Poster TH-PO0221

Real-World Effectiveness of Tenapanor (XPHOZAH) for Treatment of Hyperphosphatemia in United States Patients on Dialysis

Alan J. Fossa,[1] Dhajanae Sylvertooth,[1] Angelo Karaboyas,[1] Brian Bieber,[1] Roberto Pecoits-Filho,[1] David Rosenbaum,[2] Laura Williams,[2] Luisa Scott[2]

[1]Arbor Research Collaborative for Health, Ann Arbor, MI, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Introduction

- Tenapanor (XPHOZAH) is approved to reduce serum phosphorus in adults with chronic kidney disease on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders (PBs) or who are intolerant of any dose of PBs.[1]
- Here, we present preliminary findings from the first real-world study of tenapanor for management of serum phosphate (P), aimed at characterizing the real-world use and effectiveness of tenapanor in the United States (US) dialysis setting.

Methods

- For this preliminary analysis, data were obtained from electronic medical records of patients treated at a mid-size dialysis organization who initiated tenapanor between January and June 2024 and met the following criteria:
 - Tenapanor initiation confirmed using prescription data
 - At least 1 baseline serum P measurement
 - At least 1 serum P measurement in the 120 days following treatment initiation
 - Hyperphosphatemia at baseline (serum P ≥5.5 mg/dL)
- The primary outcome of this analysis was the within-patient change in serum P, defined as the difference between baseline and post-treatment serum P.
 - Baseline serum P measurements captured 29 to 0 days pre-tenapanor initiation
 - Post-treatment serum P measurement captured 1 to 120 days post-tenapanor initiation
 - If multiple serum P measurements were captured within these intervals, the average value was used in the analysis
- Patients were included in the primary analysis regardless of whether they discontinued tenapanor within 120 days of initiation. A sensitivity analysis was performed for persistent users who were on tenapanor for the full 120-day follow-up period.

Results

- In total, 982 patients met the inclusion criteria and were included in the primary analysis (**Table 1**; **Figure 1**). Of these patients, 700 (71%) were persistent users.

Table 1: Baseline Characteristics of All Initiators

Characteristics	All initiators (N=982)
Median age, y [Q1, Q3]	56 [46, 65]
Sex, %	
Female	38
Male	62
Race, %	
White	47
Black	22
Asian	10
Other	21
Ethnicity, %	
Hispanic/Latino	30
Non-Hispanic/Latino	70
Lab/biometric markers	
Median BMI, kg/m² [Q1, Q3]	29.7 [25.5, 35.4]
Median serum P, mg/dL [Q1, Q3]	7.4 [6.6, 8.6]
Median serum PTH, pg/mL [Q1, Q3]	558 [340, 922]
Median serum calcium, mg/dL [Q1, Q3]	8.6 [8.1, 9.1]
PB use	
Any use at baseline, %	82

BMI, body mass index; P, phosphate; PB, phosphate binder; PTH, parathyroid hormone; Q, quartile.

Figure 1: PB Use at Baseline[a]

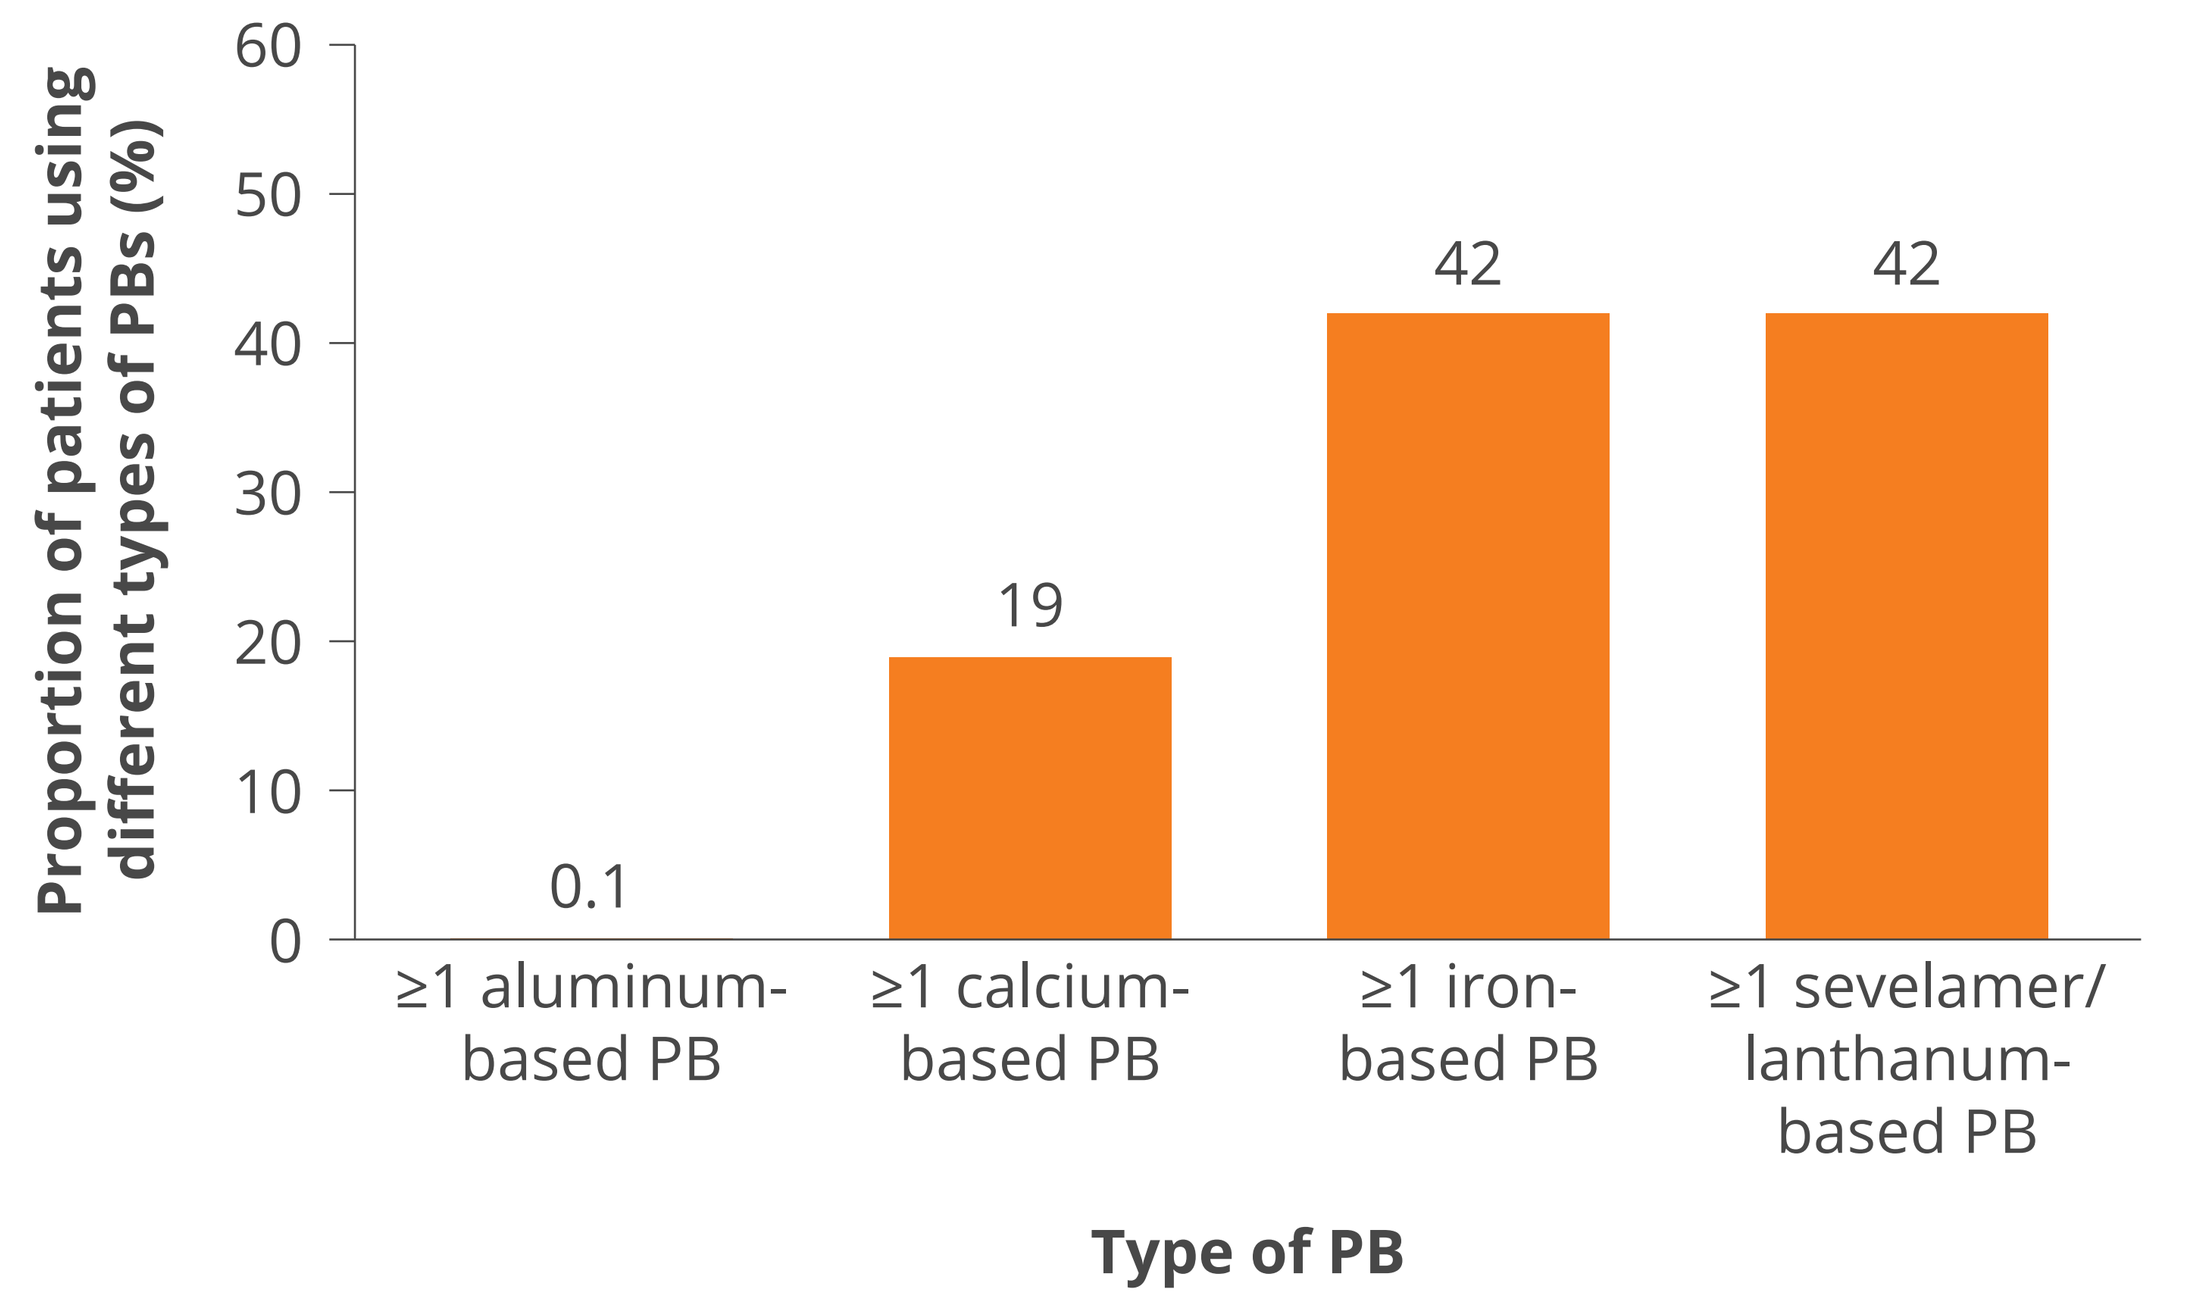
Proportion of patients using different types of PBs (%)
60
50
40
30
20
10
0
0.1
19
42
42
≥1 aluminum-based PB
≥1 calcium-based PB
≥1 iron-based PB
≥1 sevelamer/lanthanum-based PB
Type of PB

[a]Patients could have been using more than 1 type of PB (categories were not mutually exclusive).
PB, phosphate binder.

Figure 2: Mean Within-Patient Change in Serum P From Baseline at Day 120 in All Tenapanor Initiators and Persistent Users

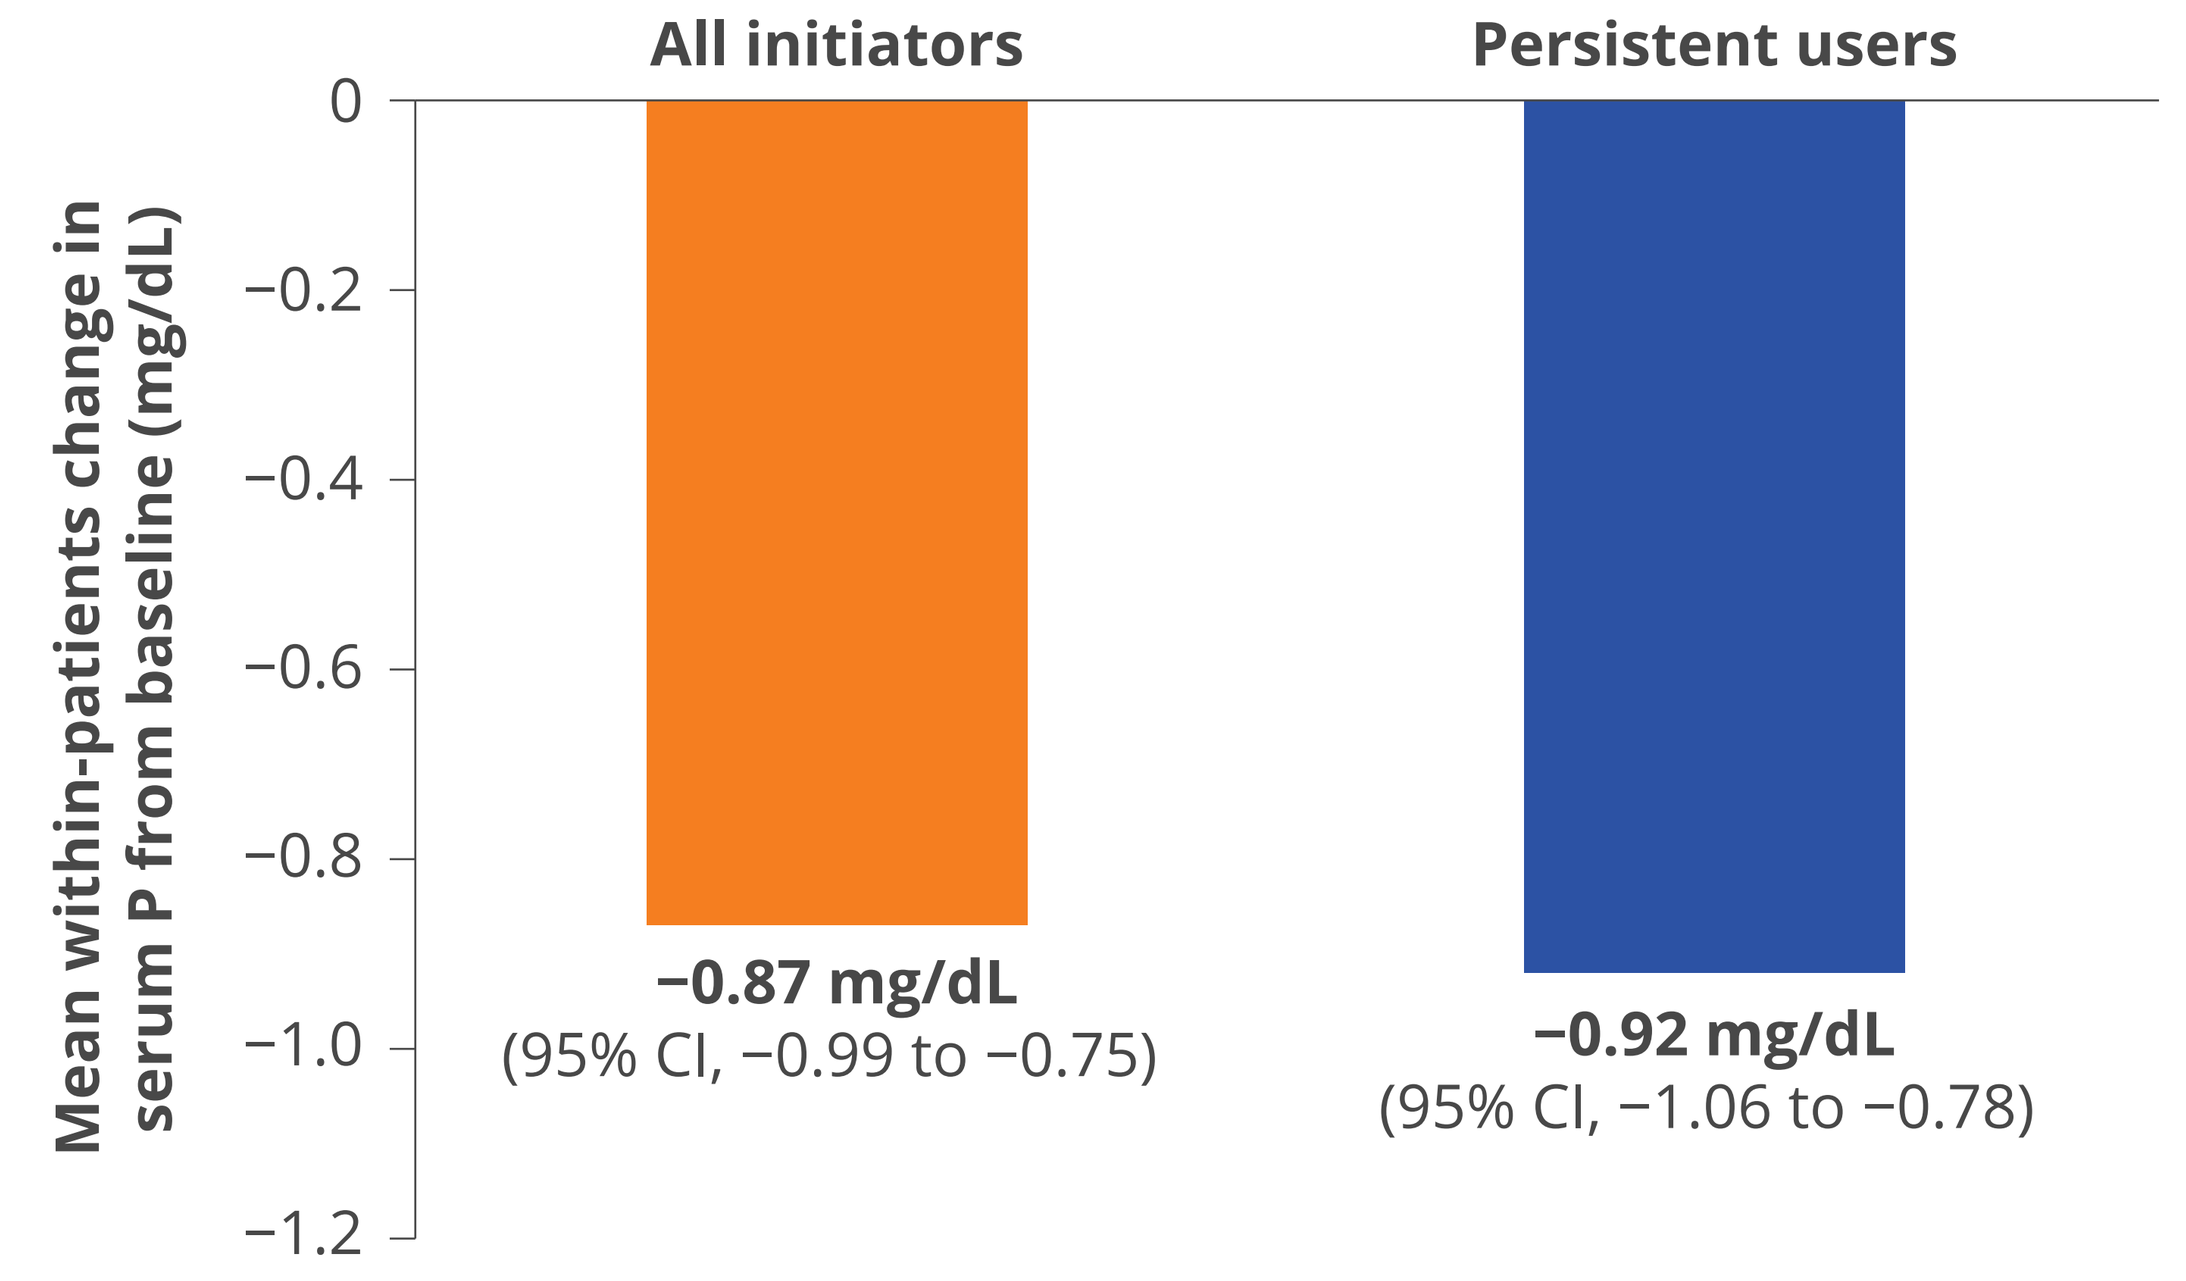
All initiators
Persistent users
Mean within-patients change in serum P from baseline (mg/dL)
0
−0.2
−0.4
−0.6
−0.8
−1.0
−1.2
−0.87 mg/dL
(95% CI, −0.99 to −0.75)
−0.92 mg/dL
(95% CI, −1.06 to −0.78)

P, phosphate.

- At 91 to 120 days after initiation, 75.5% were prescribed a dosage of 420 mg/wk.
- Mean within-patient change in serum P from baseline was −0.87 mg/dL for all initiators and −0.92 mg/dL for persistent users (**Figure 2**).
- Mean within-patient serum P decreased over time for all initiators and for persistent users (**Figure 3**).
- Median serum P decreased in all initiators from 7.4 mg/dL (interquartile range [IQR], 6.6 to 8.6) at baseline to 6.6 mg/dL (IQR, 5.6 to 8.1) at month 4 (**Figure 4**).

Figure 3: Within-Patient *Reduction* in Serum P at Each Follow-Up Interval for All Tenapanor Initiators and Persistent Users

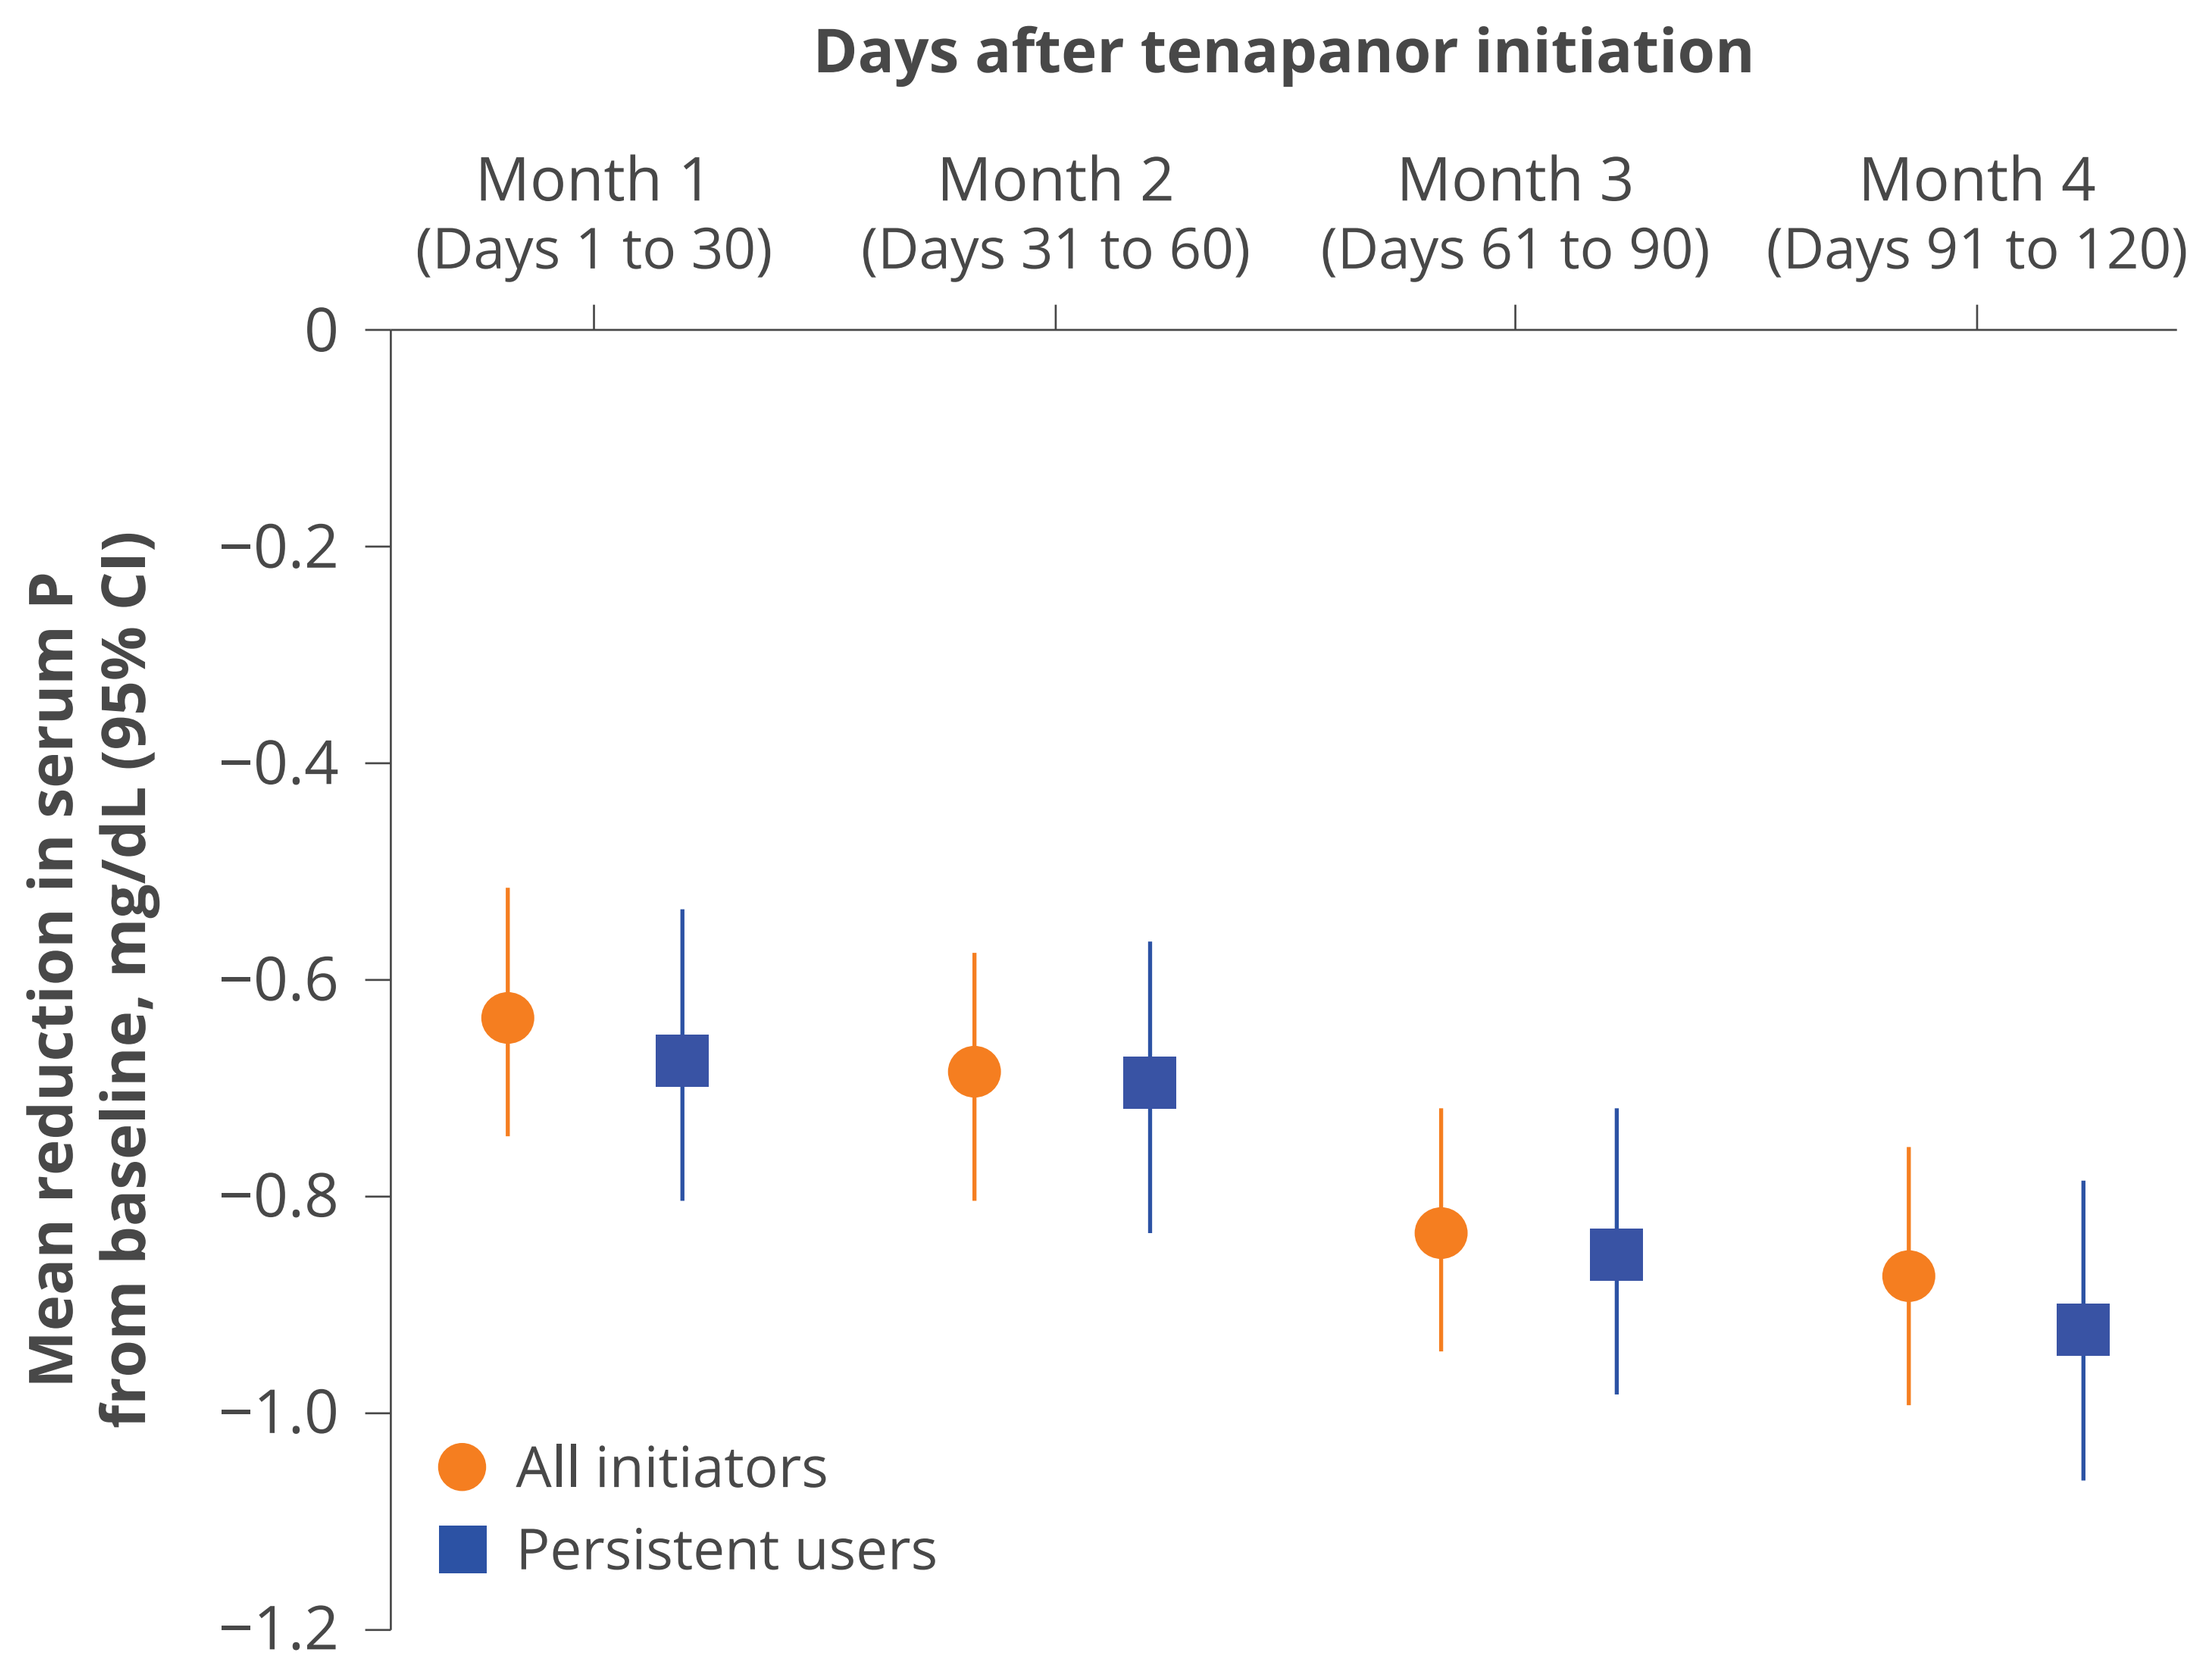
Days after tenapanor initiation
Month 1 (Days 1 to 30)
Month 2 (Days 31 to 60)
Month 3 (Days 61 to 90)
Month 4 (Days 91 to 120)
Mean reduction in serum P from baseline, mg/dL (95% CI)
0
−0.2
−0.4
−0.6
−0.8
−1.0
−1.2
All initiators
Persistent users

P, phosphate.

Figure 4: Serum P Distribution Over Time for All Tenapanor Initiators[a]

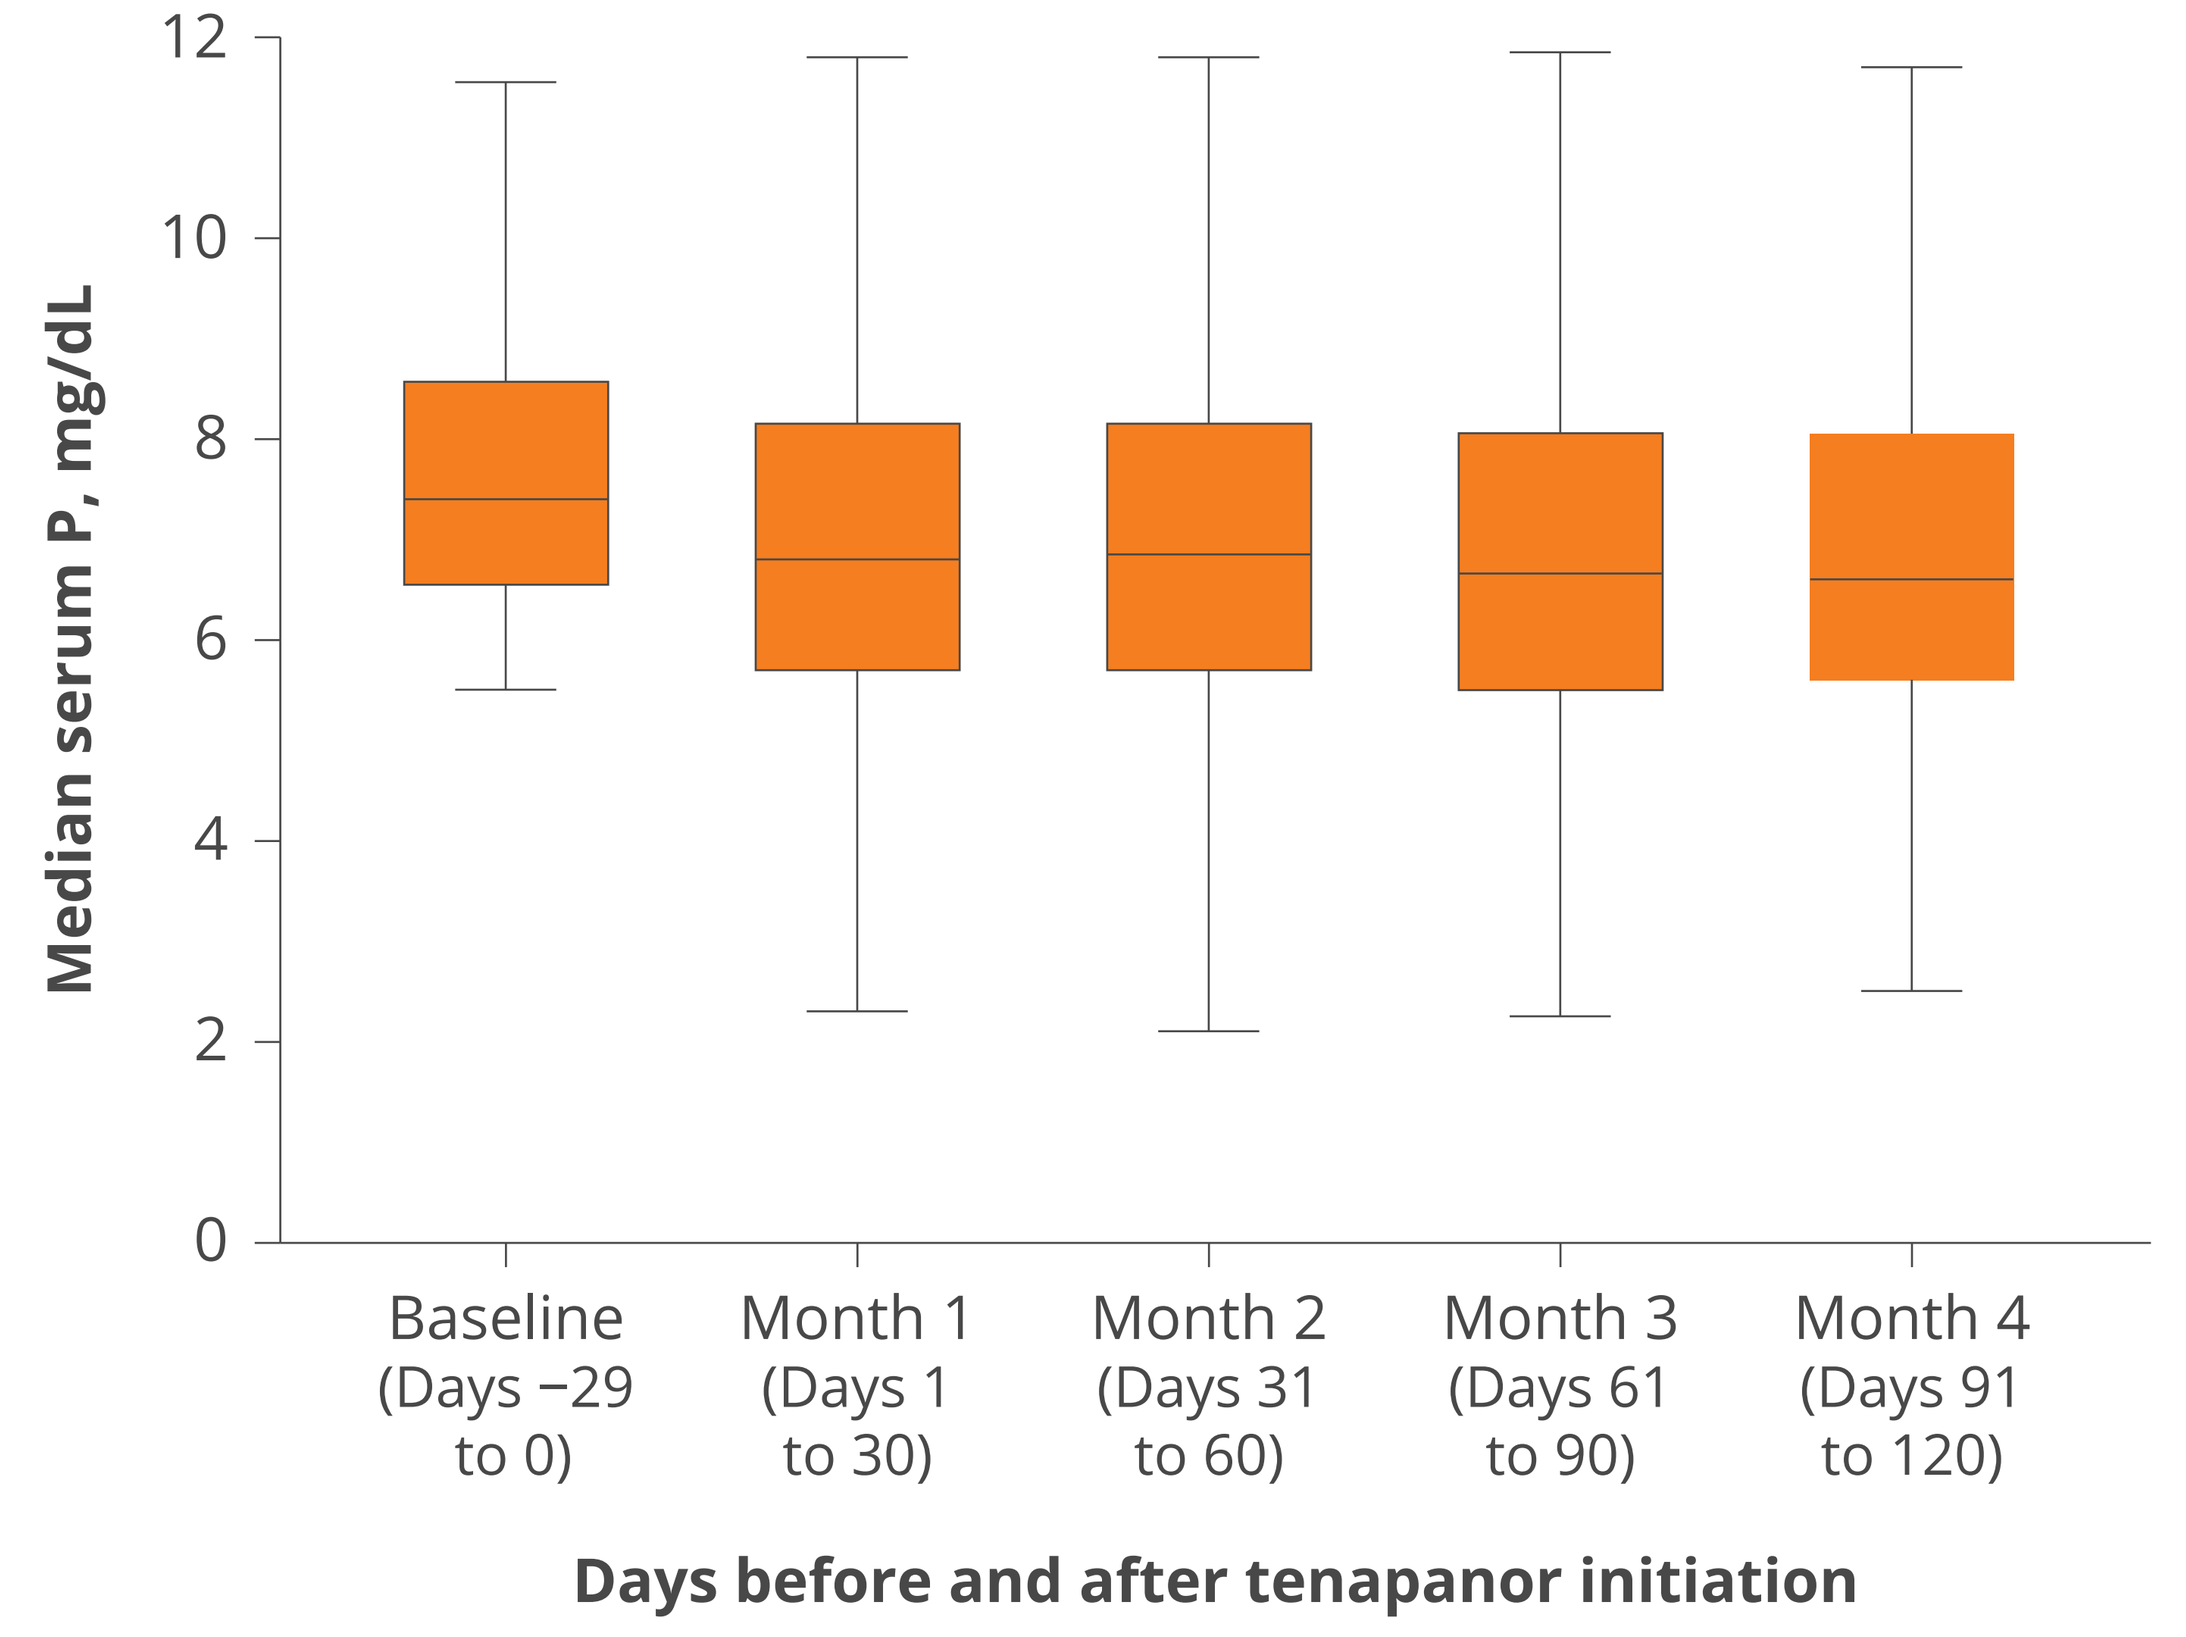
Median serum P, mg/dL
12
10
8
6
4
2
0
Baseline (Days −29 to 0)
Month 1 (Days 1 to 30)
Month 2 (Days 31 to 60)
Month 3 (Days 61 to 90)
Month 4 (Days 91 to 120)
Days before and after tenapanor initiation

[a]Box extends from the 25th to the 75th percentile with a horizonal line at the 50th percentile. Whiskers extend to the 5th and to the 95th percentiles.
P, phosphate.

- Among all initiators, the proportions of patients reaching a ≥1- or ≥2-mg/dL threshold decrease in serum P or a serum P of <5.5 mg/dL after treatment were 45%, 25%, and 23%, respectively. Results were similar when restricting to persistent users (**Figure 5**).
- At 91 to 120 days after initiation, 24% of patients had reduced their daily PB dose to some degree, and 17% of patients had reduced their daily PB dose burden by at least 50%.
- Tenapanor dosage remained relatively consistent over 4 months.

Figure 5: All Tenapanor Initiators and Persistent Users Reaching a ≥1- or ≥2-mg/dL Threshold Decrease in Serum P or a Serum P of <5.5 mg/dL After Treatment

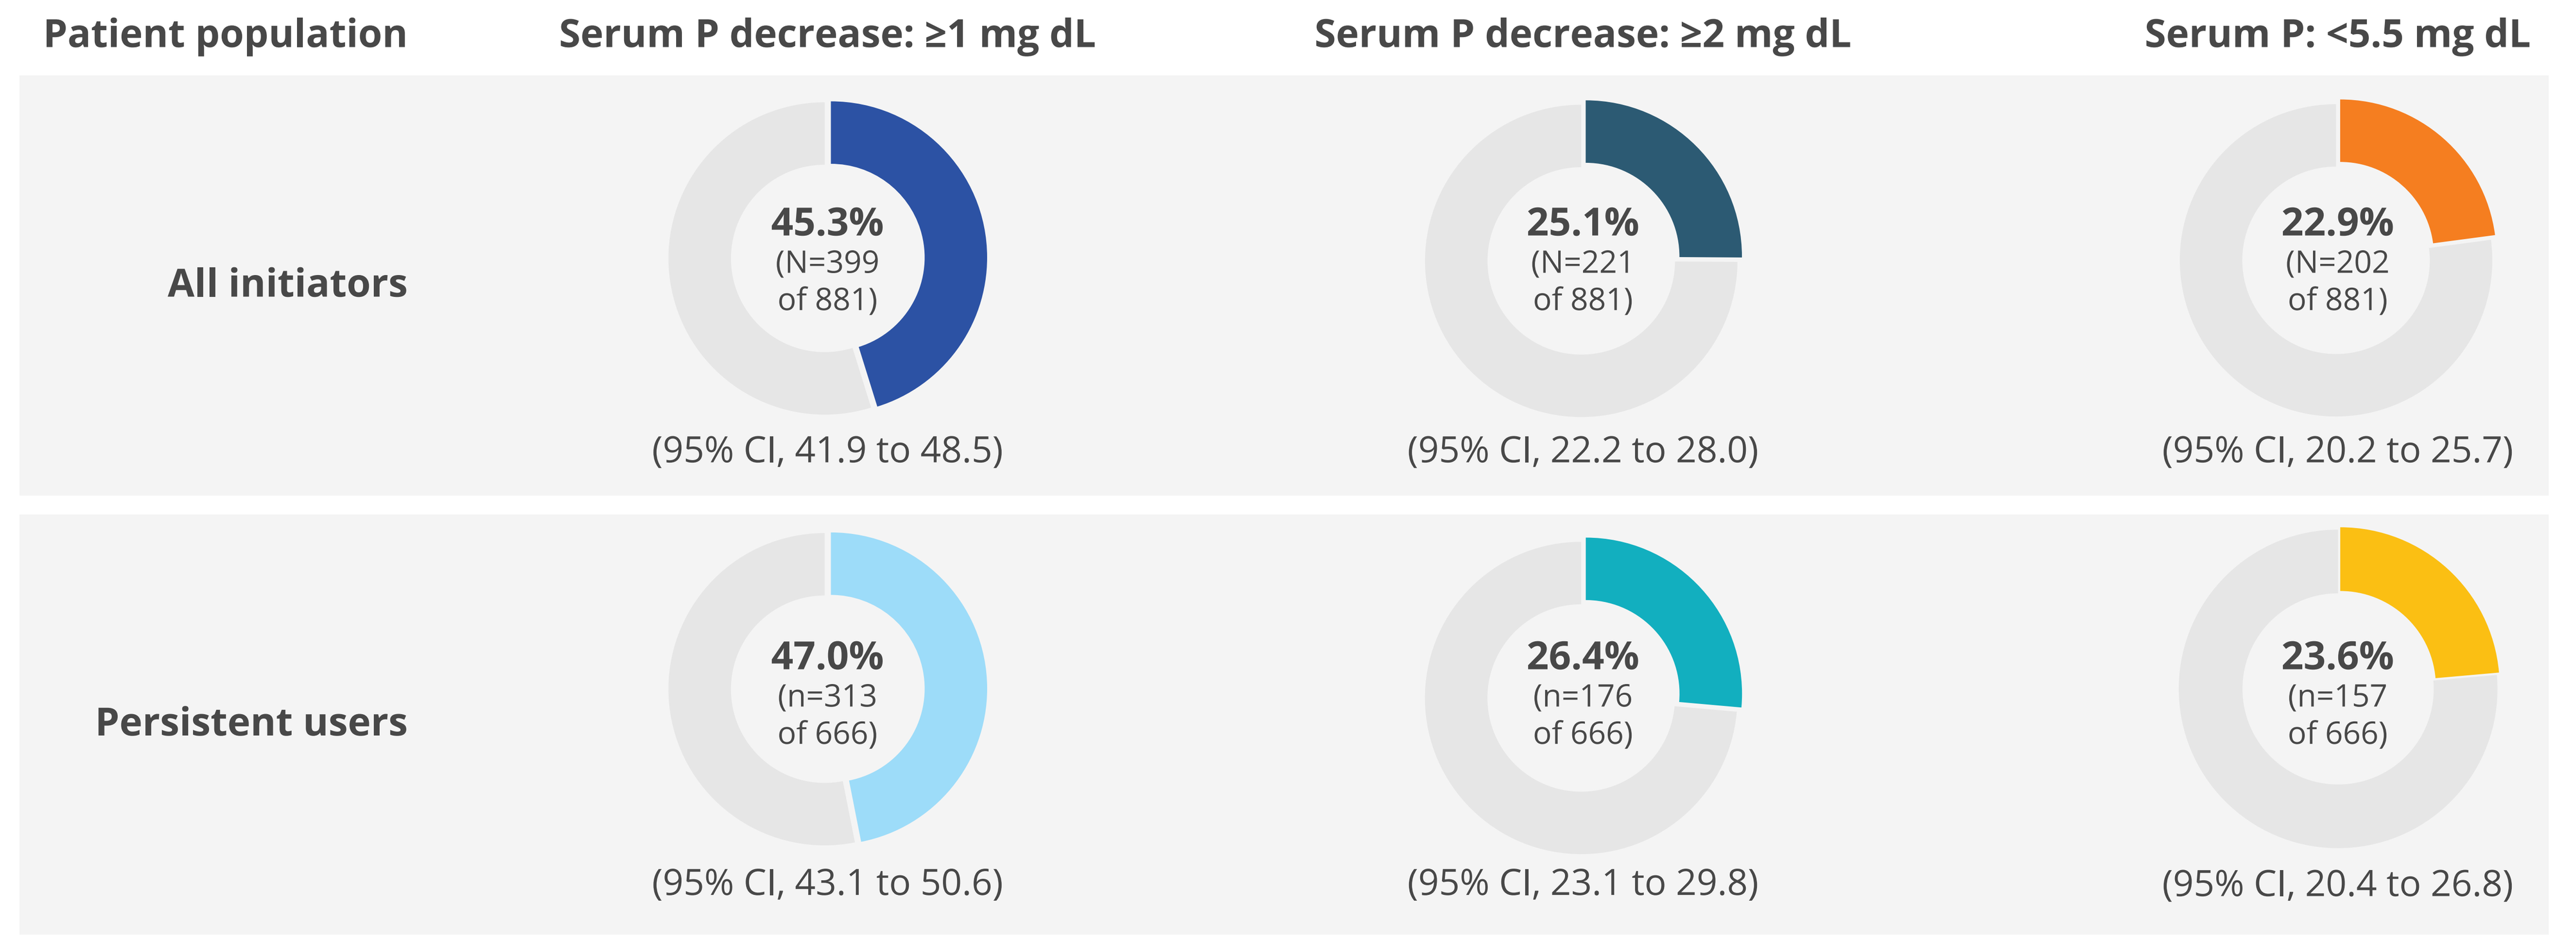
Patient population
Serum P decrease: ≥1 mg dL
Serum P decrease: ≥2 mg dL
Serum P: <5.5 mg dL
All initiators
45.3% (N=399 of 881)
(95% CI, 41.9 to 48.5)
25.1% (N=221 of 881)
(95% CI, 22.2 to 28.0)
22.9% (N=202 of 881)
(95% CI, 20.2 to 25.7)
Persistent users
47.0% (n=313 of 666)
(95% CI, 43.1 to 50.6)
26.4% (n=176 of 666)
(95% CI, 23.1 to 29.8)
23.6% (n=157 of 666)
(95% CI, 20.4 to 26.8)

P, phosphate.

Limitations

- This preliminary analysis includes patients from only 1 dialysis organization and may not be representative of all US patients on dialysis.
- Tenapanor use is based on the date of tenapanor prescription and may not reflect actual tenapanor usage.
- No comparison group was included in this analysis, limiting our ability to place these results in the broader context of pharmacological hyperphosphatemia control.

Conclusions

Patients prescribed tenapanor in this real-world study experienced a reduction in serum phosphate of nearly 1 mg/dL on average, with substantial proportions of patients experiencing ≥1- and ≥2-mg/dL reductions in serum phosphate.

Results were similar between all initiators and patients persisting on tenapanor for the entire follow-up period.

These findings support the real-world effectiveness of tenapanor for management of serum phosphate in patients with end-stage kidney disease and hyperphosphatemia.

Disclosures

This study was supported by Ardelyx, Inc. Alan J. Fossa, Dhajanae Sylvertooth, Angelo Karaboyas, Brian Bieber, and Roberto Pecoits-Filho are employees of Arbor Research; all funding was provided to Arbor Research and not to the individuals directly. David Rosenbaum, Laura Williams, and Luisa Scott are employees of Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

References

1. XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

Alan Fossa can be contacted for further information on this study at alan.fossa@arborresearch.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American Society of Nephrology (ASN) 2025 Kidney Week; November 6-9, 2025; Houston, TX, USA.

MED-US—2500507 11/2025

XPHOZAH (tenapanor) Important Safety Information and Indication

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in XPHOZAH-treated patients were reported to be mild-to-moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, click here.

CKD, chronic kidney disease.
XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only