XPHOZAH (tenapanor) Important Safety Information and Indication

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in XPHOZAH-treated patients were reported to be mild-to-moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, click here.

CKD, chronic kidney disease.

XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2023.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only
MED-US—2500214 04/2025

Poster number: G-310

Tenapanor-Treated Patients Using Over-the-Counter Antidiarrheal Agents Had Decreased Stool Frequency and Improved Stool Consistency

Steven Fishbane,[1] Suling Zhao,[2] Susan Edelstein,[3] Yang Yang,[2] and David M. Spiegel[3]

[1]Northwell Health, Great Neck, NY, USA; [2] Ardelyx, Inc., Fremont, CA, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Introduction

- Tenapanor is a first-in-class, minimally absorbed inhibitor of sodium/hydrogen exchanger isoform 3, which decreases intestinal phosphate permeability of the paracellular pathway, the primary route of gastrointestinal phosphate absorption.[1-3]
- Tenapanor is approved to reduce serum phosphorus (P) in adults with chronic kidney disease on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.[4]
- Diarrhea, the most common adverse drug reaction observed across trials, was mostly mild to moderate in severity and often resolved within approximately 2 weeks; however, it was the primary reason for discontinuation of tenapanor in clinical trials.[4-6]
- The post-registrational, randomized, open-label OPTIMIZE study (NCT04549597) evaluated tenapanor alone or in combination with phosphate binders at a reduced dose and demonstrated that tenapanor reduced serum P in patients on maintenance dialysis.[7]
- In OPTIMIZE, patients were counseled to expect changes in their stool frequency and consistency, and they were also instructed that antidiarrheal agents such as loperamide (Imodium®) could be used to address the diarrhea. Patients were further advised to discontinue use of stool softeners and laxatives prior to initiating tenapanor.[7]
- This post hoc analysis looked at tenapanor discontinuation rates and changes in stool frequency and consistency in patients who self-medicated with loperamide.

Methods

- Methods for OPTIMIZE have been previously described[7]; briefly:
 - OPTIMIZE enrolled patients on maintenance dialysis with serum P >5.5 and ≤10.0 mg/dL who were taking phosphate binders (PBs) or patients who were PB naive with serum P >4.5 and ≤10.0 mg/dL.
 - All patients initiated tenapanor treatment at a dose of 30 mg twice a day (bid) upon enrollment. PB-naive patients received tenapanor alone, and patients taking PBs were randomized to discontinue PBs and add tenapanor treatment or add tenapanor treatment with a binder dose reduction of ≥50%.
 - The tenapanor dose could be titrated any time during the study based on serum P and/or gastrointestinal tolerability. The PB dose could also be adjusted at any time based on serum P, except that dose up-titration was not allowed until week 2.
 - Serum P was measured over a 10-week efficacy period followed by an optional 16-week safety extension.
- Daily eDiary entries tracked stool frequency and consistency via the Bristol Stool Form Scale (BSFS; **Figure 1**) before and after loperamide initiation.
- Loperamide could be initiated during the study.
- The demographics, safety, and medication burden were summarized by use of loperamide.
- The eDiary results were compared for the week before and after initiating loperamide during the study.
- Only patients who initiated loperamide during the study were included, while those who were already using loperamide before the study and continued treatment throughout the study were excluded from this analysis.
- Time points were defined as follows:
 - The week before initiating loperamide: Any eDiary entry recorded within 7 days before loperamide use.
 - The week after initiating loperamide: Any eDiary entry recorded within 7 days after starting loperamide.
- Discontinuation of tenapanor was assessed via 2 categories: “Study discontinuation” and “Diarrhea leading to tenapanor discontinuation.”
 - “Study discontinuation” describes patients who prematurely discontinued from the study due to various reasons, such as adverse events (AEs), withdrawn consent, and protocol violation.
 - “Diarrhea leading to tenapanor discontinuation” describes patients reporting an AE of diarrhea that led to premature discontinuation of tenapanor treatment.

Figure 1: Bristol Stool Form Scale

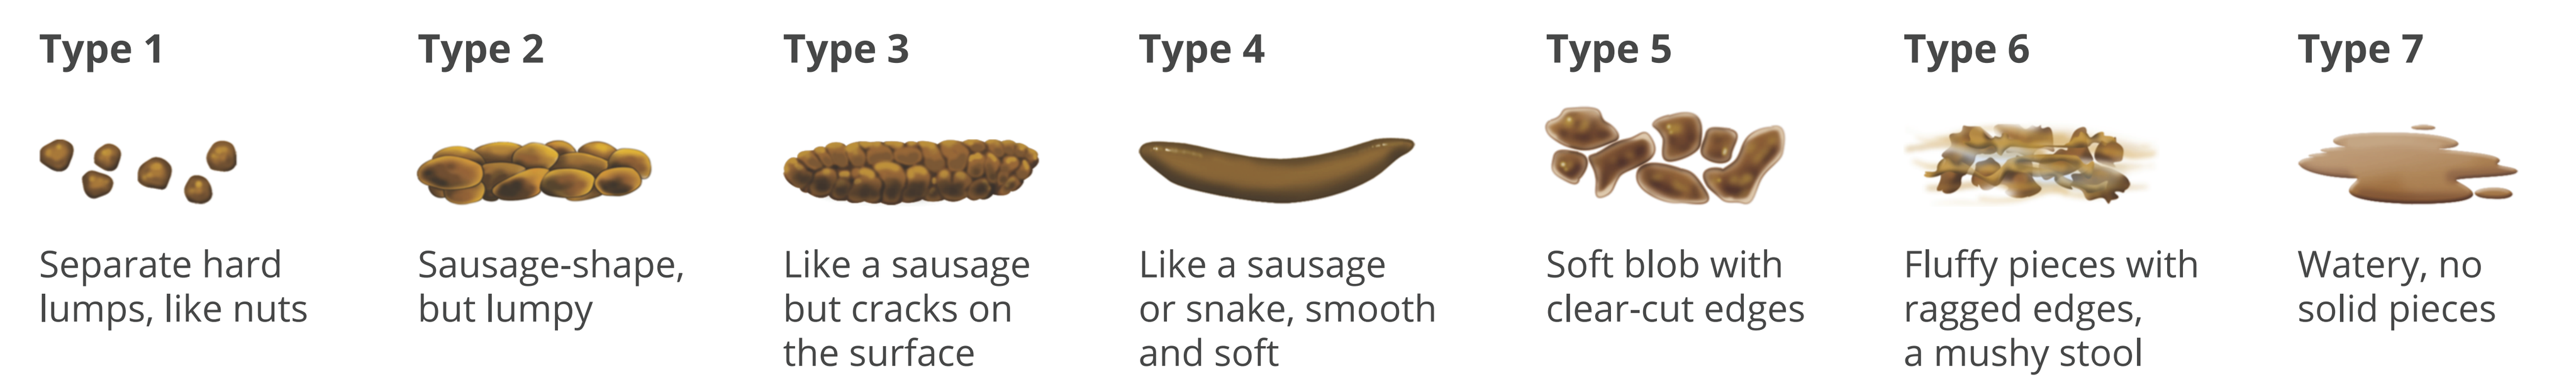

Type 1
Separate hard lumps, like nuts
Type 2
Sausage-shape, but lumpy
Type 3
Like a sausage but cracks on the surface
Type 4
Like a sausage or snake, smooth and soft
Type 5
Soft blob with clear-cut edges
Type 6
Fluffy pieces with ragged edges, a mushy stool
Type 7
Watery, no solid pieces

©2000 Rome Foundation, Inc. All Rights Reserved.

Results

Patients

- Among 333 tenapanor-treated patients in OPTIMIZE, 102 patients (30.6%) self-medicated with loperamide. Baseline demographics and characteristics of these patients are presented in **Table 1**.

Table 1: Baseline Demographics and Disease Characteristics (Safety Population)

	Use of loperamide (N=102)	No use of loperamide (N=231)	Total (N=333)
Age, mean (SD), y	53.9 (11.3)	52.7 (12.5)	53.0 (12.1)
Male, n (%)	66 (64.7)	161 (69.7)	227 (68.2)
Race, n (%)			
Asian	2 (2.0)	14 (6.1)	16 (4.8)
Black or African American	47 (46.1)	99 (42.9)	146 (43.8)
White	41 (40.2)	105 (45.5)	146 (43.8)
Other	12 (11.8)	13 (5.6)	25 (7.5)
Ethnicity, n (%)			
Non-Hispanic/Latino	78 (76.5)	162 (70.1)	240 (72.1)
Hispanic/Latino	24 (23.5)	69 (29.9)	93 (27.9)
Mean BMI, kg/m² (SD)	32.8 (8.5)	31.9 (8.3)	32.2 (8.3)
Type of dialysis, n (%)			
Hemodialysis	88 (86.3)	180 (78.0)	268 (80.5)
Peritoneal dialysis	14 (13.7)	51 (22.1)	65 (19.5)
Duration of dialysis treatment, mean (SD), mo	62.6 (51.3)	50.4 (49.2)	54.1 (50.1)
BL serum P, mean (SD), mg/dL	6.9 (1.0)	7.1 (1.1)	7.1 (1.1)

BL, baseline; BMI, body mass index; P, phosphorus.

Diarrhea Leading to Tenapanor Discontinuation

- Diarrhea was reported in 39.9% (133 of 333) of patients during the entire study.
 - Of these patients, 54.1% (72 of 133) initiated the use of loperamide.
- Among patients reporting diarrhea:
 - Study discontinuation rates in patients using loperamide (20.8%, 15 of 72) were nearly half that of patients not using loperamide (37.7%, 23 of 61; **Figure 2**).
 - Incidence of diarrhea leading to tenapanor discontinuation in patients using loperamide (9.7%, 7 of 72) was less than half that of patients not using loperamide (24.6%, 15 of 61; **Figure 2**).

Figure 2: Discontinuation Rates by Use of Loperamide (Safety Population With Reported Diarrhea)

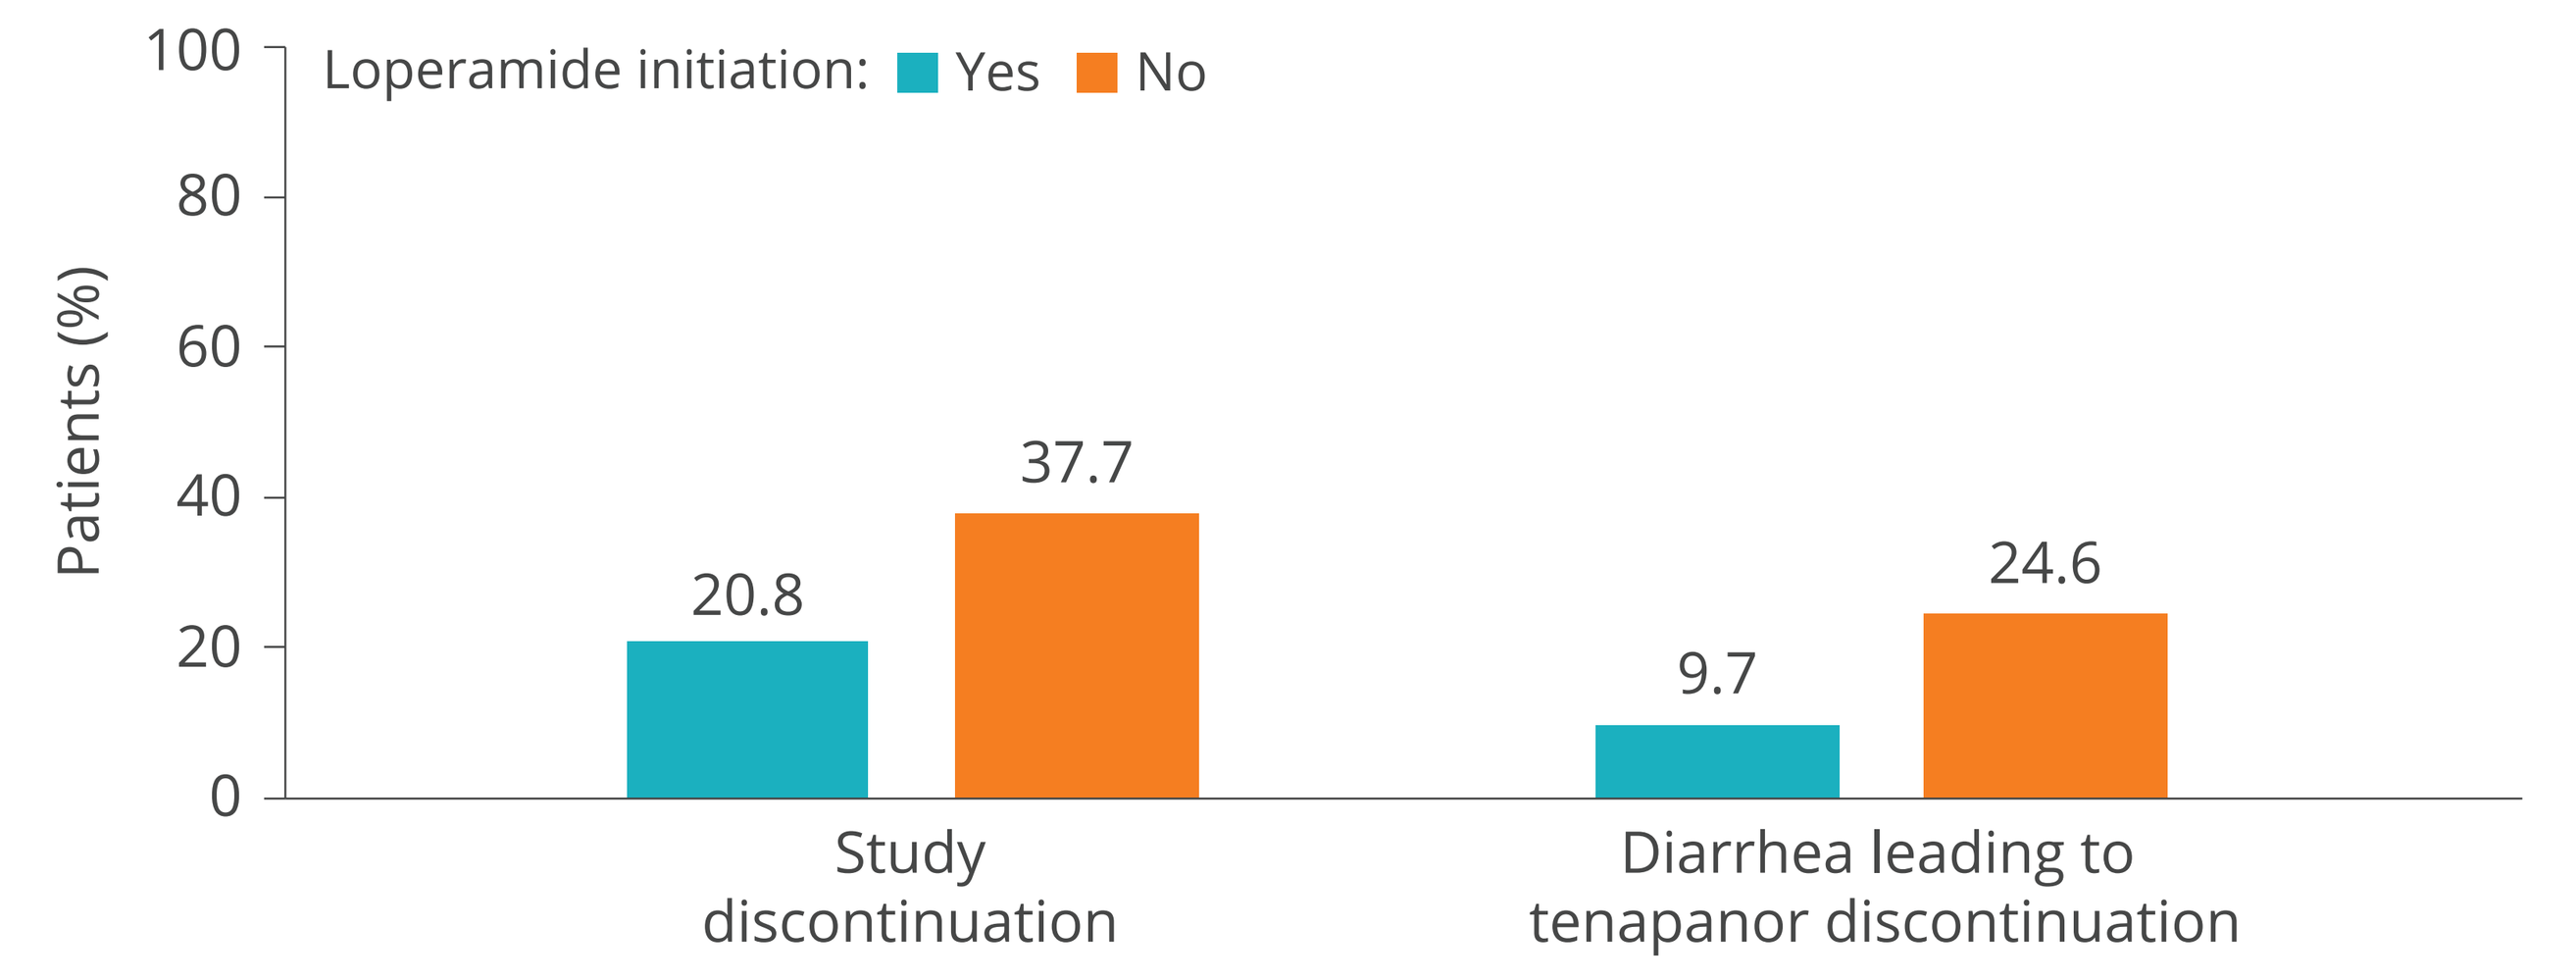

100
80
60
40
20
0
Patients (%)
Loperamide initiation: Yes No
20.8
37.7
9.7
24.6
Study discontinuation
Diarrhea leading to tenapanor discontinuation

Stool Frequency and Consistency

- Among patients who initiated loperamide during the study (n=102), 62 had sufficient daily stool frequency entries from both the week before and week after initiating loperamide for the derivation of the average weekly stool frequency (AWSF).
- Among these 62 patients, the mean (SD) AWSF decreased from 11.7 (6.6) to 11.0 (5.7) after initiating loperamide. Of these patients:
 - Fifty percent (31 of 62) had an AWSF >10 before initiating loperamide. In the AWSF >10 subset, the mean (SD) AWSF decreased from 16.9 (5.3) to 13.6 (6.6) after initiating loperamide, with a mean (SD) reduction of 3.3 (5.2) (**Figure 3A**).
 - The mean (SD) value of the average weekly stool consistency score remained stable from 5.8 (0.8) to 5.9 (0.8) before and after initiating loperamide, with a mean (SD) change of 0.1 (0.9) (**Figure 3B**).
 - Twenty-nine percent (18 of 62) had an AWSF >14 before initiating loperamide. In the AWSF >14 subset, the mean (SD) AWSF decreased from 20.1 (4.6) to 15.6 (7.1) after initiating loperamide, with a mean (SD) reduction of 4.5 (5.4) (**Figure 3A**).
 - The mean (SD) value of the average weekly stool consistency score changed from 6.0 (0.6) to 5.8 (0.9) before and after initiating loperamide, with a mean (SD) reduction of 0.3 (0.9) (**Figure 3B**).
- A total of 61 patients initiated loperamide during the study and had sufficient daily stool consistency entries from both the week before and week after initiating loperamide for the derivation of the average weekly stool consistency score.
- Among these 61 patients, the mean (SD) average weekly stool consistency score remained consistent, at 5.7 (1.0) and 5.8 (0.9) before and after initiating loperamide, respectively.
 - Of these patients, 44.3% (27 of 61) had an average weekly stool consistency score of >6 before initiating loperamide, with the mean (SD) decreased from 6.6 (0.3) to 6.0 (0.9) after initiation of loperamide.

Figure 3: (A) AWSF and (B) Consistency Before and After Loperamide Initiation (Safety Population With a High AWSF Before Initiating Loperamide)

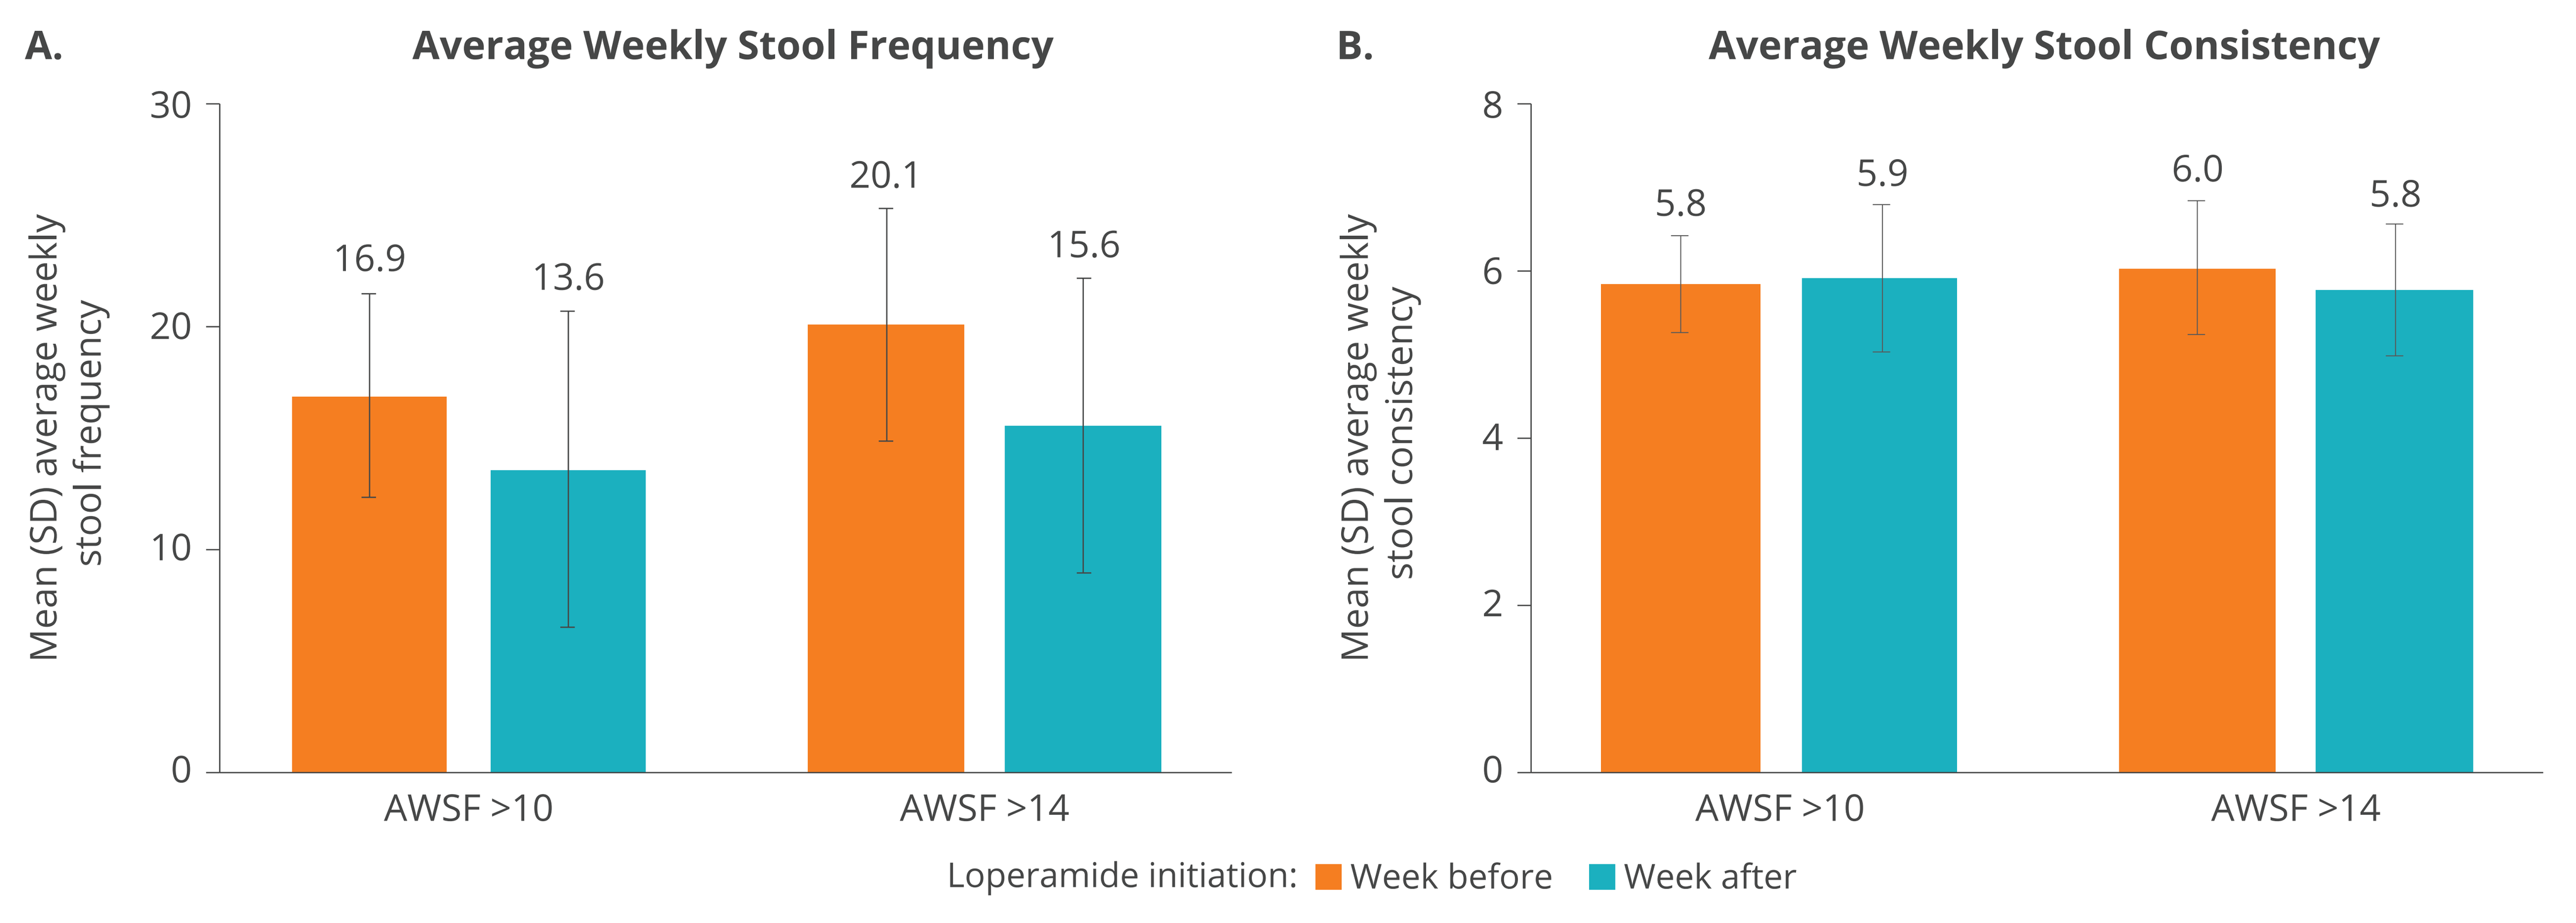

A.
Average Weekly Stool Frequency
Mean (SD) average weekly stool frequency
30
20
10
0
16.9
13.6
20.1
15.6
AWSF >10
AWSF >14
B.
Average Weekly Stool Consistency
Mean (SD) average weekly stool consistency
8
6
4
2
0
5.8
5.9
6.0
5.8
AWSF >10
AWSF >14
Loperamide initiation: Week before Week after

AWSF, average weekly stool frequency.

- Among the patients with AWSF >10 and AWSF >14 before initiating loperamide:
 - The percentage of patients with a reduced AWSF after using loperamide was 71.0% (22 of 31) and 77.8% (14 of 18), respectively (**Figure 4**).
 - The percentage of patients with a reduced average weekly stool consistency was 51.6% (16 of 31) and 72.2% (13 of 18), respectively (**Figure 4**).

Effect of Loperamide Use on Serum P

- Serum P decreased in both patient subgroups at the end of the 10-week treatment period, with mean serum P levels of 6.2 mg/dL and 6.1 mg/dL in patients with and without loperamide use, respectively.

Figure 4: Proportion of Patients With Any Response to Loperamide Initiation (Safety Population With a High Weekly Stool Frequency Before Initiating Loperamide)

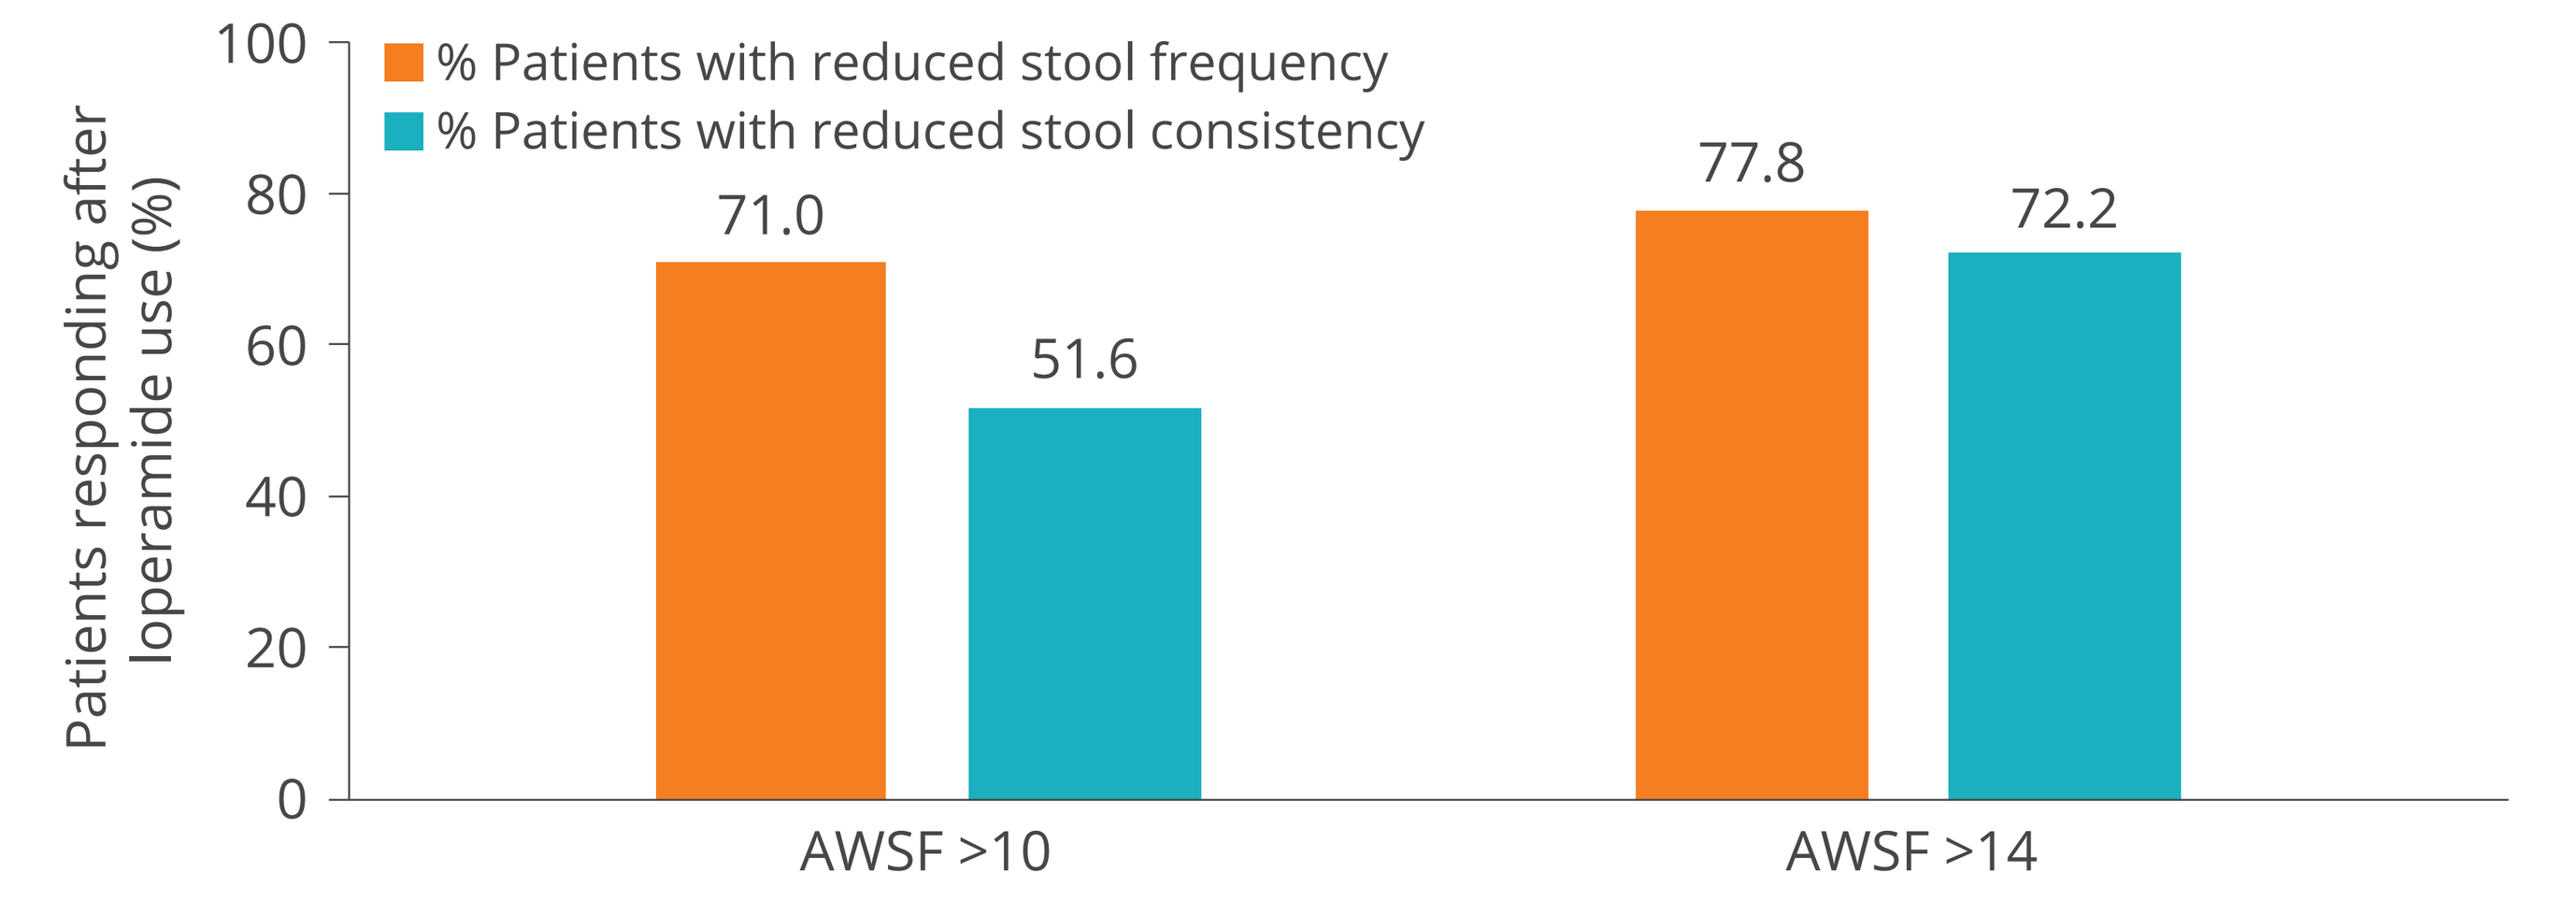

100
80
60
40
20
0
Patients responding after loperamide use (%)
% Patients with reduced stool frequency
% Patients with reduced stool consistency
71.0
51.6
77.8
72.2
AWSF >10
AWSF >14

AWSF, average weekly stool frequency.

Conclusions

- Loperamide appears to be a treatment option that reduces diarrheal symptoms that may be associated with tenapanor treatment.
- The rate of tenapanor discontinuation was lower in patients taking loperamide.
- For patients with a high weekly stool frequency, loperamide reduced average weekly stool frequency and improved stool consistency.

References

1. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
2. King AJ et al. *Sci Transl Med*. 2018;10.eaam6474.
3. Labonté ED et al. *J Am Soc Nephrol*. 2015;26:1138-49.
4. XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx Inc.; 2023.
5. Block GA et al. *Kidney360*. 2021;2:1600-10.
6. Silva AL et al. *Kidney360*. 2023;4:1580-89.
7. Sprague SM et al. *Kidney360*. 2024;5:732-42.

Acknowledgements

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc. Radha Narayan, PhD, of Ardelyx, Inc., critically reviewed the poster.

Disclosures

Steven Fishbane served as a scientific advisor for Ardelyx, Inc. Suling Zhao, Susan Edelstein, Yang Yang, and David Spiegel are full-time employees of Ardelyx, Inc.

Susan Edelstein can be contacted for further information on this study at: sedelstein@ardelyx.com

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

National Kidney Foundation – Spring Clinical Meeting 2025 (SCM25); April 9-13, 2025; Boston MA, USA

Fishbane S, et al. Am J Kidney Dis. 2025;85(4S1):S96-S97.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only
MED-US—2500214 04/2025

Study Limitations

- OPTIMIZE was an open-label study with no placebo arm; results should be interpreted accordingly.
- These data result from a post hoc analysis of OPTIMIZE and should be interpreted as hypothesis-generating.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only
MED-US—2500214 04/2025