Poster 21

Tenapanor Improves Combined Symptoms of Abdominal Bloating, Pain, and Complete Spontaneous Bowel Movement Frequency in Patients With IBS-C Experiencing Moderate to Severe Bloating

Kyle Staller,[1] Yang Yang,[2] Suling Zhao,[2] Susan Edelstein[2]

[1]Massachusetts General Hospital, Boston, MA, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Introduction

- Abdominal bloating is one of the most bothersome symptoms of irritable bowel syndrome with constipation (IBS-C),[1] but it is not included in the Rome IV diagnostic criteria.[2]
- Tenapanor is a first-in-class, inhibitor of intestinal sodium/hydrogen exchanger isoform 3 approved by the United States Food and Drug Administration for treatment of IBS-C in adults.[3]
- Previous studies have demonstrated that tenapanor improves complete spontaneous bowel movement (CSBM) frequency and abdominal symptoms in IBS-C.[4-7]
- This post hoc analysis assessed the effect of tenapanor on moderate to severe abdominal bloating, time to bloating improvement, and the combined effects on bloating, abdominal pain, and CSBM frequency in patients with IBS-C and moderate to severe bloating at baseline.

Methods

- Data were pooled from 3 randomized, placebo-controlled clinical trials: a phase 2b study (NCT01923428) and the phase 3 T3MPO-1 (NCT02621892) and T3MPO-2 (NCT02686138) studies, in which patients with IBS-C who met the Rome III criteria were randomized to receive tenapanor or placebo twice a day (bid) for 12 or 26 weeks.[5-7]
 – Study-level data from the first 12 weeks of each study were included in this post hoc analysis of the intent-to-treat analysis sets comparing the tenapanor 50 mg bid and placebo groups.
- A phone diary was used to collect data on daily abdominal bloating on a scale of 0 to 10, which was categorized as mild (<4) or moderate to severe (≥4).
- This analysis examined patients with moderate to severe bloating at baseline, calculated as the mean of average weekly bloating scores of week −2 and week −1. These patients comprised the efficacy analysis set (EAS).
 – For a valid study week (ie, a study week with at least 4 days of abdominal bloating reporting), the average weekly bloating score was calculated with the following formula: sum of daily abdominal bloating scores in that study week divided by number of days with abdominal bloating reported in that study week.
- Efficacy endpoints included change from baseline in average weekly bloating score and time to onset of achieving an abdominal bloating response defined as achieving a ≥30% reduction from baseline in average weekly bloating score.
- Six multi-symptom composite endpoints were also evaluated to assess the simultaneous improvement from baseline in abdominal bloating, abdominal pain, and CSBMs/week.
 – The response was defined using multiple thresholds: ≥30%, ≥40%, and ≥50% reduction in abdominal bloating and pain plus an increase of ≥1 or ≥2 CSBMs/week, achieved within the same week for ≥6 of 12 weeks.

Results

Patients

Table 1: Demographics and Baseline Characteristics of Patients With Moderate to Severe Bloating at Baseline (Pooled EAS Analysis Set)[a]

	Placebo bid (N=625)	Tenapanor 50 mg bid (N=628)	Overall (N=1253)
Age at informed consent, mean (SD), y	44.8 (13.5)	45.5 (12.9)	45.2 (13.2)
Sex, n (%)			
Female	523 (83.7)	520 (82.8)	1043 (83.2)
Race, n (%)			
Asian	10 (1.6)	22 (3.5)	32 (2.6)
Black or African American	201 (32.2)	187 (29.8)	388 (31.0)
White	397 (63.5)	407 (64.8)	804 (64.2)
Other[b]	17 (2.7)	12 (1.9)	29 (2.3)
Ethnicity, n (%)			
Hispanic	160 (25.6)	176 (28.0)	336 (26.8)
Non-Hispanic	465 (74.4)	452 (72.0)	917 (73.2)
Body mass index, mean (SD), kg/m²	29.9 (6.8)	30.1 (7.0)	30.0 (6.9)
Duration of IBS-C symptoms before randomization, mean (SD), y[c]	11.6 (11.8)	11.2 (11.5)	11.4 (11.7)
Baseline weekly score for abdominal bloating, mean (SD)	6.92 (1.49)	6.90 (1.53)	6.91 (1.51)

bid, twice a day; EAS, efficacy analysis set; IBS-C, irritable bowel syndrome with constipation.
[a]Average weekly abdominal bloating score ≥4 (N=1253). [b]Includes American Indian or Alaska Native, Multiple, Unknown, and Other. [c]Five patients in the T3MPO-1 study did not report the start date of their IBS-C symptoms; data reported for the following numbers of patients: placebo (N=621), tenapanor (N=627), and overall (N=1248).

Change in Abdominal Bloating Over a 12-Week Treatment Period

- Tenapanor treatment resulted in a consistently greater reduction from baseline in average weekly bloating score compared with placebo over the first 12 weeks (**Figure 1**) (all *P*≤0.0010).
- The least-squares (LS) mean change at week 8 was −2.45 for tenapanor and −1.82 for placebo, with an LS mean difference of −0.64 in week 8 (*P*<0.0001).
- At week 12, the LS mean change was −2.66 for tenapanor and −2.10 for placebo, with an LS mean difference of −0.57 (*P*=0.0003, analysis of co-variance).

Time to First Abdominal Bloating Response in a 12-Week Treatment Period

- Patients treated with tenapanor had faster improvement in bloating compared with those treated with placebo.
- The median time to onset of achieving a ≥30% reduction in weekly bloating score was 5 weeks (95% CI, 4.0-6.0) for tenapanor-treated patients versus 8 weeks (95% CI, 7.0-10.0) for placebo-treated patients (log-rank test *P*<0.0001; **Figure 2**).
- By week 5, the cumulative probability of achieving a ≥30% reduction in weekly bloating response was 51.3% with tenapanor versus 41.3% with placebo (**Figure 2**).

Multi-Symptom Composite Response After a 12-Week Treatment Period

- Across several thresholds, significantly larger proportions of patients with moderate to severe bloating treated with tenapanor achieved a multi-symptom composite response in the same week for ≥6 of 12 weeks than those treated with placebo (*P*≤0.006, Chi-square test; **Figure 3**).

Figure 1: LS Mean Change From Baseline in Average Weekly Abdominal Bloating Score in Patients With Moderate to Severe Bloating at Baseline (Pooled EAS Analysis Set)

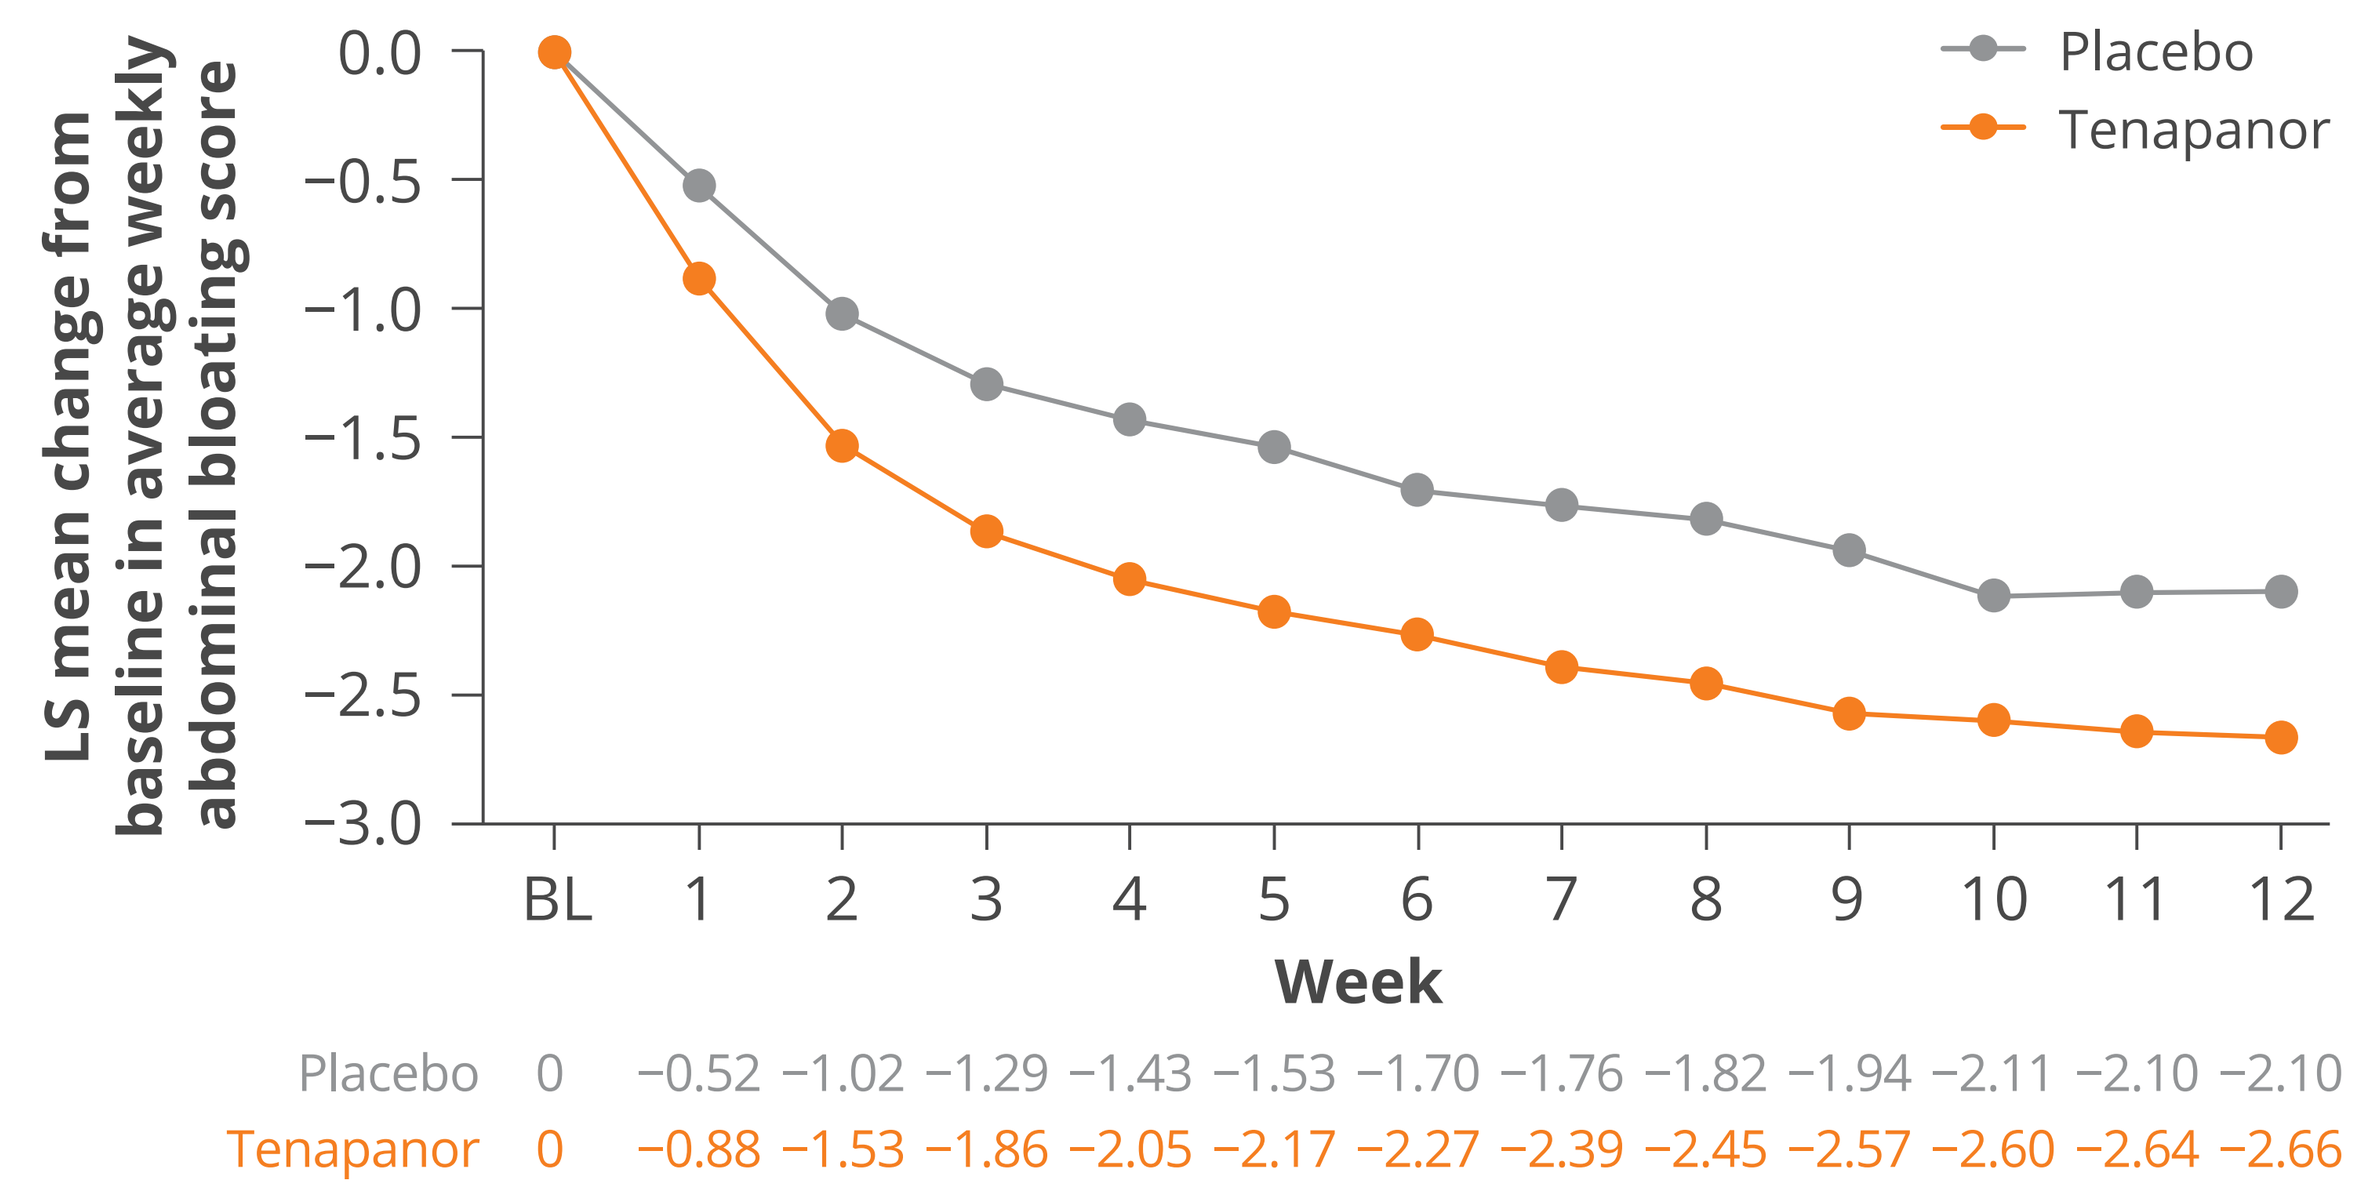
LS mean change from baseline in average weekly abdominal bloating score
0.0
−0.5
−1.0
−1.5
−2.0
−2.5
−3.0
BL 1 2 3 4 5 6 7 8 9 10 11 12
Week
Placebo
Tenapanor
Placebo 0 −0.52 −1.02 −1.29 −1.43 −1.53 −1.70 −1.76 −1.82 −1.94 −2.11 −2.10 −2.10
Tenapanor 0 −0.88 −1.53 −1.86 −2.05 −2.17 −2.27 −2.39 −2.45 −2.57 −2.60 −2.64 −2.66

BL, baseline; EAS, efficacy analysis set; LS, least-squares.

Figure 2: Time to Onset of Achieving a ≥30% Reduction in Weekly Bloating Score in Patients With Moderate to Severe Bloating at Baseline (Pooled EAS Analysis Set)

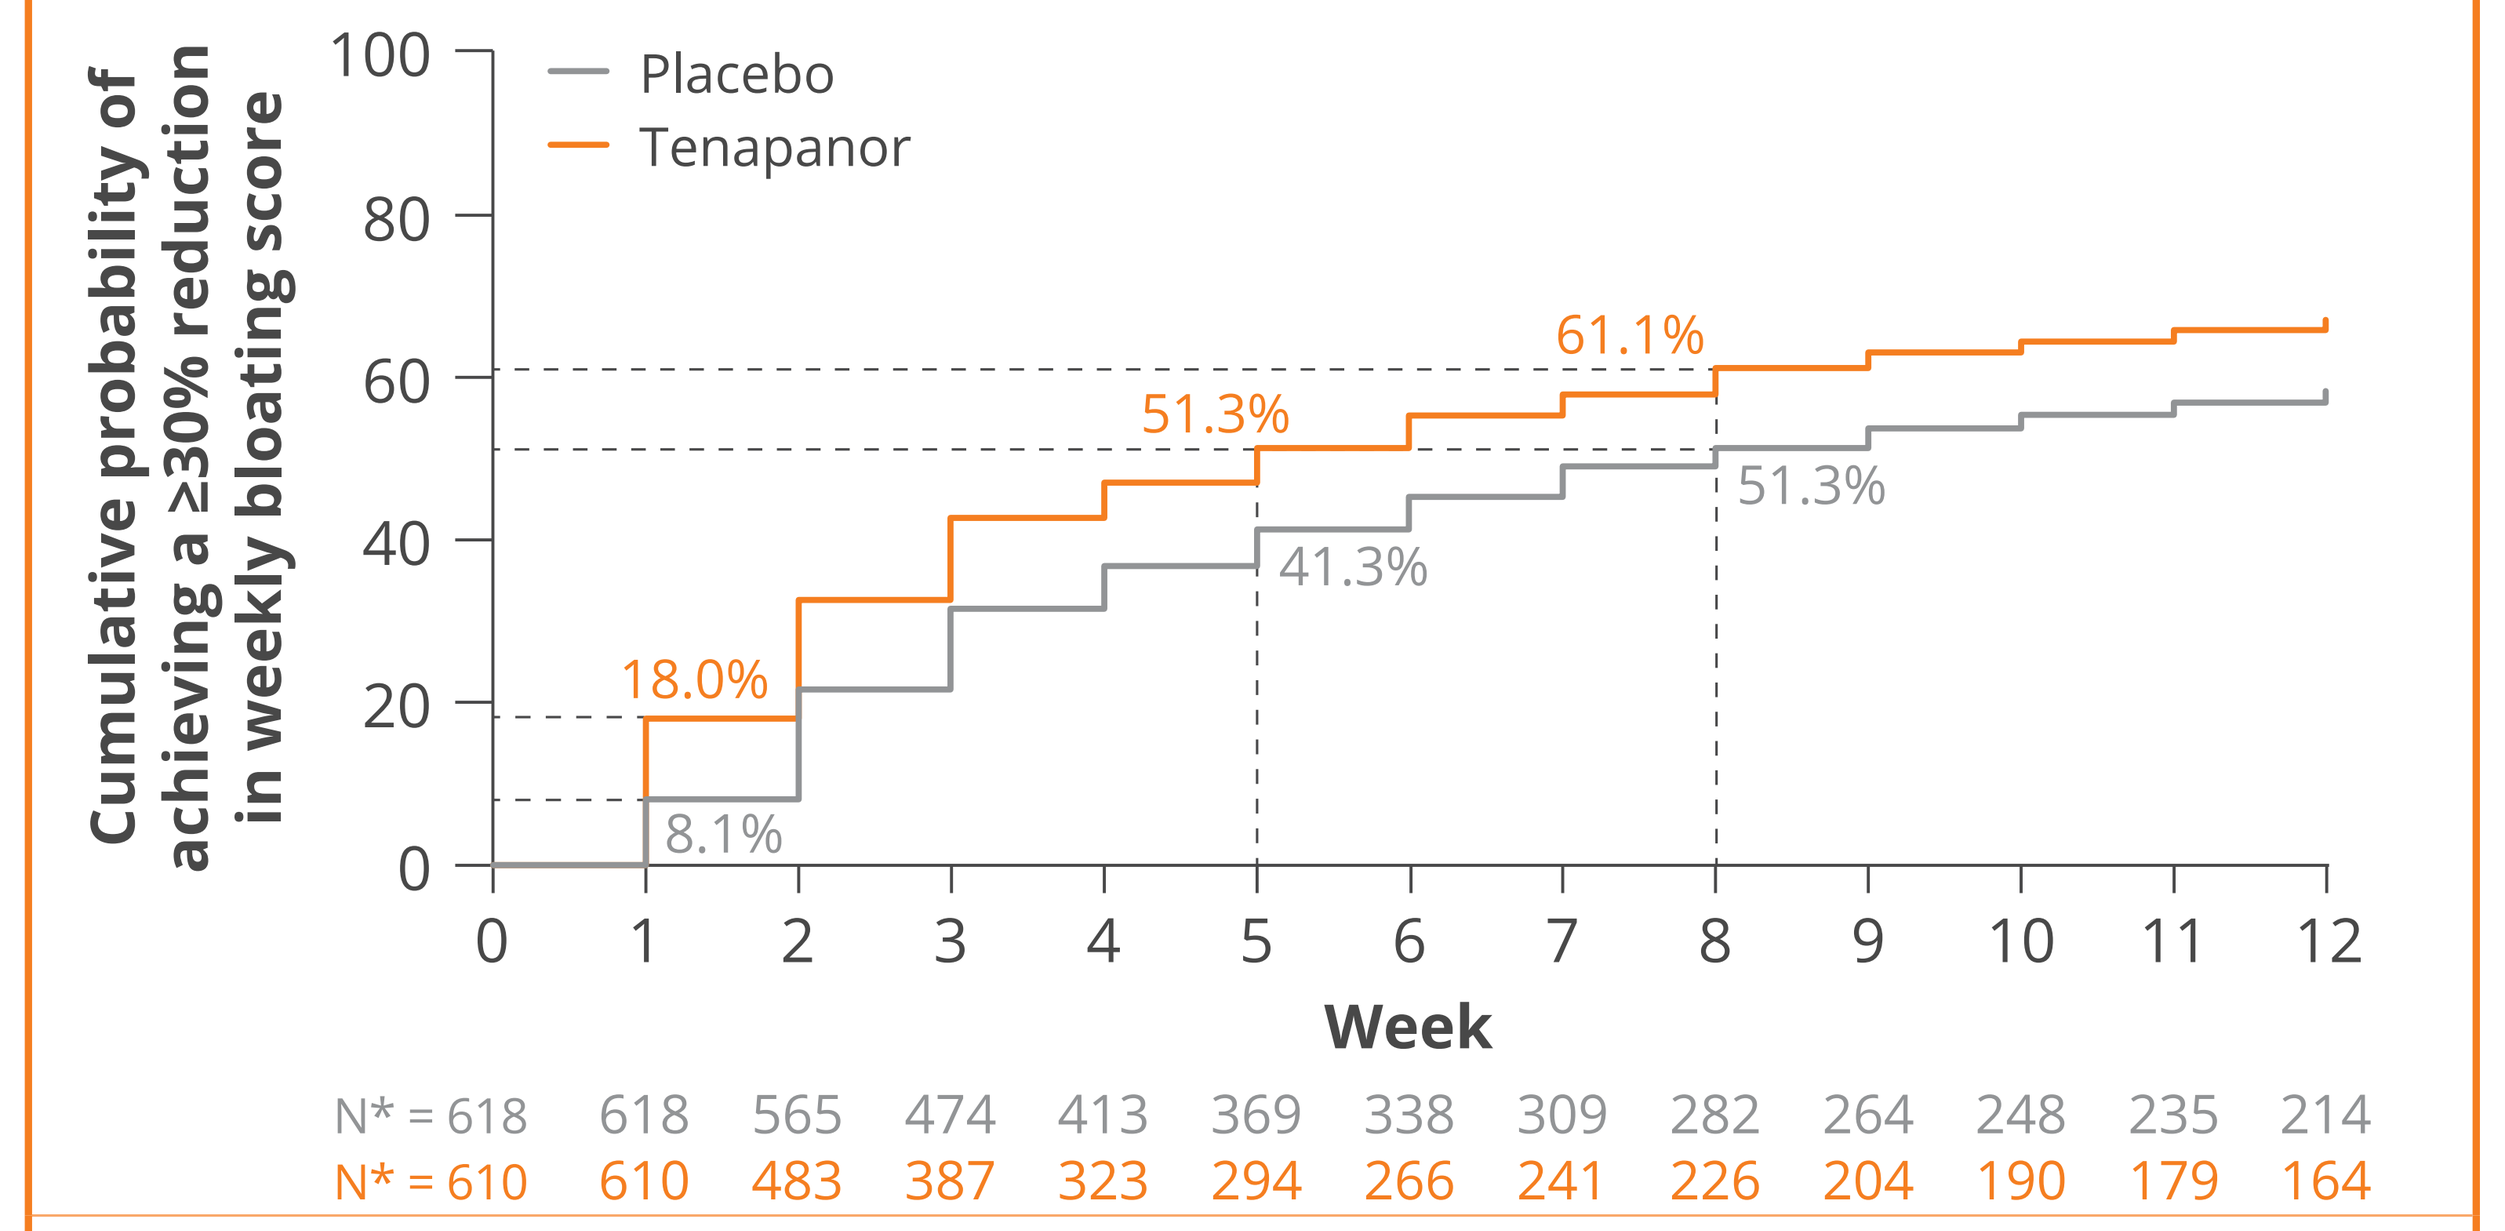
Cumulative probability of achieving a ≥30% reduction in weekly bloating score
100
80
60
40
20
0
Placebo
Tenapanor
61.1%
51.3%
51.3%
41.3%
18.0%
8.1%
0 1 2 3 4 5 6 7 8 9 10 11 12
Week
N* = 618 618 565 474 413 369 338 309 282 264 248 235 214
N* = 610 610 483 387 323 294 266 241 226 204 190 179 164

*N represents the number of patients who have not yet responded and remain under observation at the corresponding study week.
EAS, efficacy analysis set.

Figure 3: Multi-Symptom Composite Response in Patients With Moderate to Severe Bloating at Baseline (Pooled EAS Analysis Set)

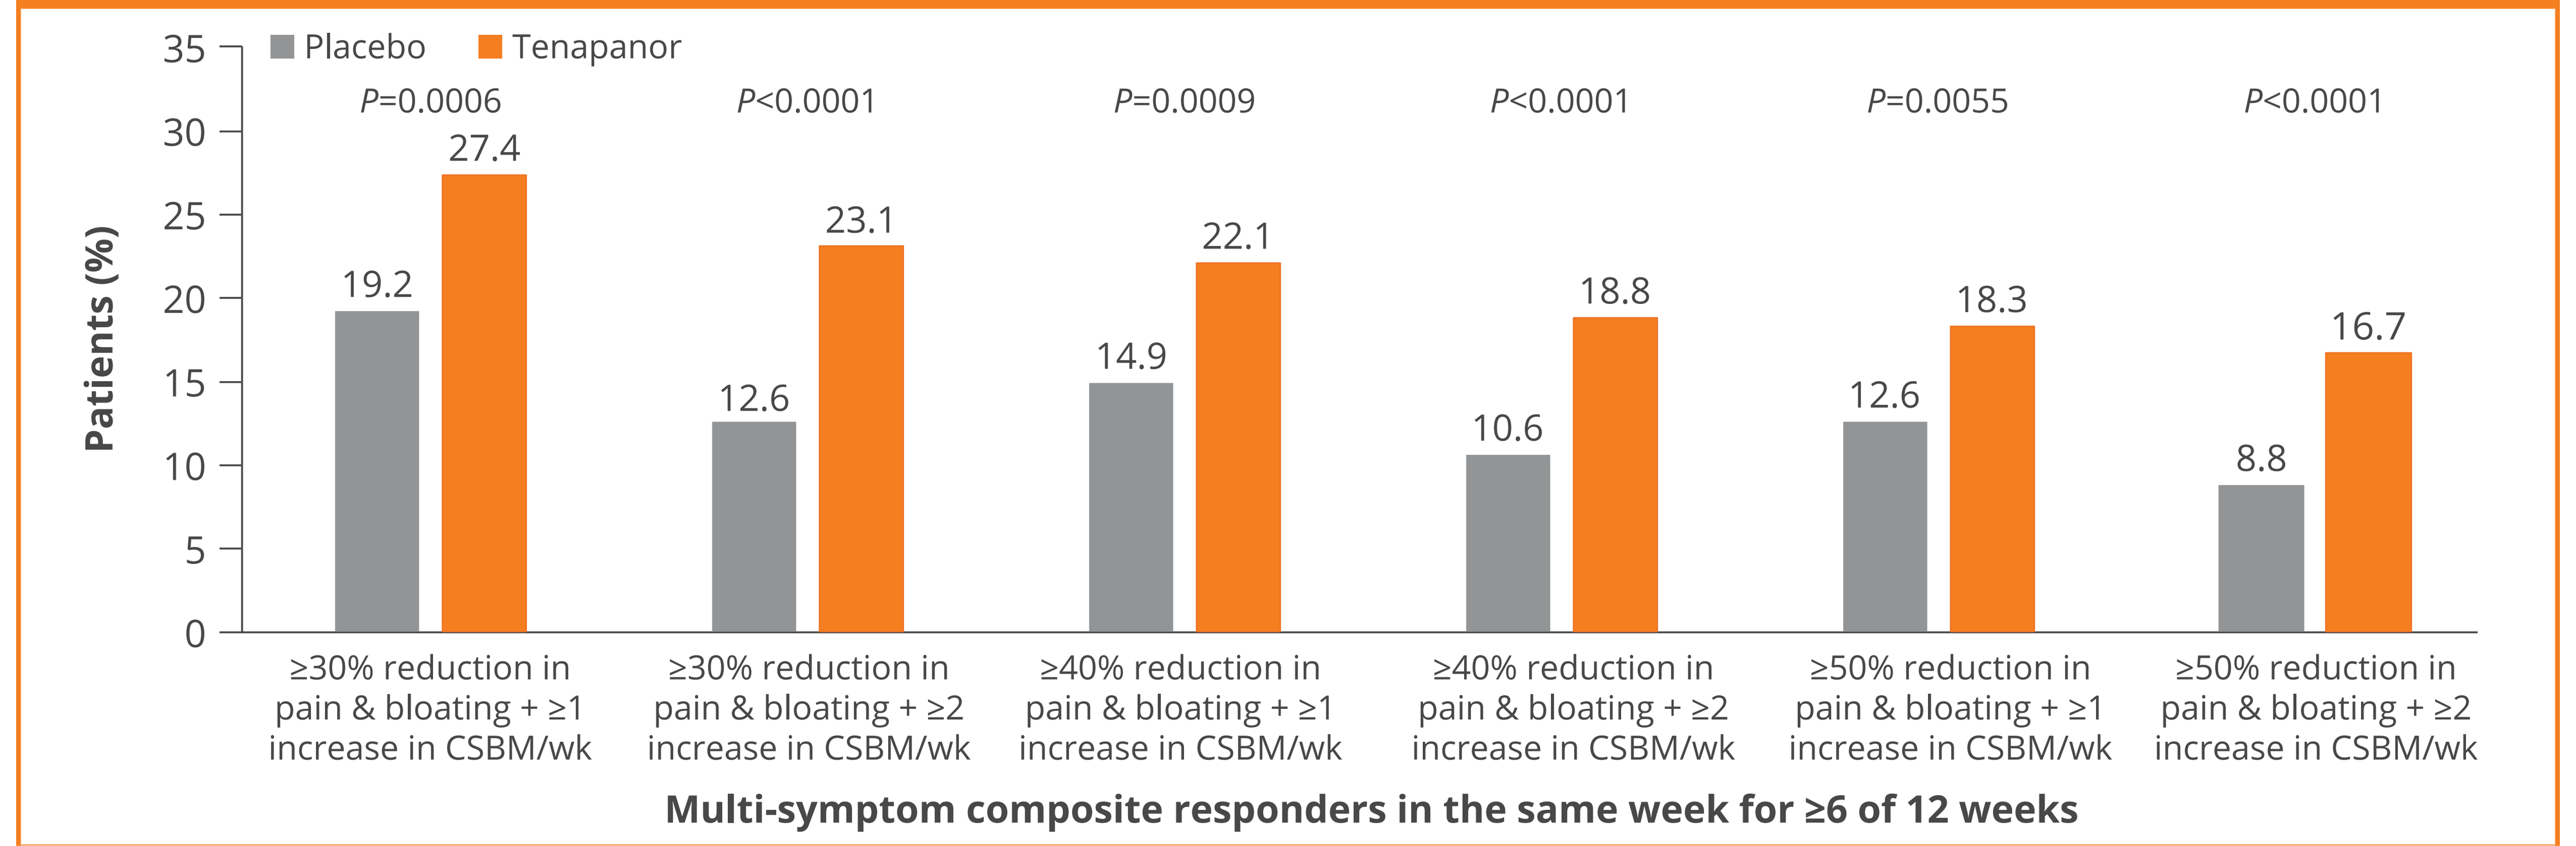
Placebo
Tenapanor
Patients (%)
35
30
25
20
15
10
5
0
P=0.0006
19.2
27.4
≥30% reduction in pain & bloating + ≥1 increase in CSBM/wk
P<0.0001
12.6
23.1
≥30% reduction in pain & bloating + ≥2 increase in CSBM/wk
P=0.0009
14.9
22.1
≥40% reduction in pain & bloating + ≥1 increase in CSBM/wk
P<0.0001
10.6
18.8
≥40% reduction in pain & bloating + ≥2 increase in CSBM/wk
P=0.0055
12.6
18.3
≥50% reduction in pain & bloating + ≥1 increase in CSBM/wk
P<0.0001
8.8
16.7
≥50% reduction in pain & bloating + ≥2 increase in CSBM/wk
Multi-symptom composite responders in the same week for ≥6 of 12 weeks

CSBM, complete spontaneous bowel movement; EAS, efficacy analysis set; wk, week.

Safety

- The adverse events occurring during the individual studies have been previously reported.[5-7]
- In this population, 37.8% of all patients experienced a treatment-emergent adverse event (TEAE). In the tenapanor group, the most common TEAE related to study drug was diarrhea (**Table 2**, 13.1%), which is consistent with previously reported diarrhea rates among tenapanor-treated patients with bloating of any severity (14.8%).[4]

Table 2: Overview of TEAEs in Patients With Moderate to Severe Bloating at Baseline (Pooled EAS Analysis Set)

TEAEs, n (%)	Placebo bid (N=625)	Tenapanor 50 mg bid (N=628)	Overall (N=1253)
Any TEAE	207 (33.1)	267 (42.5)	474 (37.8)
Study drug–related TEAEs	54 (8.6)	124 (19.7)	178 (14.2)
Serious TEAEs	7 (1.1)	8 (1.3)	15 (1.2)
Deaths	0	0	0
Study drug–related TEAEs by preferred term[a]			
Diarrhea	10 (1.6)	82 (13.1)	92 (7.3)
Flatulence	10 (1.6)	14 (2.2)	24 (1.9)
TEAEs leading to study drug discontinuation by preferred term[b]	7 (1.1)	45 (7.2)	52 (4.2)
Diarrhea	4 (0.6)	37 (5.9)	41 (3.3)

bid, twice a day; EAS, efficacy analysis set; TEAE, treatment-emergent adverse event.
[a]TEAEs by the preferred term occurring in ≥2.0% of patients in the tenapanor group and at a higher incidence than in the placebo group. [b]TEAEs leading to study drug discontinuation by the preferred term occurring in ≥2.0% of patients in the tenapanor group and at a higher incidence than in the placebo group.

Limitations

- There is currently no standardized classification for categorizing bloating severity (ie, mild, moderate, or severe).
- This post hoc analysis may not be representative of the real-world population with IBS-C due to the stringent criteria involved in the enrollment of patients with IBS-C into clinical trials.
- Patient drug adherence is usually higher in a clinical trial setting than in clinical practice.
- A high placebo response is commonly observed in clinical studies, particularly those relying on patient-reported outcomes such as pain, constipation symptoms, depression, and QoL measures.

Conclusions

Tenapanor may be effective in reducing bloating–a persistent and bothersome symptom in a large percentage of patients with IBS-C.

In patients with moderate to severe bloating, tenapanor significantly improved the combined symptoms of abdominal bloating, abdominal pain, and CSBM frequency.

Onset of a clinically meaningful reduction was seen as early as week 1 and was sustained through the duration of treatment.

Disclosures

This study was supported by Ardelyx, Inc. Kyle Staller has received research funding from Ardelyx, served as a consultant for AbbVie, Ardelyx, Atmo, Ferring, Gemelli Biotech, Laborie, Salix, and Takeda, and served on the advisory board of Mindset Health. Yang Yang, Suling Zhao, and Susan Edelstein are employees of Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

References

1. Neri L et al. *Neurogastroenterol Motil*. 2016;28:581-91.
2. Lacy BE et al. *Gastroenterology*. 2016;150:1393-407.
3. IBSRELA (tenapanor hydrochloride). Prescribing information. Ardelyx Inc.; 2025.
4. Lembo AJ et al. *Am J Gastroenterol*. 2024;119:937-45.
5. Chey WD et al. *Am J Gastroenterol*. 2017;112:763-74.
6. Chey WD et al. *Am J Gastroenterol*. 2020;115:281-93.
7. Chey WD et al. *Am J Gastroenterol*. 2021;116:1294-303.

Kyle Staller can be contacted for further information on this study at KSTALLER@mgh.harvard.edu.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American Association of Nurse Practitioners (AANP) 2026 National Conference; June 23-27, 2026; Las Vegas, NV, USA.

MED-US—2600263 06/2026

IBSRELA® (tenapanor) is indicated for treatment of irritable bowel syndrome with constipation (IBS-C) in adults

Important Safety Information

WARNING: RISK OF SERIOUS DEHYDRATION IN PEDIATRIC PATIENTS

- **IBSRELA is contraindicated in patients less than 6 years of age; in nonclinical studies in young juvenile rats, administration of tenapanor caused deaths presumed to be due to dehydration. *[see PI Contraindications (4), Use in Specific Populations (8.4)]*.**
- **Avoid use of IBSRELA in patients 6 years to less than 12 years of age. *[see PI Warnings and Precautions (5.1), Use in Specific Populations (8.4)]*.**
- **The safety and effectiveness of IBSRELA have not been established in pediatric patients less than 18 years of age. *[see PI Use in Specific Populations (8.4)]*.**

CONTRAINDICATIONS

IBSRELA is contraindicated in patients less than 6 years of age due to the risk of serious dehydration.

IBSRELA is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Risk of Serious Dehydration in Pediatric Patients

IBSRELA is contraindicated in patients below 6 years of age. The safety and effectiveness of IBSRELA in patients less than 18 years of age have not been established. In young juvenile rats (less than 1 week old; approximate human age equivalent of less than 2 years of age), decreased body weight and deaths occurred, presumed to be due to dehydration, following oral administration of tenapanor. There are no data available in older juvenile rats (human age equivalent 2 years to less than 12 years).

Avoid the use of IBSRELA in patients 6 years to less than 12 years of age. Although there are no data in older juvenile rats, given the deaths in younger rats and the lack of clinical safety and efficacy data in pediatric patients, avoid the use of IBSRELA in patients 6 years to less than 12 years of age.

Diarrhea

Diarrhea was the most common adverse reaction in two randomized, double-blind, placebo-controlled trials of IBS-C. Severe diarrhea was reported in 2.5% of IBSRELA-treated patients. If severe diarrhea occurs, suspend dosing and rehydrate patient.

MOST COMMON ADVERSE REACTIONS

The most common adverse reactions in IBSRELA-treated patients (incidence ≥2% and greater than placebo) were: diarrhea (16% vs 4% placebo), abdominal distension (3% vs <1%), flatulence (3% vs 1%) and dizziness (2% vs <1%).

For additional safety information, including the Boxed Warning, please see full Prescribing Information: click here.

IBS-C, irritable bowel syndrome with constipation.

IBSRELA (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025. Available at: https://www.ibsrela-hcp.com/PI

© Ardelyx, Inc., 2026. All rights reserved. Ardelyx and IBSRELA are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only