Patient Perception of Phosphate-Lowering Treatment Regimen Improves Adherence to Therapy

Amy Mosman,[1] Susan Edelstein,[2] David M. Spiegel,[2] Suling Zhao,[2] and David Rosenbaum[2]

[1]SLUCare Physician Group, St. Louis, MO, USA; [2]Ardelyx, Inc., Waltham, MA, USA

Background

- The 2017 Kidney Disease Improving Global Outcomes guidelines recommend that for patients with chronic kidney disease receiving maintenance dialysis, hyperphosphatemia should be managed by reducing serum phosphate (P) concentrations towards the normal range (2.5-4.5 mg/dL).[1-3]
- Patient adherence to phosphate-lowering medication is critical, but difficult to achieve due to the large pill burden (number and dosing frequency) and size of most phosphate binders.[4-6]
- Tenapanor is not a phosphate binder, but rather a first-in-class, minimally absorbed phosphate absorption inhibitor. Tenapanor targets the primary pathway of phosphate absorption in the gastrointestinal tract, the paracellular pathway, by selectively inhibiting sodium hydrogen exchanger isoform 3 (NHE3).[7-9]
- Tenapanor is approved as add-on therapy to reduce serum phosphorus in patients with CKD on dialysis who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.[10]
- Tenapanor is dosed as one 12-mm (half-inch) oval pill taken orally twice a day (bid).[10] These characteristics may improve patient adherence.
- Here, we present results from our evaluation of the effect of patients' perceptions of their phosphate-lowering treatment regimen on treatment adherence and reductions in serum P concentrations during Part A (ie, a 10-week treatment period) of the OPTIMIZE study.

Methods

- OPTIMIZE (NCT04549597) was a post-registrational, randomized, open-label, 2-part study that evaluated different ways of initiating tenapanor therapy in patients with end-stage kidney disease receiving maintenance dialysis with hyperphosphatemia despite the use of phosphate binders (**Figure 1**).

Figure 1: Study Design of the PB-Experienced Arms of the OPTIMIZE Study (Part A Only)

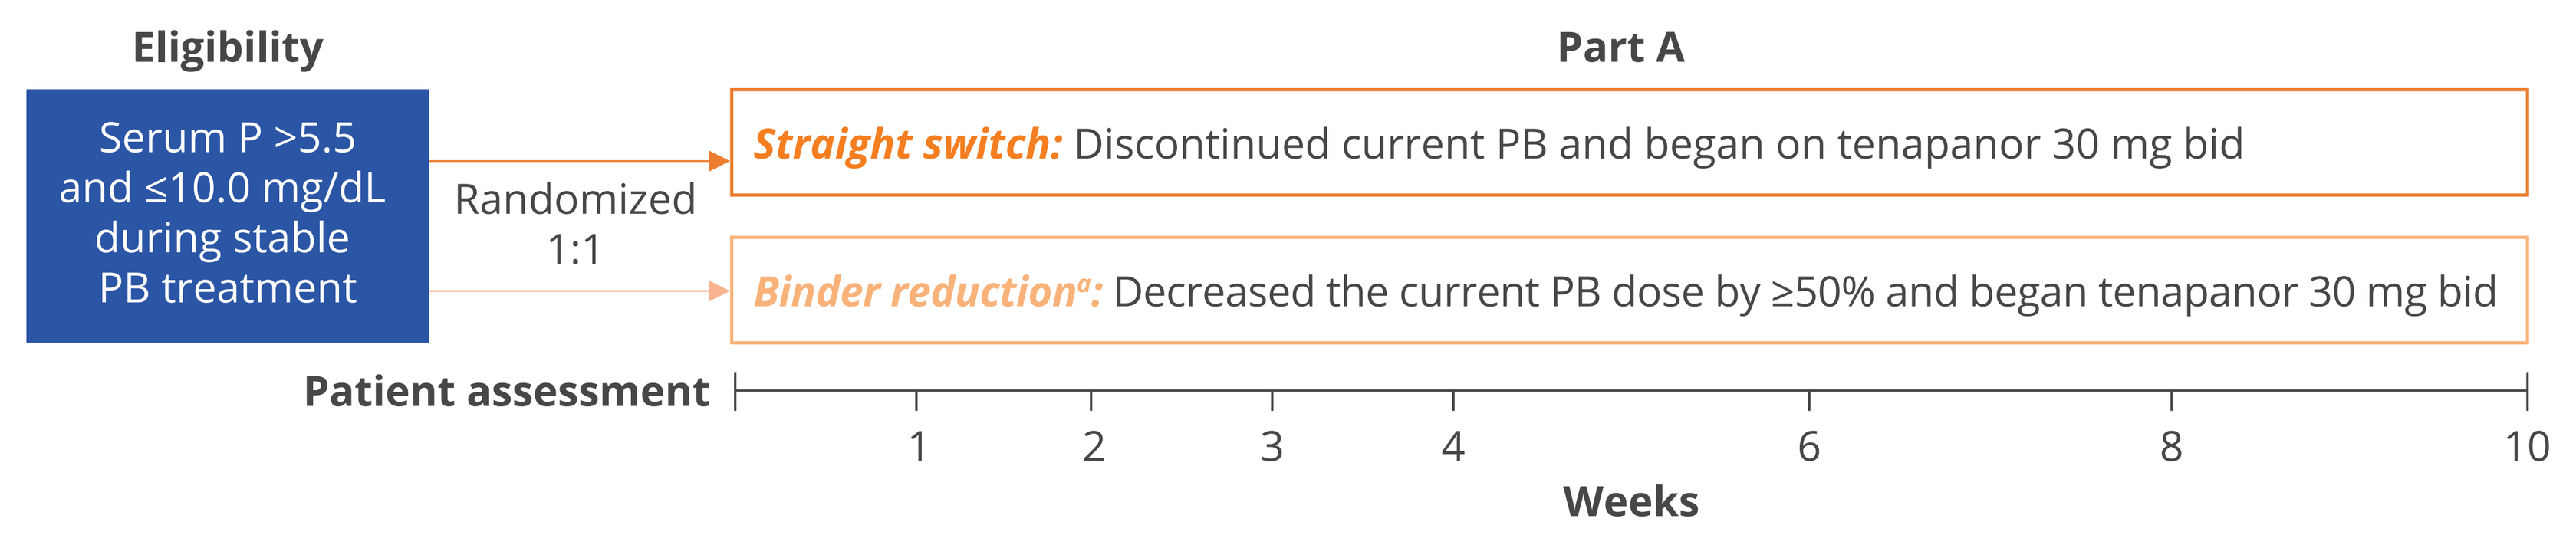
Eligibility
Part A
Serum P >5.5 and ≤10.0 mg/dL during stable PB treatment
Randomized 1:1
Straight switch: Discontinued current PB and began on tenapanor 30 mg bid
Binder reduction[a]: Decreased the current PB dose by ≥50% and began tenapanor 30 mg bid
Patient assessment
1
2
3
4
6
8
10
Weeks

A third cohort of patients was included in the OPTIMIZE study, however, patients from that cohort were excluded from this analysis because they were naive to treatment with PBs.
[a]PB reduction was >50% if patient was taking an odd number of pills; eg, a patient taking 3 PB pills/day would reduce their dose to 1 PB pill/day, a patient taking 5 PB pills/day would reduce their dose to 2 PB pills/day.
bid, twice a day; P, phosphate; PB, phosphate binder.

- Patients already receiving stable treatment with phosphate binders were enrolled in a 10-week treatment period (Part A) and randomized to 1 of 2 cohorts:
 – Cohort 1: patients added tenapanor 30 mg bid and stopped their phosphate binder (straight switch).
 – Cohort 2: patients added tenapanor 30 mg bid and reduced their phosphate binder dose by ≥50% (binder reduction).
 – After week 2, phosphate binders could be dose adjusted to achieve a serum P concentration ≤5.5 mg/dL.
- At the end of Part A (the week 10 visit or the patient's last visit if they discontinued prior to week 10), patients were asked to complete an electronic questionnaire to evaluate their perception of their serum P–lowering treatment regimen during the study versus before the study started (**Figure 2**).

Figure 2: Patient Experience Questionnaire

Q1. How would you characterize your feelings about your phosphate management routine during the study compared to how you felt about your phosphate management routine prior to this study?

Very much improved	Much improved	Minimally improved	No change	Minimally worse	Much worse	Very much worse

Q2. Which of the following changes do you think most likely contributed to the change in how you felt with your phosphate management routine during the study?

Size of phosphate-lowering pills	Number of phosphate-lowering pills	Number of times/ day I had to take phosphate-lowering pills	Number of phosphate-lowering medicines I have to take	Frequency of bowel movements	Form of bowel movements	Other stomach/ GI changes (eg, nausea, bloating, etc)	Other

GI, gastrointestinal.

- We evaluated tenapanor adherence and reductions in serum P concentrations in patients with positive versus less favorable perceptions of their regimen.

Results

Patients

- In OPTIMIZE, 303 binder-treated patients entered Part A: 151 in Cohort 1 and 152 in Cohort 2. Baseline demographic and disease characteristics for these patients have been published previously[11] and are summarized in **Table 1**.

Table 1: Baseline Demographics and Disease Characteristics (Safety Population)

	Total (N=303)
Mean age, years (SD)	52.8 (11.6)
Male, n (%)	209 (69.0)
Race, n (%)	
White	126 (41.6)
Black	137 (45.2)
Other[a]	37 (12.2)
Unknown	3 (1.0)
Mean BMI, kg/m² (SD)	32.3 (8.4)
Mean duration of ESKD, years (SD)	5.4 (5.1)
Hemodialysis, n (%)	243 (80.2)
Type of phosphate binder taken at screening, n (%)	
Sevelamer binder	118 (38.9)
Calcium-based binder	43 (14.2)
Iron-based binder	80 (26.4)
Other non-sevelamer binder (eg, lanthanum carbonate)	10 (3.3)
Combined	52 (17.2)

[a]Includes Asian, Native American or Alaskan, Native Hawaiian or Pacific Islander, and other.
BMI, body mass index; ESKD, end-stage kidney disease.

Treatment Regimen Perception and Adherence

- Of the 303 patients enrolled in Cohorts 1 and 2 of the study, 243 answered Question 1, which characterized their feelings about their treatment regimen.
 – By the end of Part A, 205 of 243 (84.4%) patients believed their regimen had improved (either very much, much, or minimally), 30 of 243 (12.3%) patients felt it was unchanged, and 8 of 243 (3.3%) patients believed it had worsened (either minimally or very much) (**Figure 3**).
- Adherence to tenapanor was greater in patients who felt their regimen was improved (study discontinuation rate in Part A: 6 of 205 patients [2.9%]) versus those who believed there was no change or a worsening (study discontinuation rate from Part A: 12 of 38 patients [31.6%]) (**Figure 4**).

Figure 3: Patients' Perceptions of Their Phosphate Management Regimen

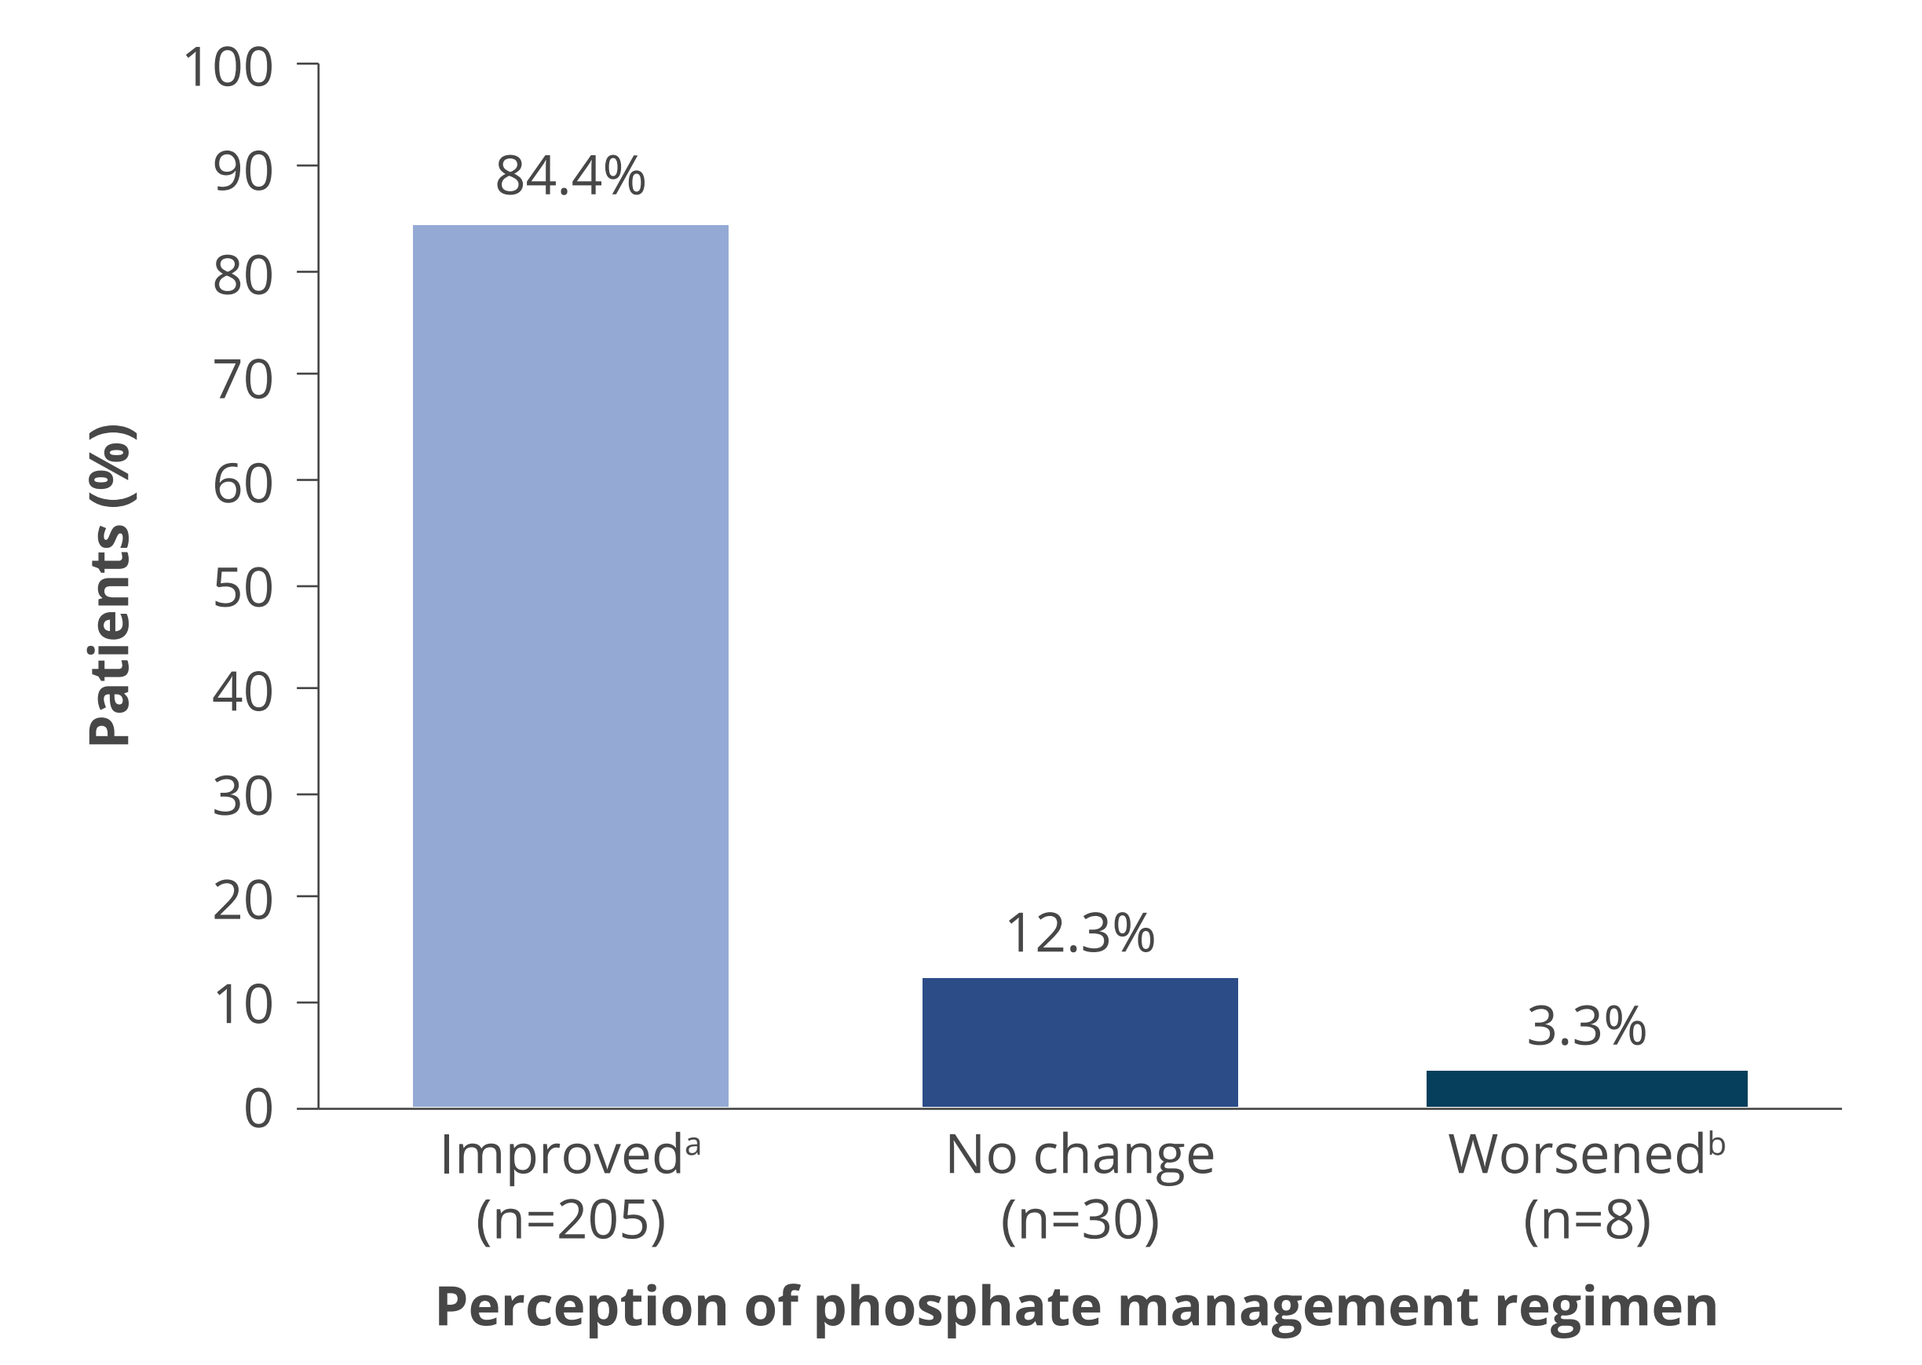
Patients (%)
100
90
80
70
60
50
40
30
20
10
0
84.4%
12.3%
3.3%
Improved[a] (n=205)
No change (n=30)
Worsened[b] (n=8)
Perception of phosphate management regimen

N=243.
[a]Patients who responded that their phosphate management regimen had "very much improved," "much improved," or "minimally improved" in the Patient Experience Questionnaire (Q1).
[b]Patients who responded that their phosphate management regimen had "minimally worsened" or "very much worsened" in the Patient Experience Questionnaire (Q1).

Figure 4: Study Discontinuation Rates in Part A by Patients' Perceptions of Their Phosphate Management Regimen

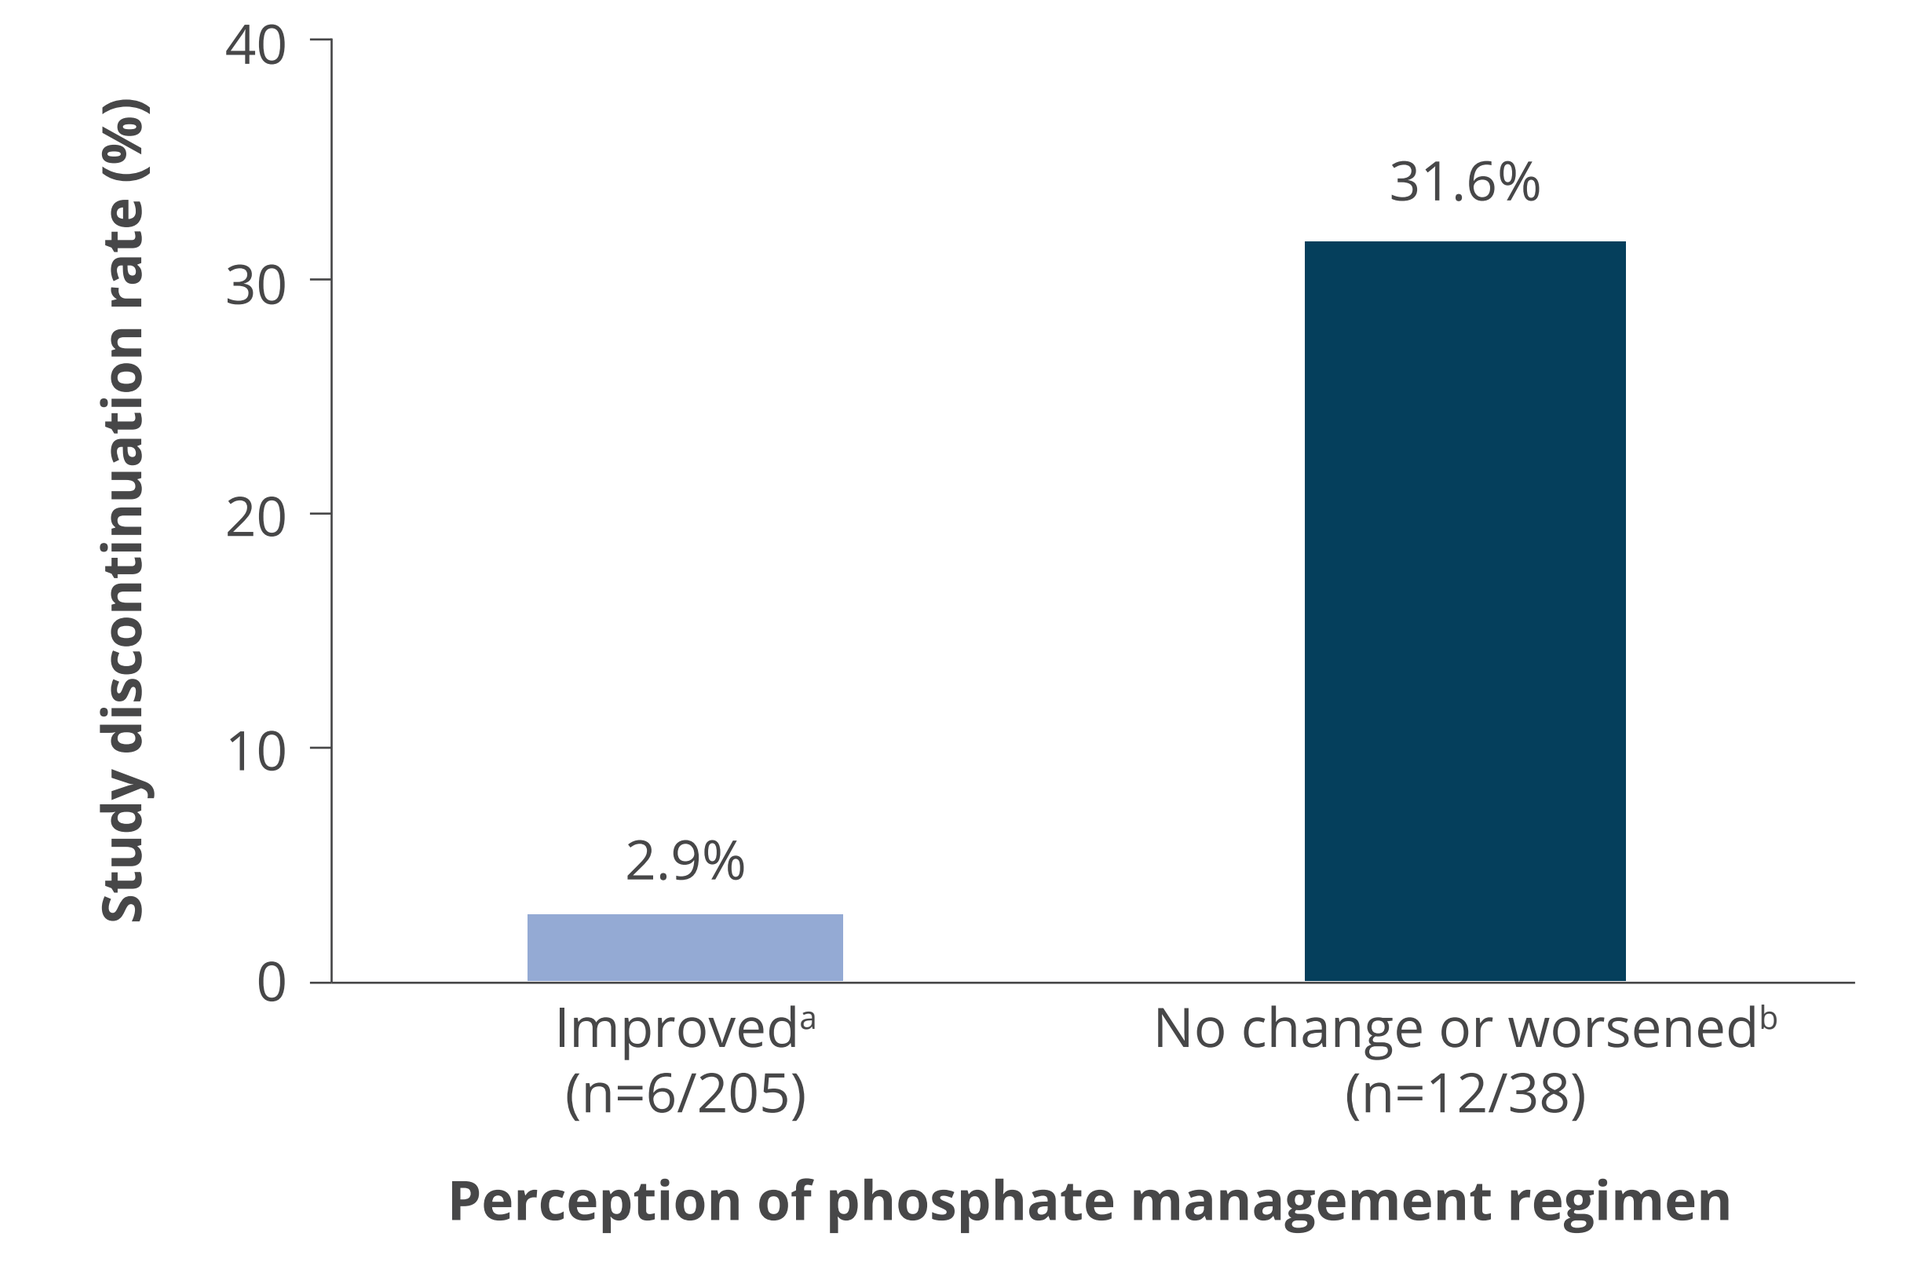
Study discontinuation rate (%)
40
30
20
10
0
2.9%
31.6%
Improved[a] (n=6/205)
No change or worsened[b] (n=12/38)
Perception of phosphate management regimen

Total number of patients who discontinued from Part A: 18 of 243 (7.4%).
[a]Patients who responded that their phosphate management regimen had "very much improved," "much improved," or "minimally improved" in the Patient Experience Questionnaire (Q1). [b]Patients who responded that their phosphate management regimen had "no change," "minimally worsened," or "very much worsened" in the Patient Experience Questionnaire (Q1).

Treatment Regimen Perception and Reductions in Serum P Concentrations

- Of the 205 patients who reported an improvement in their serum P–lowering treatment regimen, approximately half (90 of 205 [43.9%]) had a reduction in their mean serum P concentration of ≥1.2 mg/dL.
- Of the 38 patients who reported no change or worsening in their serum P management routine, a smaller percentage (13 of 38 [34.2%]) of patients had a reduction in their mean serum P concentration of ≥1.2 mg/dL.
- At the end of Part A, patients who reported an improved perception of their phosphate management regimen had a greater mean serum P reduction from baseline (1.0 mg/dL) compared with those who reported no change or worsening (0.4 mg/dL) (**Figure 5**).
 – The greatest mean serum P reduction from baseline (1.5 mg/dL) was observed in the 69 patients who responded that their phosphate management was "very much improved."

Figure 5: Mean Serum P Changes at the End of Part A by Patients' Perceptions of Their Phosphate Management Regimen

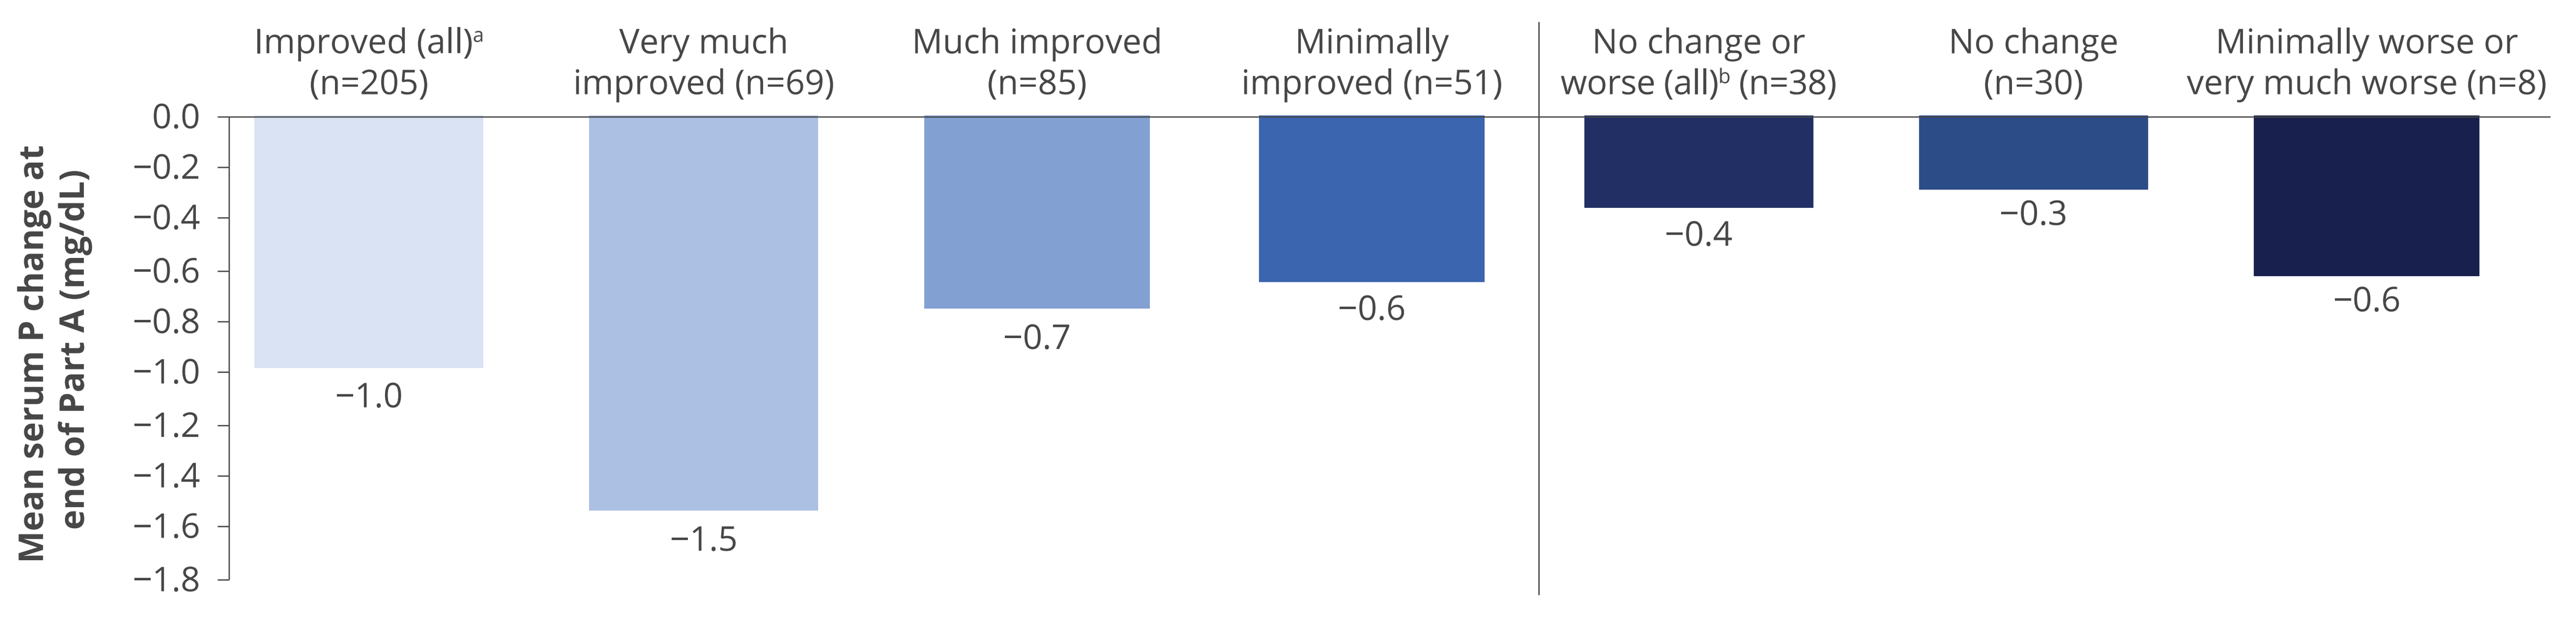
Mean serum P change at end of Part A (mg/dL)
Improved (all)[a] (n=205)
Very much improved (n=69)
Much improved (n=85)
Minimally improved (n=51)
No change or worse (all)[b] (n=38)
No change (n=30)
Minimally worse or very much worse (n=8)
0.0
−0.2
−0.4
−0.6
−0.8
−1.0
−1.2
−1.4
−1.6
−1.8
−1.0
−1.5
−0.7
−0.6
−0.4
−0.3
−0.6

N=243.
No patients responded "much worse" to Q1 of the Patient Experience Questionnaire; therefore, this response has been omitted from the figure.
[a]Combined mean change from baseline in serum P for patients who responded "very much improved," "much improved," or "minimally improved" to Q1 of the Patient Experience Questionnaire. [b]Combined mean change from baseline in serum P for patients who responded "no change," "minimally worse," or "very much worse" to Q1 of the Patient Experience Questionnaire.
P, phosphate.

Patient-Cited Reasons for an Improved Phosphate Management Routine

- Approximately two-thirds of patients reporting an improvement in their phosphate management routine (132 of 205 patients [64%]) attributed it to a better phosphate-lowering treatment regimen (reduced pill size/ number), while approximately one-third (64 of 205 [31%]) attributed it to improvements in bowel function (either improvement in bowel movement frequency or stool form).
 – These were the 2 changes selected most frequently by patients as reasons for having an improved perception of their phosphate management routine.
- The remaining patients (9 of 205 [4.4%]) stated other reasons for having an improved perception of their phosphate management routine.
- The mean serum P reductions from baseline were 1.1 mg/dL and 0.9 mg/dL for patients reporting improvements due to changes in their bowel movements (n=64) or medication burden (n=132), respectively. The mean serum P reduction for patients reporting worsening due to changes in their bowel movements (n=7) was 0.9 mg/dL (**Figure 6**).

Figure 6: Mean Serum P Changes From Baseline at the End of Part A in Patients Citing Bowel Movement or Medication Burden as the Main Reason for the Change in Their Perception

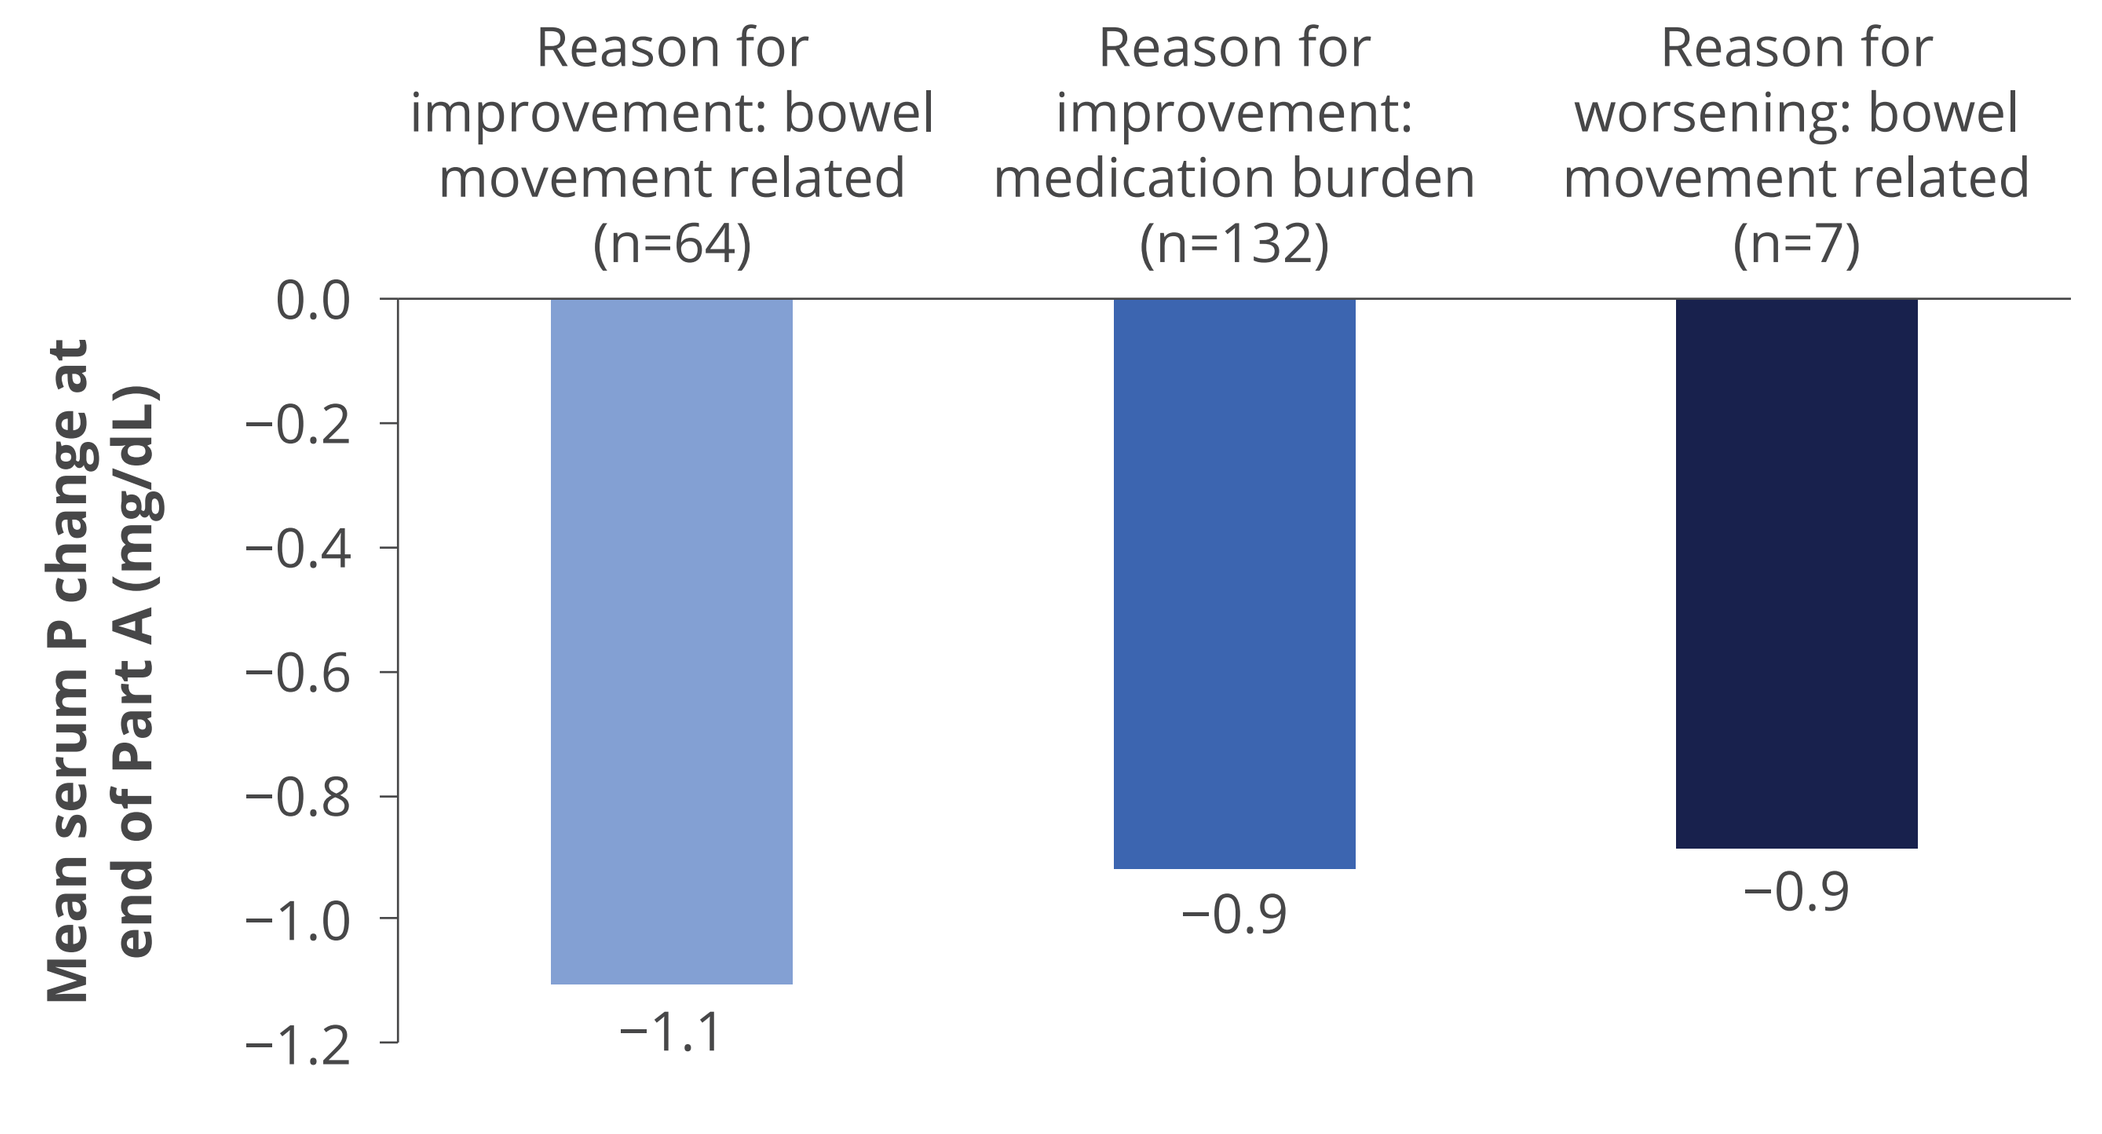
Mean serum P change at end of Part A (mg/dL)
Reason for improvement: bowel movement related (n=64)
Reason for improvement: medication burden (n=132)
Reason for worsening: bowel movement related (n=7)
0.0
−0.2
−0.4
−0.6
−0.8
−1.0
−1.2
−1.1
−0.9
−0.9

Patients who selected a different reason as the main reason for a change in their perception of their phosphate management routine were excluded from plot due to small sample size.
P, phosphate.

Conclusions

An improvement in patients' perception of their phosphate-lowering treatment regimen may improve adherence to treatment and promote a greater serum P reduction at the end of treatment.

More research is needed to validate these relationships and determine the impact of other contributing factors.

References

1. KDIGO 2017. *Kidney Int Suppl* (2011). 2017;7(1):1-59.
2. National Insitutes of Health. Phosphorus fact sheet for health professionals. 2023. Available from: https://ods.od.nih.gov/factsheets/Phosphorus-HealthProfessional/#disc
3. KDIGO CKD Work Group. *Kidney Int*. 2024;105:S117-314.
4. Barreto FC et al. *Kidney Int Rep*. 2019;4:1043-56.
5. Fissell RB et al. *Hemodial Int*. 2016;20:38-49.
6. Forfang D et al. *Kidney Med*. 2022;4:100437.
7. Jacobs JW et al. *ACS Med Chem Lett*. 2022;13:1043-51.
8. King AJ et al. *Sci Transl Med*. 2018;10:eaam6474.
9. Labonté ED et al. *J Am Soc Nephrol*. 2015;26:1138-49.
10. XPHOZAH. Prescribing information. Ardelyx Inc.; 2023. Available from: https://dailymed.nlm.nih.gov/dailymed/druginfo.cfm?setid=c57e279c-0f1b-4238-81b7-4e0dc0cc60c5
11. Sprague SM et al. *Kidney360*. 2024. Epub ahead of print.

Disclosures

Amy Mosman has no conflicts of interest to disclose. Susan Edelstein, David Spiegel, Suling Zhao, and David Rosenbaum are employees of Ardelyx, Inc.

Acknowledgements

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Amy Mosman, PA-C, can be contacted for further information on this study at Amy.Mosman@slucare.ssmhealth.com.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

National Kidney Foundation – Spring Clinical Meetings 2024 (SCM24); May 14-18, 2024; Long Beach, CA, USA

MED-US—2400176 07/2025

INDICATION

XPHOZAH (tenapanor) is indicated to reduce serum phosphorus in adults with chronic kidney disease (CKD) on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders or who are intolerant of any dose of phosphate binder therapy.

IMPORTANT SAFETY INFORMATION

CONTRAINDICATIONS

XPHOZAH is contraindicated in patients under 6 years of age.

XPHOZAH is contraindicated in patients with known or suspected mechanical gastrointestinal obstruction.

WARNINGS AND PRECAUTIONS

Diarrhea

Patients may experience severe diarrhea. Treatment with XPHOZAH should be discontinued in patients who develop severe diarrhea.

MOST COMMON ADVERSE REACTIONS

Diarrhea, which occurred in 43% to 53% of patients, was the only adverse reaction reported in at least 5% of XPHOZAH-treated patients with CKD on dialysis across trials. The majority of diarrhea events in the XPHOZAH-treated patients were reported to be mild to moderate in severity and resolved over time or with dose reduction. Diarrhea was typically reported soon after initiation but could occur at any time during treatment with XPHOZAH. Severe diarrhea was reported in 5% of XPHOZAH-treated patients in these trials.

For additional safety information, please see full Prescribing Information, available here.

CKD, chronic kidney disease.
XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc; 2025.

© Ardelyx, Inc., 2025. All rights reserved. Ardelyx and XPHOZAH are registered trademarks of Ardelyx, Inc.
Do not copy, modify, or disseminate without permission. For educational purposes only