Poster TH-PO169

Tenapanor Reduces Serum Phosphate With Similar Efficacy and Tolerability Profiles When Added to Various Phosphate Binders

Stuart M. Sprague,[1] David P. Tietjen,[2] Jesslyn M. Roesch,[3] Yang Yang,[4] Suling Zhao,[4] Susan A. Edelstein,[4] David P. Rosenbaum,[4] David M. Spiegel[4]

[1]NorthShore University Health System, Evanston, IL, USA; [2]Nephrology Consultants LLC, Huntsville, AL, USA; [3]Apogee Clinical Research LLC, Huntsville, AL, USA; [4]Ardelyx, Inc., Waltham, MA, USA

Introduction

- Tenapanor is approved to reduce serum phosphorus in adults with chronic kidney disease on dialysis as add-on therapy in patients who have an inadequate response to phosphate binders (PBs) or who are intolerant of any dose of PB therapy.[1]
- PBs alone are often insufficient treatment to achieve target-directed serum phosphate (serum P). Moreover, PBs often require numerous pills each day and can cause gastrointestinal side effects.[2-4]
- Typically, these side effects may include bloating, nausea, abdominal pain, constipation, and diarrhea (of varying severity), depending on the type of PB regimen used.[2]
 - For example, calcium-based binders and sevelamer can result in constipation, while iron-based binders are associated with increased rates of diarrhea.[2]
- Tenapanor is a first-in-class, minimally absorbed inhibitor of sodium/hydrogen exchanger 3, which decreases the permeability of the paracellular phosphate pathway, the primary gastrointestinal phosphate absorption pathway, thus providing a mechanism of action distinct from PBs.[5]
- We carried out a post hoc analysis of 2 clinical trials, AMPLIFY (NCT03824587) and OPTIMIZE (NCT04549597), to further evaluate the efficacy and tolerability of tenapanor when added to different types of PBs.

Methods

- In the Ardelyx-supported AMPLIFY and OPTIMIZE trials, patients on maintenance dialysis with uncontrolled serum P (defined as ≥5.5 and >5.5 mg/dL, respectively), despite receiving stable doses of PBs, were initiated on tenapanor.[6,7]
- In AMPLIFY, patients received an initial dose of 30 mg tenapanor or placebo twice a day (bid); during the first 2 weeks of the treatment period, tenapanor could have been titrated down (or back up) based on tolerability and serum P levels.[6]
 - Tenapanor was added to baseline PB therapy, and the latter remained stable during the 4-week treatment period (**Figure 1A**).[5]
- In OPTIMIZE, in the binder reduction arm (Cohort 2), tenapanor (initial dose: 30 mg bid) was added to a baseline PB, and the PB was reduced by ≥50% (**Figure 1B**).[7]

Figure 1: Study Designs of AMPLIFY (A) and OPTIMIZE (B)[a,b]

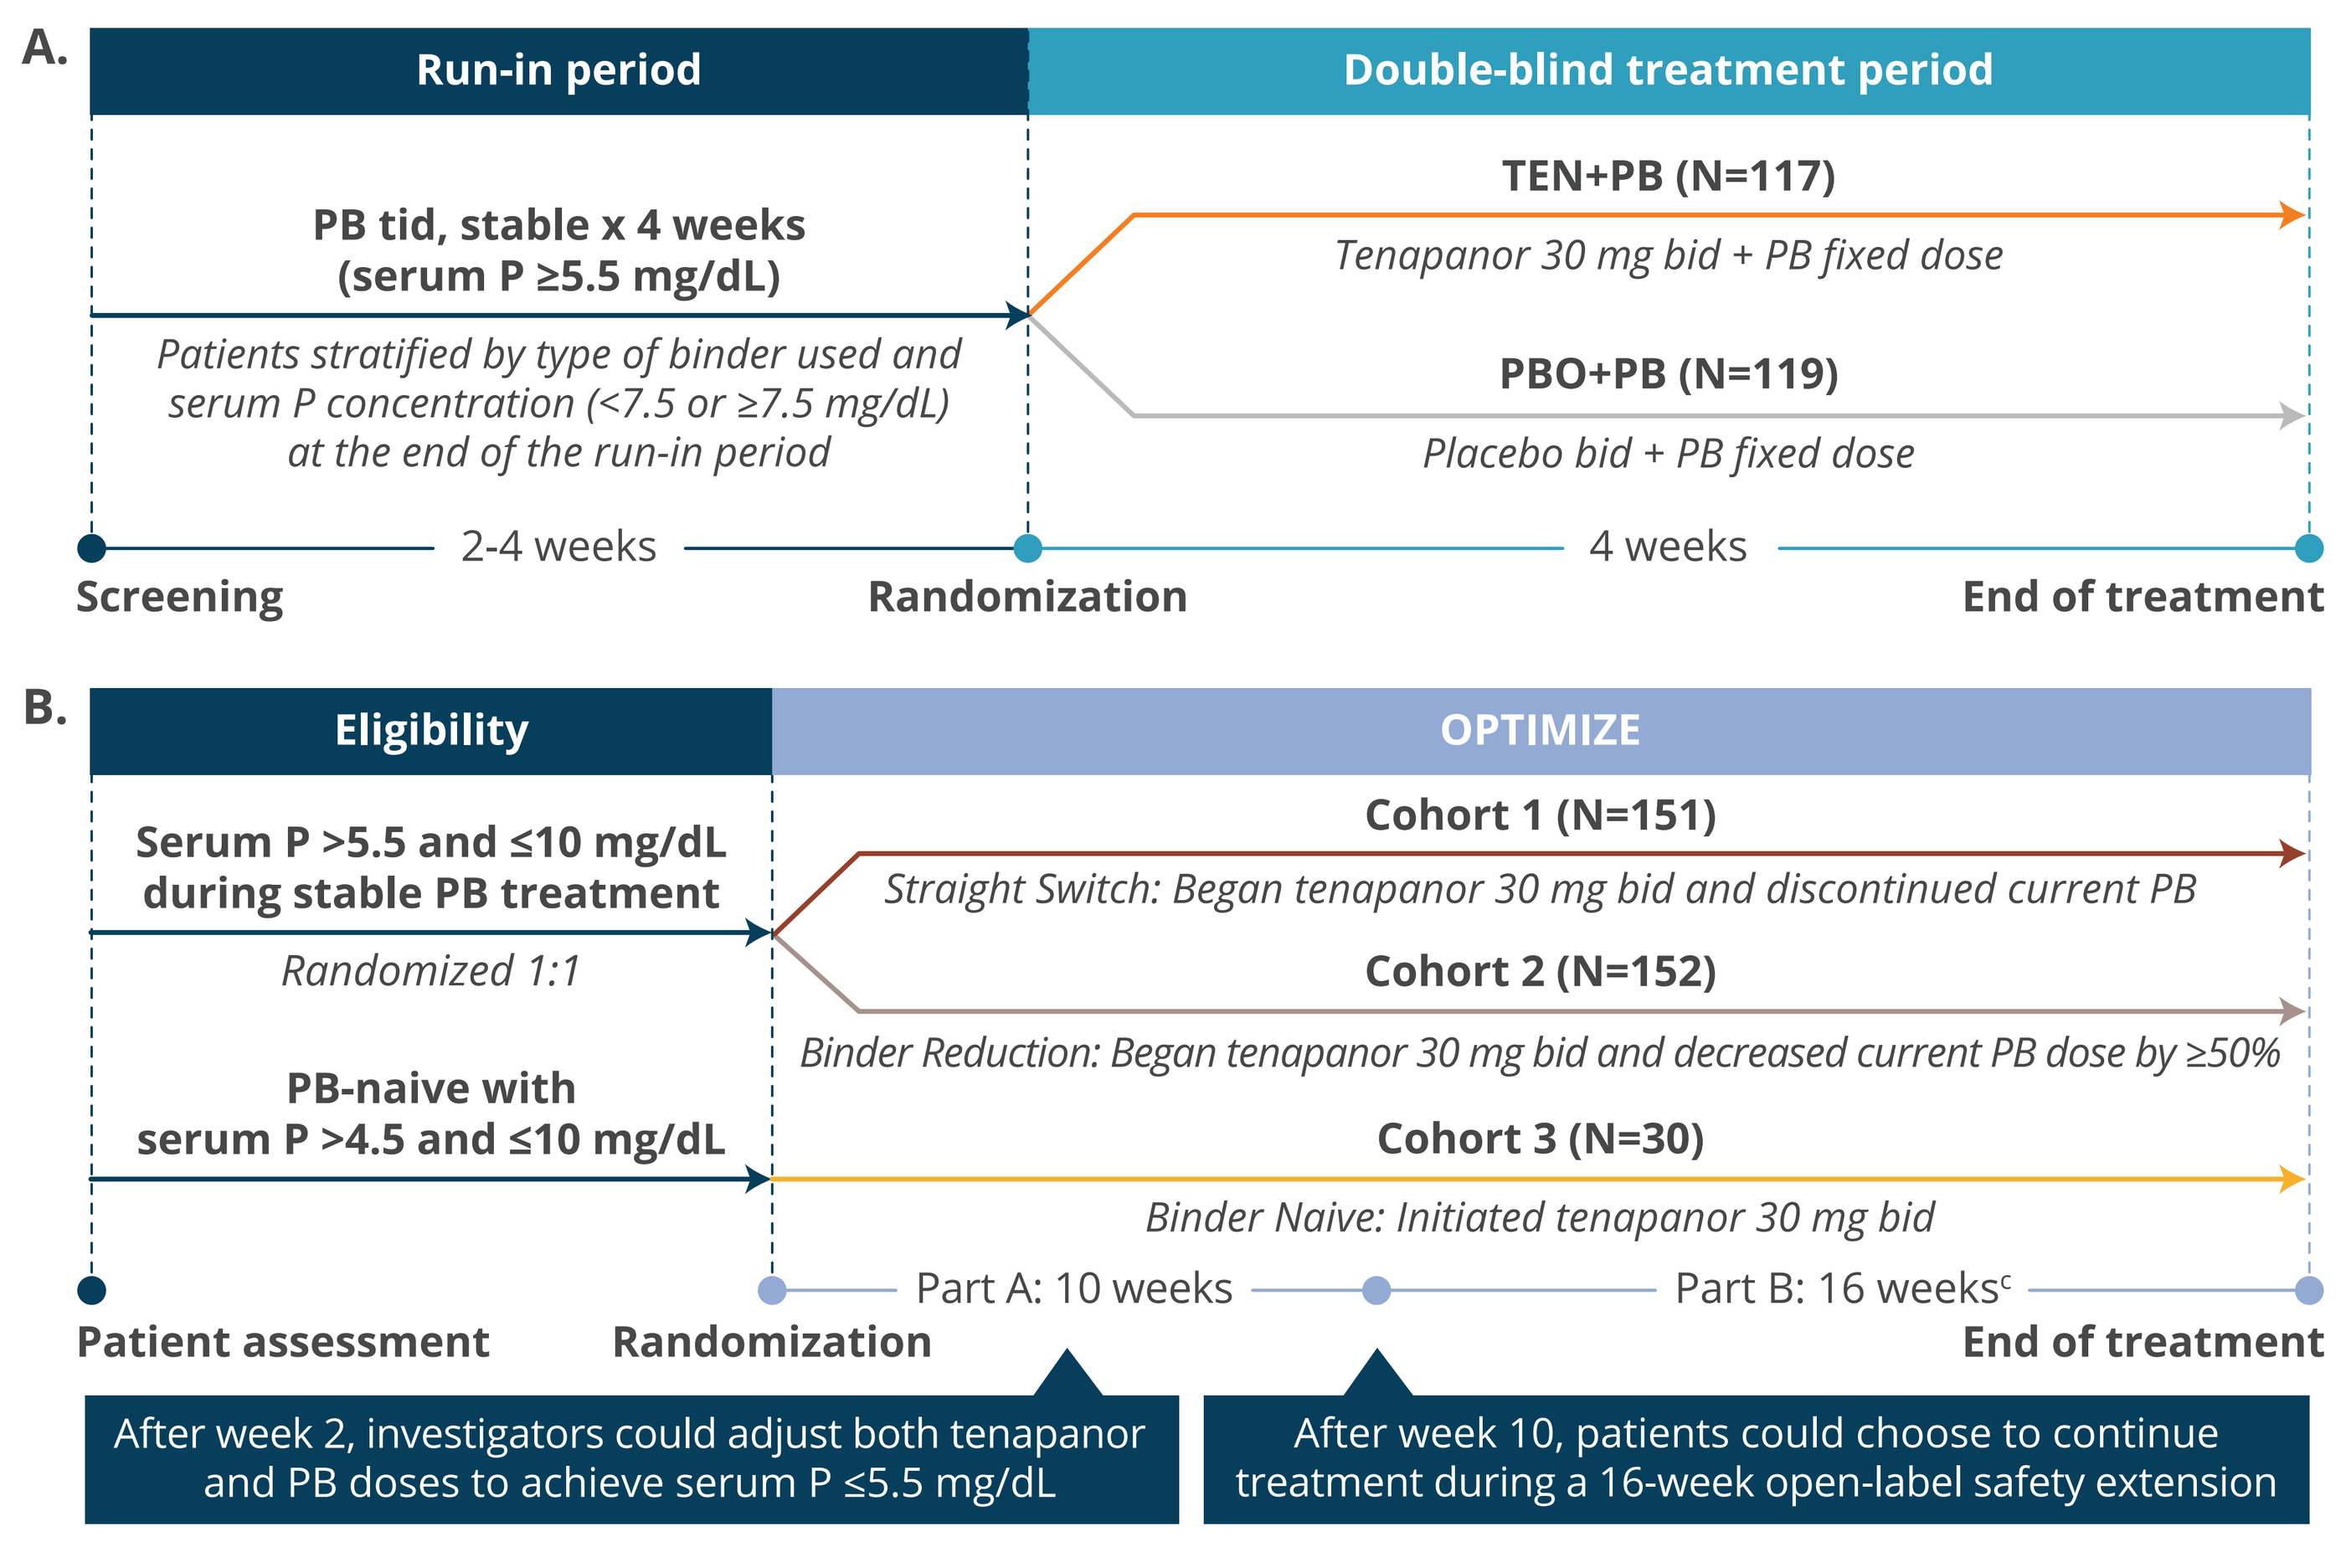
A.
Run-in period
Double-blind treatment period
PB tid, stable x 4 weeks
(serum P ≥5.5 mg/dL)
Patients stratified by type of binder used and serum P concentration (<7.5 or ≥7.5 mg/dL) at the end of the run-in period
TEN+PB (N=117)
Tenapanor 30 mg bid + PB fixed dose
PBO+PB (N=119)
Placebo bid + PB fixed dose
2-4 weeks
4 weeks
Screening
Randomization
End of treatment
B.
Eligibility
OPTIMIZE
Serum P >5.5 and ≤10 mg/dL during stable PB treatment
Randomized 1:1
PB-naive with serum P >4.5 and ≤10 mg/dL
Cohort 1 (N=151)
Straight Switch: Began tenapanor 30 mg bid and discontinued current PB
Cohort 2 (N=152)
Binder Reduction: Began tenapanor 30 mg bid and decreased current PB dose by ≥50%
Cohort 3 (N=30)
Binder Naive: Initiated tenapanor 30 mg bid
Part A: 10 weeks
Part B: 16 weeks[c]
Patient assessment
Randomization
End of treatment
After week 2, investigators could adjust both tenapanor and PB doses to achieve serum P ≤5.5 mg/dL
After week 10, patients could choose to continue treatment during a 16-week open-label safety extension

[a]Only patients from Cohort 2 in OPTIMIZE (binder reduction cohort) were intended to be evaluated for the effect of tenapanor in combination with PBs and were included in this analysis. Patients in Cohort 1 were excluded as they discontinued PBs at the initiation of tenapanor, and patients in Cohort 3 were excluded as they were naive to PB treatment at baseline. [b]If patients randomized to Cohort 2 of OPTIMIZE were taking an odd number of binder pills at baseline, they decreased the number of pills by more than 50% (eg, 3 pills were decreased to 1, 5 pills were decreased to 2). [c]Part B was optional.
bid, twice a day; P, phosphate; PB, phosphate binder; PBO, placebo; TEN, tenapanor; tid, three times a day.

- In this post hoc analysis, serum P reductions from baseline were evaluated at the end of the 4-week treatment period in tenapanor-treated patients in AMPLIFY (N=116) and at the end of the 10-week treatment period (ie, part A) in Cohort 2 patients in OPTIMIZE (N=152).
- In the efficacy analysis and summary of baseline demographics and disease characteristics by study, these patients (ie, post hoc efficacy analysis set) were categorized into 2 subgroups based on their baseline PB regimen:
 - Sevelamer-based PB regimen subgroup: patients who were receiving either sevelamer alone or sevelamer plus non-sevelamer PB(s) at baseline.
 - Non–sevelamer based PB regimen subgroup: patients who were receiving non-sevelamer PB(s) only at baseline.
- Safety and tolerability were assessed by baseline PB regimen with more refined categorization (ie, sevelamer alone, calcium-based PB alone, iron-based PB alone, lanthanum carbonate alone, and multiple PB types) using pooled data obtained during the 4-week treatment period in AMPLIFY and from the entire 26-week study in OPTIMIZE.
 - The analysis set for this post hoc safety analysis comprised 269 tenapanor-treated patients from AMPLIFY and OPTIMIZE Cohort 2 (ie, post hoc safety analysis set).

Results

Patients

- Baseline demographics and disease characteristics for the 268 patients in the post hoc efficacy analysis set are presented in **Table 1**.
 - For patients receiving a sevelamer PB regimen at baseline, 46 of 60 (76.7%) patients in AMPLIFY received sevelamer alone, which was comparable to the 61 of 82 (74.4%) patients in OPTIMIZE Cohort 2. The remaining patients received sevelamer in combination with non-sevelamer PB(s).
 - For patients receiving a non-sevelamer PB regimen at baseline, iron-based PB use was more prevalent in OPTIMIZE Cohort 2 (41 of 70 [58.6%]) compared with AMPLIFY (14 of 56 [25.0%]), in which the most commonly used PBs at baseline were calcium-based (27 of 56 [48.2%]).

Table 1: Baseline Demographics and Disease Characteristics (Post Hoc Efficacy Analysis Set[a])

	AMPLIFY N=116		OPTIMIZE Cohort 2 N=152	
Characteristic	SEV-based PB regimen N'=60	Non–SEV based PB regimen N'=56	SEV-based PB regimen N'=82	Non–SEV based PB regimen N'=70
Age, mean (SD), y	54.9 (11.3)	54.2 (13.6)	55.0 (11.2)	51.2 (12.9)
Female, n (%)	23 (38.3)	28 (50.0)	29 (35.4)	21 (30.0)
Race, n (%)				
Asian	1 (1.7)	1 (1.8)	2 (2.4)	1 (1.4)
Black or African American	23 (38.3)	28 (50.0)	40 (48.8)	31 (44.3)
Native American or Alaskan	3 (5.0)	1 (1.8)	5 (6.1)	1 (1.4)
Native Hawaiian or Pacific Islander	1 (1. 7)	0	1 (1.2)	3 (4.3)
White	31 (51.7)	26 (46.4)	32 (39.0)	30 (42.9)
Other or unknown	1 (1.7)	0	2 (2.4)	4 (5.7)
Ethnicity, n (%)				
Hispanic or Latino	22 (36.7)	15 (26.8)	17 (20.7)	21 (30.0)
Non-Hispanic or non-Latino	38 (63.3)	40 (71.4)	65 (79.3)	49 (70.0)
Not reported	0	1 (1.8)	0	0
BMI, mean (SD), kg/m²	33.4 (6.7)	33.4 (8.7)	31.8 (8.3)	32.1 (8.4)
Duration since first dialysis at baseline, mean (SD), mo	63.1 (55.4)	50.0 (39.6)	60.1 (53.5)	54.9 (44.5)
Binder type at baseline, n (%)				
Sevelamer binder alone	46 (76.7)	0	61 (74.4)	0
Calcium-based binder alone	0	27 (48.2)	0	20 (28.6)
Iron-based binder alone	0	14 (25.0)	0	41 (58.6)
Lanthanum carbonate–based binder alone	0	3 (5.4)	0	4 (5.7)
Multiple PB types	14 (23.3)	12 (21.4)	21 (25.6)	5 (7.1)
PB pills per day at baseline, median [min, max], pills	N/A	N/A	n=78 9.0 (6, 23)	n=70 8.5 (3, 18)
Total daily PB dose at baseline, median [min, max], mg	6401 [2400, 21600]	4001 [1500, 12006]	n=78 7200 [3030, 18001]	n=70 3000 [1260, 12006]
Serum P at baseline, mean (SD), mg/dL	6.8 (1.3)	6.6 (1.4)	7.1 (1.1)	7.2 (1.1)

[a]One patient in AMPLIFY received tenapanor but prematurely discontinued from the study without any post-baseline serum P measurements. This patient was therefore included in the post hoc safety analysis set but excluded from the post hoc efficacy analysis set.
BMI, body mass index; N/A, not applicable; P, phosphate, PB, phosphate binder; SEV sevelamer.

Efficacy

- In AMPLIFY, serum P consistently decreased from baseline throughout the 4-week treatment period when tenapanor was added to sevelamer-based and non–sevelamer based PB regimens, with no appreciable difference in the mean serum P reductions from baseline at the end of the 4-week treatment period between sevelamer-based regimen (0.9 mg/dL [SD: 1.3]) and non–sevelamer based regimen (0.8 mg/dL [SD: 1.4]) subgroups (**Figure 2A**).
- In OPTIMIZE Cohort 2, serum P levels consistently decreased from baseline throughout the 10-week treatment period when tenapanor was added to sevelamer-based and non–sevelamer based PB regimens, with no appreciable difference in the mean serum P reductions from baseline at the end of the 10-week treatment period between sevelamer-based regimen (1.0 mg/dL [SD: 1.8]) and non–sevelamer based regimen (0.9 mg/dL [SD: 1.5]) subgroups (**Figure 2B**).

Figure 2: Raw Mean Serum P Changes From Baseline to the End of the 4-Week Treatment Period in AMPLIFY (A) and From Baseline to the End of the 10-Week Treatment Period in OPTIMIZE (B) (Post Hoc Efficacy Analysis Set)

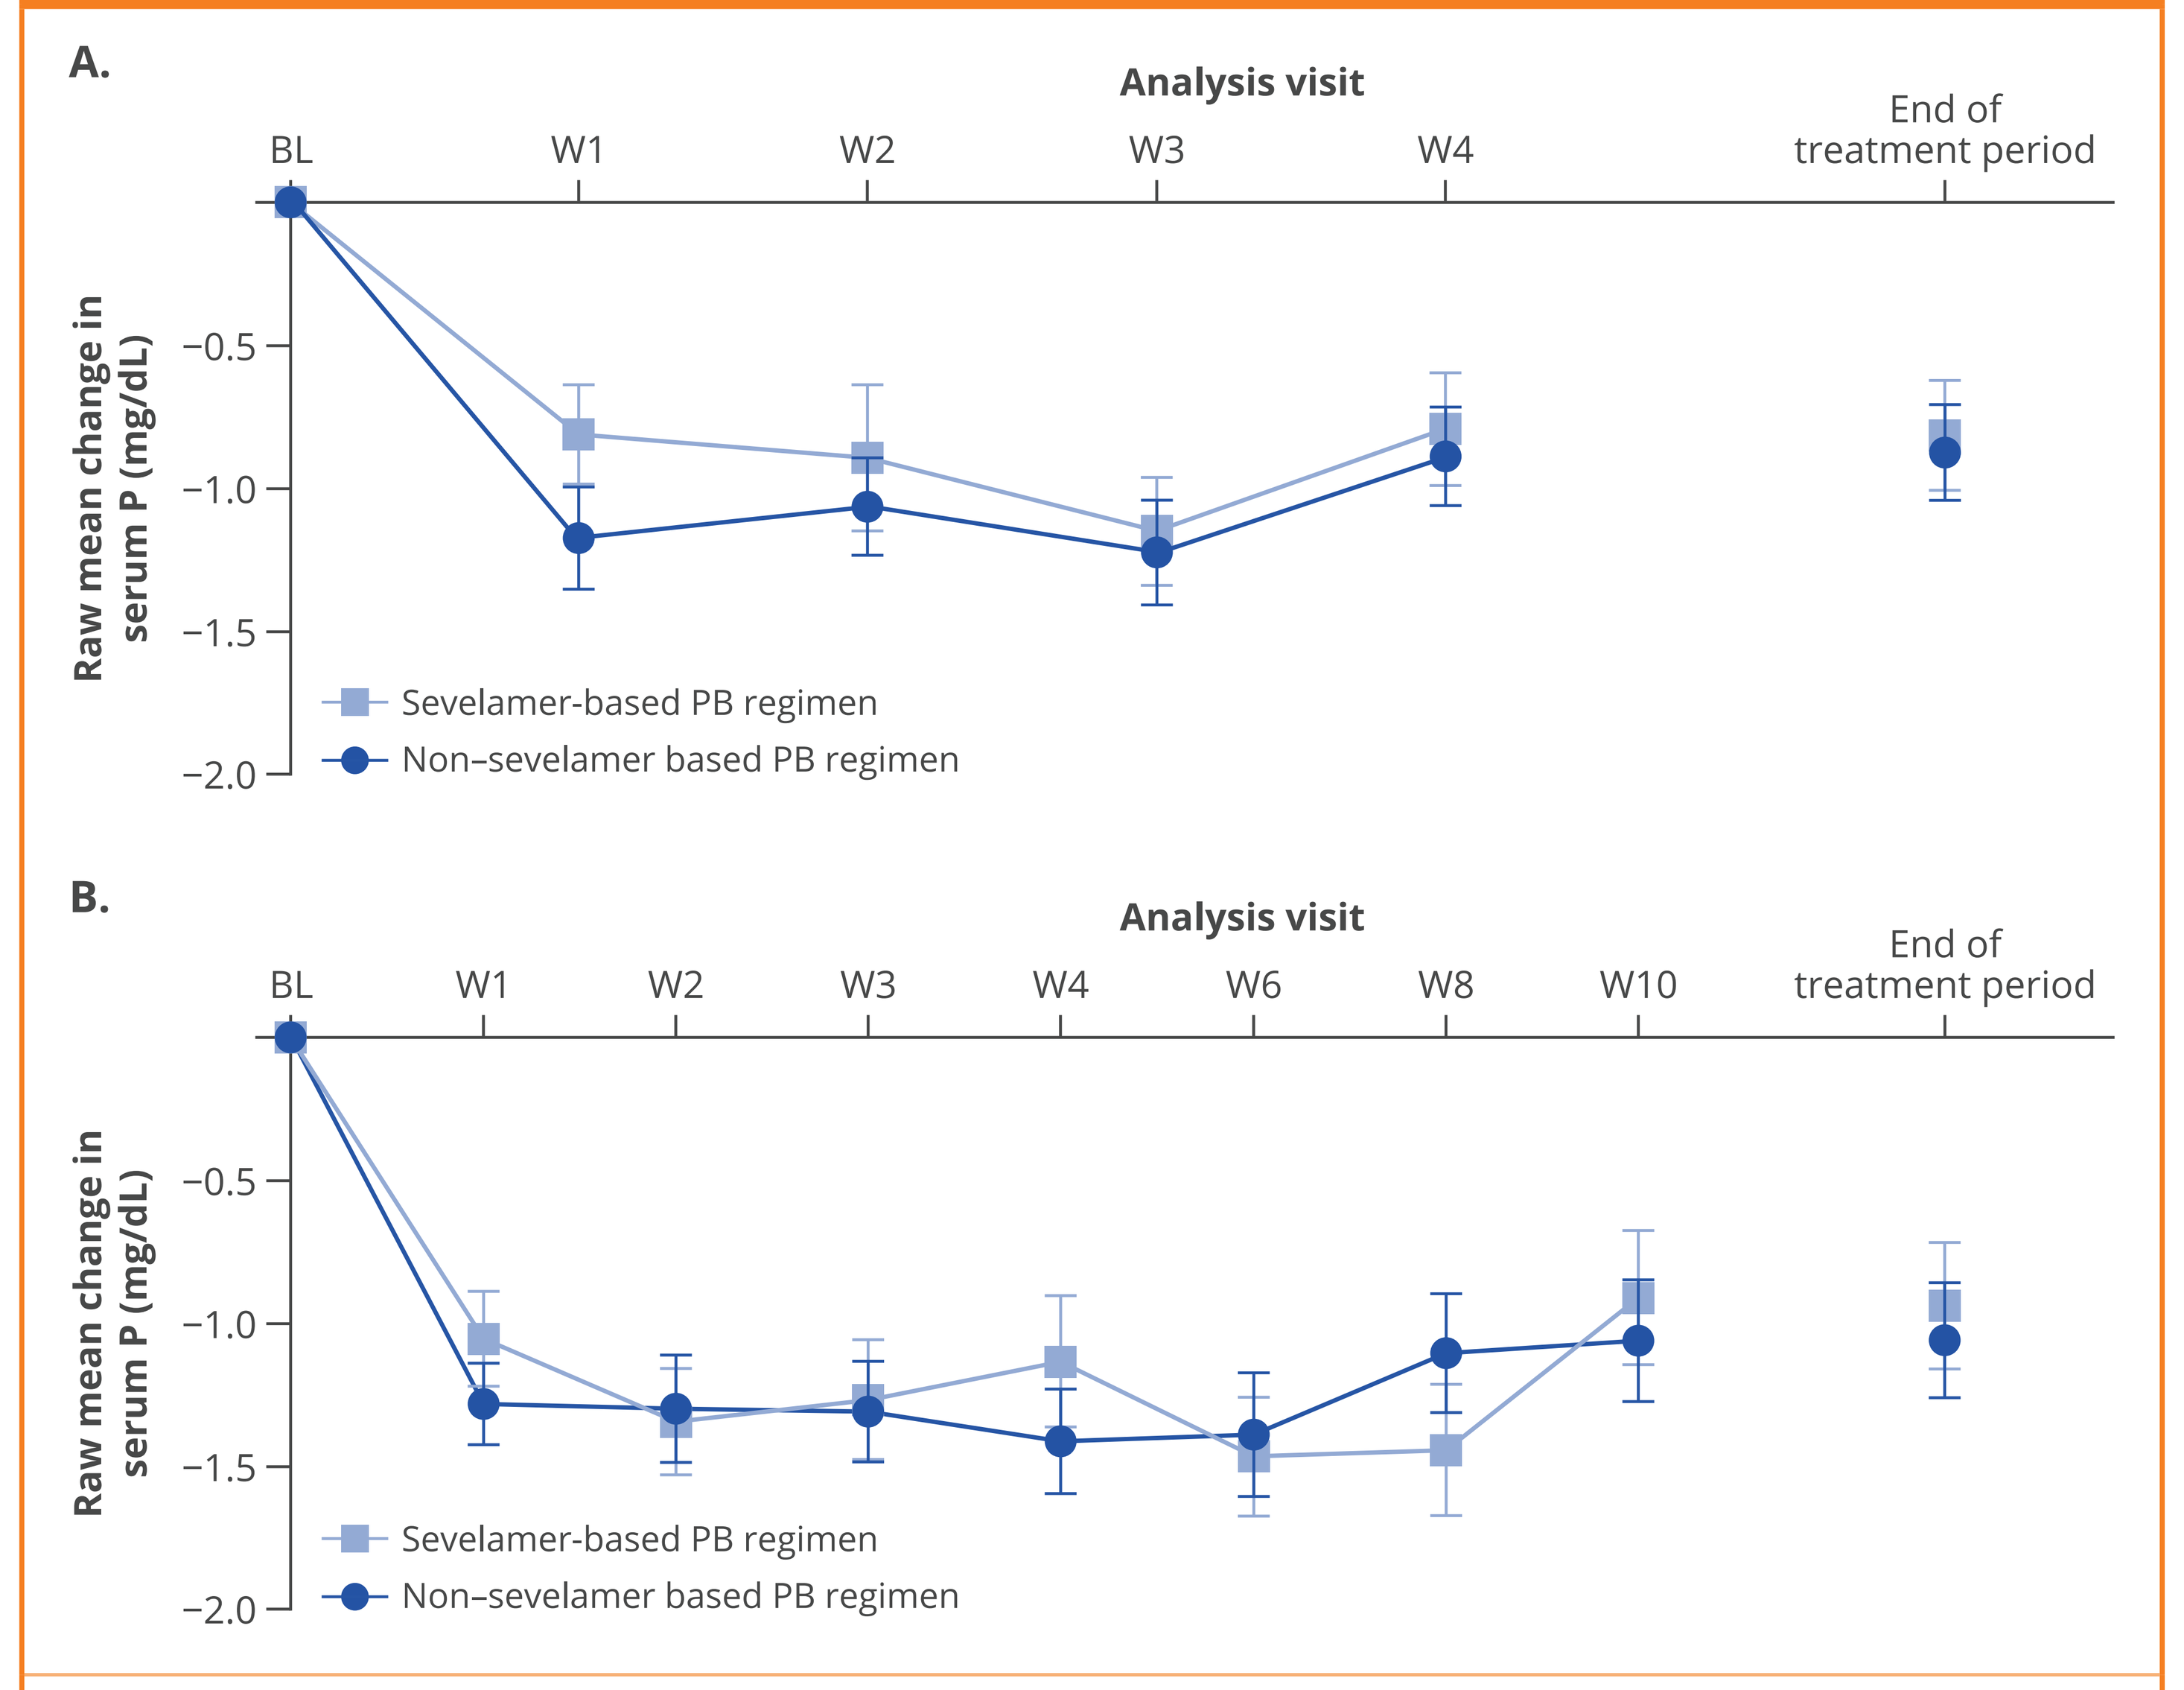
A.
Analysis visit
BL
W1
W2
W3
W4
End of treatment period
Raw mean change in serum P (mg/dL)
−0.5
−1.0
−1.5
−2.0
Sevelamer-based PB regimen
Non–sevelamer based PB regimen
B.
Analysis visit
BL
W1
W2
W3
W4
W6
W8
W10
End of treatment period
Raw mean change in serum P (mg/dL)
−0.5
−1.0
−1.5
−2.0
Sevelamer-based PB regimen
Non–sevelamer based PB regimen

Error bars represent ± standard error of the mean.
BL, baseline; P, phosphate; PB, phosphate binder.

Safety and Tolerability

- Data from the 269 patients treated with tenapanor in the post hoc safety analysis set were analyzed for safety and tolerability based on their baseline PB regimen.
- The overall incidence rate of treatment-emergent adverse events (TEAEs) ranged from 42.9% (3 of 7 patients receiving tenapanor plus lanthanum carbonate) to 68.1% (32 of 47 patients receiving tenapanor plus calcium-based PB) (**Figure 3**).
 - No unexpected TEAEs were reported.
- The highest incidence of suspected adverse reactions (SARs) was observed in patients who received tenapanor with multiple PB types (29 of 53 [54.7%]); however, this subgroup had the lowest incidence of serious adverse events and TEAEs leading to tenapanor discontinuation among all baseline PB-type subgroups (**Figure 3**).
- Overall, the only SAR with tenapanor that occurred in more than 5% of patients was diarrhea (108 of 269 [40.1%]).
 - Incidence rates of diarrhea were similar when tenapanor was added to sevelamer alone (38.9% [42 of 108]), calcium-based PB alone (46.8% [22 of 47]), iron-based PB alone (33.3% [18 of 54]), lanthanum carbonate alone (28.6% [2 of 7]), and multiple PB types (56.6% [30 of 53]).

Figure 3: Overall Summary of TEAEs by Baseline PB Type (Post Hoc Safety Analysis Set)

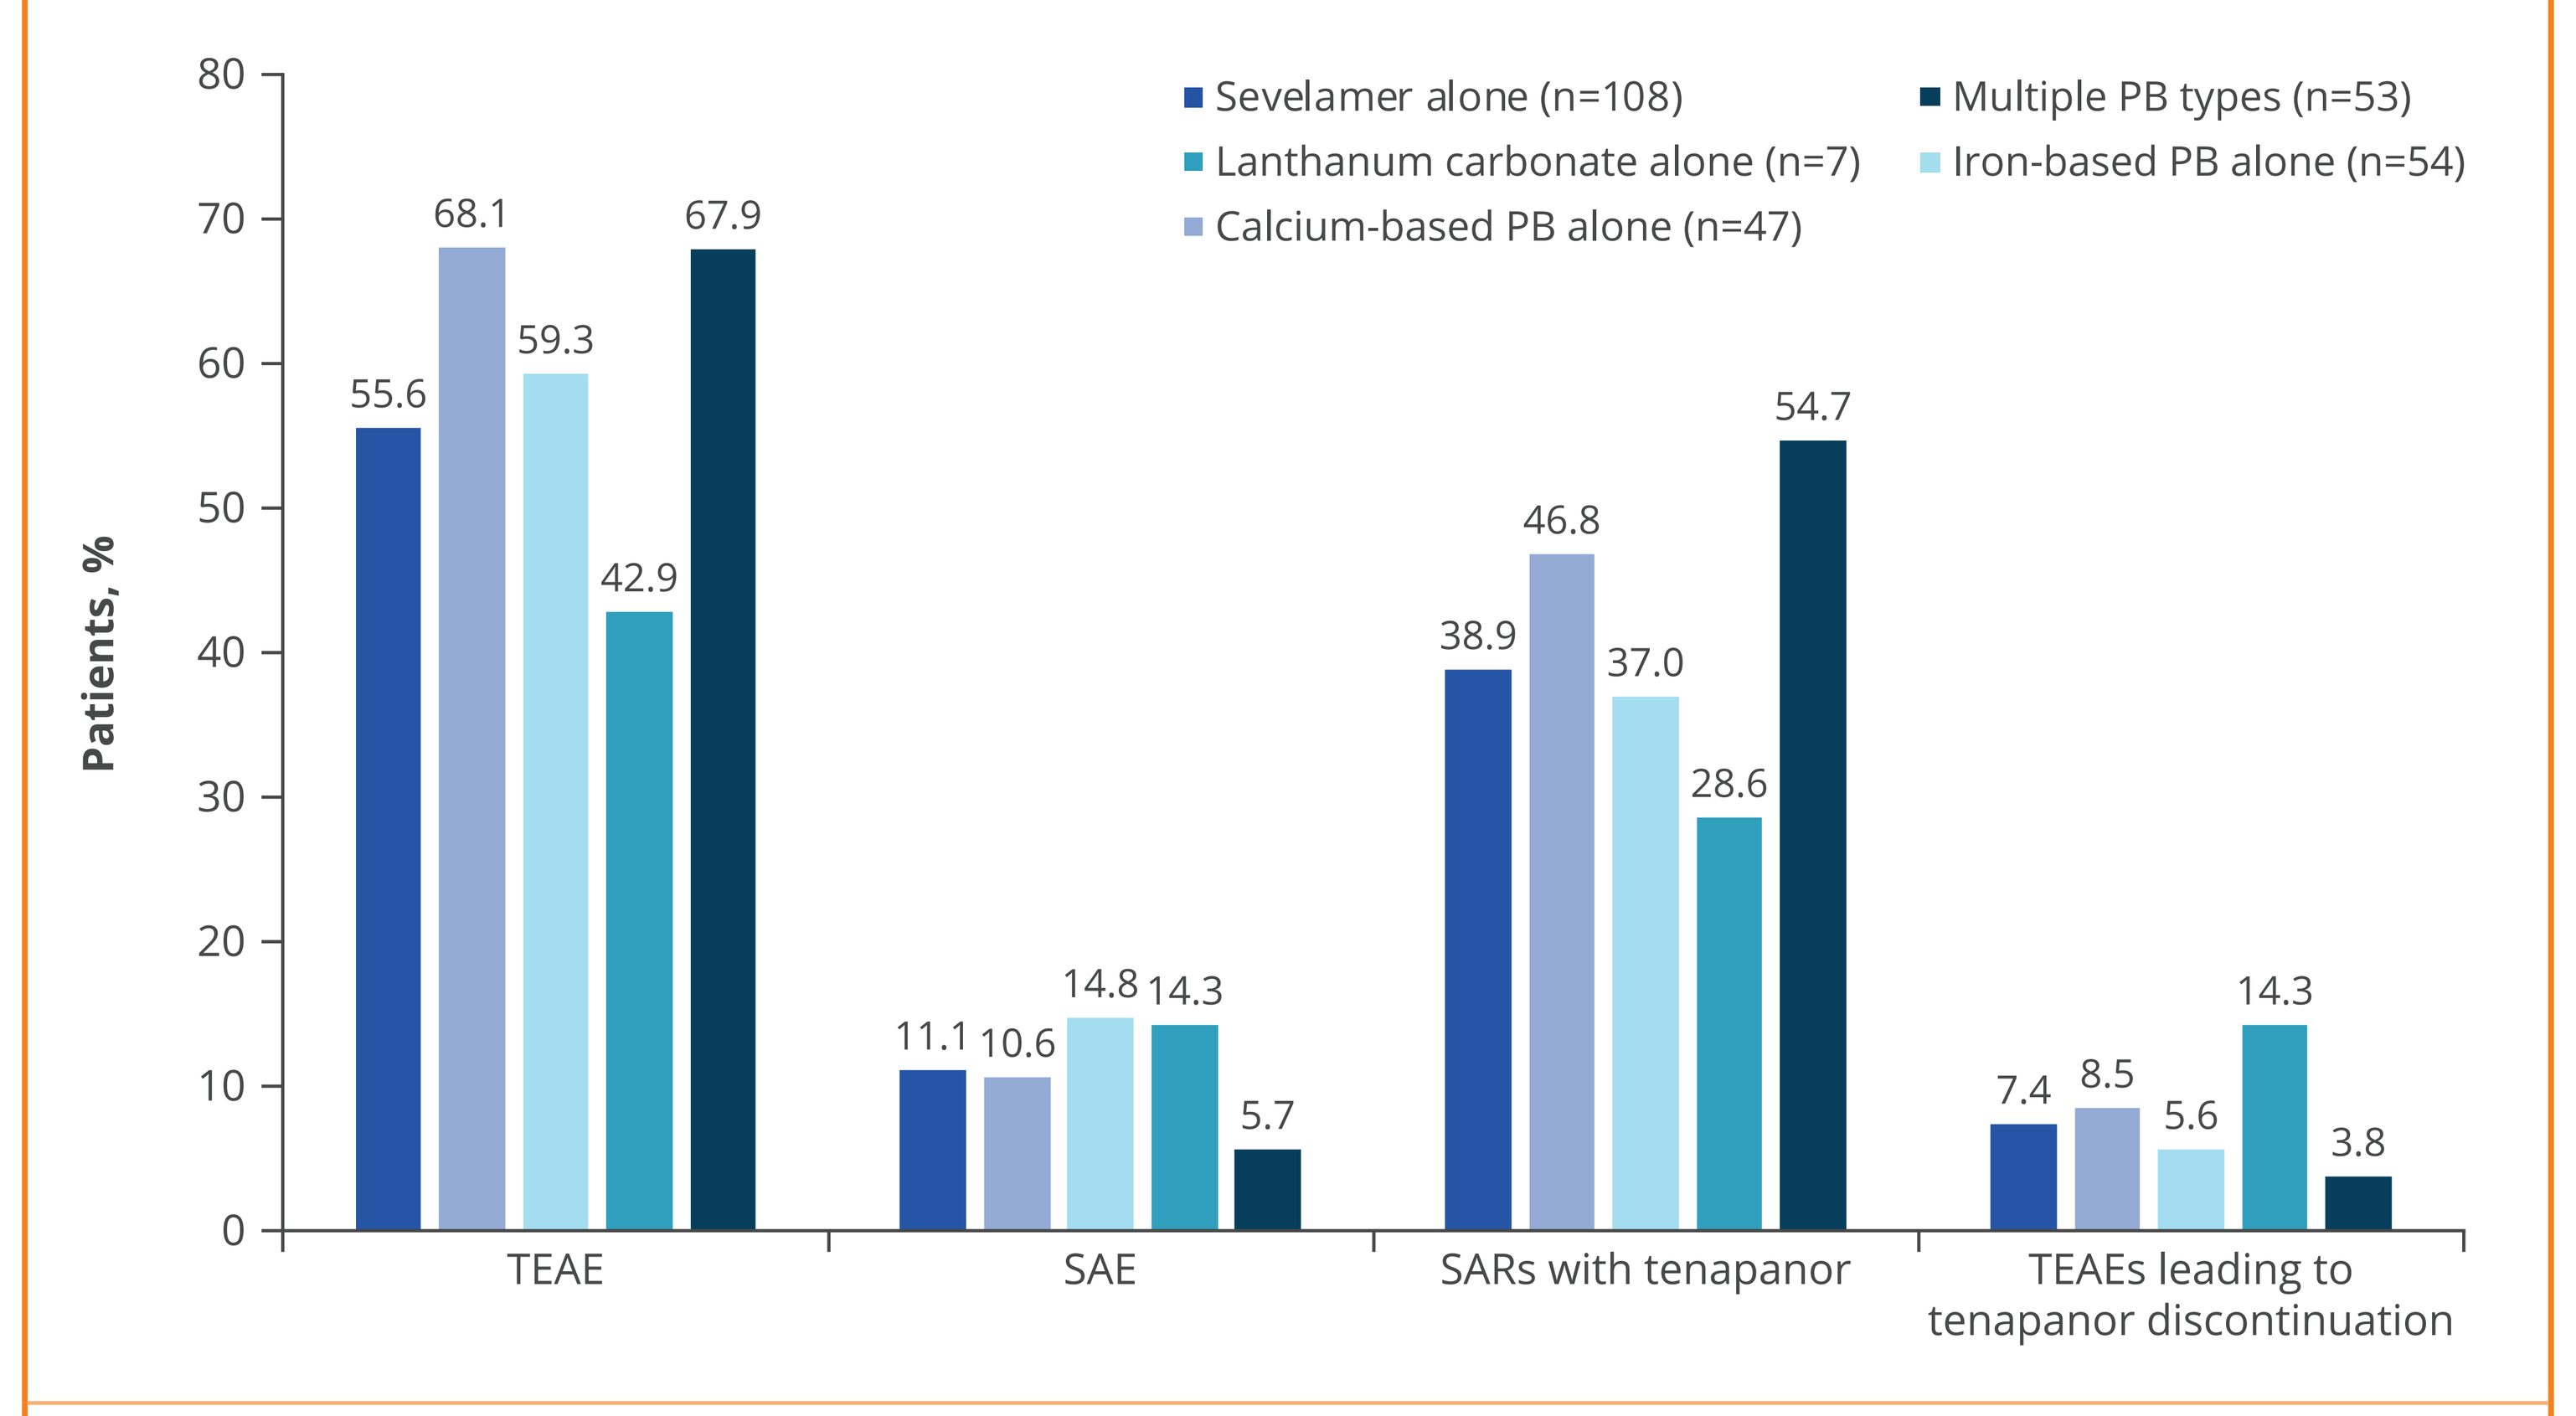
Patients, %
80
70
60
50
40
30
20
10
0
Sevelamer alone (n=108)
Lanthanum carbonate alone (n=7)
Calcium-based PB alone (n=47)
Multiple PB types (n=53)
Iron-based PB alone (n=54)
55.6
68.1
59.3
42.9
67.9
11.1
10.6
14.8
14.3
5.7
38.9
46.8
37.0
28.6
54.7
7.4
8.5
5.6
14.3
3.8
TEAE
SAE
SARs with tenapanor
TEAEs leading to tenapanor discontinuation

PB, phosphate binder; SAE, serious adverse event; SAR, suspected adverse reaction; TEAE, treatment-emergent adverse event.

- Study discontinuation rates were similar when tenapanor was added to sevelamer alone (13.0% [14 of 108]), calcium-based PB alone (8.5% [4 of 47]), iron-based PB alone (16.7% [9 of 54]), lanthanum carbonate alone (14.3% [1 of 7]), and multiple PB types (9.4% [5 of 53]).

Conclusion

Tenapanor provides a clinically meaningful serum P reduction with similar efficacy and tolerability when added to PBs, regardless of the background PB type.

Disclosures

This study was supported by Ardelyx, Inc. Yang Yang, Suling Zhao, Susan Edelstein, David Rosenbaum, and David Spiegel are employees of Ardelyx, Inc. Stuart M. Sprague receives research grants from and is a consultant for Ardelyx, Inc. David Tietjen and Jesslyn M. Roesch have nothing to disclose.

References

1. XPHOZAH (tenapanor hydrochloride). Prescribing information. Ardelyx, Inc.; 2023.
2. Biruete A et al. *J Ren Nutr*. 2020;30:4-10.
3. Chiu YW et al. *Clin J Am Soc Nephrol*. 2009;4:1089-96.
4. Robinson J et al. Poster presented at: American Society of Nephrology; October 24-27, 2020; San Diego, CA.
5. Silva AL et al. *Kidney360*. 2023;4:1580-9.
6. Pergola PE et al. *J Am Soc Nephrol*. 2021;32:1465-73.
7. Sprague SM et al. *Kidney360*. 2024;5:732-42.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr Sprague can be contacted for further information on this study at stuartmsprague@gmail.com.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American Society of Nephrology: Kidney Week 2024; October 23–27, 2024; San Diego, CA, USA.