Poster FR-PO0254

Etelcalcetide Utilization Rates Decreased and Parathyroidectomy Rates Increased Following the Incorporation of Calcimimetics Into the End-Stage Kidney Disease Bundle

Brian Bieber,[1] Angelo Karaboyas,[1] Roberto Pecoits-Filho,[1] Kevin J. Martin,[2] Stuart M. Sprague[3]

[1]Arbor Research Collaborative for Health, Ann Arbor, MI, USA; [2]Saint Louis University School of Medicine, Division of Nephrology, St. Louis, MO, USA; [3]NorthShore University Health System, Division of Nephrology and Hypertension, Evanston, IL, USA, and University of Chicago Pritzker School of Medicine, Chicago, IL, USA

Introduction

- Secondary hyperparathyroidism (SHPT) is a complication of end-stage kidney disease (ESKD) with severe and persistent cases requiring invasive parathyroidectomy (PTX). SHPT is common and has an especially high prevalence among Black patients.[1,2]
- Calcimimetics, such as cinacalcet and etelcalcetide (ETEL), are used to manage SHPT and reduce the risk of PTX among patients on dialysis.[2]
- The Transitional Drug Add-On Payment Adjustment (TDAPA) policy established temporary reimbursement for new ESKD treatments, like ETEL, to support adoption.[3]
- TDAPA payments for ETEL were provided from January 2018 through December 2020, after which ETEL was incorporated into the ESKD bundle that includes a single standardized payment for all patients on dialysis, regardless of use.[3]
- This study, designed as a natural experiment, examines the trends in calcimimetic prescriptions and PTX rates among patients on hemodialysis (HD) during and after TDAPA, stratified by race and dialysis organization (DO) size, to better understand the policy impact on patient outcomes.

Methods

- Calcimimetic prescriptions and PTX events (based on codes from the International Classification of Diseases, 10th Revision, and the Healthcare Common Procedure Coding System) were identified for patients on HD enrolled in fee-for-service Medicare from 2016-2021.
- Data were supplied by the United States Renal Data System (USRDS).

Results

Patients

- Of the 244,394 patients included in this study, 58.2% were male, 32.0% were Black, and the mean age was 64.1 years.

Overall Calcimimetic Prescription Prevalence and PTX rates

- Following the introduction of ETEL in 2018, its prescription rate increased to 9.5% during the TDAPA period, then declined to 3.8% by March 2021 after the TDAPA period ended (**Figure 1**).
- Cinacalcet prescriptions rates declined from 25.7% to 18.0% during the TDAPA period and rebounded to 24.1% in June 2021 after the TDAPA period.
- PTX rates, measured per 1000 patient-years, declined from 6.8 in 2016 to 3.4 in 2020, and increased again to 4.4 in 2021, coinciding with the increase and subsequent decrease in ETEL use.

Figure 1: PTX Rate and ETEL Use Over Time Among US Patients on HD on FFS Medicare

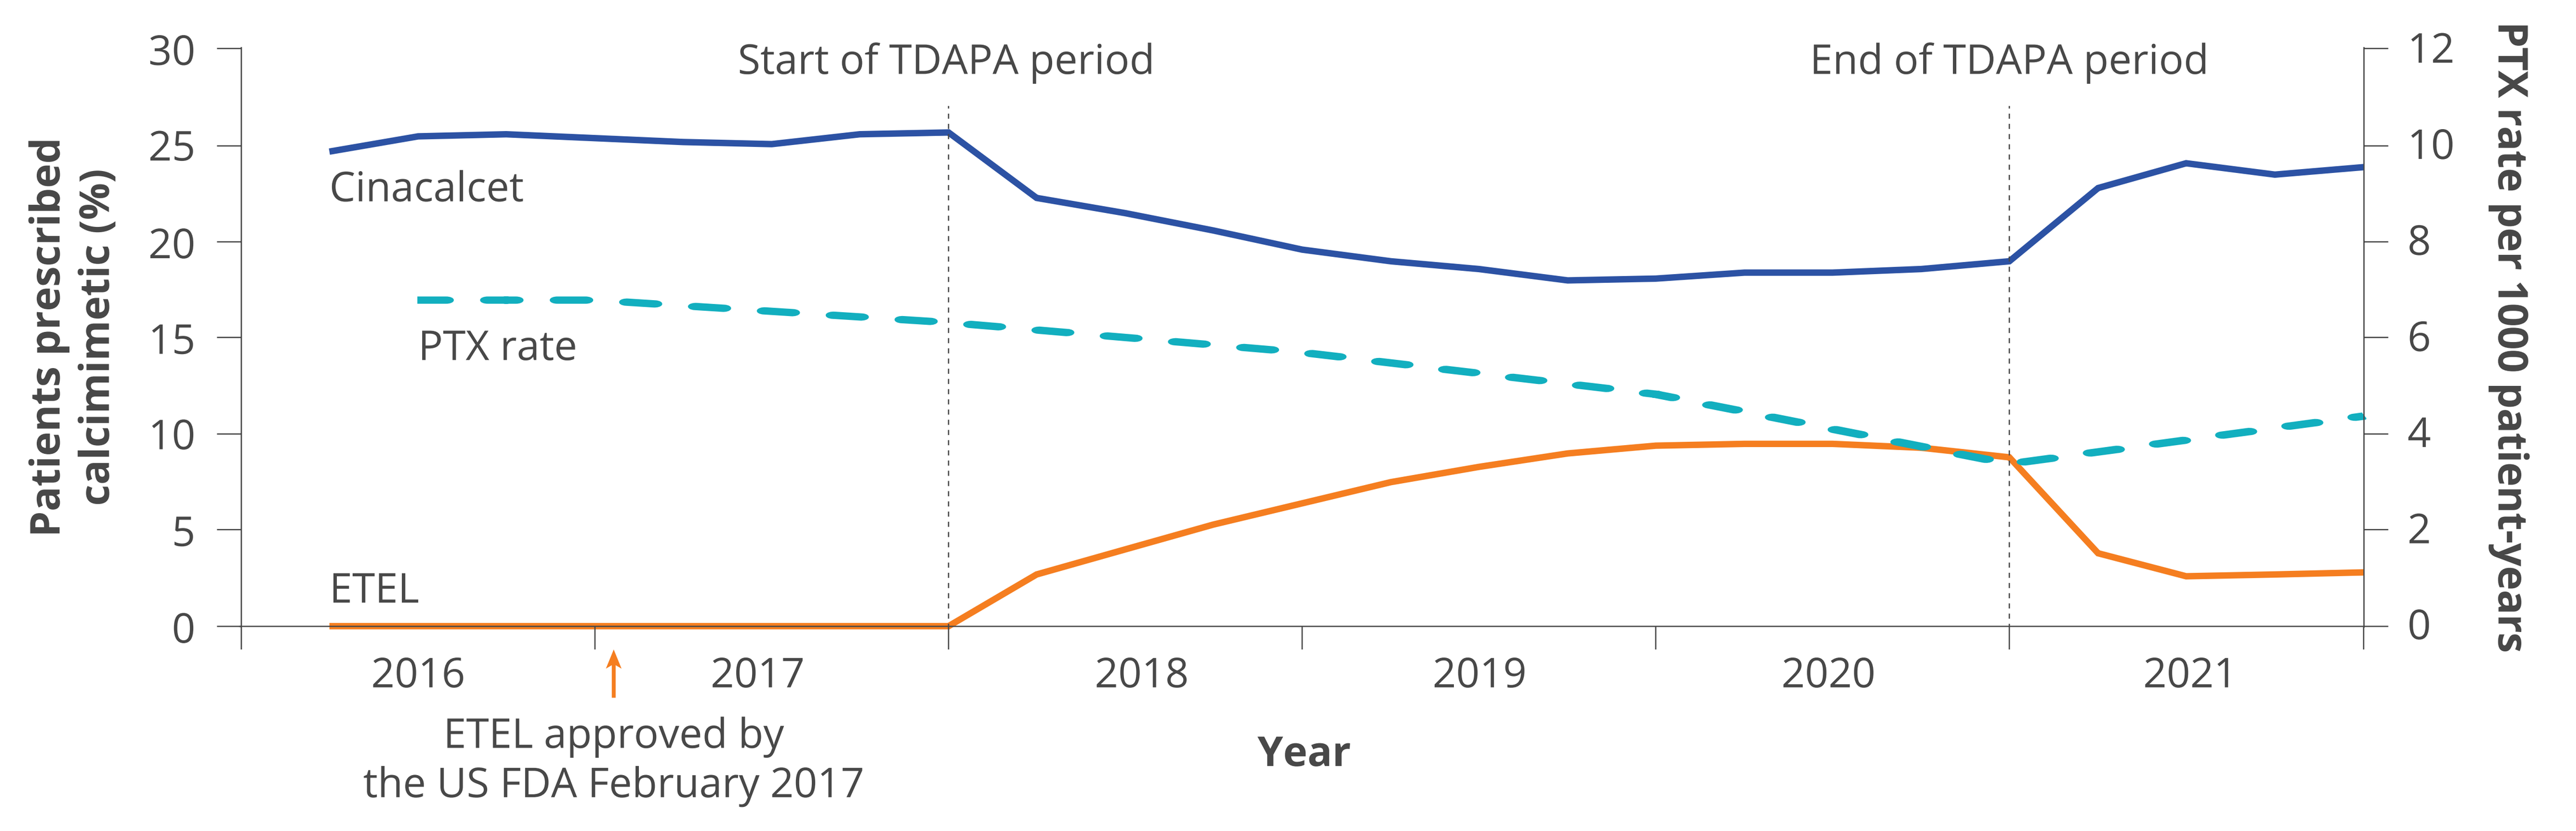

Patients prescribed calcimimetic (%)
PTX rate per 1000 patient-years
Start of TDAPA period
End of TDAPA period
Cinacalcet
PTX rate
ETEL
ETEL approved by the US FDA February 2017
Year

ETEL, etelcalcetide; FFS, fee-for-service; HD, hemodialysis; PTX, parathyroidectomy; TDAPA, Transitional Drug Add-On Payment Adjustment; US FDA, United States Food and Drug Administration.

ETEL Prescriptions and PTX Rates Stratified by Race

- During the TDAPA period, ETEL prescriptions increased from 3.1% in March 2018 to 12.8% in Black patients and from 2.4% to 7.6% in non-Black patients, but sharply declined to 5.5% and 2.9%, respectively, after the end of TDAPA coverage (**Figure 2A**).
- The decline in the PTX rates per 1000 patient-years during the TDAPA period (8.5 to 4.6) and increase after (4.6 to 6.5) were numerically greater among Black patients than in non-Black patients (4.1 to 2.7 to 3.3; **Figure 2B**).

Figure 2: (A) ETEL Use and (B) PTX Rate by Race Among US Patients on HD on FFS Medicare

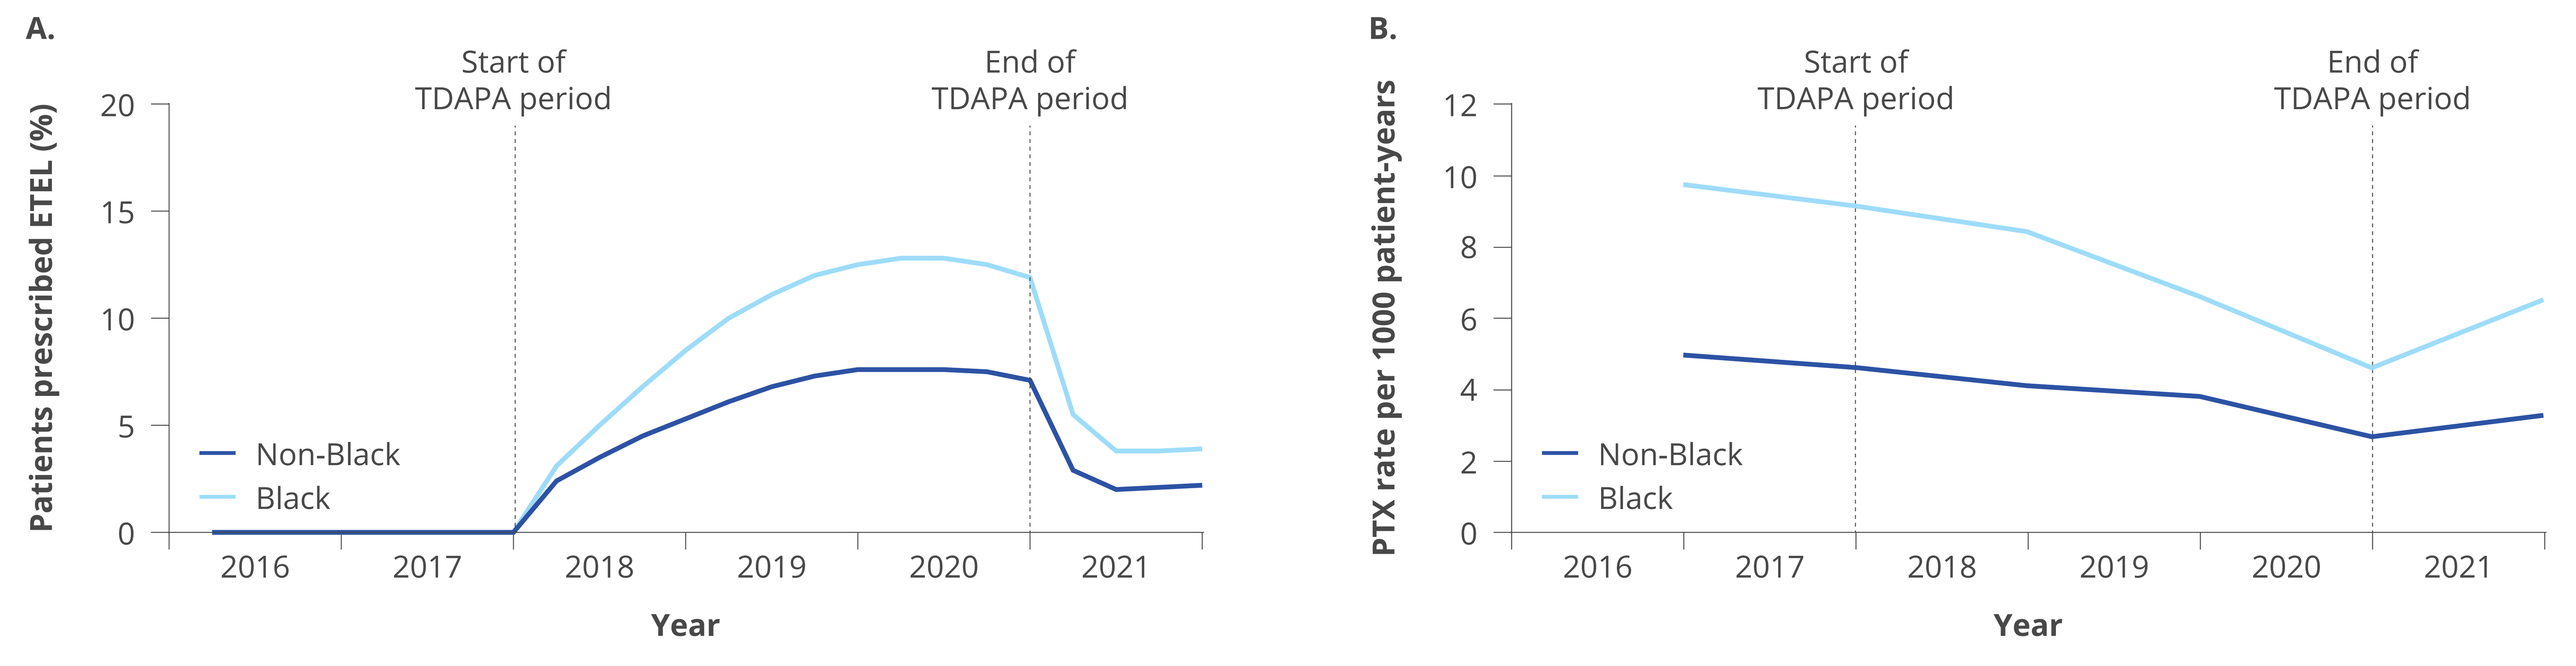

A.
Patients prescribed ETEL (%)
B.
PTX rate per 1000 patient-years
Start of TDAPA period
End of TDAPA period
Non-Black
Black
Year

ETEL, etelcalcetide; FSS, fee-for-service; HD, hemodialysis; PTX, parathyroidectomy; TDAPA, Transitional Drug Add-On Payment Adjustment.

ETEL Use and PTX Rates Stratified by DO Size

- In small/medium-sized DOs (S/MDOs) and unaffiliated dialysis facilities (UDFs), uptake of ETEL peaked at 18.5% and 17.6%, respectively, during the TDAPA period and declined sizably to 0.6% and 6.3% after, while in large DOs (LDOs), ETEL use peaked at 7.1% during and declined to 3.9% after TDAPA ended (**Figure 3A**).
- During the TDAPA period, the decrease in PTX rates was 6.1 to 2.9 per 1000 patient-years in S/MDOs, 5.6 to 3.9 per 1000 patient-years in UDFs, and 5.8 to 3.4 per 1000 patient-years in LDOs (**Figure 3B**), which was followed by a post-TDAPA increase from 2.9 to 4.8 per 1000 patient-years in the S/MDOs, 3.9 to 4.3 per 1000 patient-years in the UDFs, and 3.4 to 4.4 per 1000 patient-years in the LDOs.
- Differences in calcimimetic use by DO size were not driven by differences in patient demographics.

Figure 3: (A) ETEL Use and (B) PTX Rate by DO Size Among US Patients on HD on FFS Medicare

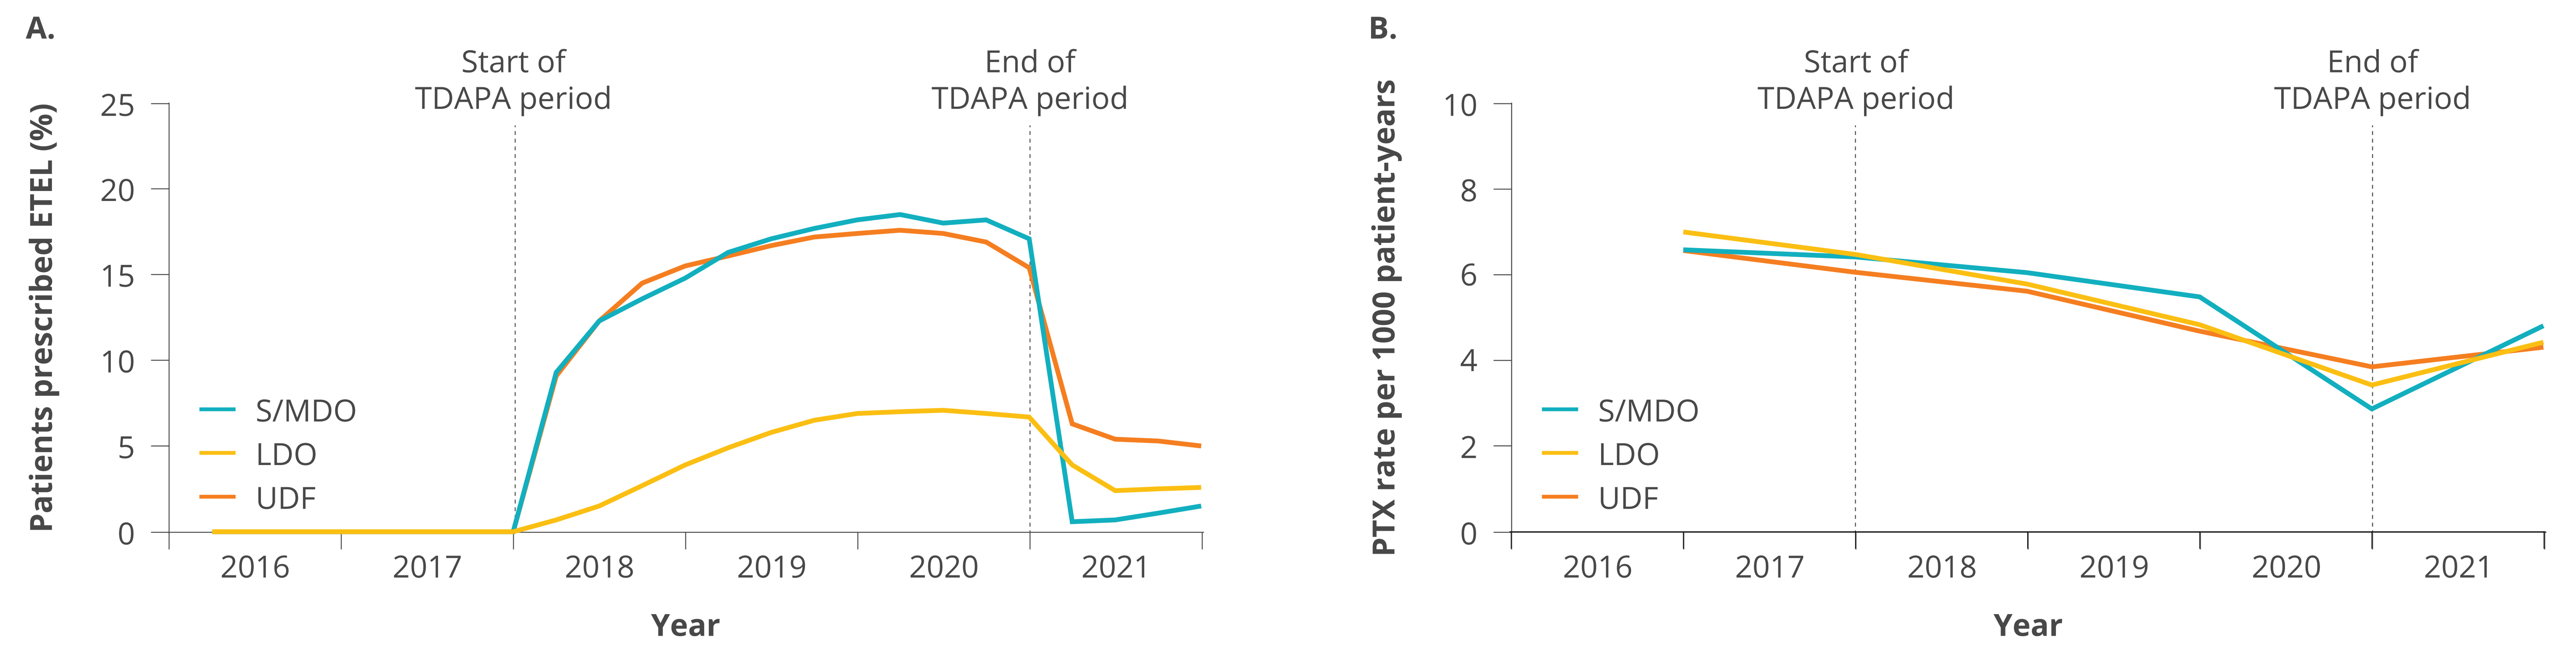

A.
Patients prescribed ETEL (%)
B.
PTX rate per 1000 patient-years
Start of TDAPA period
End of TDAPA period
S/MDO
LDO
UDF
Year

ETEL, etelcalcetide; FFS, fee-for-service; LDO, large dialysis organization; PTX, parathyroidectomy; S/MDO, small/medium dialysis organization; TDAPA, Transitional Drug Add-On Payment Adjustment; UDF, unaffiliated dialysis facility.

Limitations

- These findings are established using ecological associations of patient groups in the data extracted from the USRDS.
- PTX rates may have been influenced by the COVID pandemic with fewer elective surgeries being performed.
- Medicare Advantage patients were not represented in this analysis due to data availability.

Conclusions

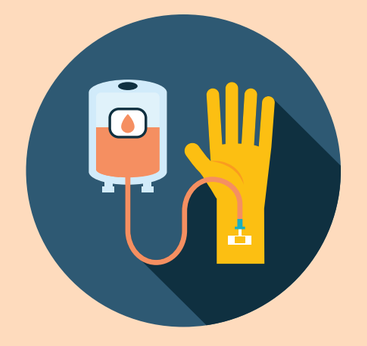

ETEL prescriptions increased during the TDAPA period and then decreased following its incorporation into the bundle, coinciding with a marked decrease—and then increase—in PTX rates over the same time. These patterns were amplified among Black patients and S/MDOs or UDFs where ETEL uptake was highest.

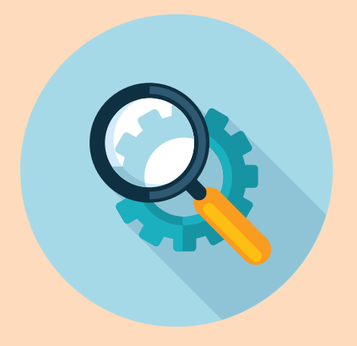

These findings highlight the impact of reimbursement policy on clinical practice and patient outcomes, underlining the unintended consequences of reducing access to effective therapies, increasing health disparities, and potentially suppressing future therapeutic innovation.

Disclosures

This study was supported by Ardelyx, Inc. Brian Bieber, Angelo Karaboyas, and Roberto Pecoits-Filho are employees of Arbor Research Collaborative for Health, which administers the DOPPS Programs. All funding is provided to Arbor Research and not to the employees directly. Kevin J. Martin is a paid consultant for Ardelyx, Inc., and participates in the data safety monitoring board or advisory board for Applied Therapeutics, LG Chem, and Renibus. Stuart M. Sprague receives research grants from and is a consultant for Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc. The data reported here have been supplied by the USRDS. The interpretation and reporting of these data are the responsibility of the authors and in no way should be seen as an official policy or interpretation of the US government.

References

1. Jovanovich A et al. *Clin J Am Soc Nephrol*. 2012;7:640-7.
2. Friedl C et al. *Drug Dev Des Ther*. 2018;12:1589-98.
3. Karaboyas A et al. *Clin J Am Soc Nephrol*. 2024;20:218-28.

Brian Bieber can be contacted for further information on this study at Brian.Bieber@ArborResearch.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American Society of Nephrology (ASN) 2025 Kidney Week; November 6-9, 2025; Houston, TX, USA.

MED-US—2500504 11/2025

XPHOZAH (tenapanor) Important Safety Information and Indication

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in XPHOZAH-treated patients were reported to be mild-to-moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, click here.

CKD, chronic kidney disease.
XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only