Poster P5327

High Symptom Severity Burden Is Associated With Reduced Quality of Life in United States Adults With Irritable Bowel Syndrome With Constipation

Megan Riehl,[1] Taya Westfield,[2] Alan Fossa,[2] Zachary Babcock,[3] Johannah Ruddy,[3] Luisa L. Scott[3]

[1]University of Michigan, Ann Arbor, MI, USA; [2]Arbor Research Collaborative for Health, Ann Arbor, MI, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Background

- IBS with constipation (IBS-C) is associated with considerable disease burden and reduced HRQoL.[1] However, the relationship among symptom severity, treatment satisfaction, and quality of life in adults remains incompletely characterized.
- This online survey aims to further describe the burden of IBS-C in United States (US) adults and examine associations between key patient-reported outcomes (HRQoL and role functioning), symptom severity, treatment satisfaction, patient characteristics, and social determinants of health.

Methods

- Eligible patients completed a Health Union IBS-C survey (October 21-November 26, 2025) and self-reported an IBS diagnosis with constipation as their primary bowel symptom.

IBS Symptom Severity

- Symptom severity was measured using the IBS Severity Scoring System (IBS-SSS).
 - IBS-SSS scores range from 0-500, with higher scores indicating greater severity.
 - Patient scores were analyzed both as continuous numeric scores and by severity category.

Current Treatment Satisfaction

- Patients reported current or past 6-month use of prescription medications, over-the-counter medications, and other forms of IBS treatment.
- Patients rated their satisfaction with their current IBS-C treatment using a 7-point Likert scale ranging from "Extremely satisfied" to "Extremely dissatisfied," with an option to select "Have never used any treatment."

HRQoL

- Mental and physical aspects of HRQoL were evaluated using 4 continuous 36-Item Short Form Health Survey (SF-36) scales/subscales: Mental Component Summary (MCS), Physical Component Summary (PCS) scores, and role limitations due to physical health problems (RP) and emotional problems (RE).
 - These scales have been used in patients with gastrointestinal (GI) disorders, including irritable bowel syndrome (IBS).[2-4]
 - Possible scores for each of these SF-36 scales/subscales range from 0-100, with higher scores indicating fewer health-related limitations.

Analyses

- Descriptive statistics were used to summarize patient characteristics, treatment use and satisfaction, IBS symptom severity, and HRQoL.
- Associations between MCS and PCS and predictors of interest (treatment satisfaction and IBS symptom severity) were examined using linear regression.
- Associations between RP scores and RE scores and predictors of interest (treatment satisfaction and symptom severity) were examined using logistic regression modeling with a score of <100 as the outcome of interest.
 - All regression models were adjusted for demographic, socioeconomic, community, and comorbidity characteristics.
- Associations between continuous IBS-SSS and patient characteristics were examined using a multivariable linear regression model adjusted for covariates similar to those included in the HRQoL models.

Results

Patients (N=374)

- More than half of respondents (55%) were diagnosed 1-5 years ago, 35% were diagnosed more than 5 years ago, and 10% were diagnosed within the past year. Respondents most commonly saw gastroenterologists (49%) and primary care physicians (38%) for IBS management.
- Three-quarters of respondents (75%) had ≥1 comorbid condition (**Table 1**), including mental health (54%), chronic pain/musculoskeletal (40%), and other GI conditions (33%); 48% rated their overall health as good.

IBS Symptom Severity

- Seventy-three percent reported moderate to severe IBS-C symptoms (mean IBS-SSS score: 240.9 [SD=105.1]; **Figure 1**).
- In a multivariable linear regression model adjusting for patient characteristics, IBS-SSS scores were, on average, 35 points higher (more severe) for unemployed versus employed respondents, and 35 and 33 points higher among respondents living in urban or rural areas (compared with suburban areas), respectively (**Figure 2**).

Figure 1: Distributions of IBS-SSS Scores for Survey Respondents

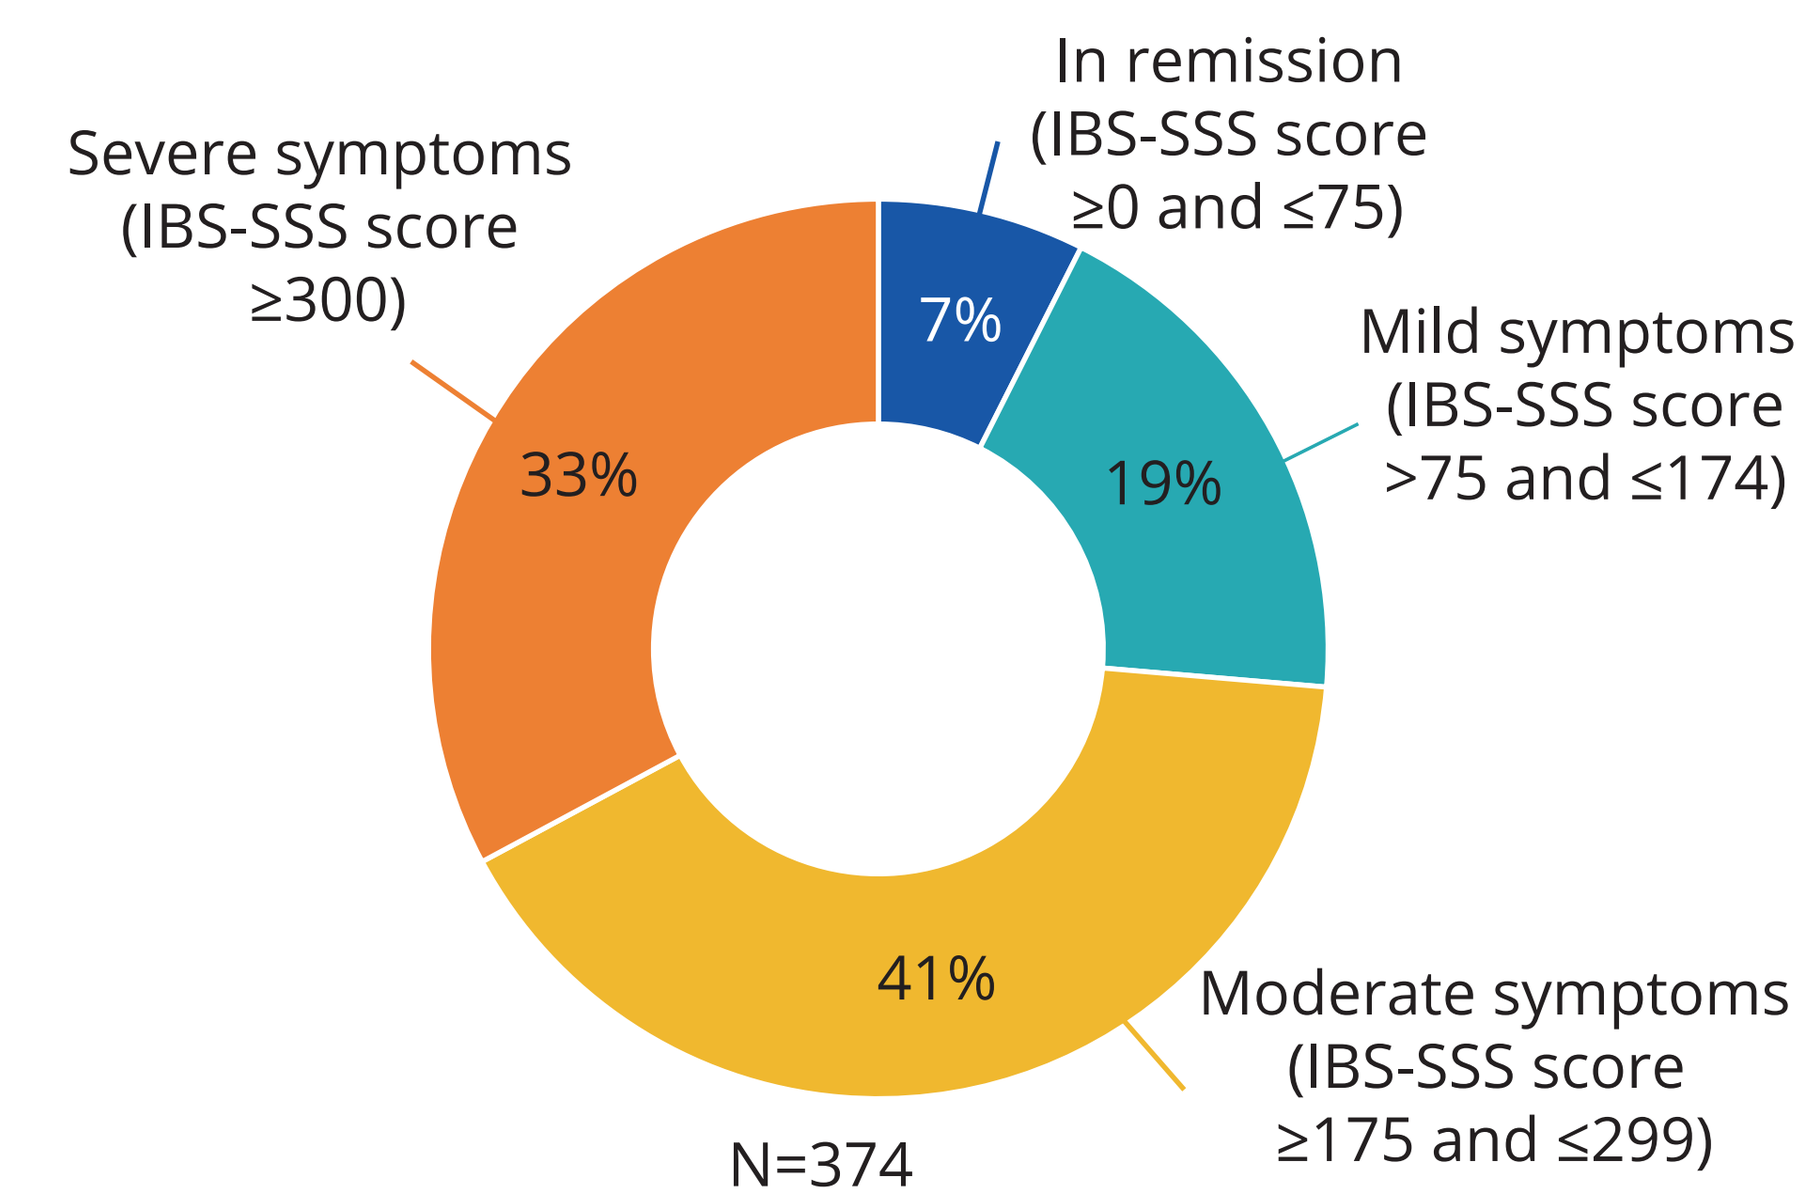
In remission (IBS-SSS score ≥0 and ≤75)
7%
Mild symptoms (IBS-SSS score >75 and ≤174)
19%
Moderate symptoms (IBS-SSS score ≥175 and ≤299)
41%
Severe symptoms (IBS-SSS score ≥300)
33%
N=374

IBS-SSS, Irritable Bowel Syndrome Severity Scoring System.

Figure 2: Associations of Employment Status and Urbanicity on IBS-SSS Scores[a]

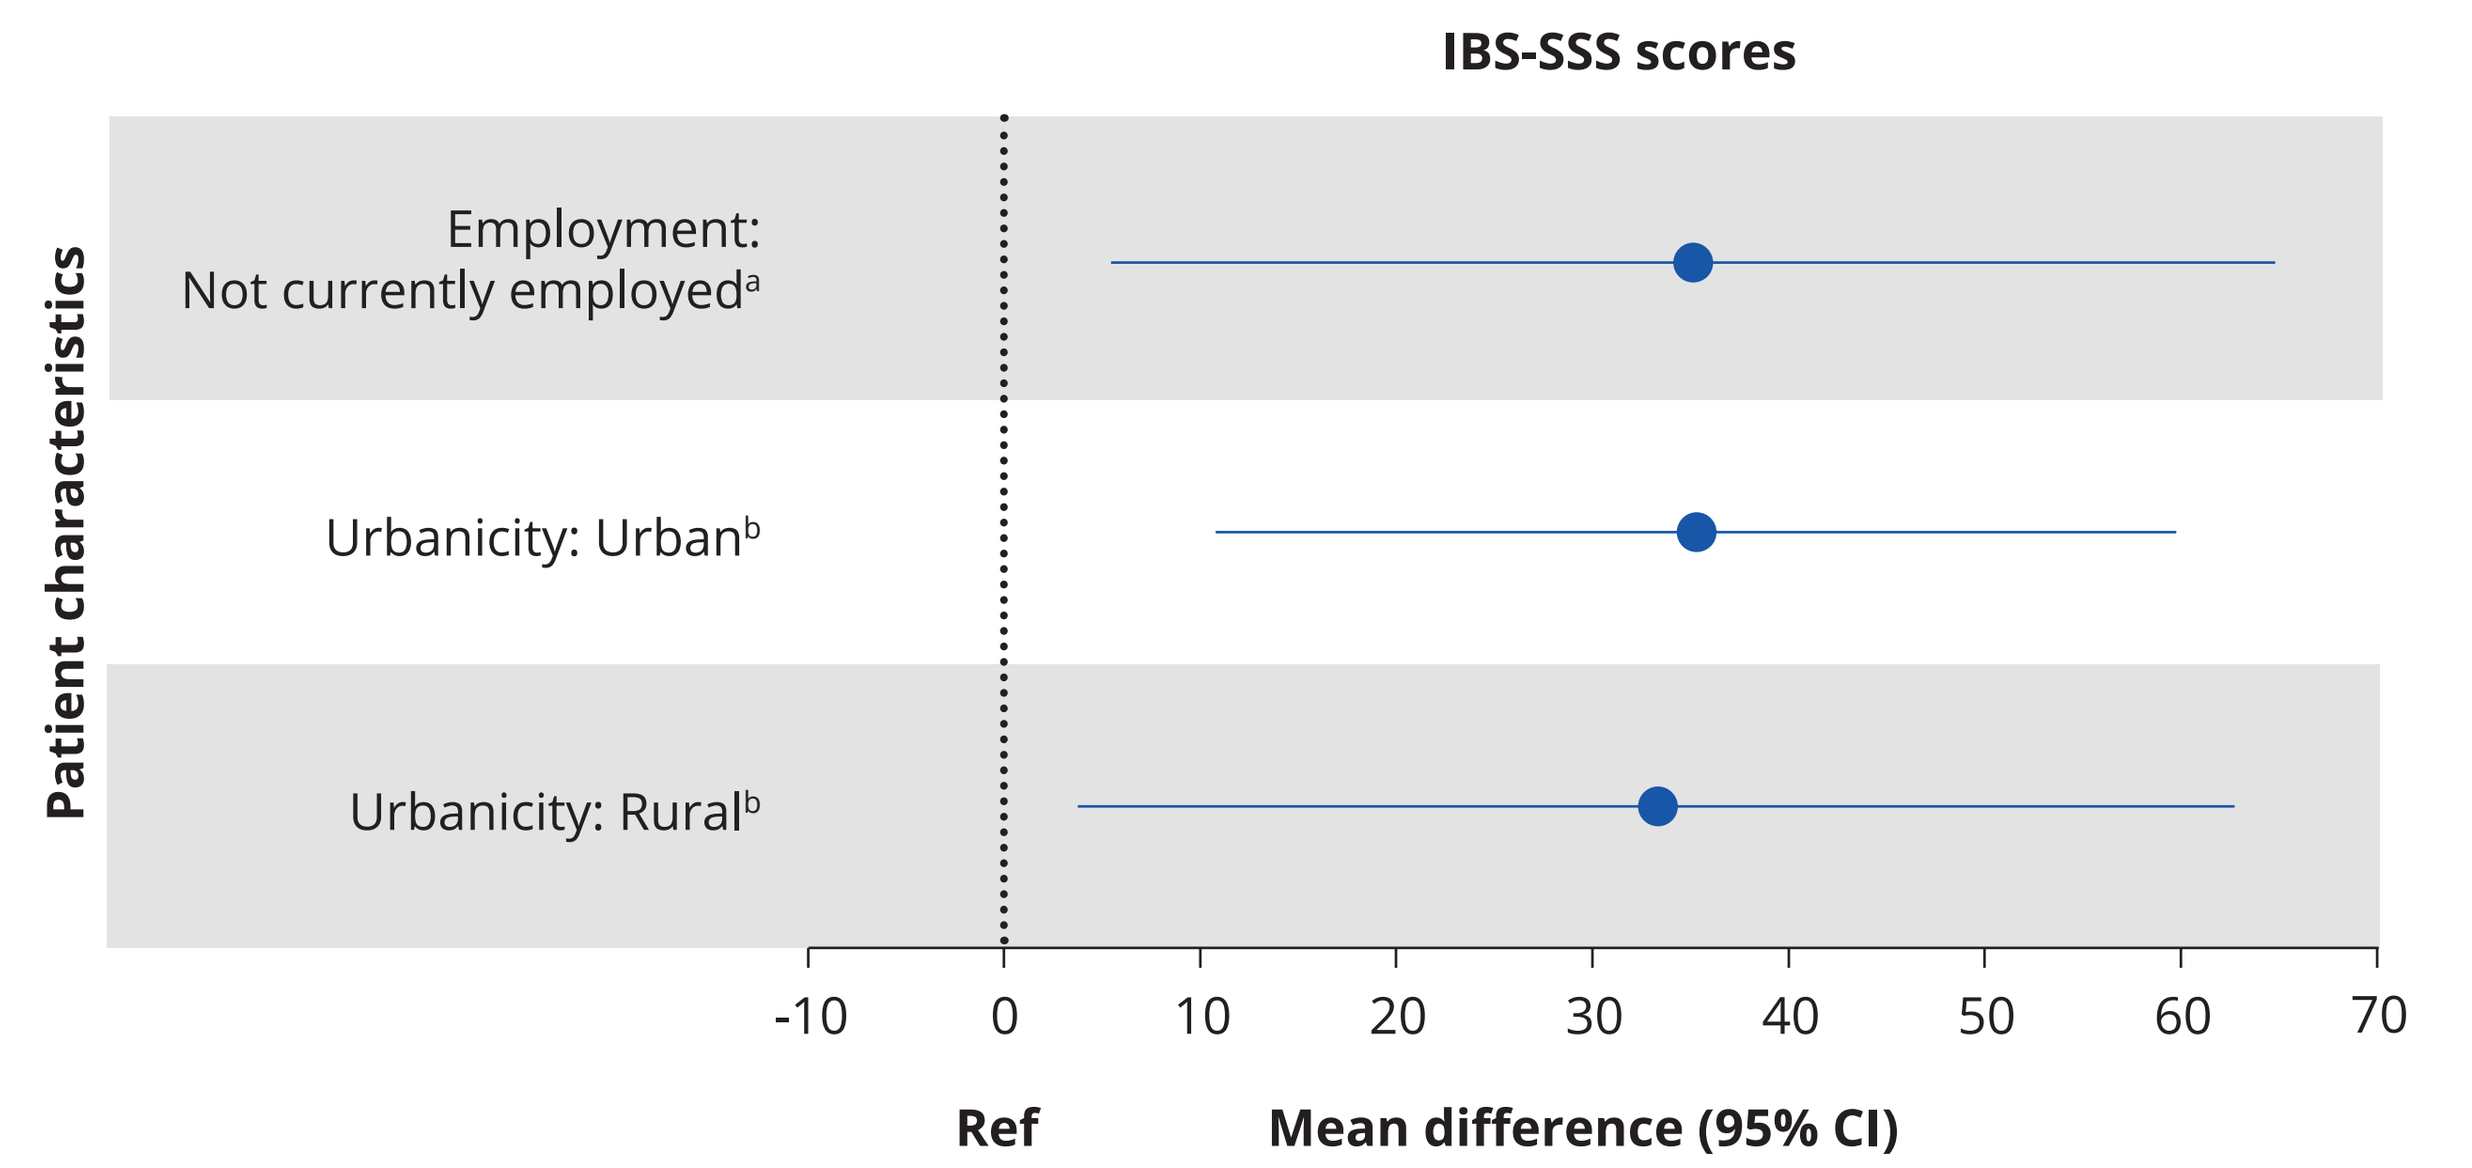
IBS-SSS scores
Patient characteristics
Employment: Not currently employed[a]
Urbanicity: Urban[b]
Urbanicity: Rural[b]
-10
0
10
20
30
40
50
60
70
Ref
Mean difference (95% CI)

[a]Referent = Employed; [b]Referent = Suburban.
CI, confidence interval; IBS-SSS; Irritable Bowel Syndrome Severity Scoring System; Ref, referent.

Treatment Satisfaction and Medication Use Patterns

- Overall, 57% of respondents reported being at least "satisfied" with their current treatment (**Figure 3**).
- Fewer than half (48%) of respondents were not currently using any prescription medications for IBS symptom management (**Table 1**).
- Only 16% of patients were currently taking an FDA-approved IBS-C prescription medication: linaclotide (11%), plecanatide (2%), lubiprostone (1%), and tenapanor (1%) (**Table 1**).

Table 1: Patient Characteristics Overall (N=374)

Characteristic	N=374
Mean age, y (SD)	50 (15)
Female, n (%)	235 (63)
Race, n (%)	
BIPOC	108 (29)
Non-BIPOC	265 (71)
IBS-C treatment group, n (%)[a]	
Any prescription medications (FDA-approved IBS-C medications or other prescription medications)	195 (52)
FDA-approved IBS-C medications[b,c]	58 (16)
Any other prescription medications[c,d]	168 (45)
No prescription medications	179 (48)
Urbanicity, n (%)	
Suburban	169 (45)
Urban	129 (35)
Rural	76 (20)
Employed	220 (59)
Living with ≥1 comorbid condition	280 (75)

Table excludes the row for a response of "prefer not to respond" to the question on race/ethnicity (N=1).
[a]Patients may be on more than 1 prescription medication currently; counts and proportions for individual treatment groups are not mutually exclusive.
[b]Includes tenapanor, plecanatide, lubiprostone, linaclotide, and tegaserod.
[c]Categories are not mutually exclusive; patients may have been taking both FDA-approved IBS-C medications and other medications.
[d]Includes prescription antiemetics, antispasmodics/anticholinergics, prescription muscle relaxants, prescription NSAIDs, prescription antacid medications, prescription analgesics, neuromodulators/antidepressants, rifaximin, prucalopride, alosetron, or eluxadoline.
BIPOC, Black, Indigenous, and People of Color; CHAMP-VA, Civilian Health and Medical Program of the Department of Veterans Affairs; FDA, Food and Drug Administration; IBS-C, irritable bowel syndrome with constipation; NSAID, non-steroidal anti-inflammatory drugs.

HRQoL

- In the general US population, mean (SD) PCS and MCS scores are 50 (10),[5] and a 5-point difference is considered clinically meaningful.[6]
- Patients with irritable bowel syndrome have mean SF-36 PCS and MCS scores 7 points lower than the general US population norms, representing clinically meaningful impairments.[7]
- Respondents reported lower HRQoL than the general US population; the mean (SD) MCS and PCS scores of 39.3 (11.6) and 40.1 (10.3) were approximately 1 SD below the mean score for the general US population.
- The physical and mental HRQoL burden of IBS-C was comparable to that observed in other chronic diseases, including cardiovascular disease, ulcerative colitis, and Crohn's disease (**Figure 4A, B**).
- For RP scores, 35% of patients reported the lowest score (0) and 25% reported the highest score (100). Similarly, for RE, 34% and 33% reported scores of 0 and 100, respectively (**Figure 4C, D**).

Figure 3: Distribution of Current Treatment Satisfaction

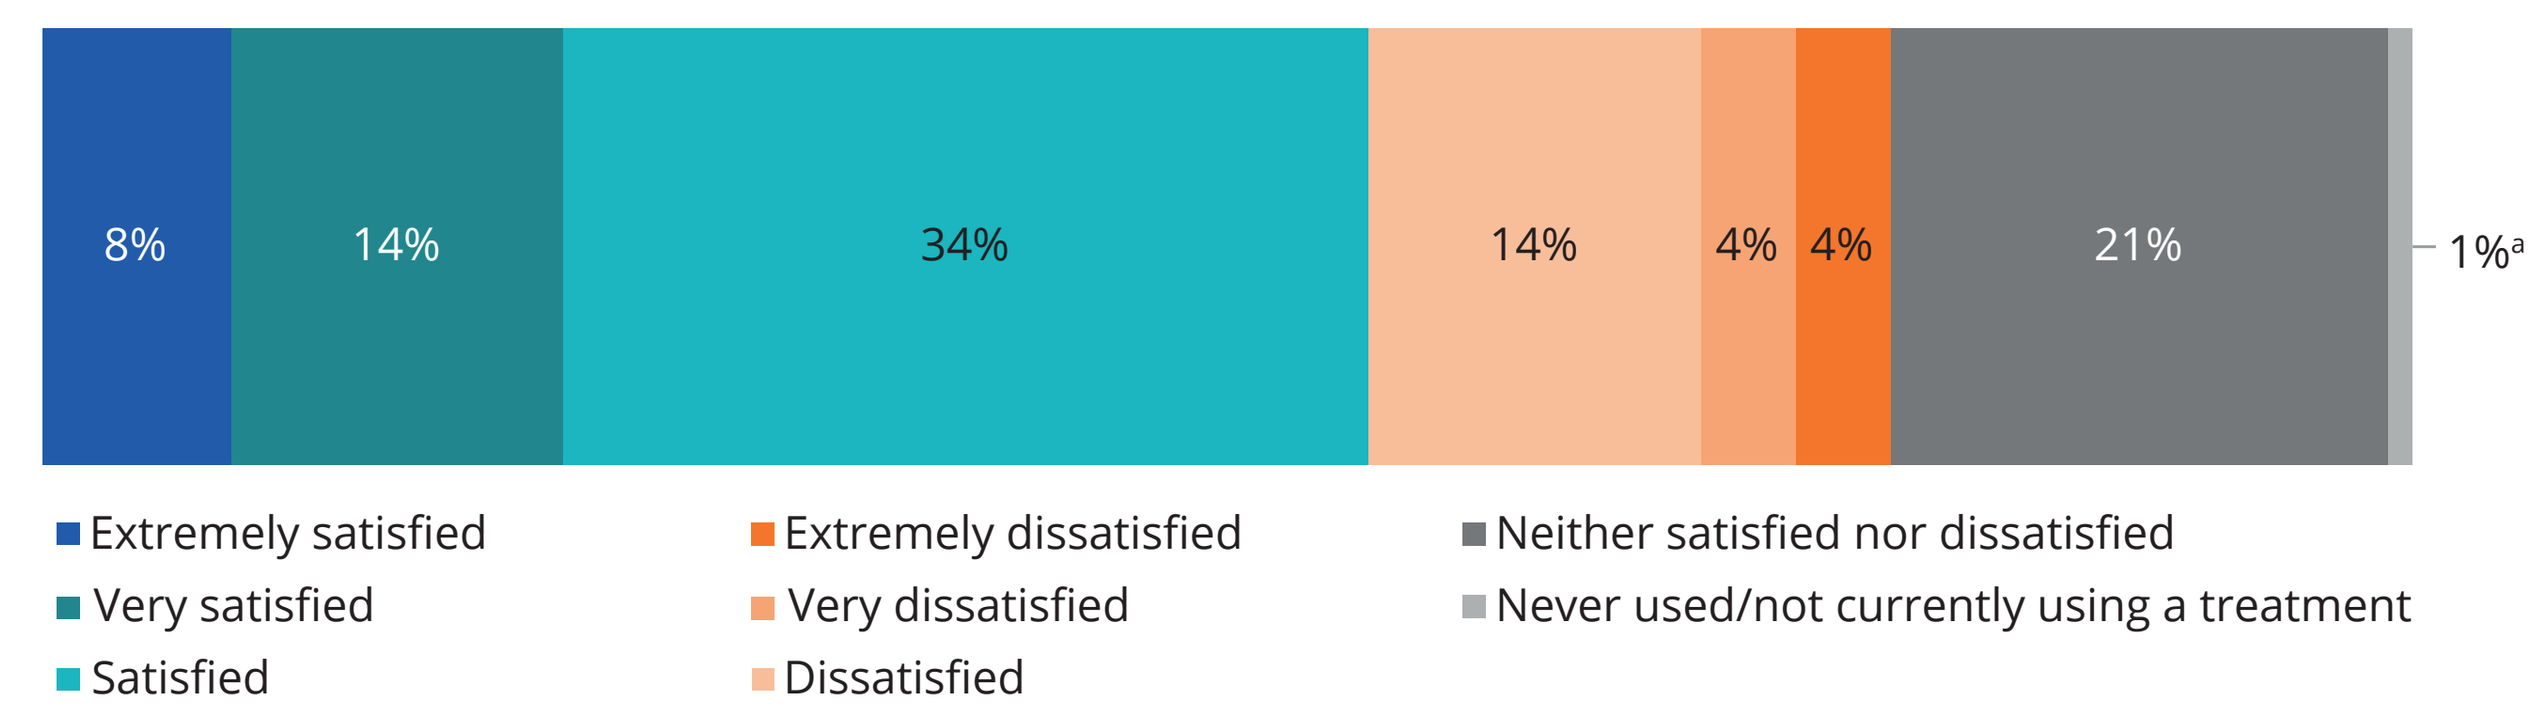
8%
14%
34%
14%
4%
4%
21%
1%[a]
Extremely satisfied
Very satisfied
Satisfied
Extremely dissatisfied
Very dissatisfied
Dissatisfied
Neither satisfied nor dissatisfied
Never used/not currently using a treatment

Four respondents (1%) were excluded; percentages were calculated among the 370 respondents with available data.
[a]Never used/not currently using any treatment.

Figure 4: (A) SF-36 MCS and (B) SF-36 PCS Across Disease States and Distributions of (C) SF-36 RP and (D) SF-36 RE Scores for Survey Responders

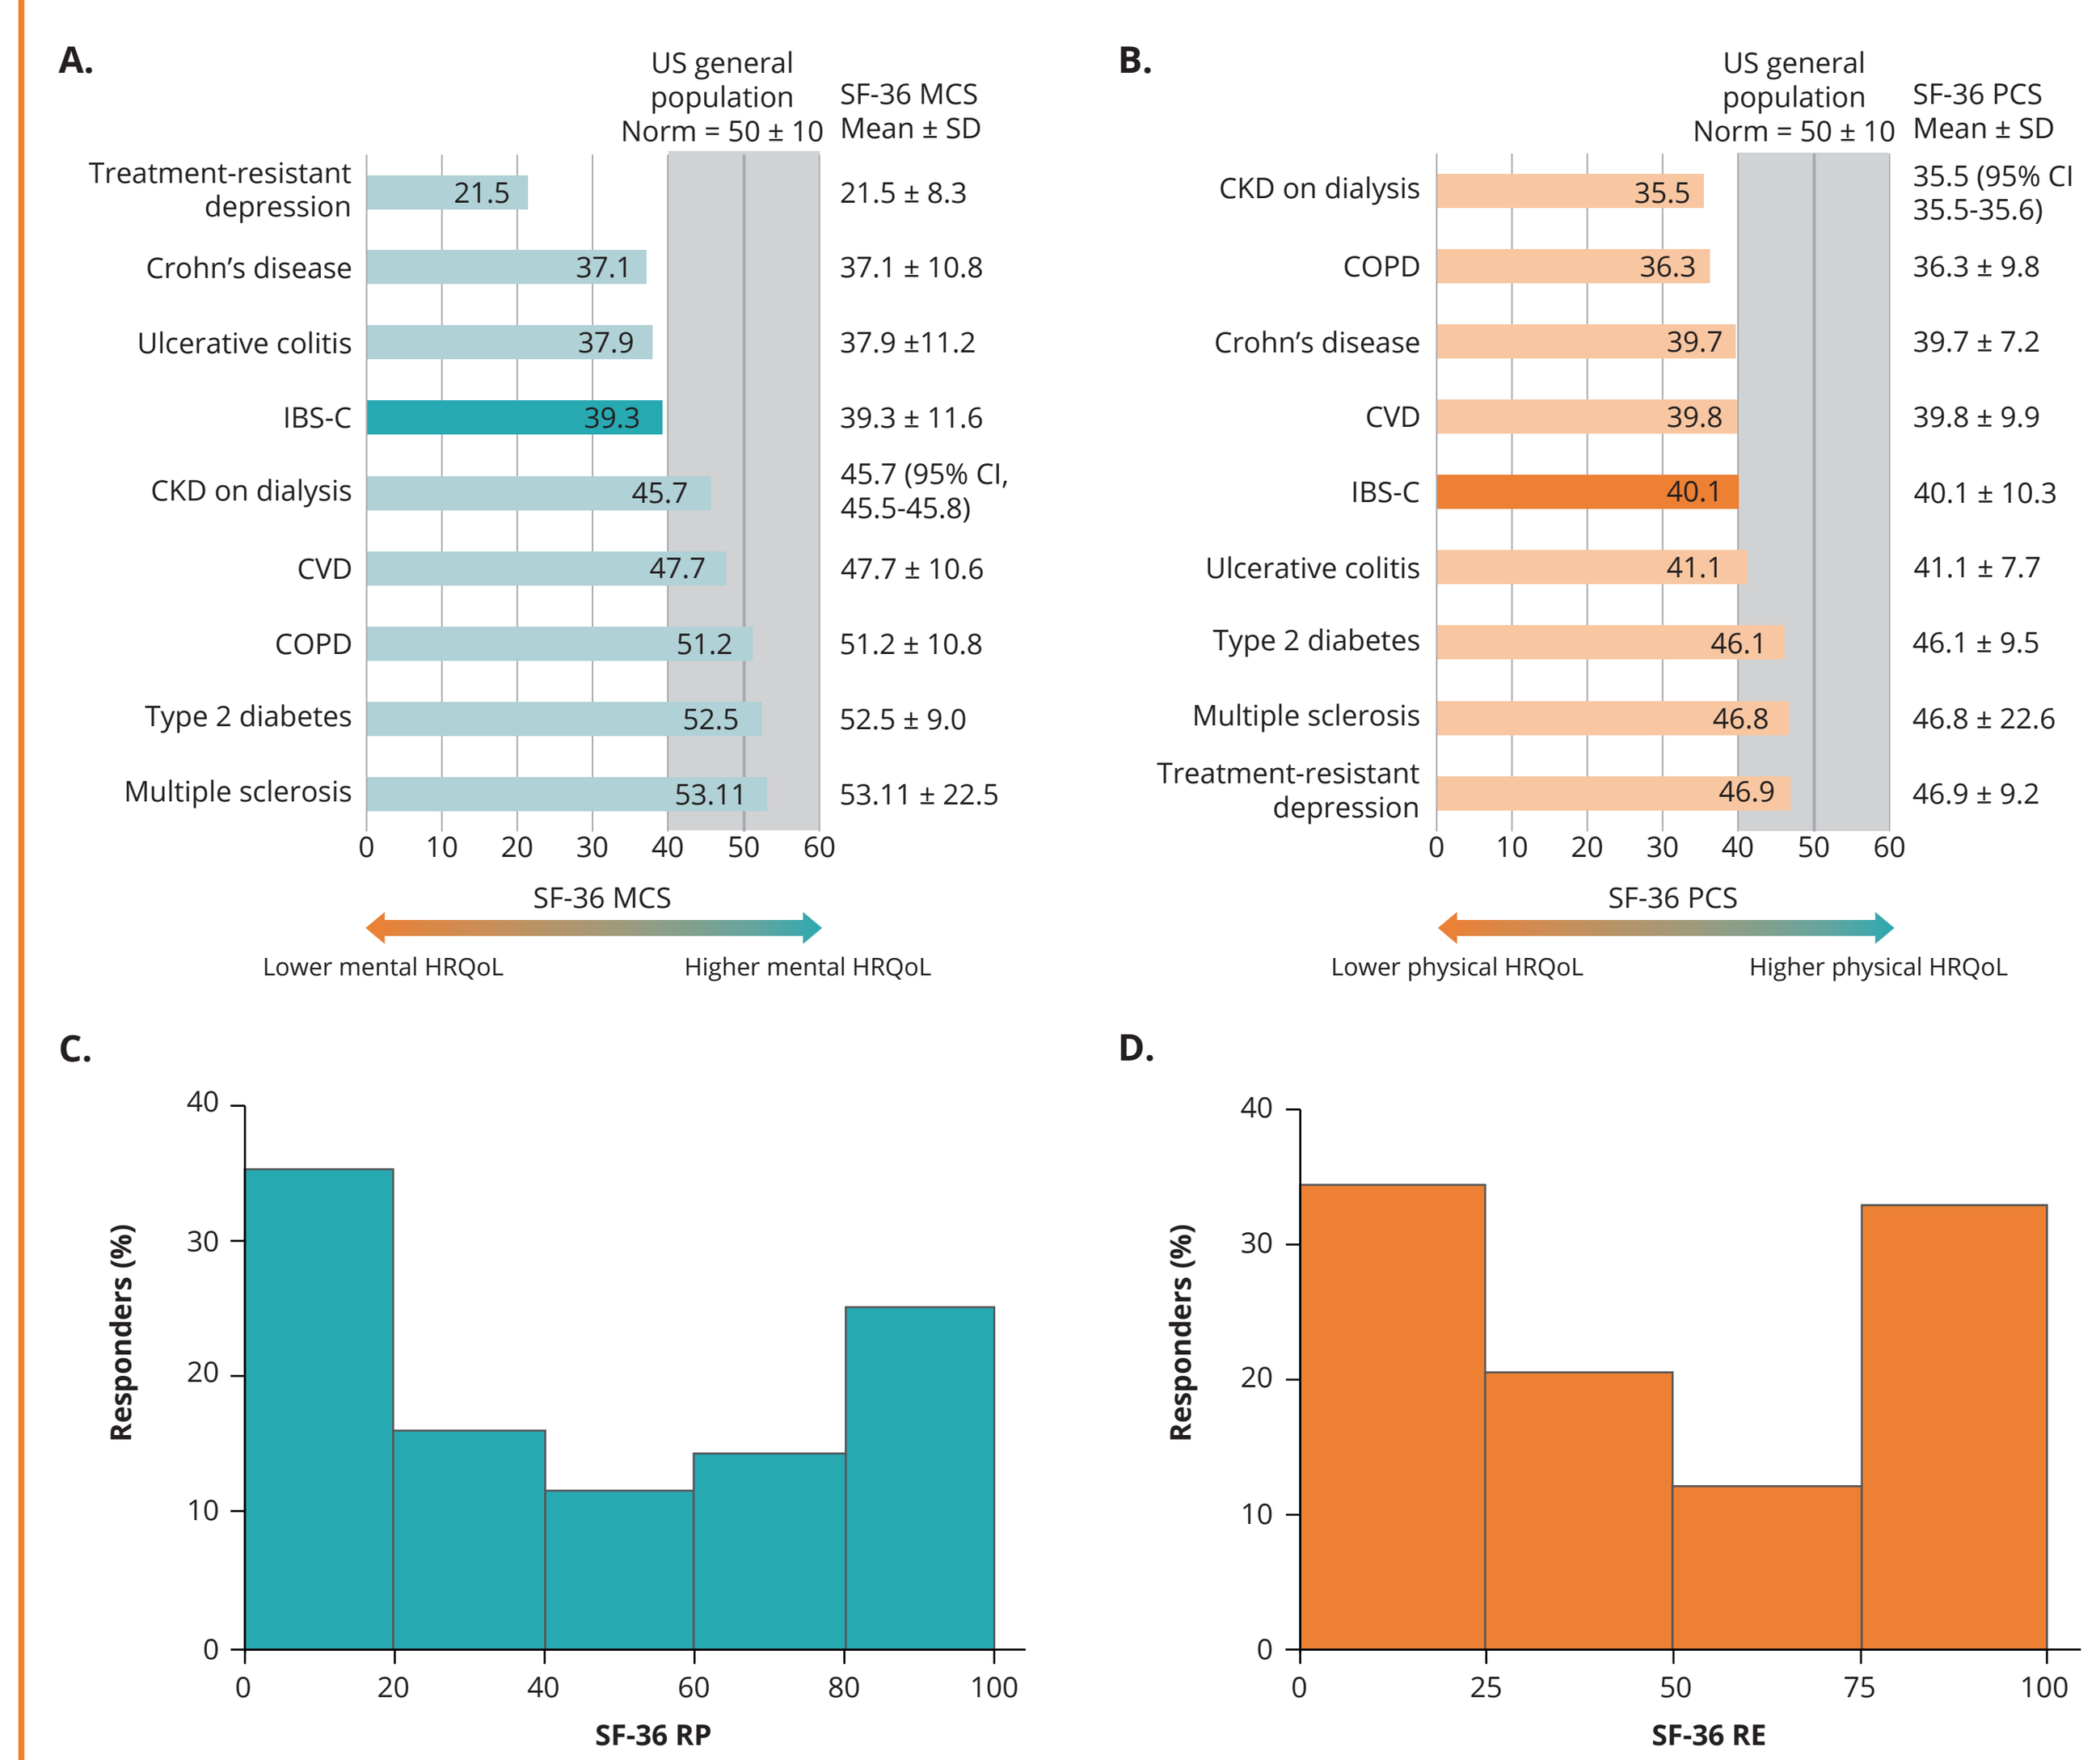
A.
US general population Norm = 50 ± 10
SF-36 MCS Mean ± SD
Treatment-resistant depression 21.5 21.5 ± 8.3
Crohn's disease 37.1 37.1 ± 10.8
Ulcerative colitis 37.9 37.9 ±11.2
IBS-C 39.3 39.3 ± 11.6
CKD on dialysis 45.7 45.7 (95% CI, 45.5-45.8)
CVD 47.7 47.7 ± 10.6
COPD 51.2 51.2 ± 10.8
Type 2 diabetes 52.5 52.5 ± 9.0
Multiple sclerosis 53.11 53.11 ± 22.5
SF-36 MCS
Lower mental HRQoL
Higher mental HRQoL
B.
US general population Norm = 50 ± 10
SF-36 PCS Mean ± SD
CKD on dialysis 35.5 35.5 (95% CI 35.5-35.6)
COPD 36.3 36.3 ± 9.8
Crohn's disease 39.7 39.7 ± 7.2
CVD 39.8 39.8 ± 9.9
IBS-C 40.1 40.1 ± 10.3
Ulcerative colitis 41.1 41.1 ± 7.7
Type 2 diabetes 46.1 46.1 ± 9.5
Multiple sclerosis 46.8 46.8 ± 22.6
Treatment-resistant depression 46.9 46.9 ± 9.2
SF-36 PCS
Lower physical HRQoL
Higher physical HRQoL
C.
Responders (%)
SF-36 RP
D.
Responders (%)
SF-36 RE

CI, confidence interval; CKD, chronic kidney disease; COPD, chronic obstructive pulmonary disease; CVD, cardiovascular disease; HRQoL, Health-related quality of life; IBS-C, irritable bowel syndrome with constipation; MCS, mental component score; PCS, physical component score; RE, role limitations due to emotional problems; RP, role limitations due to physical health problems; SF-36, 36-Item Short Form Health Survey; US, United States.

- Regression analyses showed that more severe IBS symptoms were associated with worse HRQoL relative to moderate symptoms, including lower MCS and PCS scores and higher odds of physical and emotional role limitations (**Table 2** and **Table 3**).
- Relative to those who were neither satisfied nor dissatisfied with their current treatment, dissatisfaction was associated with lower PCS scores, while satisfaction was associated with higher MCS scores (**Table 2**).
- Patients whose IBS symptoms were in remission (IBS-SSS ≥0 and ≤75) had 20% lower odds of experiencing physical limitations than those with moderate symptoms (**Table 3**).
- Compared with moderate symptoms, remission (IBS-SSS ≥0 and ≤75) was associated with 27% lower odds of emotional limitations, while severe symptoms were associated with 22% higher odds (**Table 3**).

Table 2: Predictors of MCS and PCS

	Predictor	Difference in means (adjusted)[a]	95% CI
MCS	**Current treatment satisfaction[b]**		
	Dissatisfied (ref: Neither satisfied nor dissatisfied)	−0.62	−4.0, 2.7
	Satisfied (ref: Neither satisfied nor dissatisfied)	4.6	1.8, 7.5
	Symptom severity[c]		
	In remission (ref: Moderate symptoms)	7.2	3.0, 11.0
	Mild symptoms (ref: Moderate symptoms)	2.8	−0.11, 5.6
	Severe symptoms (ref: Moderate symptoms)	−4.1	−6.6, −1.7
PCS	**Current treatment satisfaction[b]**		
	Dissatisfied (ref: Neither satisfied nor dissatisfied)	−3.3	−6.4, −0.23
	Satisfied (ref: Neither satisfied nor dissatisfied)	0.24	−2.4, 2.8
	Symptom severity[c]		
	In remission (ref: Moderate symptoms)	6.5	2.9, 10.0
	Mild symptoms (ref: Moderate symptoms)	5.1	2.6, 7.6
	Severe symptoms (ref: Moderate symptoms)	−5.3	−7.4, −3.2

[a]Adjusted for patient characteristics including age, sex, race/ethnicity, current employment status, educational attainment, insurance type, urbanicity, and presence of mental health (depression, anxiety, eating disorders, or PTSD), non-IBS GI (GERD, Celiac disease, functional dyspepsia, and IBD), and somatic (fibromyalgia, chronic pelvic pain, chronic back/neck pain, TMD, EDS, POTS, MCAS, and chronic fatigue) comorbidities.
[b]Treatment satisfaction was collapsed: satisfied includes "satisfied," "very satisfied," and "extremely satisfied." Dissatisfied includes "dissatisfied," "very dissatisfied," and "extremely dissatisfied."
[c]Symptom severity was defined according to the IBS-SSS score: In remission (0-75), mild (76-174), moderate (175-299), and severe (≥300).
CI, confidence interval; EDS, Ehlers-Danlos syndrome; GERD, gastroesophageal reflux disease; GI, gastrointestinal; IBD, inflammatory bowel disease; IBS, irritable bowel syndrome; IBS-SSS, Irritable Bowel Syndrome Severity Scoring System; MCAS, mast cell activation syndrome; MCS, mental component score; PCS, physical component score; POTS, postural orthostatic tachycardia syndrome; PTSD, post-traumatic stress disorder; TMD, temporomandibular joint disorder.

Table 3: Predictors of RP and RE (<100)

	Predictor	OR (adjusted)[a,b]	95% CI
RP	**Current treatment satisfaction**		
	Dissatisfied (ref: Neither satisfied nor dissatisfied)	1.31	0.55, 3.12
	Satisfied (ref: Neither satisfied nor dissatisfied)	0.83	0.42, 1.62
	Symptom severity[c]		
	In remission (ref: Moderate symptoms)	0.80	0.68, 0.95
	Mild symptoms (ref: Moderate symptoms)	0.89	0.80, 1.00
	Severe symptoms (ref: Moderate symptoms)	1.16	1.05, 1.28
RE	**Current treatment satisfaction**		
	Dissatisfied (ref: Neither satisfied nor dissatisfied)	1.13	0.50, 2.55
	Satisfied (ref: Neither satisfied nor dissatisfied)	0.53	0.27, 1.01
	Symptom severity[c]		
	In remission (ref: Moderate symptoms)	0.73	0.61, 0.86
	Mild symptoms (ref: Moderate symptoms)	0.92	0.82, 1.04
	Severe symptoms (ref: Moderate symptoms)	1.22	1.10, 1.35

[a]Adjusted for patient characteristics including age, sex, race/ethnicity, current employment status, educational attainment, insurance type, urbanicity, and presence of mental health (depression, anxiety, eating disorders, or PTSD), non-IBS GI (GERD, Celiac disease, functional dyspepsia, and IBD), and somatic (fibromyalgia, chronic pelvic pain, chronic back/neck pain, TMD, EDS, POTS, MCAS, and chronic fatigue) comorbidities.
[b]After adjusting for patient characteristics and social determinants of health, ORs represent the odds of reporting any role limitations due to physical or emotional health problems (RP <100 and RE <100) compared with the reference group.
[c]Symptom severity was defined according to the IBS-SSS score: In remission (0-75), mild (76-174), moderate (175-299), and severe (≥300).
CI, confidence interval; EDS, Ehlers-Danlos syndrome; GERD, gastroesophageal reflux disease; GI, gastrointestinal; IBD, inflammatory bowel disease; IBS, irritable bowel syndrome; IBS-SSS, Irritable Bowel Syndrome Severity Scoring System; MCAS, mast cell activation syndrome; OR, odds ratio; POTS, postural orthostatic tachycardia syndrome; PTSD, post-traumatic stress disorder; RE, role limitations due to emotional problems; RP, role limitations due to physical health problems; TMD, temporomandibular joint disorder.

Limitations

- Causality cannot be inferred from the associations identified in this analysis and will require further investigation.
- Treatment satisfaction could not be attributed to a specific treatment because satisfaction was measured globally, rather than being measured separately for each individual treatment.

Conclusions

- Greater IBS-C symptom severity was associated with residence in rural or urban settings, unemployment, and poorer HRQoL; assessing both symptoms and HRQoL may help better evaluate response and inform treatment approach.
- These findings highlight the importance of improving symptom control and increasing appropriate use of targeted therapies for IBS-C.
- Despite almost three-quarters of patients reporting moderate to severe IBS symptoms, only approximately one-sixth of patients were taking an FDA-approved IBS-C prescription medication.

Disclosures

This study was supported by Ardelyx, Inc. Megan Riehl has served as a consultant for Ardelyx, Calm.com, GI Psychology, and Mindset Health, and has served on advisory boards for Mindset Health and Sanofi. Taya Westfield and Alan Fossa are employees of Arbor Research Collaborative for Health, which received funding for this study from Ardelyx, Inc. All funding was provided to Arbor Research Collaborative for Health and not directly to Taya Westfield and Alan Fossa. Luisa Scott is an employee of Ardelyx, Inc.

References

1. Lacy BE et al. *Neurogastroenterol Motil*. 2024:36:e14903.
2. Ware JE Jr et al. *J Clin Epidemiol*. 1998;51:903-12.
3. Brugnera A et al. *Healthcare (Basel)*. 2024;12:757.
4. Kaplan AI et al. *Neurogastroenterol Motil*. 2023;35:e14524.
5. Maglinte GA et al. *J Clin Epidemiol*. 2012;65:497-502.
6. Norman GR et al. *Med Care*. 2003;41:582-92.
7. Gralnek IM et al. *Gastroenterology*. 2000;119:654-60.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield Medcomms, an Inizio company, and funded by Ardelyx, Inc.

Contact information

Megan Riehl can be contacted for further information on this study at mriehl@med.umich.edu.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American College of Gastroenterology (ACG) Conference; October 9-14, 2026; Nashville, TN, USA. In Person and Virtual.

MED-US–2600361 10/2026

IBSRELA® (tenapanor) is indicated for treatment of irritable bowel syndrome with constipation (IBS-C) in adults

Important Safety Information

WARNING: RISK OF SERIOUS DEHYDRATION IN PEDIATRIC PATIENTS

- **IBSRELA is contraindicated in patients less than 6 years of age; in nonclinical studies in young juvenile rats, administration of tenapanor caused deaths presumed to be due to dehydration. *[see PI Contraindications (4), Use in Specific Populations (8.4)]*.**
- **Avoid use of IBSRELA in patients 6 years to less than 12 years of age. *[see PI Warnings and Precautions (5.1), Use in Specific Populations (8.4)]*.**
- **The safety and effectiveness of IBSRELA have not been established in pediatric patients less than 18 years of age. *[see PI Use in Specific Populations (8.4)]*.**

CONTRAINDICATIONS

IBSRELA is contraindicated in patients less than 6 years of age due to the risk of serious dehydration.

IBSRELA is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Risk of Serious Dehydration in Pediatric Patients

IBSRELA is contraindicated in patients below 6 years of age. The safety and effectiveness of IBSRELA in patients less than 18 years of age have not been established. In young juvenile rats (less than 1 week old; approximate human age equivalent of less than 2 years of age), decreased body weight and deaths occurred, presumed to be due to dehydration, following oral administration of tenapanor. There are no data available in older juvenile rats (human age equivalent 2 years to less than 12 years).

Avoid the use of IBSRELA in patients 6 years to less than 12 years of age. Although there are no data in older juvenile rats, given the deaths in younger rats and the lack of clinical safety and efficacy data in pediatric patients, avoid the use of IBSRELA in patients 6 years to less than 12 years of age.

Diarrhea

Diarrhea was the most common adverse reaction in two randomized, double-blind, placebo-controlled trials of IBS-C. Severe diarrhea was reported in 2.5% of IBSRELA-treated patients. If severe diarrhea occurs, suspend dosing and rehydrate patient.

MOST COMMON ADVERSE REACTIONS

The most common adverse reactions in IBSRELA-treated patients (incidence ≥2% and greater than placebo) were: diarrhea (16% vs 4% placebo), abdominal distension (3% vs <1%), flatulence (3% vs 1%) and dizziness (2% vs <1%).

For additional safety information, including the Boxed Warning, please see full Prescribing Information: click here.

IBS-C, irritable bowel syndrome with constipation.
IBSRELA (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.
Available at: https://www.ibsrela-hcp.com/PI

© Ardelyx, Inc., 2026. All rights reserved. Ardelyx and IBSRELA are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only