Poster TH-PO0224

Treatment Satisfaction With Tenapanor (XPHOZAH): Real-World Survey of Patients With End-Stage Renal Disease and Hyperphosphatemia

Jessica Kendrick,[1] Luisa Scott,[2] Belinda Gist,[3] Laura Williams[2]

[1]Division of Renal Disease and Hypertension, University of Colorado Anschutz Medical Campus, Aurora, CO, USA; [2]Ardelyx, Inc., Waltham, MA, USA; [3]Employee of Ardelyx, Inc. Waltham, MA, USA, at the time of the study

Introduction

- Tenapanor (XPHOZAH), the first-in-class, minimally absorbed, sodium/hydrogen exchanger isoform 3 inhibitor, is approved to reduce serum phosphorus in adults with chronic kidney disease on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders (PBs) or who are intolerant of any dose of PB therapy.[1,2]
- Tenapanor is mechanistically distinct from PBs. It acts by reducing the permeability through the paracellular pathway, the primary pathway of phosphate absorption.[2,3]
- While clinical studies have shown the efficacy and safety of tenapanor,[4-8] real-world data on patient experiences with tenapanor are limited.
- Here, we report results from a real-world treatment satisfaction survey of patients with hyperphosphatemia and end-stage renal disease taking tenapanor.

Methods

- The survey was distributed via text messaging through the ArdelyxAssist program and coupled to pharmacy data, including dispense data and limited demographic information. Fill adherence was calculated using the following formulas:
 - $\text{Days dispensed} = \dfrac{\sum \text{Quantity of tenapanor dispensed across all dispenses}}{2}$
 - $\text{Fill adherence (\%)} = \left(\dfrac{\text{Days dispensed}}{\text{Duration of use (days)}}\right) \times 100$
- Patients on hemodialysis or peritoneal maintenance dialysis were eligible.
- Patients had to have initiated tenapanor ≥7 weeks prior to the survey and had tenapanor last dispensed in the prior 12 weeks.
- Logistic regression analysis was used to assess:
 - The change in likelihood of reporting "I don't know" for serum phosphate over time.
 - The reported change in serum phosphate from "a little" to "much better" over time.
- Chi-square analysis was used to assess whether patients with longer durations of use (≥180 days) were more likely to respond "a little better" or "much better" and less likely to respond "I don't know" when asked about their change in serum phosphate control.

Results

Patients

- Survey invites were sent to 2904 individuals who met the inclusion criteria, and 163 responders (5.6%) consented to participate in the survey.

Table 1: Demographics of Survey Participants

	Participants (N=163)
Age, median (min, max), y[a]	60 (30, 85)
Male, n (%)[a]	71 (60)
Duration of tenapanor use, median (min, max), d	133 (54, 329)
Tenapanor fill adherence, median (min, max), %	94 (32, 164)
PAP patients, n (%)	116 (71)

[a]Missing data in 45 patients.
PAP, patient assistance program.

Serum Phosphate Control

- When asked how they describe their current phosphate control with tenapanor, 39% patients (63 of 161; **Figure 1A**) responded "I don't know."
 - Excluding these, 74% (72 of 98) participants rated their current phosphate control as good or very good.
- Similarly, 57% (79 of 138) of patients did not know how their phosphate levels had changed since starting tenapanor (**Figure 1B**).
 - Excluding these, 63% (37 of 59) reported their phosphate levels were a little or much better since starting tenapanor.
- For each additional month of tenapanor use, the odds of changing their response from "a little better" to "much better" increased by approximately 21% (odds ratio [OR]=1.211, 95% CI, 1.023-1.434, P=0.026), and the odds of responding "I don't know" decreased by approximately 24% (OR=0.762, 95% CI, 0.657-0.883, P<0.001) (**Figure 1C**).

Figure 1: Patient Perspectives on Serum Phosphate Control and Improvement Since Starting Tenapanor[a]

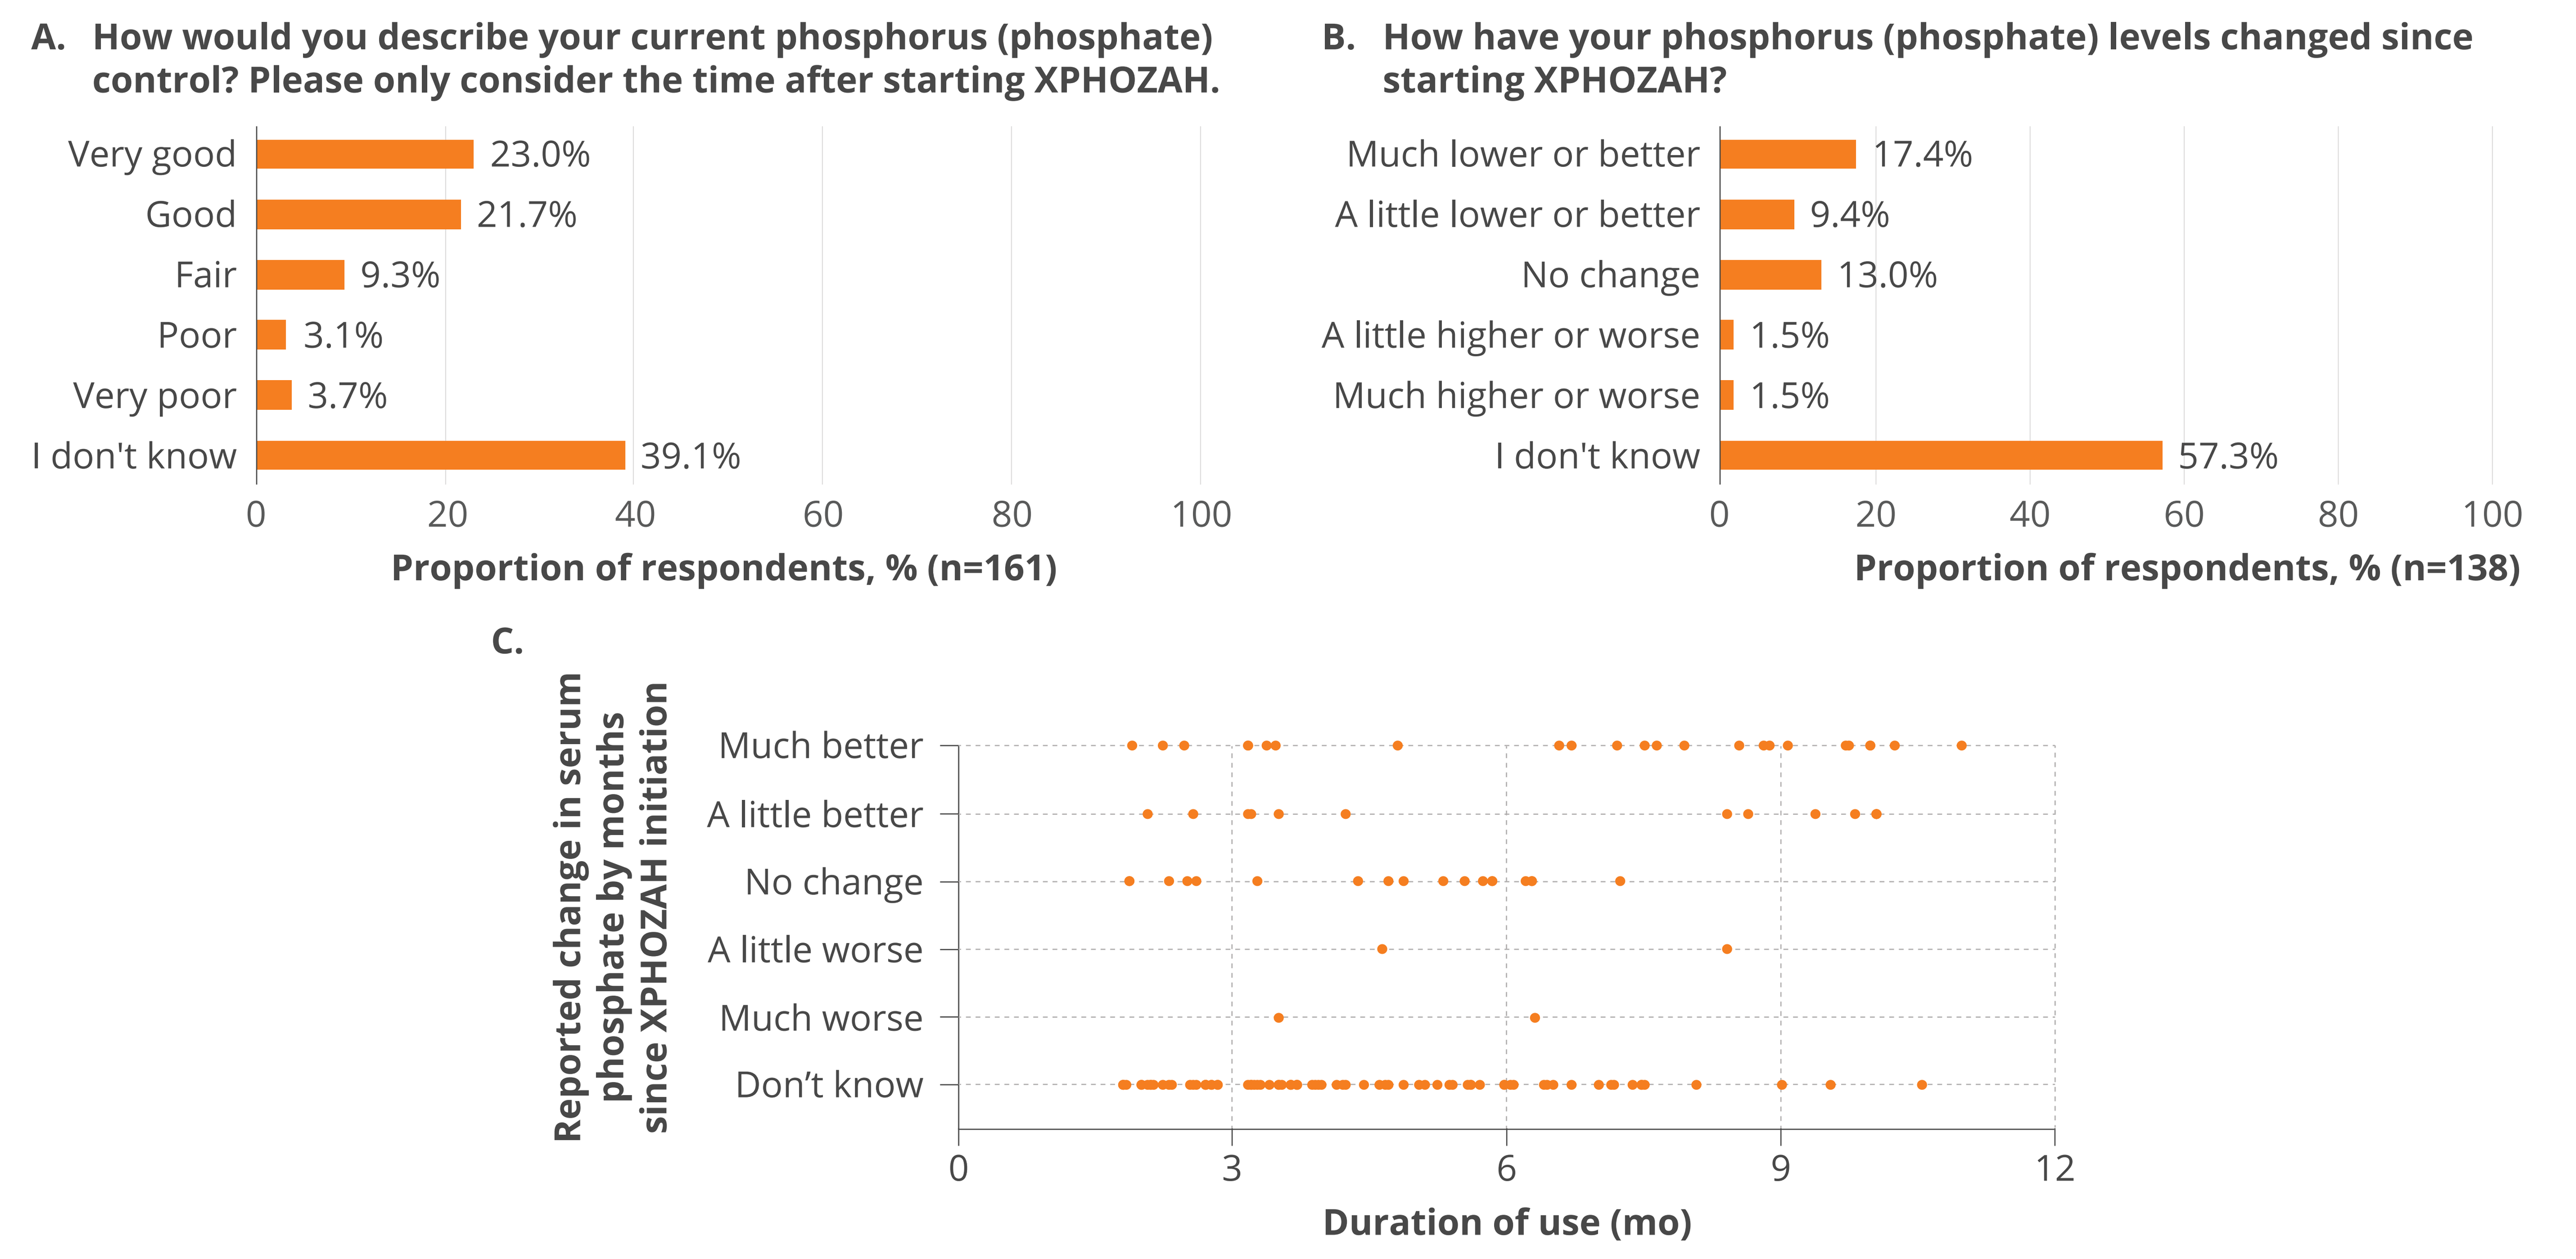
A. How would you describe your current phosphorus (phosphate) control? Please only consider the time after starting XPHOZAH.
Very good 23.0%
Good 21.7%
Fair 9.3%
Poor 3.1%
Very poor 3.7%
I don't know 39.1%
0 20 40 60 80 100
Proportion of respondents, % (n=161)
B. How have your phosphorus (phosphate) levels changed since starting XPHOZAH?
Much lower or better 17.4%
A little lower or better 9.4%
No change 13.0%
A little higher or worse 1.5%
Much higher or worse 1.5%
I don't know 57.3%
0 20 40 60 80 100
Proportion of respondents, % (n=138)
C.
Reported change in serum phosphate by months since XPHOZAH initiation
Much better
A little better
No change
A little worse
Much worse
Don't know
0 3 6 9 12
Duration of use (mo)

[a]Participants were allowed to skip questions after a prompt to respond.

Tenapanor Ease of Use and Fill Adherence

- Seventy-one percent (69 of 97) of patients reported that it was easy or very easy to take tenapanor as prescribed (**Figure 2A**).
- Fill adherence, the percentage of days during which the patient has medication available over the duration of use, was high, with a median of 94% that remained stable over time (**Figure 2B**).

Figure 2: (A) Patient Perspectives on Ease of Use and (B) Fill Adherence[a,b,c] of Tenapanor

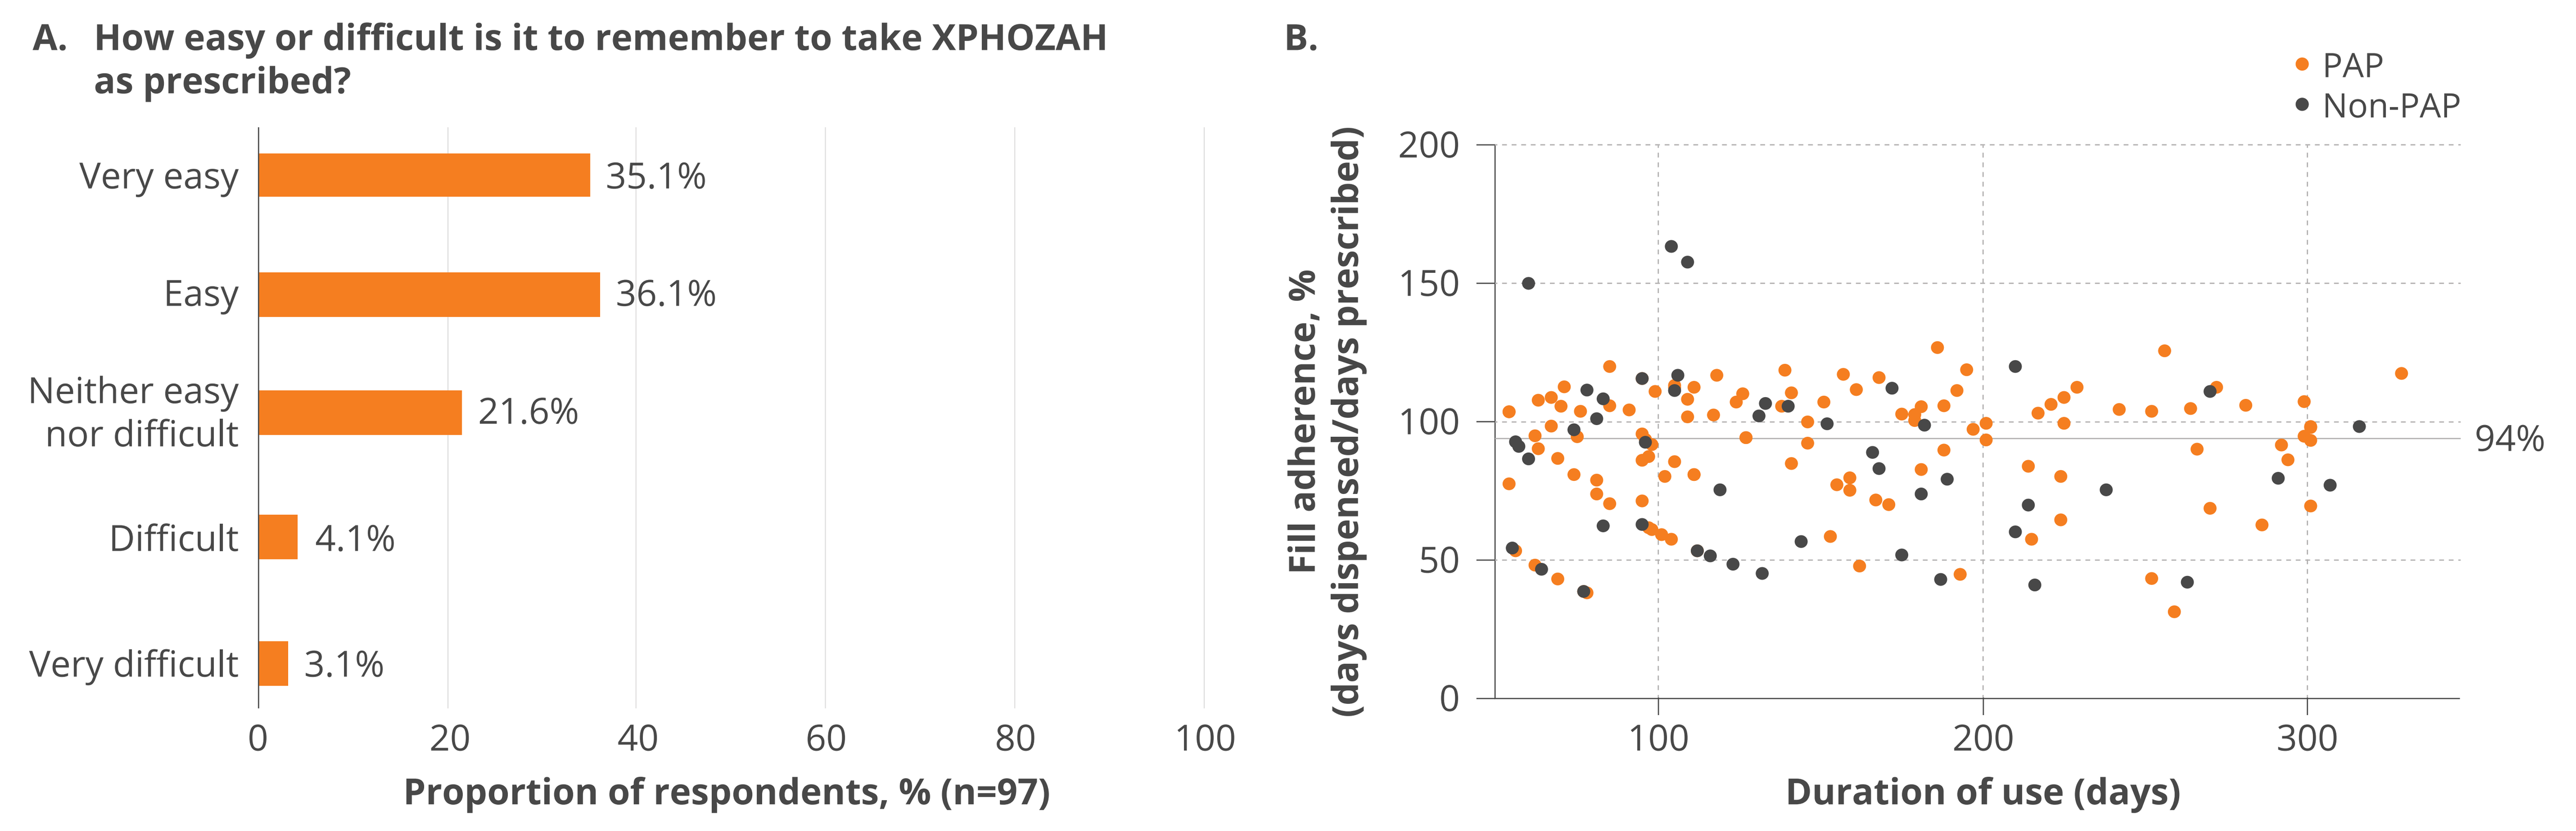
A. How easy or difficult is it to remember to take XPHOZAH as prescribed?
Very easy 35.1%
Easy 36.1%
Neither easy nor difficult 21.6%
Difficult 4.1%
Very difficult 3.1%
0 20 40 60 80 100
Proportion of respondents, % (n=97)
B.
PAP
Non-PAP
Fill adherence, % (days dispensed/days prescribed)
0 50 100 150 200
94%
100 200 300
Duration of use (days)

[a]Participants were allowed to skip questions. [b]Fill adherence is shown as the fraction of days dispensed/days prescribed. PAP, n=116; non-PAP, n=47. [c]Fill adherence greater than 100% can occur when patients refill their prescriptions earlier than scheduled and may also result from differences in data reporting vs data extraction methods.
PAP, patient assistance program.

Outlook on Serum Phosphate Control

- Of the patients who reported a change in serum phosphate levels, 69% (37 of 54) indicated that their outlook on serum phosphate control was a little or much better (**Figure 3A**).
 - In this subset, 44% (16 of 36) attributed the change to better serum phosphate control, 39% (14 of 36) to improved bowel movements, and 14% (5 of 36) to lower pill burden (**Figure 3B**).
- Of the patients who did not know if their serum phosphate levels had changed, 45% (21 of 47) reported that their outlook on serum phosphate control was a little or much better (**Figure 3A**).
 - In this subset, 73% (11 of 15) attributed this outlook to improved bowel movements and 13% (2 of 15) to lower pill burden (**Figure 3B**).

Figure 3: (A) Patient Outlooks on Serum Phosphate Control and (B) Reasons for Improved Outlook Since Starting Tenapanor[a]

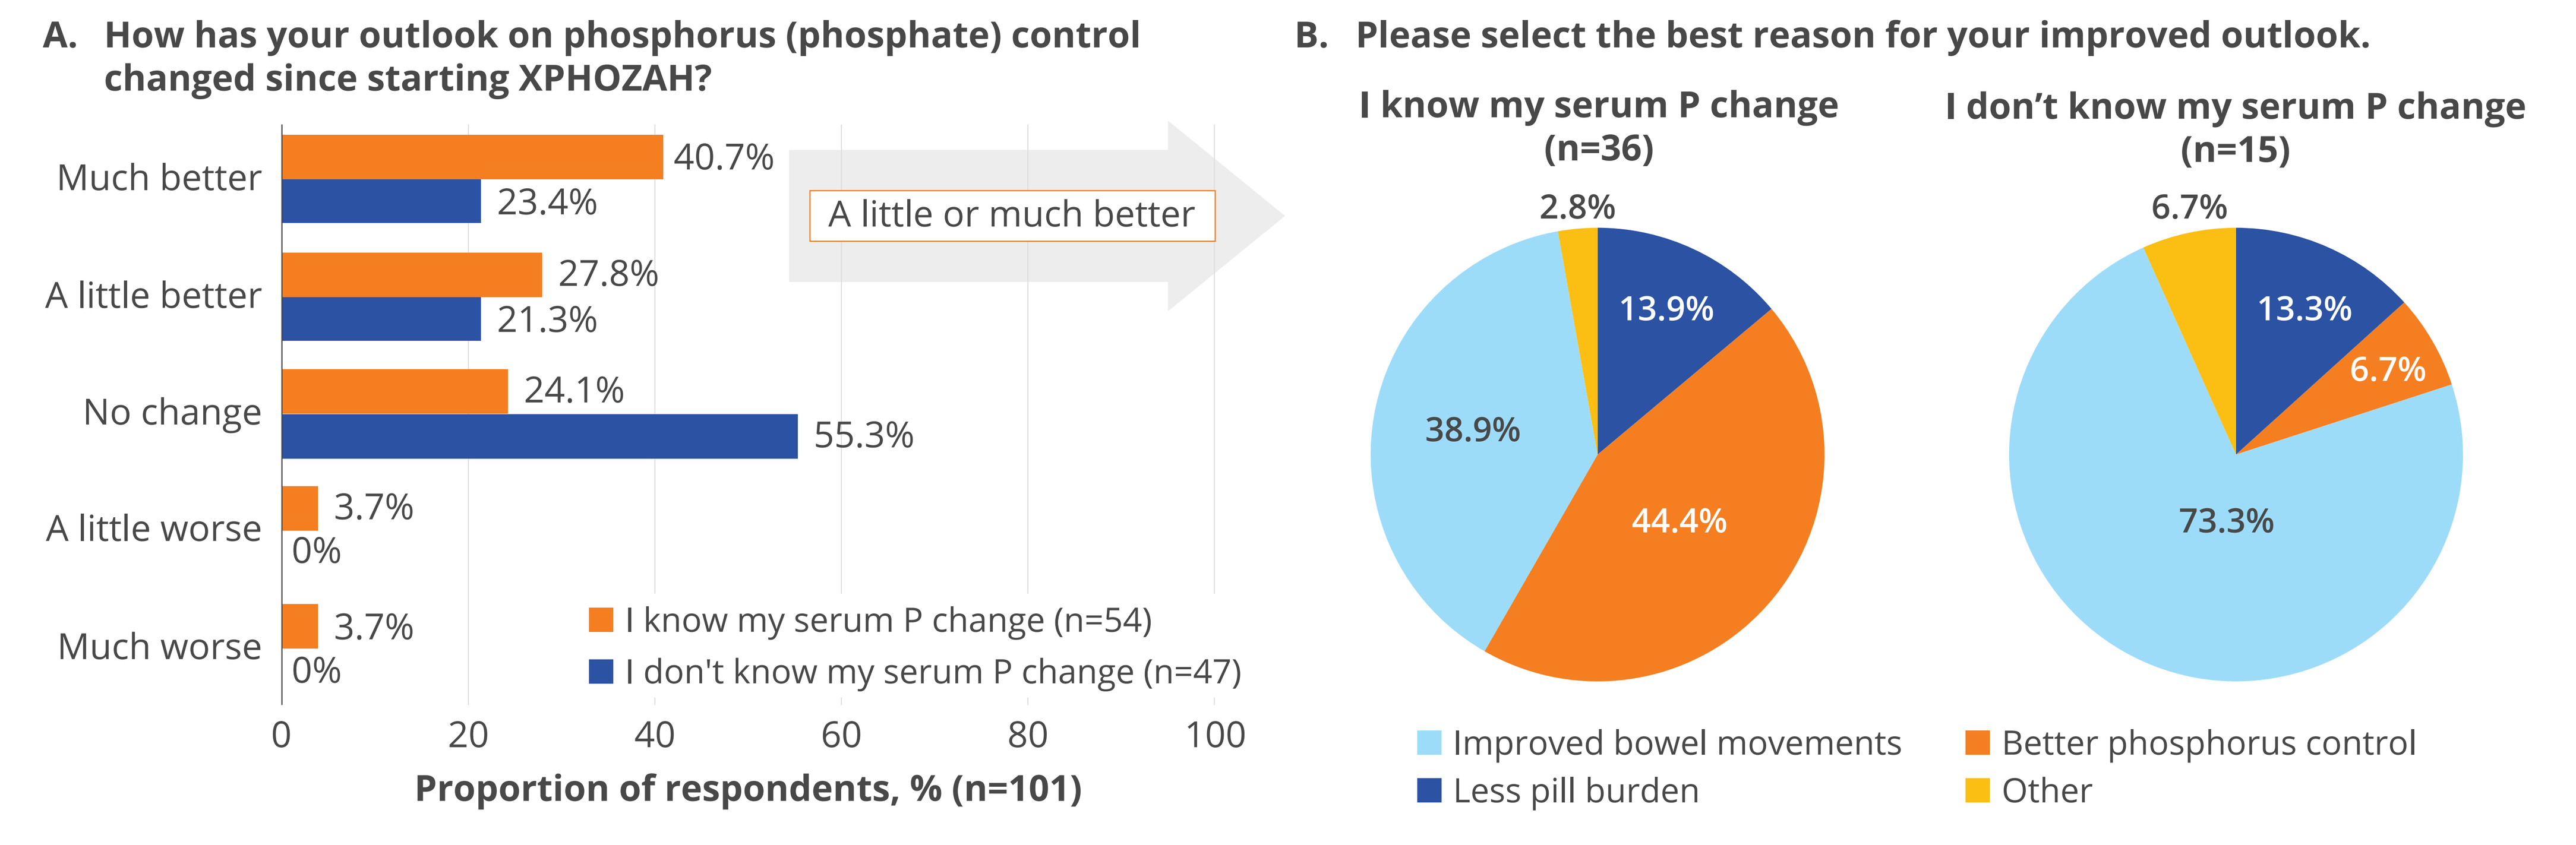
A. How has your outlook on phosphorus (phosphate) control changed since starting XPHOZAH?
Much better 40.7% 23.4%
A little better 27.8% 21.3%
No change 24.1% 55.3%
A little worse 3.7% 0%
Much worse 3.7% 0%
A little or much better
I know my serum P change (n=54)
I don't know my serum P change (n=47)
0 20 40 60 80 100
Proportion of respondents, % (n=101)
B. Please select the best reason for your improved outlook.
I know my serum P change (n=36)
2.8%
13.9%
38.9%
44.4%
I don't know my serum P change (n=15)
6.7%
13.3%
6.7%
73.3%
Improved bowel movements
Better phosphorus control
Less pill burden
Other

[a]Participants were allowed to skip questions.
P, phosphate.

Limitations

- Bias may have been introduced due to participants receiving tenapanor at no cost and the potential for self-selection by individuals with more favorable experiences.
- The survey included only current users of tenapanor with no control group and the assessment tool was not validated.

Conclusions

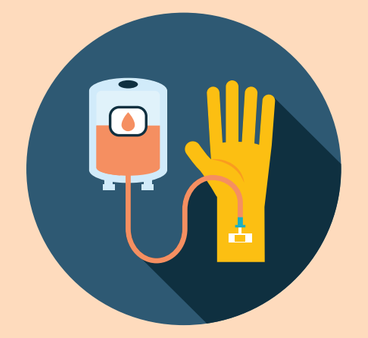

Patients report positive experiences with tenapanor in the real world, including improved serum phosphate control, bowel movements, and lower pill burden.

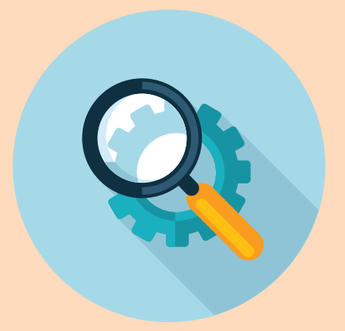

These results also demonstrate the need for greater patient education.

Disclosures

This study was supported by Ardelyx, Inc. Jessica Kendrick served as an advisory board member for Ardelyx, Inc. Luisa Scott and Laura Williams are employees of Ardelyx, Inc. Belinda Gist was an employee of Ardelyx, Inc., at the time of the study.

References

1. XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc.; 2025.
2. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
3. Fishbane SN et al. *Kidney Med*. 2021;3:1057-64.
4. Block GA et al. *J Am Soc Nephrol*. 2019;30:641-52.
5. Block GA et al. *Kidney360*. 2021;2:1600-10.
6. Silva AL et al. *Kidney360*. 2023;4:1580-89.
7. Pergola PE et al. *J Am Soc Nephrol*. 2021;32:1465-73.
8. Sprague SM et al. *Kidney360*. 2024;5:732-42.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Jessica Kendrick can be contacted for further information on this study at Jessica.Kendrick@cuanschutz.edu.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American Society of Nephrology (ASN) 2025 Kidney Week; November 6-9, 2025; Houston, TX, USA.

MED-US—2500505 11/2025

XPHOZAH (tenapanor) Important Safety Information and Indication

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in XPHOZAH-treated patients were reported to be mild-to-moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, click here.

CKD, chronic kidney disease.
XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only