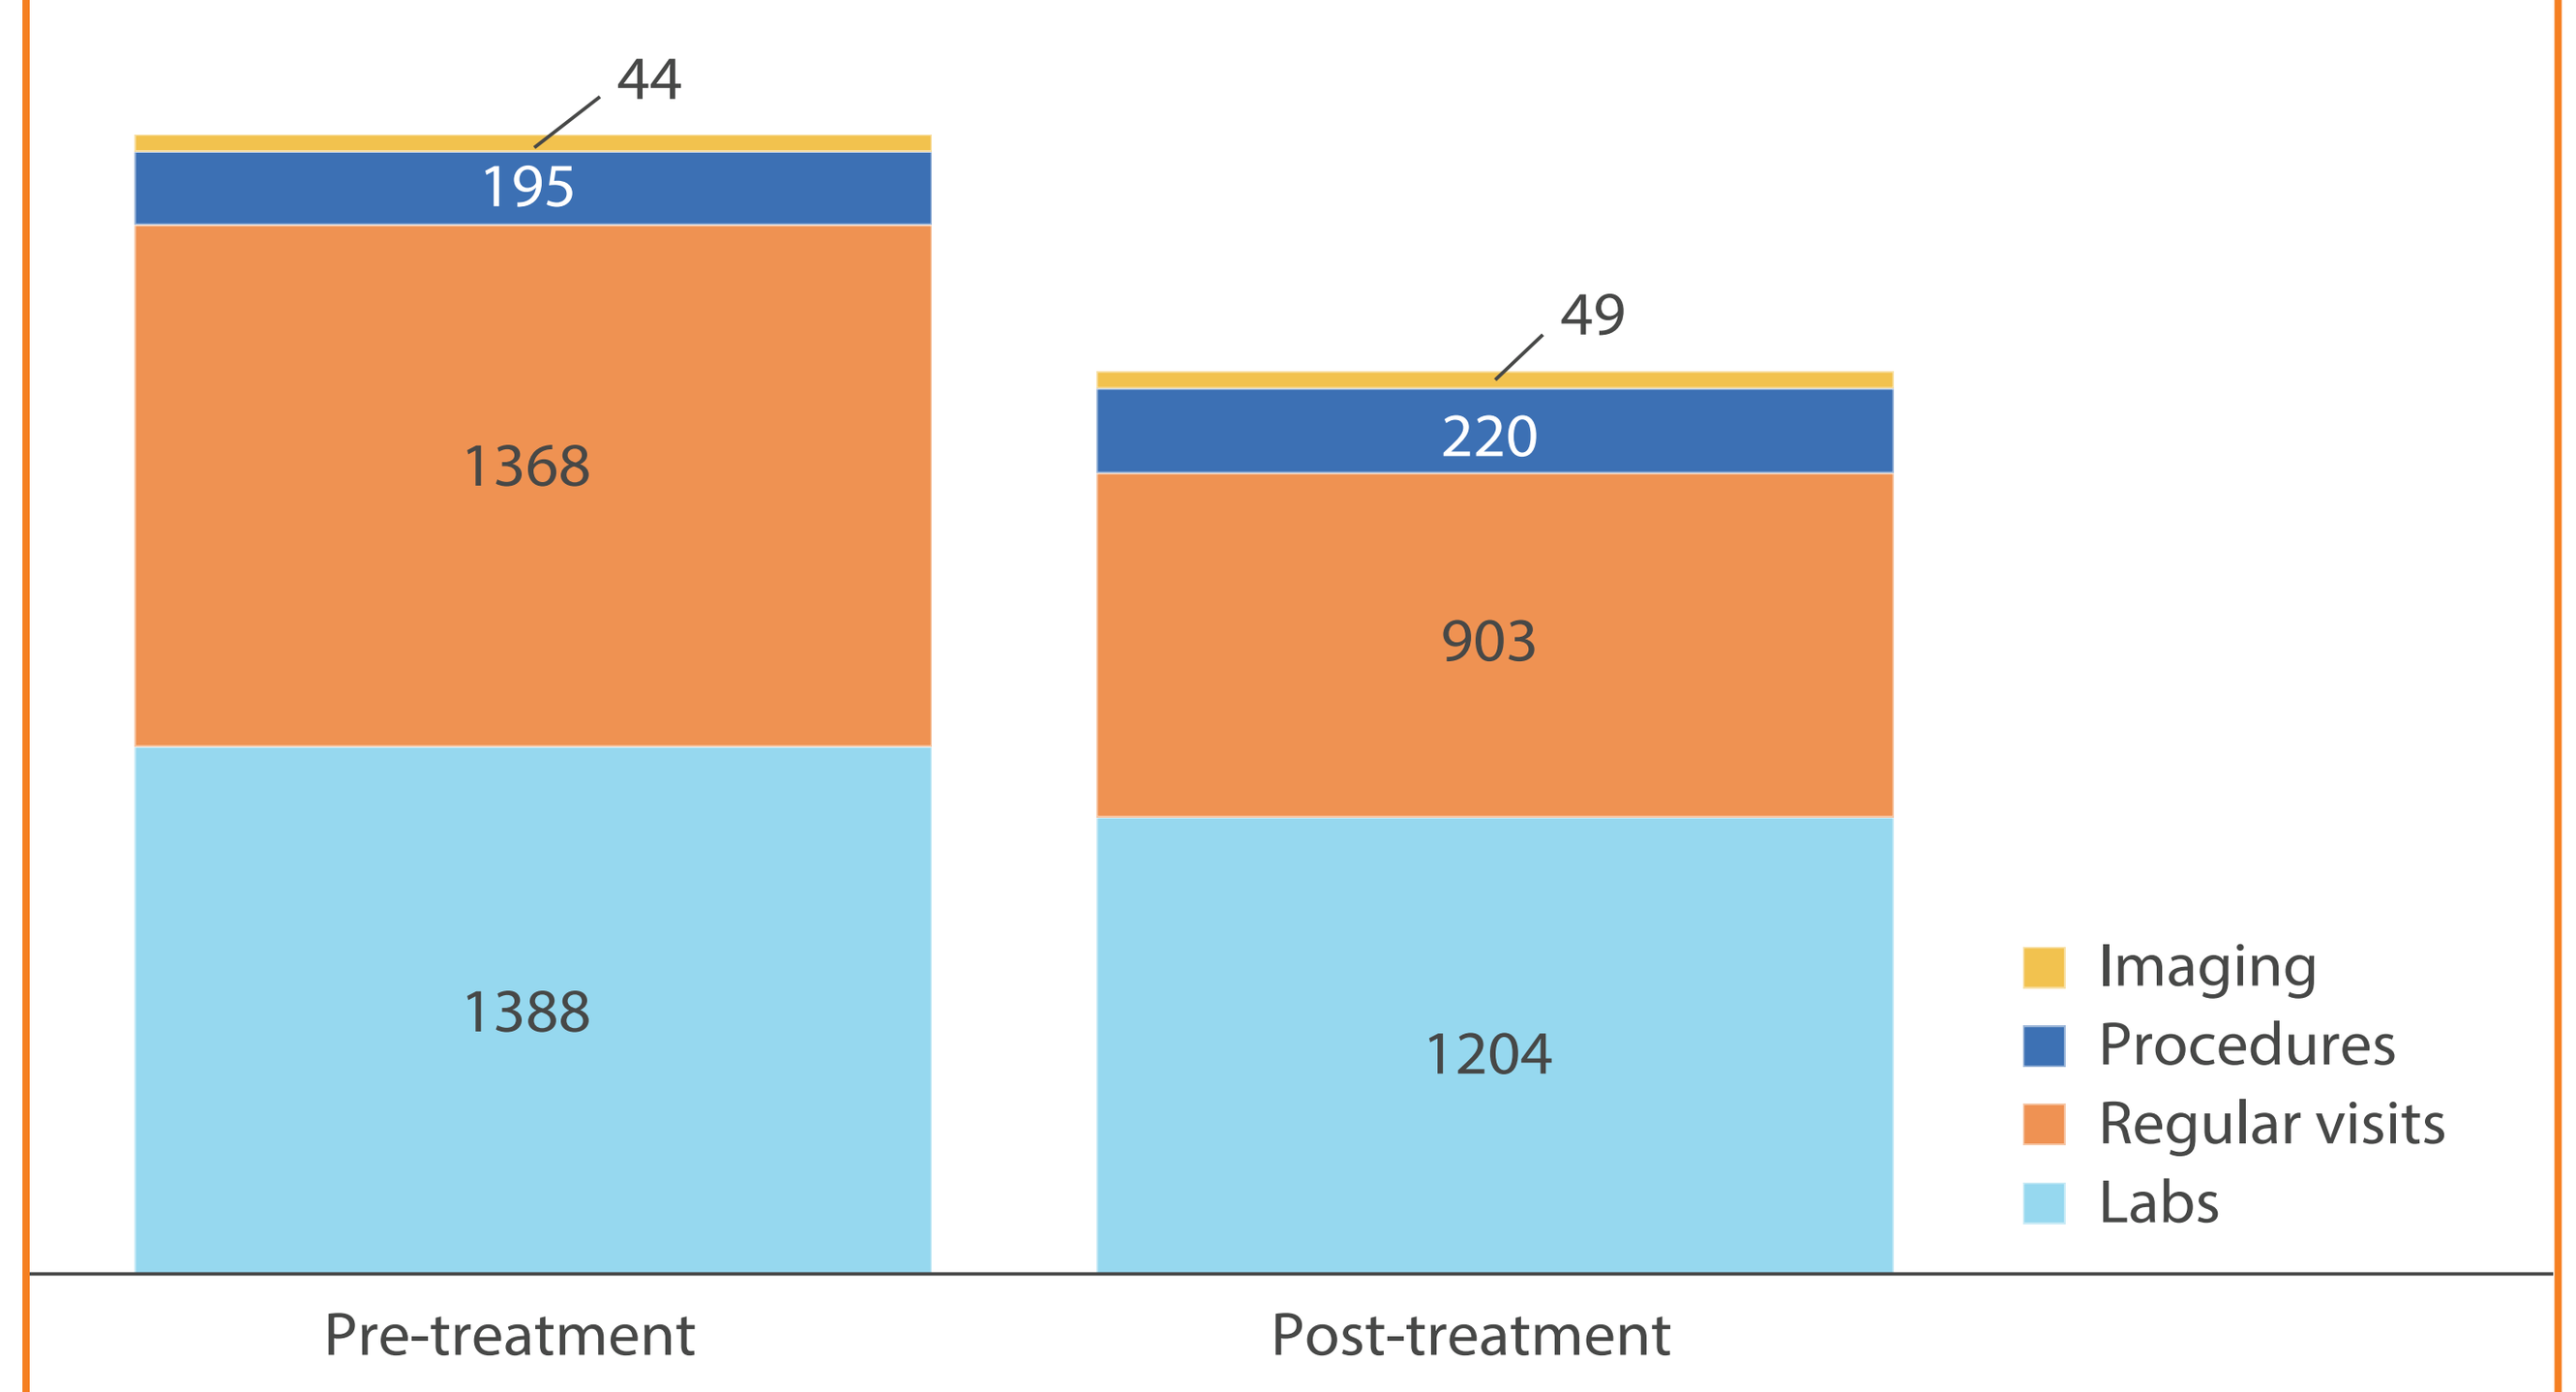
44
195
1368
1388
49
220
903
1204
Imaging
Procedures
Regular visits
Labs
Pre-treatment
Post-treatment

Pre-treatment period: 365 days prior to tenapanor initiation (inclusive of initiation date). Post-treatment period: 365 days after initiation. HCRU, health care resource utilization.

- GI-HCRU was reduced in patients who had used ≥1 IBS-C medication in the previous year (**Figure 2A**).
 - The previously treated subgroup had a baseline median HCRU of 5.0 (IQR: 3.0, 9.0) encounters per year and a median change of −2.0 [−5.0, 1.0] encounters per year following tenapanor initiation ($P<0.001$).
- HCRU was lower after tenapanor initiation regardless of whether patients had a lower or higher BMI (**Figure 2B**) or CCI score (**Figure 2C**).

Figure 2: HCRU Stratified by Pre- and Post-Treatment Period and (A) Number of Medications Used Before Treatment (0 or ≥1), (B) BMI Group (BMI <30 or ≥30), and (C) CCI Group (CCI Score <2 or ≥2)

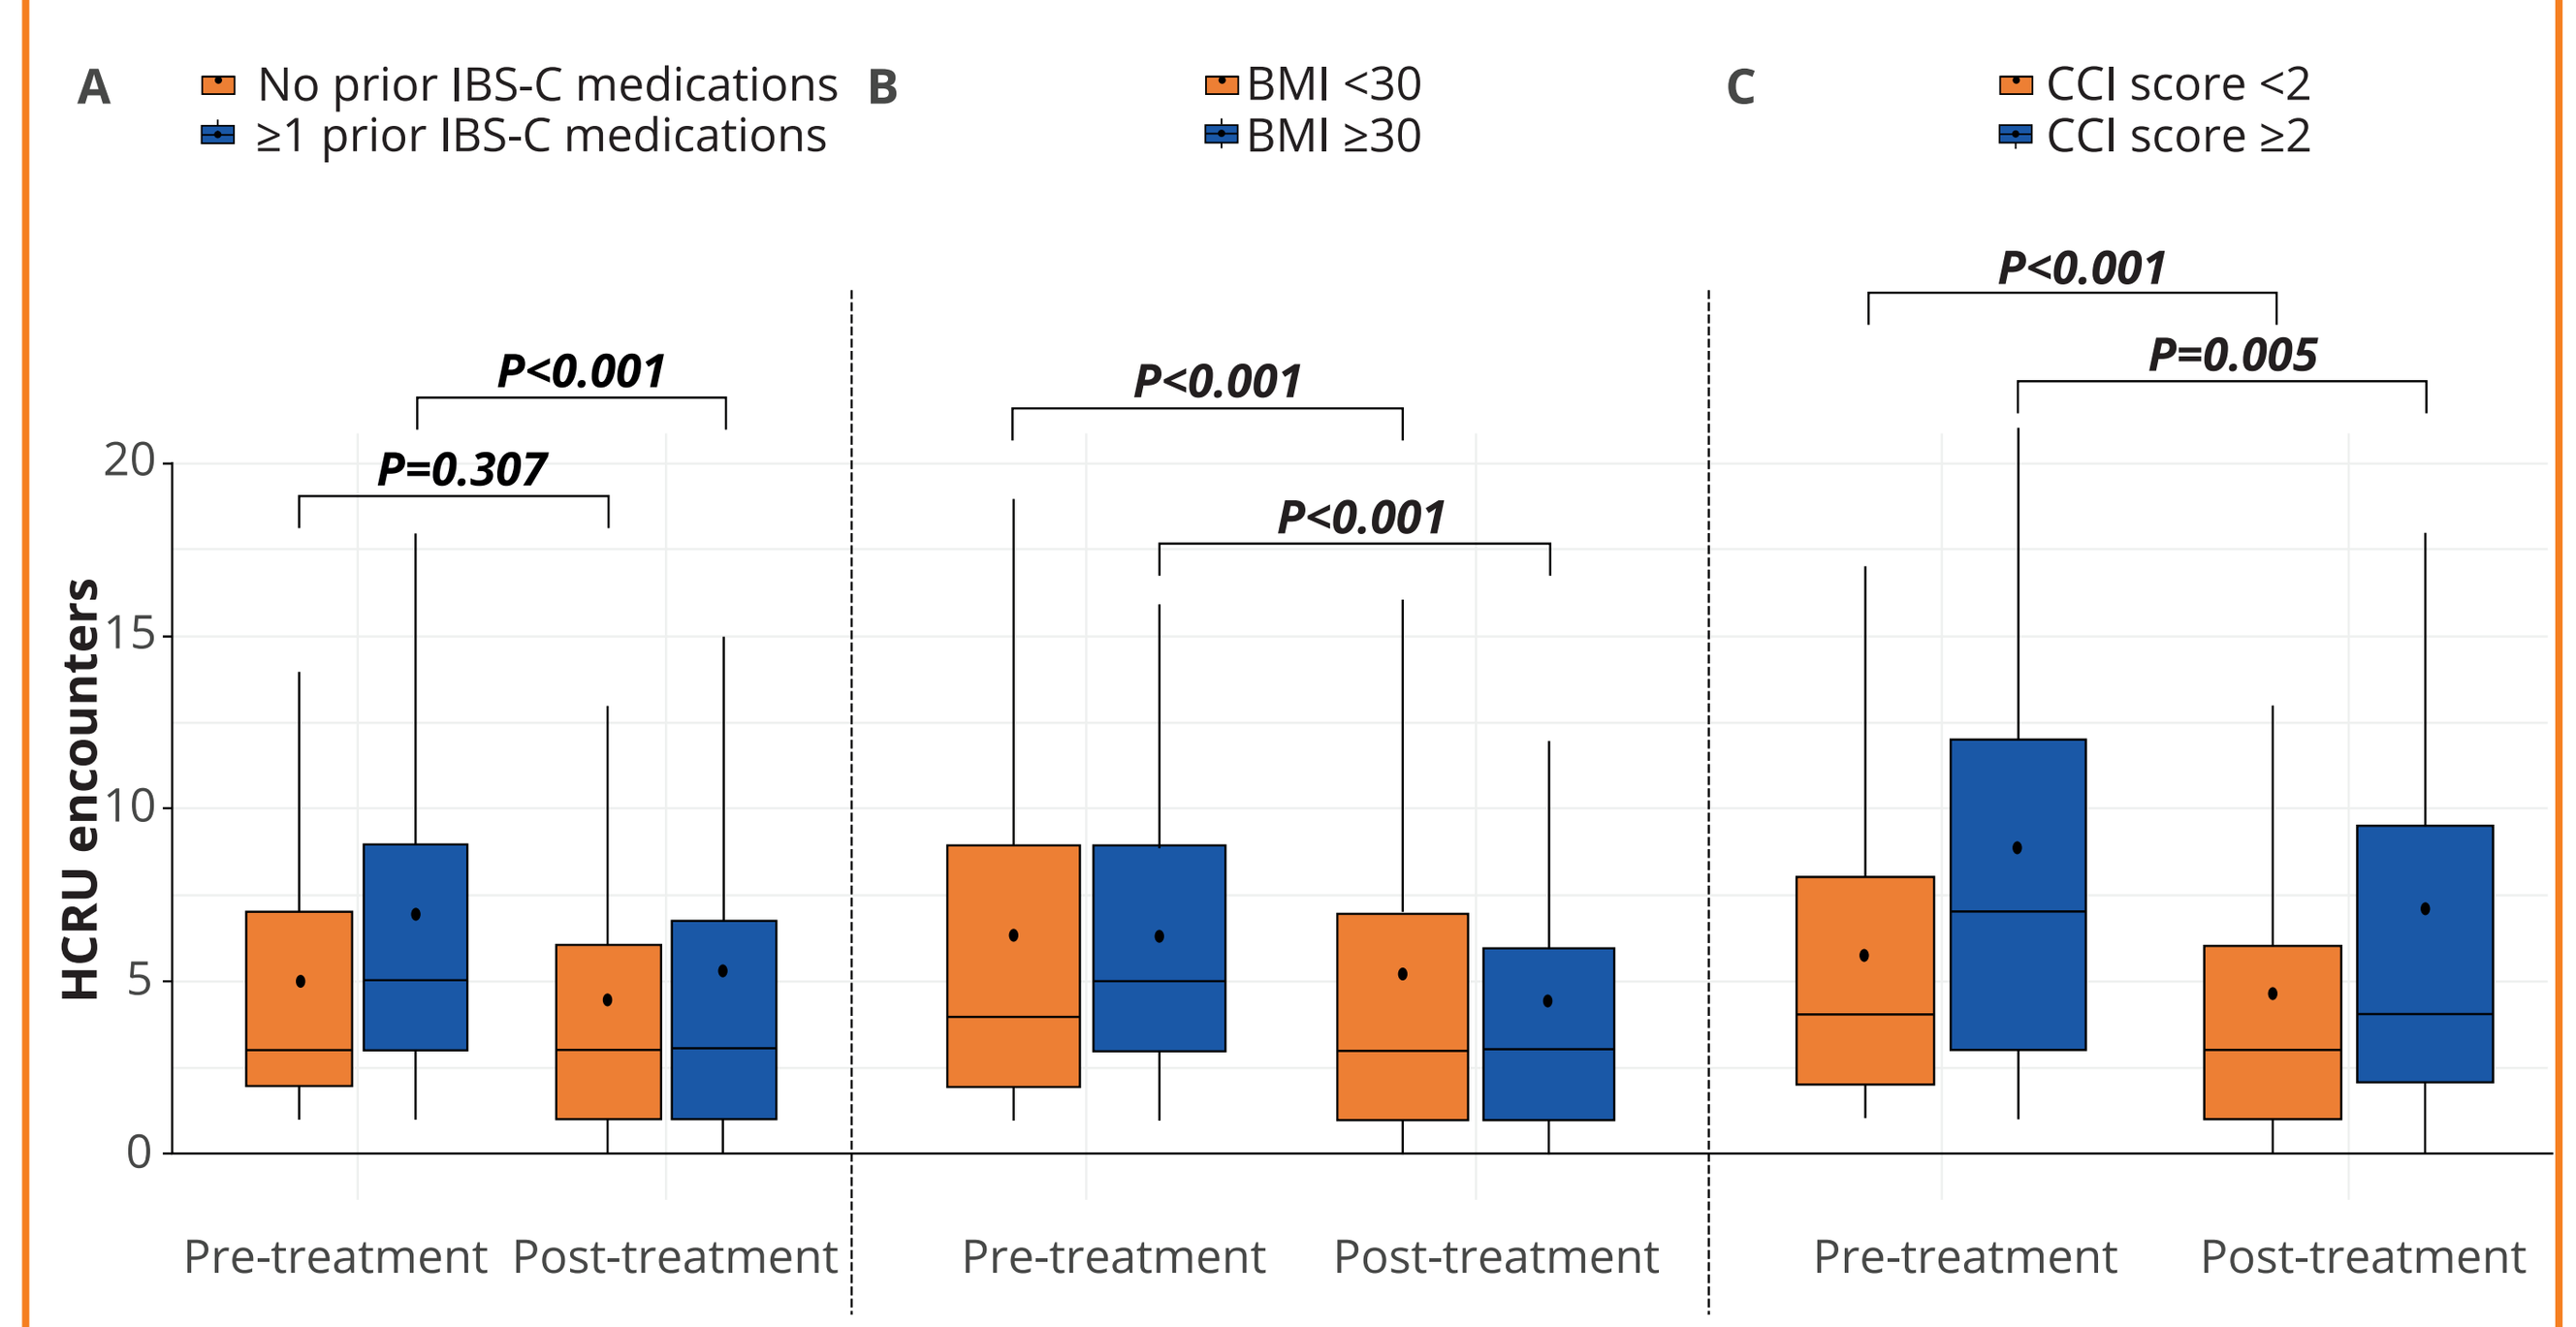
A
No prior IBS-C medications
≥1 prior IBS-C medications
B
BMI <30
BMI ≥30
C
CCI score <2
CCI score ≥2
HCRU encounters
0
5
10
15
20
P<0.001
P=0.307
P<0.001
P<0.001
P<0.001
P=0.005
Pre-treatment
Post-treatment
Pre-treatment
Post-treatment
Pre-treatment
Post-treatment

P-values are for pre- versus post-treatment comparisons of median HCRU. Black circles indicate mean values. Pre-treatment period: 365 days prior to tenapanor initiation (inclusive of initiation date). Post-treatment period: 365 days after initiation. BMI, body mass index; CCI, Charlson Comorbidity Index; HCRU, health care resource utilization; IBS-C, irritable bowel syndrome with constipation.

Tenapanor Discontinuation

- The 1-year cumulative incidence of tenapanor discontinuation overall was 23% (95% CI, 19%-26%).
- Cumulative incidence of tenapanor discontinuation at 1 year was similar across all patient groups, ranging from 19% (13%-25%) in patients with a BMI ≥30 to 27% (21%-33%) for patients who had not previously used IBS-C medications (**Figure 3A-C**).

Figure 3: Tenapanor Discontinuation by (A) Number of Medications Used Before Treatment (0 or ≥1), (B) BMI Group (BMI <30 or ≥30), and (C) CCI Group (CCI Score <2 or ≥2)

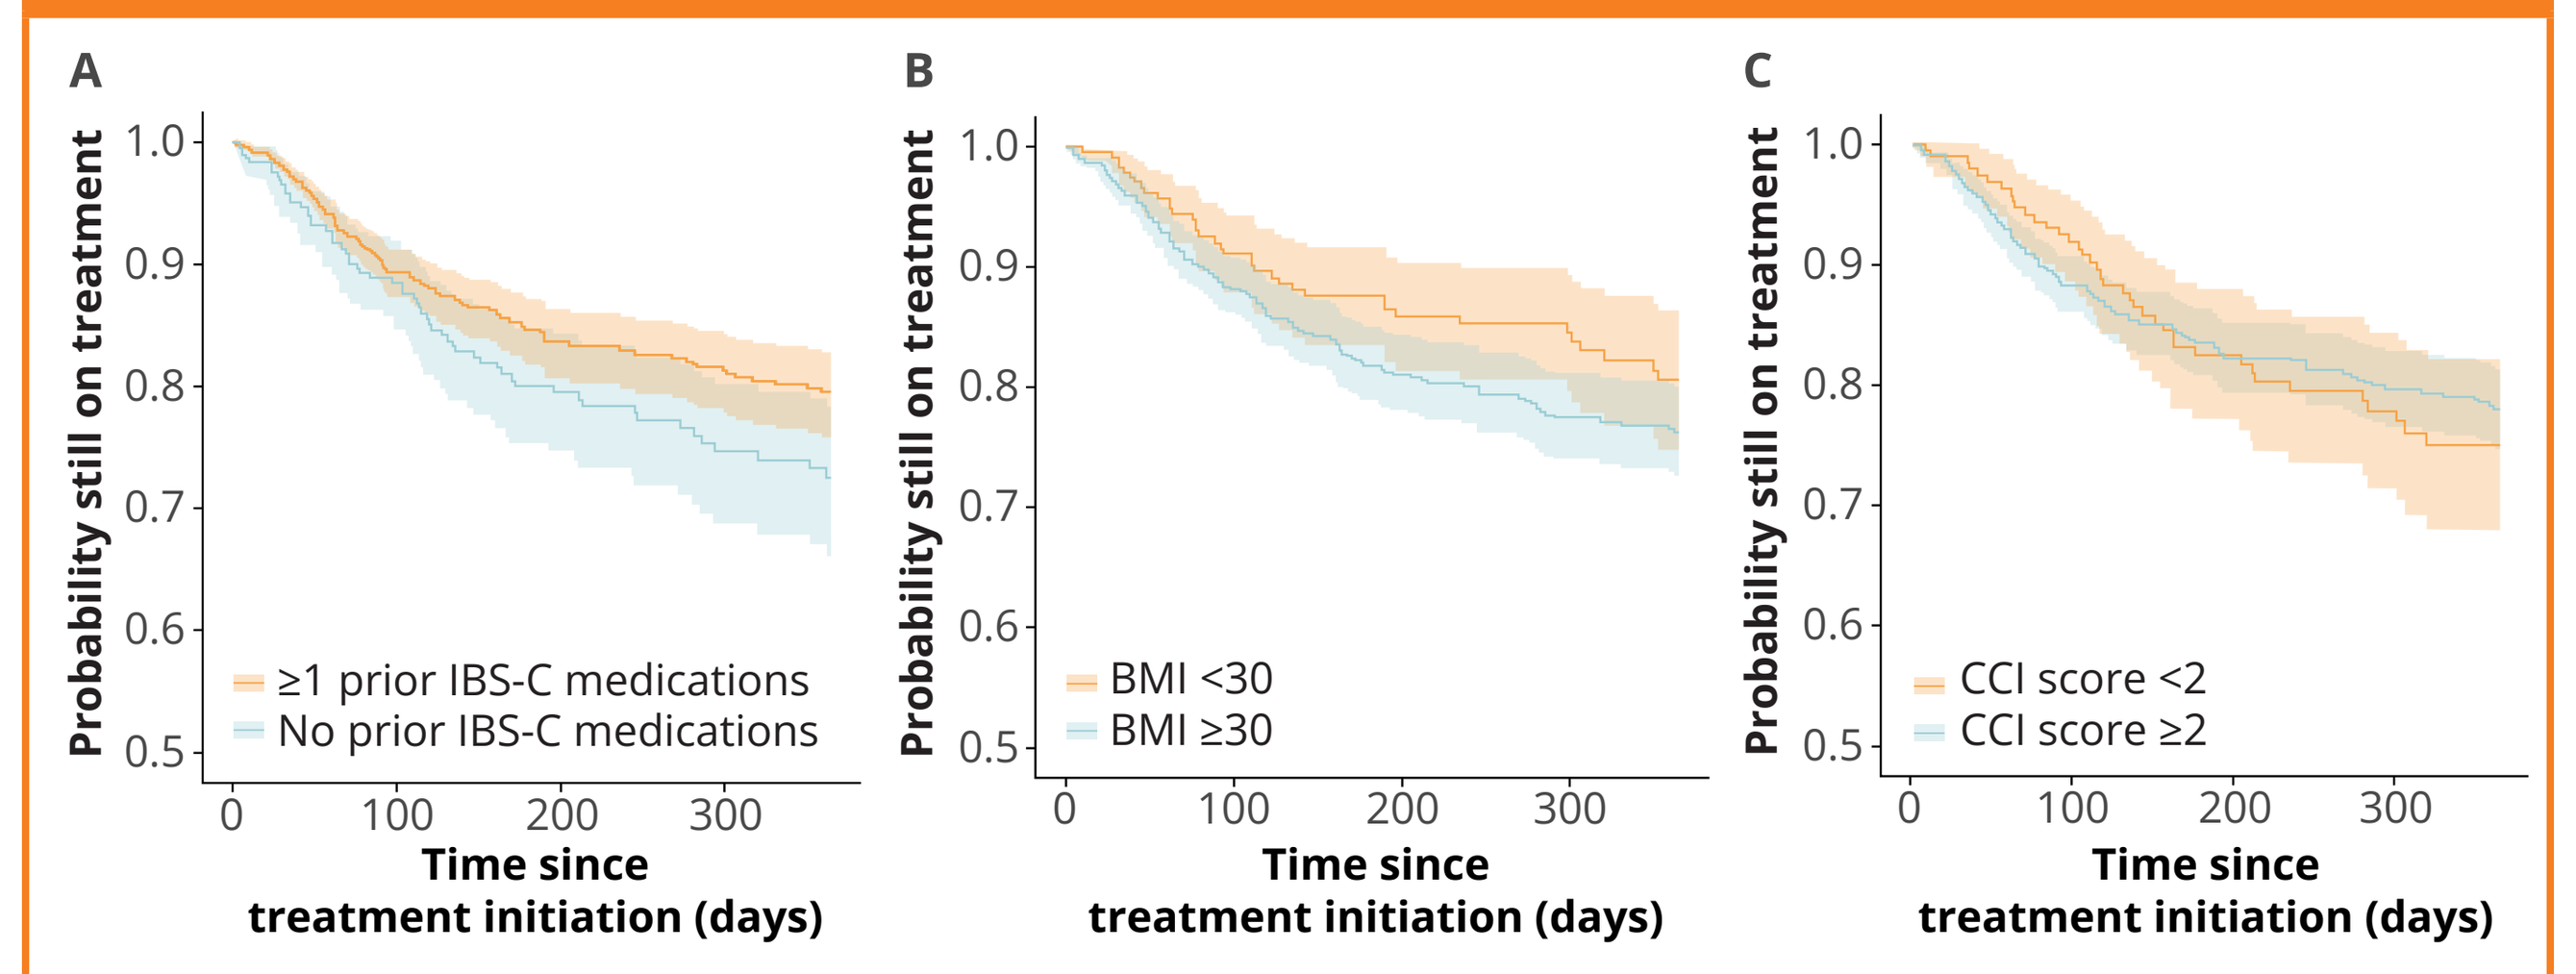
A
B
C
Probability still on treatment
1.0
0.9
0.8
0.7
0.6
0.5
≥1 prior IBS-C medications
No prior IBS-C medications
BMI <30
BMI ≥30
CCI score <2
CCI score ≥2
0
100
200
300
Time since
treatment initiation (days)

BMI, body mass index; CCI, Charlson Comorbidity Index.

Table 2: Patient Characteristics by Stratification Factors: IBS-C Treatment Use Before Tenapanor Initiation, BMI, and CCI scores

Variable	Number of prior IBS-C medications		BMI		CCI score	
	0 (n=140)	≥1 (n=330)	<30 (n=349)	≥30 (n=121)	<2 (n=375)	≥2 (n=95)
Mean age (SD), y	57.2 (18.2)	56.3 (15.9)	57 (17.1)	55.4 (14.9)	55.7 (17.2)	59.7 (13.4)
Female gender	118 (84.3)	286 (86.7)	299 (85.7)	105 (86.8)	325 (86.7)	79 (83.2)
Race/ethnicity						
Asian	1 (0.7)	4 (1.2)	4 (1.1)	1 (0.8)	4 (1.1)	1 (1.1)
Black or African American	4 (2.9)	34 (10.3)	18 (5.2)	20 (16.5)	27 (7.2)	11 (11.6)
Declined/ unspecified/ other	39 (27.9)	104 (31.5)	103 (29.5)	40 (33.1)	120 (32.0)	23 (24.2)
Hispanic	9 (6.4)	16 (4.8)	21 (6.0)	4 (3.3)	19 (5.1)	6 (6.3)
Multiple races	15 (10.7)	35 (10.6)	39 (11.2)	11 (9.1)	32 (8.5)	18 (18.9)
White	72 (51.4)	137 (41.5)	164 (47.0)	45 (37.2)	173 (46.1)	36 (37.9)
BMI <30 kg/m²	117 (83.6)	232 (70.3)	–	–	284 (75.7)	65 (68.4)
Medicare[a]	67 (48.2)	139 (42.4)	160 (46.1)	46 (38.3)	158 (42.5)	48 (50.5)
Not Medicaid[a]	138 (99.3)	327 (99.7)	345 (99.4)	120 (100.0)	371 (99.7)	94 (89.9)
CCI, mean (SD)	0.8 (1.7)	0.8 (1.5)	0.8 (1.6)	1 (1.5)	–	–
No psychiatric comorbidities[b]	114 (81.4)	264 (80.0)	280 (80.2)	98 (81.0)	307 (81.9)	71 (74.7)
No somatic comorbidities[c]	123 (87.9)	293 (88.8)	307 (88.0)	109 (90.1)	340 (90.7)	76 (80.0)
Number of IBS-C medications used before tenapanor treatment initiation						
0	–	–	117 (33.5)	23 (19.0)	115 (30.7)	25 (26.3)
≥1	–	–	232 (66.5)	98 (81.0)	260 (69.3)	70 (73.7)

Units are n (%) unless indicated otherwise.
[a]Descriptive statistics were calculated using non-missing values. N's may not sum to column totals due to missing data.
[b]Psychiatric comorbidities include anxiety, depression, bipolar disorder, and post-traumatic stress disorder.
[c]Somatic comorbidities include chronic pain, fibromyalgia, chronic fatigue syndrome, chronic back pain, migraines, chronic pelvic pain, and temporomandibular joint disorder.
BMI, body mass index; CCI, Charlson Comorbidity Index; IBS-C, irritable bowel syndrome with constipation.

Limitations

- The analysis lacked a comparator group (ie, patients who initiated other IBS-C medications). As such, within-patient changes in HCRU and treatment discontinuation rates associated with tenapanor cannot be appraised in the wider context of available treatments for IBS-C.
- It is not possible to determine with certainty that the initiation of tenapanor was solely responsible for any observed changes in HCRU in any given group, as the findings may have been influenced by additional confounding factors.
- Treatment discontinuation data were derived from provider documentation (within the gastroenterology EHR) rather than pharmacy dispensing data; therefore, treatment discontinuation may represent ongoing provider management of therapy rather than true medication persistence.
- Reductions in HCRU may be interpreted as reflecting improvements in patients' clinical status or well-being, but HCRU is a surrogate outcome; thus, this relationship cannot be assumed in the absence of direct patient-reported or clinical outcome measures.

Conclusions

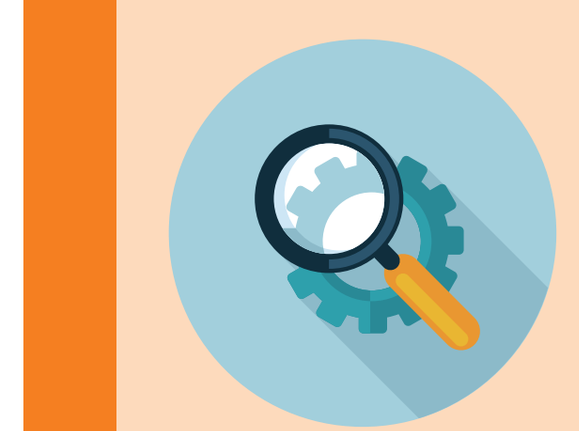

Tenapanor was associated with decreased GI-HCRU, even in patients who are typically more complex, like those who have tried IBS-C medications, patients with a BMI ≥30, or those who have chronic morbidities.

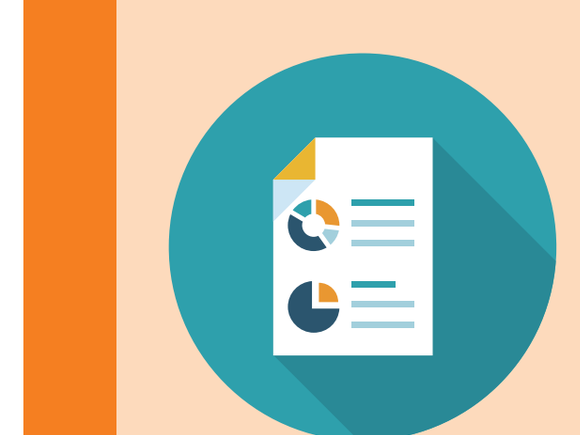

Real-world persistence with tenapanor was favorable, with more than three-quarters of patients continuing treatment through 1 year. Persistence was maintained regardless of previous IBS-C treatment experience, BMI, or chronic comorbidities.

Disclosures

Kyle Staller has received research funding from Ardelyx, has served as a consultant for AbbVie, Ardelyx, Atmo, Ferring, Gemelli Biotech, Laborie, Salix, and Takeda, and served on the advisory board of Mindset Health. Alan Fossa and Megan O'Byrne are employees of Arbor Research Collaborative for Health, which received funding for this study from Ardelyx, Inc. All funding was provided to Arbor Research Collaborative for Health and not directly to Alan Fossa or Megan O'Byrne. Luisa L. Scott is an employee of Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Contact Information

Kyle Staller can be contacted for further information on this study at KSTALLER@mgh.harvard.edu

References

1. Lacy BE et al. *Am J Gastroenterol*. 2021;116:17-44.
2. Shin A, Xu H. *Am J Gastroenterol*. 2024;119:1571-9.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American College of Gastroenterology (ACG) Conference; October 9-14, 2026; Nashville, TN, USA; In Person and Virtual.

MED-US–2600360 10/2026

IBSRELA® (tenapanor) is indicated for treatment of irritable bowel syndrome with constipation (IBS-C) in adults

Important Safety Information

WARNING: RISK OF SERIOUS DEHYDRATION IN PEDIATRIC PATIENTS

- **IBSRELA is contraindicated in patients less than 6 years of age; in nonclinical studies in young juvenile rats, administration of tenapanor caused deaths presumed to be due to dehydration. *[see PI Contraindications (4), Use in Specific Populations (8.4)]*.**
- **Avoid use of IBSRELA in patients 6 years to less than 12 years of age. *[see PI Warnings and Precautions (5.1), Use in Specific Populations (8.4)]*.**
- **The safety and effectiveness of IBSRELA have not been established in pediatric patients less than 18 years of age. *[see PI Use in Specific Populations (8.4)]*.**

CONTRAINDICATIONS

IBSRELA is contraindicated in patients less than 6 years of age due to the risk of serious dehydration.

IBSRELA is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Risk of Serious Dehydration in Pediatric Patients

IBSRELA is contraindicated in patients below 6 years of age. The safety and effectiveness of IBSRELA in patients less than 18 years of age have not been established. In young juvenile rats (less than 1 week old; approximate human age equivalent of less than 2 years of age), decreased body weight and deaths occurred, presumed to be due to dehydration, following oral administration of tenapanor. There are no data available in older juvenile rats (human age equivalent 2 years to less than 12 years).

Avoid the use of IBSRELA in patients 6 years to less than 12 years of age. Although there are no data in older juvenile rats, given the deaths in younger rats and the lack of clinical safety and efficacy data in pediatric patients, avoid the use of IBSRELA in patients 6 years to less than 12 years of age.

Diarrhea

Diarrhea was the most common adverse reaction in two randomized, double-blind, placebo-controlled trials of IBS-C. Severe diarrhea was reported in 2.5% of IBSRELA-treated patients. If severe diarrhea occurs, suspend dosing and rehydrate patient.

MOST COMMON ADVERSE REACTIONS

The most common adverse reactions in IBSRELA-treated patients (incidence ≥2% and greater than placebo) were: diarrhea (16% vs 4% placebo), abdominal distension (3% vs <1%), flatulence (3% vs 1%) and dizziness (2% vs <1%).

For additional safety information, including the Boxed Warning, please see full Prescribing Information: click here.

IBS-C, irritable bowel syndrome with constipation.

IBSRELA (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.
Available at: https://www.ibsrela-hcp.com/PI

© Ardelyx, Inc., 2026. All rights reserved. Ardelyx and IBSRELA are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only