Poster TH-PO0223

Improvement in Bowel Movements Seen in Patients With End-Stage Kidney Disease and Mild to Severe Constipation After Treatment With Tenapanor

Steven Fishbane,[1] Suling Zhao,[2] Yang Yang,[2] Susan Edelstein,[2] David M. Spiegel[2]

[1]Zucker School of Medicine at Hofstra & Northwell Health, Great Neck, NY, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Introduction

- Tenapanor is a first-in-class, minimally absorbed inhibitor of sodium/hydrogen exchanger isoform 3, which decreases intestinal phosphate permeability through the paracellular pathway, the primary route of gastrointestinal phosphate absorption.[1,2]
- Tenapanor is approved to reduce serum phosphorus in adults with chronic kidney disease on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.[3]
- Constipation is commonly reported among patients with end-stage kidney disease (ESKD) receiving dialysis, and while uncomfortable for all, it often impairs the effectiveness of peritoneal dialysis.[4,5]
- The randomized, placebo-controlled phase 3 BLOCK study (NCT02675998) evaluated tenapanor in adult patients with hyperphosphatemia receiving maintenance hemodialysis and demonstrated that tenapanor reduced serum phosphate (P).[6] This post hoc analysis of the BLOCK study examined changes in stool frequency and consistency in patients treated with tenapanor who experienced constipation at baseline.

Methods

- This post hoc analysis was based on the data from the 8-week randomized treatment period (RTP) of the BLOCK study (**Figure 1**).[6] Patients with an increase in serum P of ≥1.5 mg/dL and absolute values from 6.0 mg/dL to <10.0 mg/dL after the phosphate binder washout period were eligible for randomization.

Figure 1: Study Design of BLOCK[6]

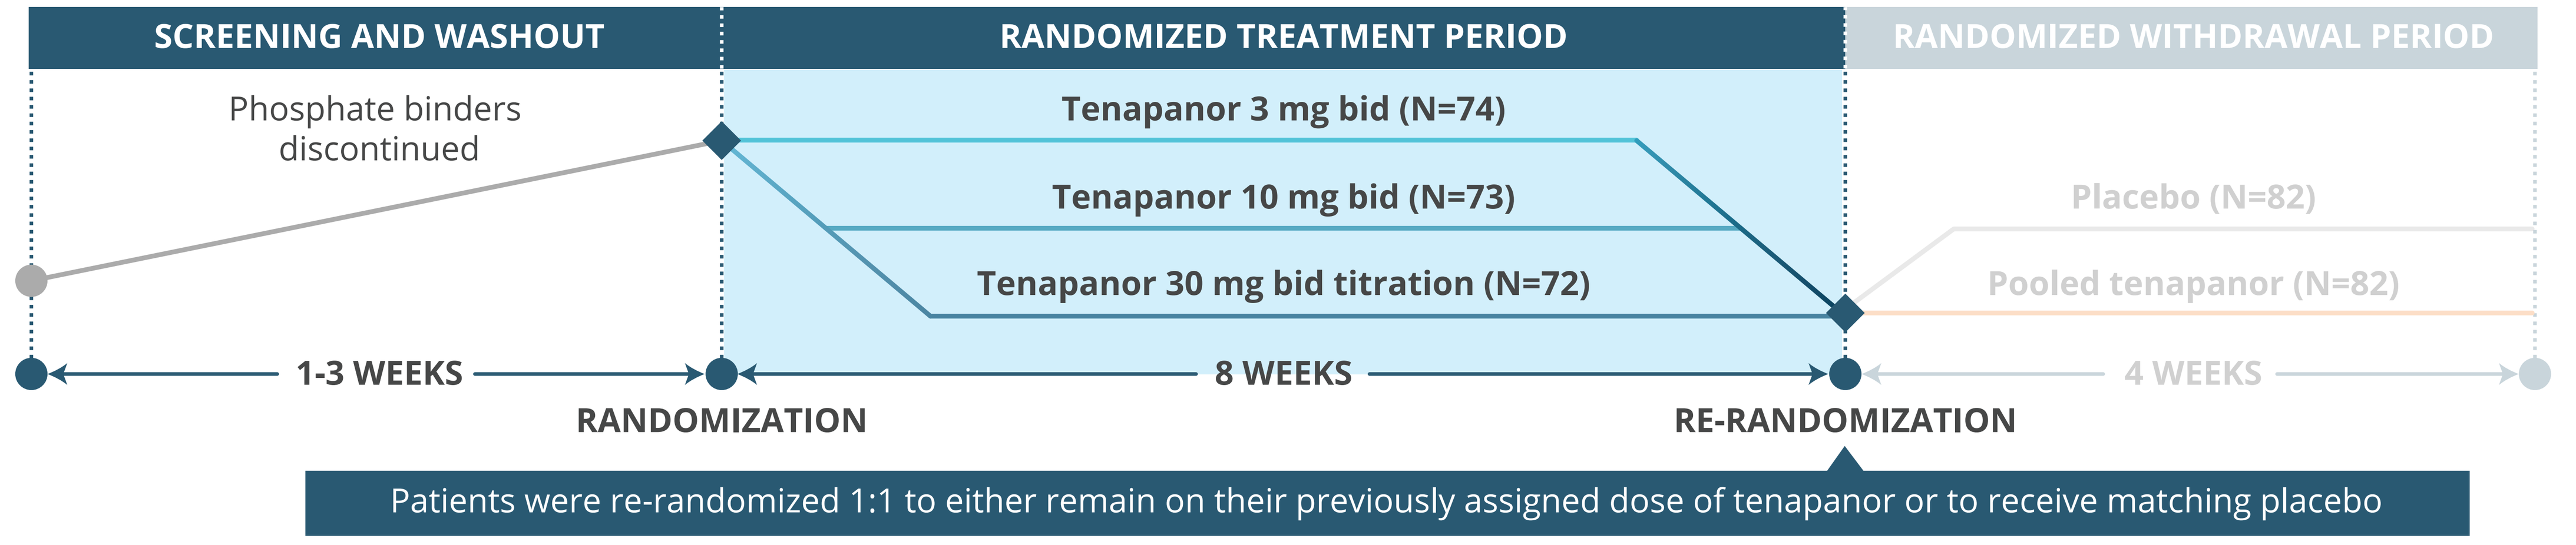
SCREENING AND WASHOUT
RANDOMIZED TREATMENT PERIOD
RANDOMIZED WITHDRAWAL PERIOD
Phosphate binders discontinued
Tenapanor 3 mg bid (N=74)
Tenapanor 10 mg bid (N=73)
Tenapanor 30 mg bid titration (N=72)
Placebo (N=82)
Pooled tenapanor (N=82)
1-3 WEEKS
8 WEEKS
4 WEEKS
RANDOMIZATION
RE-RANDOMIZATION
Patients were re-randomized 1:1 to either remain on their previously assigned dose of tenapanor or to receive matching placebo

bid, twice a day.

- Patients from all 3 tenapanor dose groups were included in this analysis.
- Daily eDiary entries were used to assess stool frequency and consistency via the Bristol Stool Form Scale (BSFS; **Figure 2**). Stool consistency between type 3 and type 5 is considered normal.[7]
- Patients were categorized for the purposes of this post hoc analysis into 2 constipation subgroups based on baseline weekly stool frequency (BWSF): ≤6 (constipation) and ≤3 (severe constipation).
- The BWSF was derived as the mean of the weekly stool frequency values across all valid study weeks during the screening period. A study week was considered valid if it had at least 4 days of reporting on spontaneous bowel movement (SBM). For a valid study week, the weekly stool frequency was calculated using the following formula:

$$\text{Weekly stool frequency} = \left(\frac{\sum \text{Daily SBM frequencies in that study week}}{\text{Number of days with reporting of SBM in that study week}}\right) \times 7$$

- The baseline weekly stool consistency score was derived as the mean of the weekly stool consistency scores across all valid study weeks during the screening period. For a valid study week, the weekly stool consistency score was calculated using the following formula:

$$\text{Weekly stool consistency score} = \frac{\sum \text{Stool consistency scores from valid SBMs in that study week}}{\text{Number of valid SBMs + number of days with 0 valid SBMs in that study week}}$$

Figure 2: Bristol Stool Form Scale

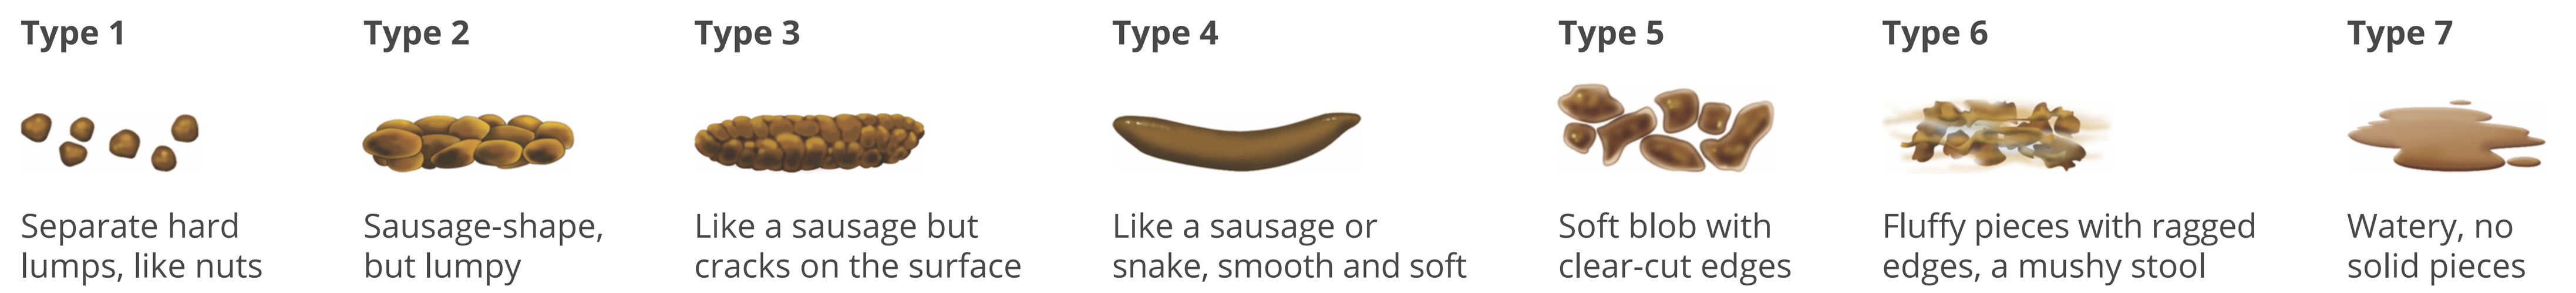
Type 1
Separate hard lumps, like nuts
Type 2
Sausage-shape, but lumpy
Type 3
Like a sausage but cracks on the surface
Type 4
Like a sausage or snake, smooth and soft
Type 5
Soft blob with clear-cut edges
Type 6
Fluffy pieces with ragged edges, a mushy stool
Type 7
Watery, no solid pieces

©2000 Rome Foundation. All Rights Reserved.

Results

Patients

Table 1: Baseline Demographics and Disease Characteristics

	BWSF ≤6 at BL (N=59)	BWSF ≤3 at BL (N=8)
Mean (SD) age, y	53.5 (10.4)	48.3 (11.0)
Male, n (%)	35 (59.3)	7 (87.5)
Race, n (%)		
Black or African American	35 (59.3)	3 (37.5)
White	23 (39)	4 (50)
Other	1 (1.7)	1 (12.5)
Ethnicity, n (%)		
Non-Hispanic/Latino	51 (86.4)	5 (62.5)
Hispanic/Latino	8 (13.6)	3 (37.5)
Mean (SD) BMI, kg/m²	34.2 (8.4)	32.9 (7.9)
Mean (SD) duration of dialysis treatment, y	4.7 (4.9)	4.8 (3.5)
Mean (SD) serum P, mg/dL	7.4 (1.4)	7.3 (1.3)

BL, baseline; BMI, body mass index; BWSF, baseline weekly stool frequency; P, phosphate.

Stool Frequency and Consistency

- In patients with BWSF ≤6 (N=59):
 - Mean weekly stool frequency increased from a mean (SE) of 4.7 (0.2) at baseline to 8.2 (0.7) (**Figure 3A**), and mean weekly stool consistency score improved from a mean (SE) of 2.7 (0.1) to 4.0 (0.2) (**Figure 3B**) during the end-of-treatment (EOT) week of the RTP.
 - The mean (SE) change from baseline to EOT of the RTP in mean weekly stool frequency was 3.5 (0.7) (*P*<0.001; **Figure 3C**) and in mean weekly stool consistency was 1.4 (0.2) (*P*<0.001; **Figure 3D**).
- In patients with BWSF ≤3 (N=8):
 - Mean weekly stool frequency increased from severe constipation at baseline (mean [SE] of 2.3 [0.3]) to within the normal range (6.3 [1.9]; **Figure 3A**), and mean weekly stool consistency score improved from a mean (SE) BSFS of 1.4 (0.1) at baseline to 3.3 (0.7) during the EOT week of the RTP (**Figure 3B**).
 - The mean (SE) change from baseline to EOT of the RTP in mean weekly stool frequency was 4.0 (2.1) (*P*<0.05; **Figure 3C**) and in mean weekly stool consistency was 1.9 (0.7) (*P*=0.02; **Figure 3D**).

Figure 3: Mean Weekly and Mean Change in Weekly (A, C) Stool Frequency and (B, D) Stool Consistency in Patients With a BWSF ≤6 (N=59) and ≤3 (N=8)

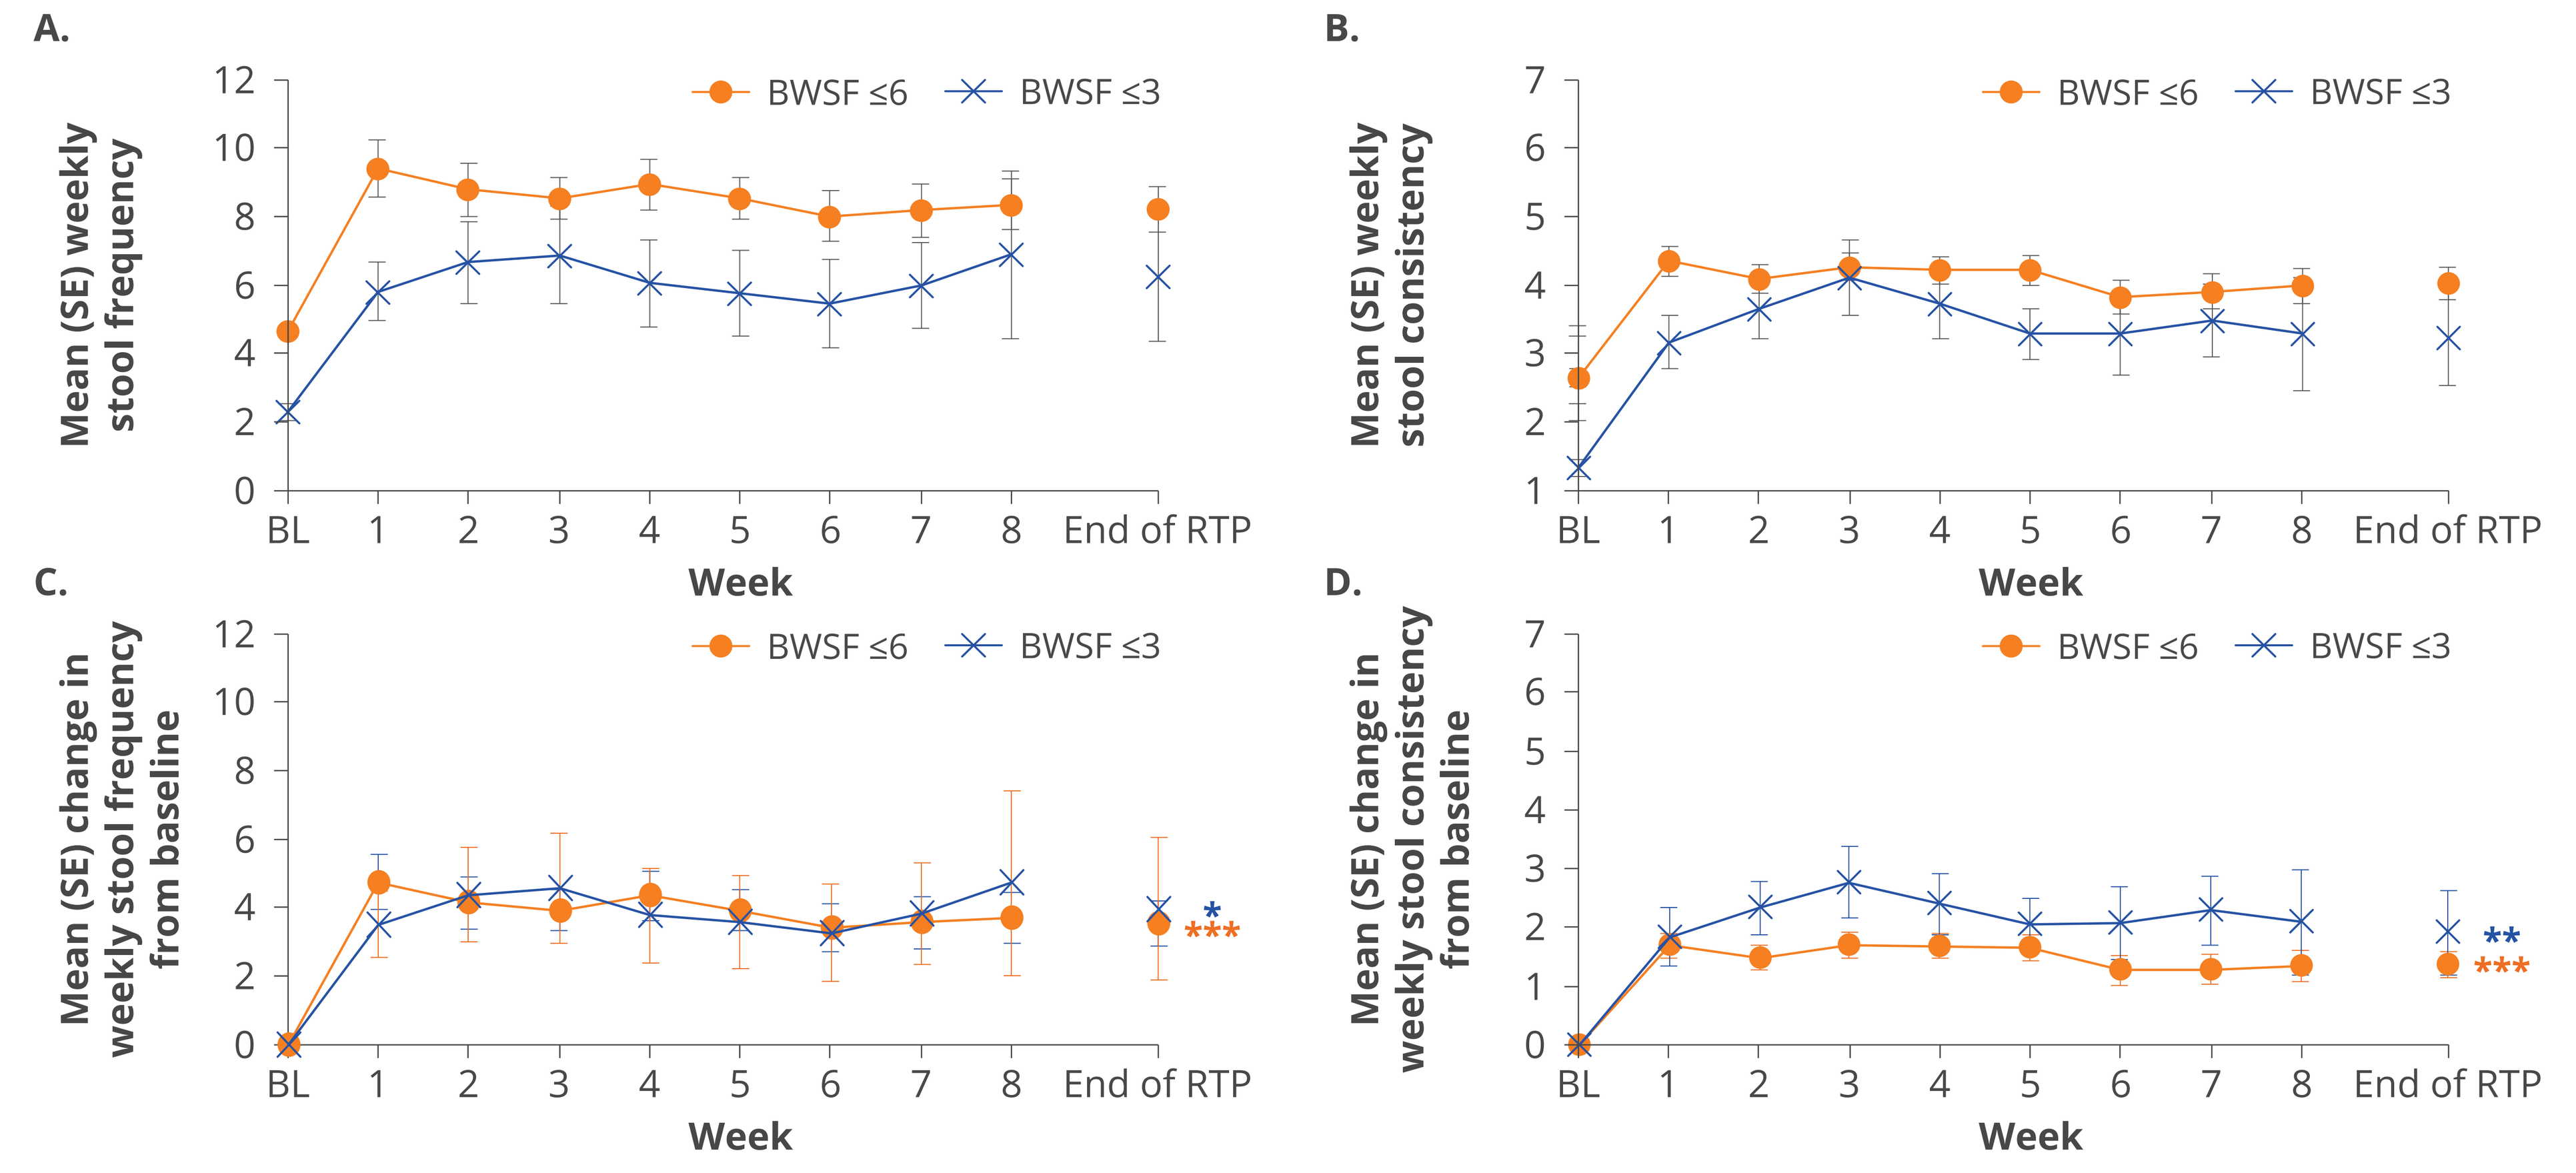
A.
Mean (SE) weekly stool frequency
BWSF ≤6
BWSF ≤3
Week
BL 1 2 3 4 5 6 7 8 End of RTP
B.
Mean (SE) weekly stool consistency
C.
Mean (SE) change in weekly stool frequency from baseline
*

D.
Mean (SE) change in weekly stool consistency from baseline
**

*P<0.05; **P=0.02; ***P<0.001. *P* values represent a pairwise t-test to evaluate the change from baseline to the end of RTP.
BL, baseline; BWSF, baseline weekly stool frequency; RTP, randomized treatment period.

Serum P

- After the 8-week RTP, similar reductions in serum P (mean [SE]) from baseline were observed in both constipation subgroups (BWSF ≤6, 1.0 [0.2] mg/dL; BWSF ≤3, 1.1 [0.7]), which was consistent with serum P reduction observed in the overall population in the BLOCK study.

Safety

- Safety outcomes in BLOCK have been previously reported[6]; the safety profile of tenapanor in this population was consistent with that among the overall population **(Table 2**).

Table 2: Overview of Treatment-Emergent Adverse Events

TEAE, n (%)	BWSF ≤6 (N=59)	BWSF ≤3 (N=8)
Patients with any TEAE	38 (64.4)	6 (75.0)
Serious TEAE	7 (11.9)	1 (12.5)
TEAEs experienced by ≥5% patients		
Diarrhea	24 (40.7)	5 (62.5)
Abdominal discomfort	3 (5.1)	1 (12.5)
Flatulence	3 (5.1)	2 (25.0)
Hyperphosphatemia	4 (6.8)	N/A
Hypocalcemia	3 (5.1)	N/A
TEAEs leading to study drug discontinuation by preferred term[a]	12 (20.3)	0
Diarrhea	9 (15.3)	0

[a]TEAEs leading to study drug discontinuation by the preferred term occurring in ≥2.0% of tenapanor-treated patients.
BWSF, baseline weekly stool frequency; TEAE, treatment-emergent adverse event.

Limitations

- In the BLOCK study, data on the use of rescue medication for constipation were not collected from the patients.
- No placebo control was included in the RTP.
- Due to the small sample size, the data should be viewed as exploratory in nature.

Conclusion

The mean weekly stool frequency increased, and the mean weekly stool consistency improved from constipation at baseline to normal in patients with ESKD who were taking tenapanor to reduce serum P.

Disclosures

This study was supported by Ardelyx, Inc. Steven Fishbane served as a scientific advisor for Ardelyx, Inc. Suling Zhao, Yang Yang, Susan Edelstein, and David M. Spiegel are employees of Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

References

1. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
2. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
3. XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx Inc.; 2025.
4. Sumida K et al. *Kidney Int Rep*. 2019;5:121-34.
5. Kosmadakis G et al. *Perit Dial Int*. 2019;39:399-404.
6. Block GA et al. *J Am Soc Nephrol*. 2019;30:641-52.
7. Blake MR et al. *Aliment Pharmacol Ther*. 2016;44:693-703.

Steven Fishbane can be contacted for further information on this study at Sfishbane@northwell.edu.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American Society of Nephrology (ASN) 2025 Kidney Week; November 6-9, 2025; Houston, TX, USA.

MED-US—2500506 11/2025

XPHOZAH (tenapanor) Important Safety Information and Indication

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in XPHOZAH-treated patients were reported to be mild-to-moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, click here.

CKD, chronic kidney disease.
XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only