Poster P2335

Patients With Irritable Bowel Syndrome With Constipation From the IBS In America 2024 Real-World Survey Experience Burdensome Symptoms Beyond Constipation

Baharak Moshiree,[1] Johannah Ruddy,[2] Belinda Gist,[2] Elizabeth Stremke,[2] Laura Williams,[2] Eric Shah[3]

[1]Wake Forest Medical University, Charlotte, NC, USA; [2]Ardelyx, Inc., Waltham, MA, USA; [3]University of Michigan, Ann Arbor, MI, USA

Background

- Irritable bowel syndrome (IBS) is a debilitating disorder of gut-brain interaction characterized by symptoms of recurring abdominal pain and disordered defecation.[1]
- IBS with constipation (IBS-C) accounts for approximately one-third of IBS cases and is characterized by hard and lumpy stools.[2,3]
- Patients with IBS-C also experience abdominal symptoms, such as bloating, discomfort, cramping, excessive gas, and fullness, that also contribute to the disease burden.[4,5]
- In 2024, the IBS In America survey was conducted to gain insight into the real-life experiences of patients with IBS.
- Here, we report on the symptoms described by patients with IBS-C beyond that of abdominal pain and constipation in the IBS In America 2024 survey.

Methods

- IBS In America 2024 was a 15-minute, real-world, online survey fielded by Health Union from January 15 to April 14, 2024, in United States residents aged ≥18 years, covering demographics, comorbidities, quality of life (QoL)/impact, treatments, and health care provider (HCP) interactions.
- Patients who completed the survey and met the following criteria were invited to participate in an institutional review board–approved extension survey and are included in this analysis:
 - Diagnosed with IBS-C by an HCP.
 - Currently seeing an HCP to treat their IBS-C.
 - Had ever used an over-the-counter or prescription treatment for their IBS-C.
- Four questions addressed patients' menstrual status and the impact of menstruation on constipation.

Results

Patients

- Respondents with IBS-C (N=284) had a mean age of 51.4 years and were predominantly White (87%) and female (92%); 48% of females were postmenopausal (**Table 1**).
- Most respondents (90%) had been living with IBS-C for ≥2 years, with 80% reporting IBS episodes occurring weekly or daily over the past year (**Table 1**).

Table 1: Demographics and Baseline Characteristics of Patients With IBS-C Who Completed the Survey

	IBS-C respondents (N=284)
Age, mean (range), y	51.4 (18–86)
Female, n (%)	262 (92)
Menstrual status[a] (n=262)	
Currently having menstrual cycles	81 (31)
Perimenopausal	24 (9)
Menopausal	31 (12)
Postmenopausal	126 (48)
Race,[b] n (%)	
Asian or South Asian	3 (1)
Black or African American	28 (10)
Native Hawaiian or other Pacific Islander	1 (0.4)
White	247 (87)
Other[c]	11 (3)
Ethnicity	
Hispanic or Latino	11 (4)
Non-Hispanic	273 (96)
Duration since IBS-C diagnosis, y	
≥15 y	88 (31)
10–15 y	43 (15)
5–10 y	60 (21)
2–5 y	65 (23)
Frequency of IBS episodes over the past year	
Daily	102 (36)
Weekly	125 (44)

[a]Self-described menstrual status. [b]Participants were instructed to select all that apply, permitting them to identify with multiple races/ethnicities. [c]Includes Native American or Alaska Native, "Prefer not to answer," and Other races/ethnicities.
IBS, irritable bowel syndrome; IBS-C, irritable bowel syndrome with constipation.

Symptoms

- Within the previous 7 days:
 - Eighty-six percent of patients with IBS-C experienced hard or lumpy stools at least once, which bothered 51% of these patients "quite a bit" or "very much" (**Figure 1A**).
 - Ninety-five percent needed to strain while trying to have bowel movements (BMs), with 49% of the patients straining "quite a bit" or "very much" (**Figure 1B**).
 - Eighty-five percent of patients experienced pain in the rectum or anus while trying to have a BM, of whom 27% described the pain as "quite bad" or "very bad" (**Figure 1C**).

Figure 1: Proportion of Patients With IBS-C Who Experienced (A) Hard or Lumpy Stools, (B) Strain, and/or (C) Pain in the Rectum or Anus While Trying to Have a Bowel Movement in the Previous 7 Days

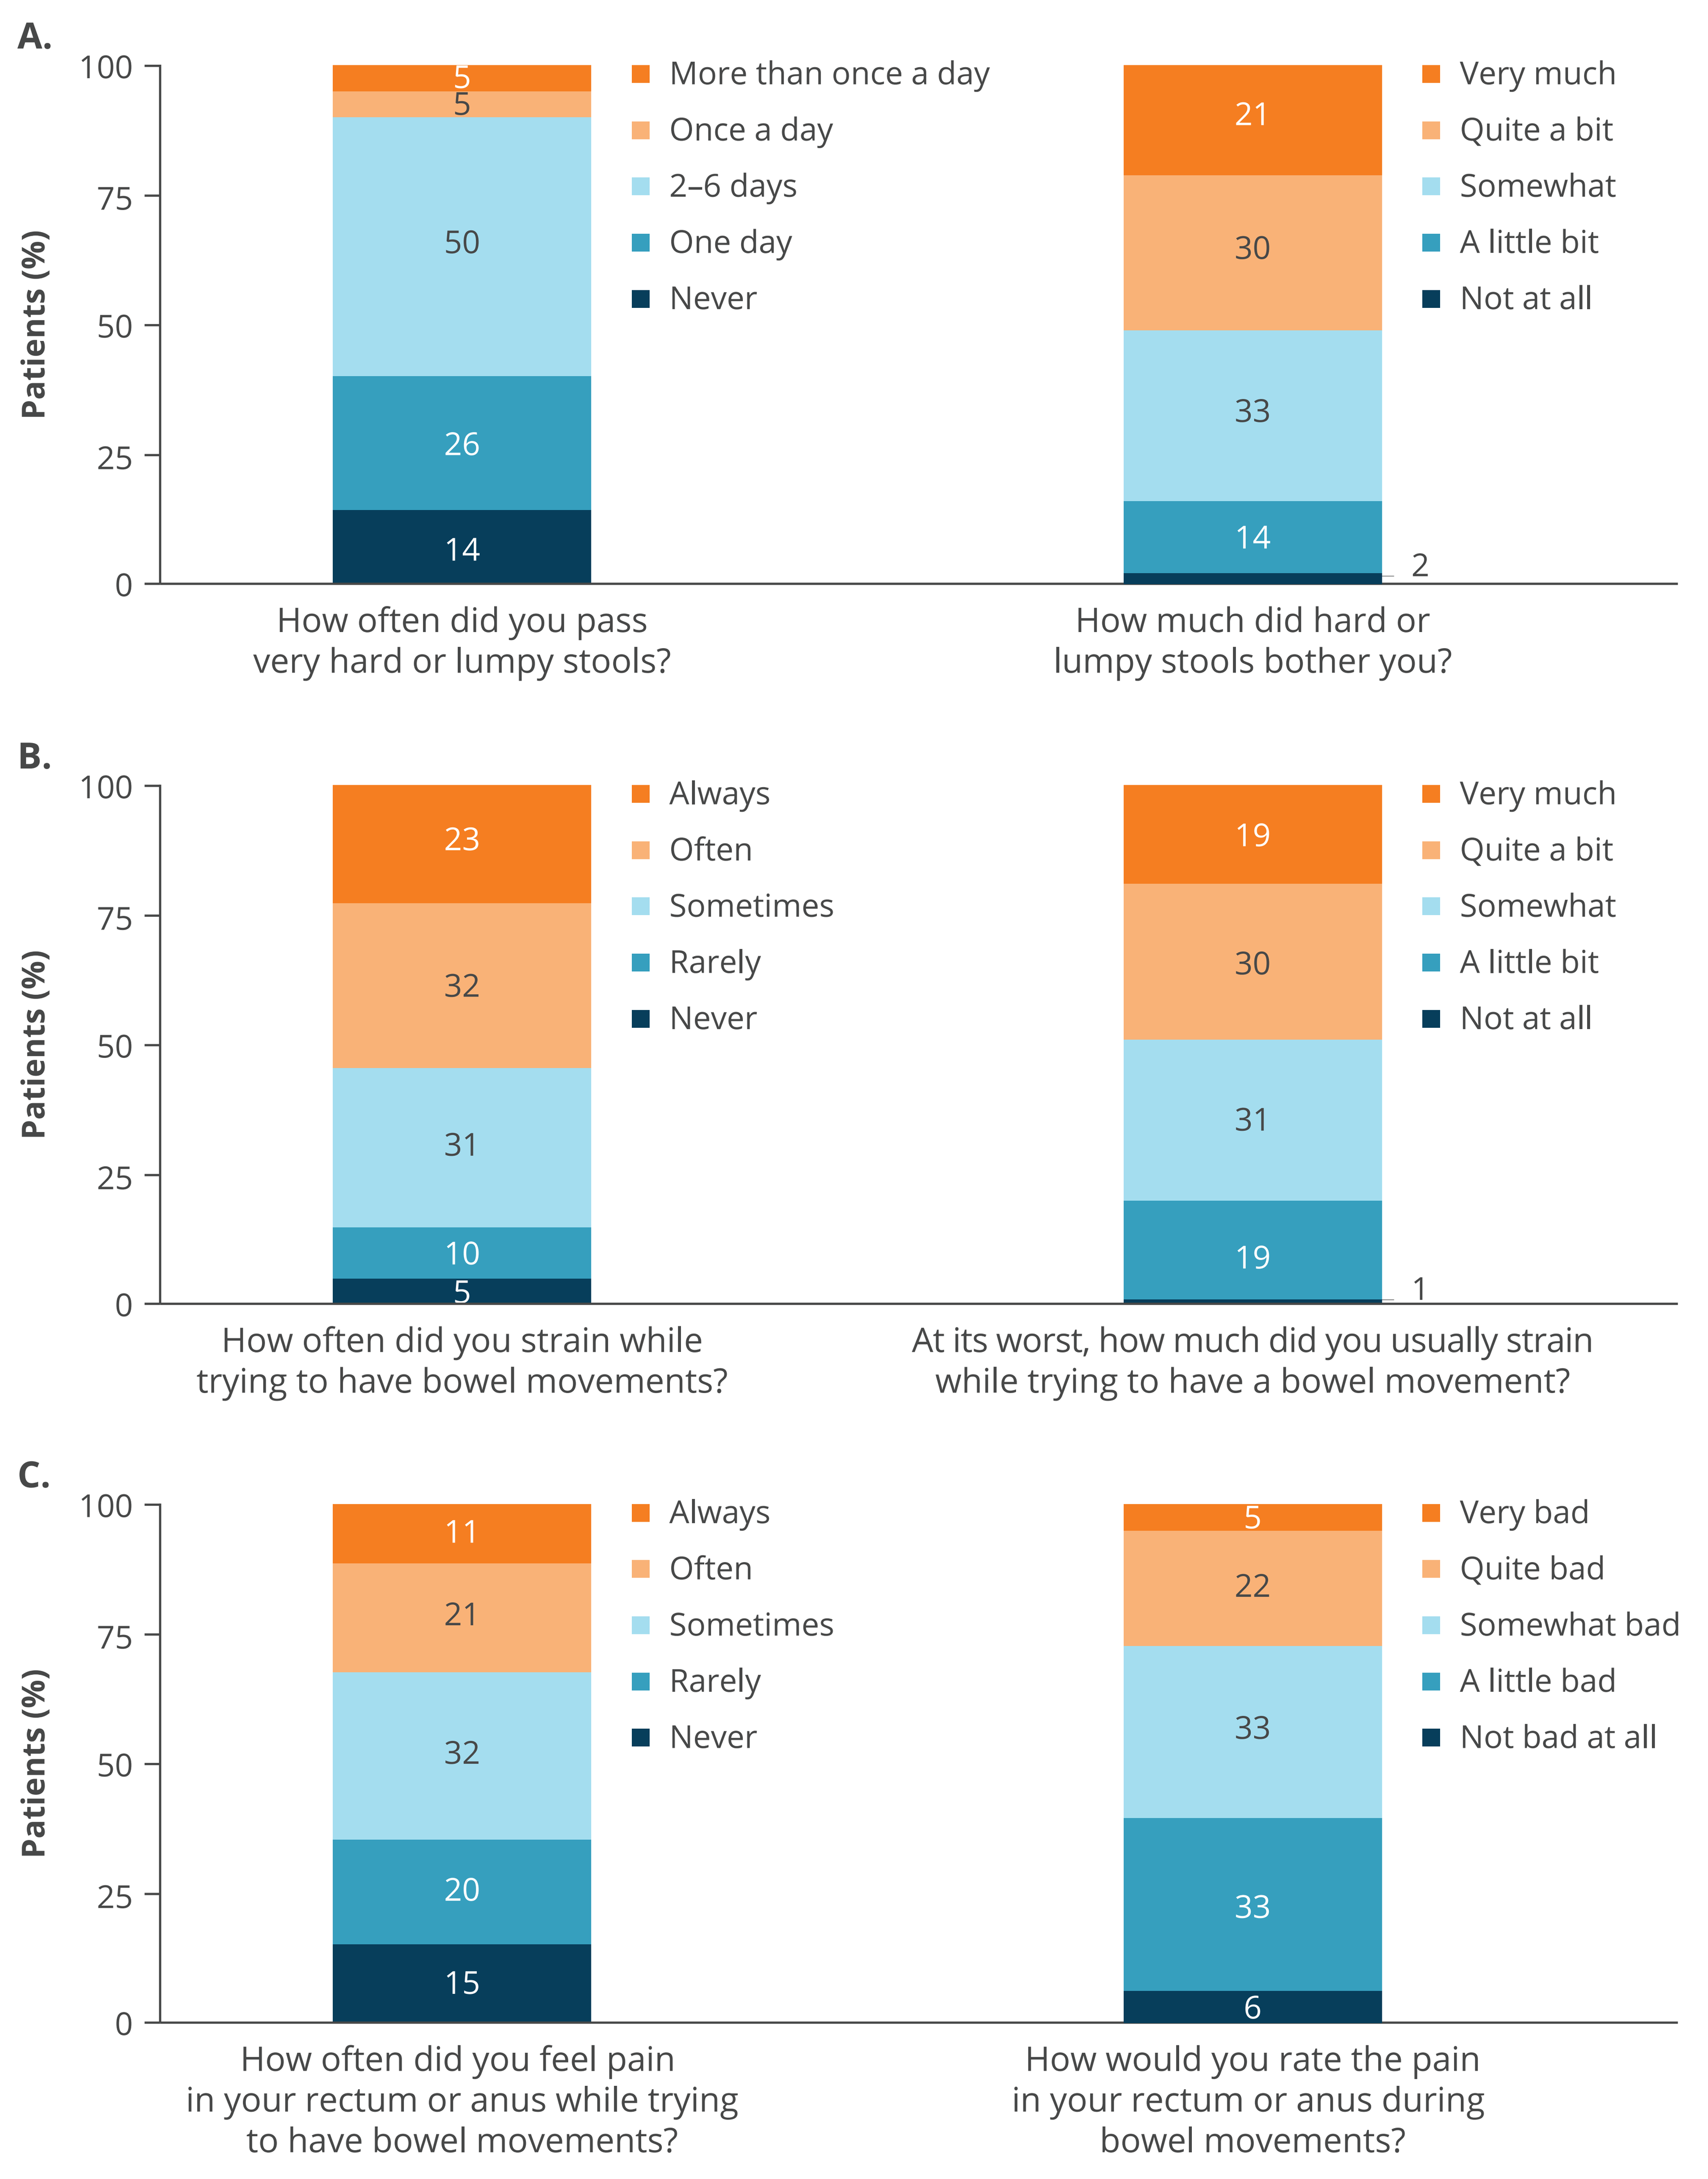

A.
Patients (%)
100
75
50
25
0
5
5
50
26
14
More than once a day
Once a day
2–6 days
One day
Never
How often did you pass very hard or lumpy stools?
21
30
33
14
2
Very much
Quite a bit
Somewhat
A little bit
Not at all
How much did hard or lumpy stools bother you?
B.
23
32
31
10
5
Always
Often
Sometimes
Rarely
Never
How often did you strain while trying to have bowel movements?
19
30
31
19
1
Very much
Quite a bit
Somewhat
A little bit
Not at all
At its worst, how much did you usually strain while trying to have a bowel movement?
C.
11
21
32
20
15
Always
Often
Sometimes
Rarely
Never
How often did you feel pain in your rectum or anus while trying to have bowel movements?
5
22
33
33
6
Very bad
Quite bad
Somewhat bad
A little bad
Not bad at all
How would you rate the pain in your rectum or anus during bowel movements?

IBS-C, irritable bowel syndrome with constipation.

- A feeling of an incomplete BM was reported "often" or "always" by 56% of patients with IBS-C in the previous 7 days, and 47% had to manually extract stool (**Figure 2**).

Figure 2: Proportion of Patients With IBS-C Who Experienced Feelings of an Incomplete BM and/or Had to Manually Extract Stool in the Previous 7 Days

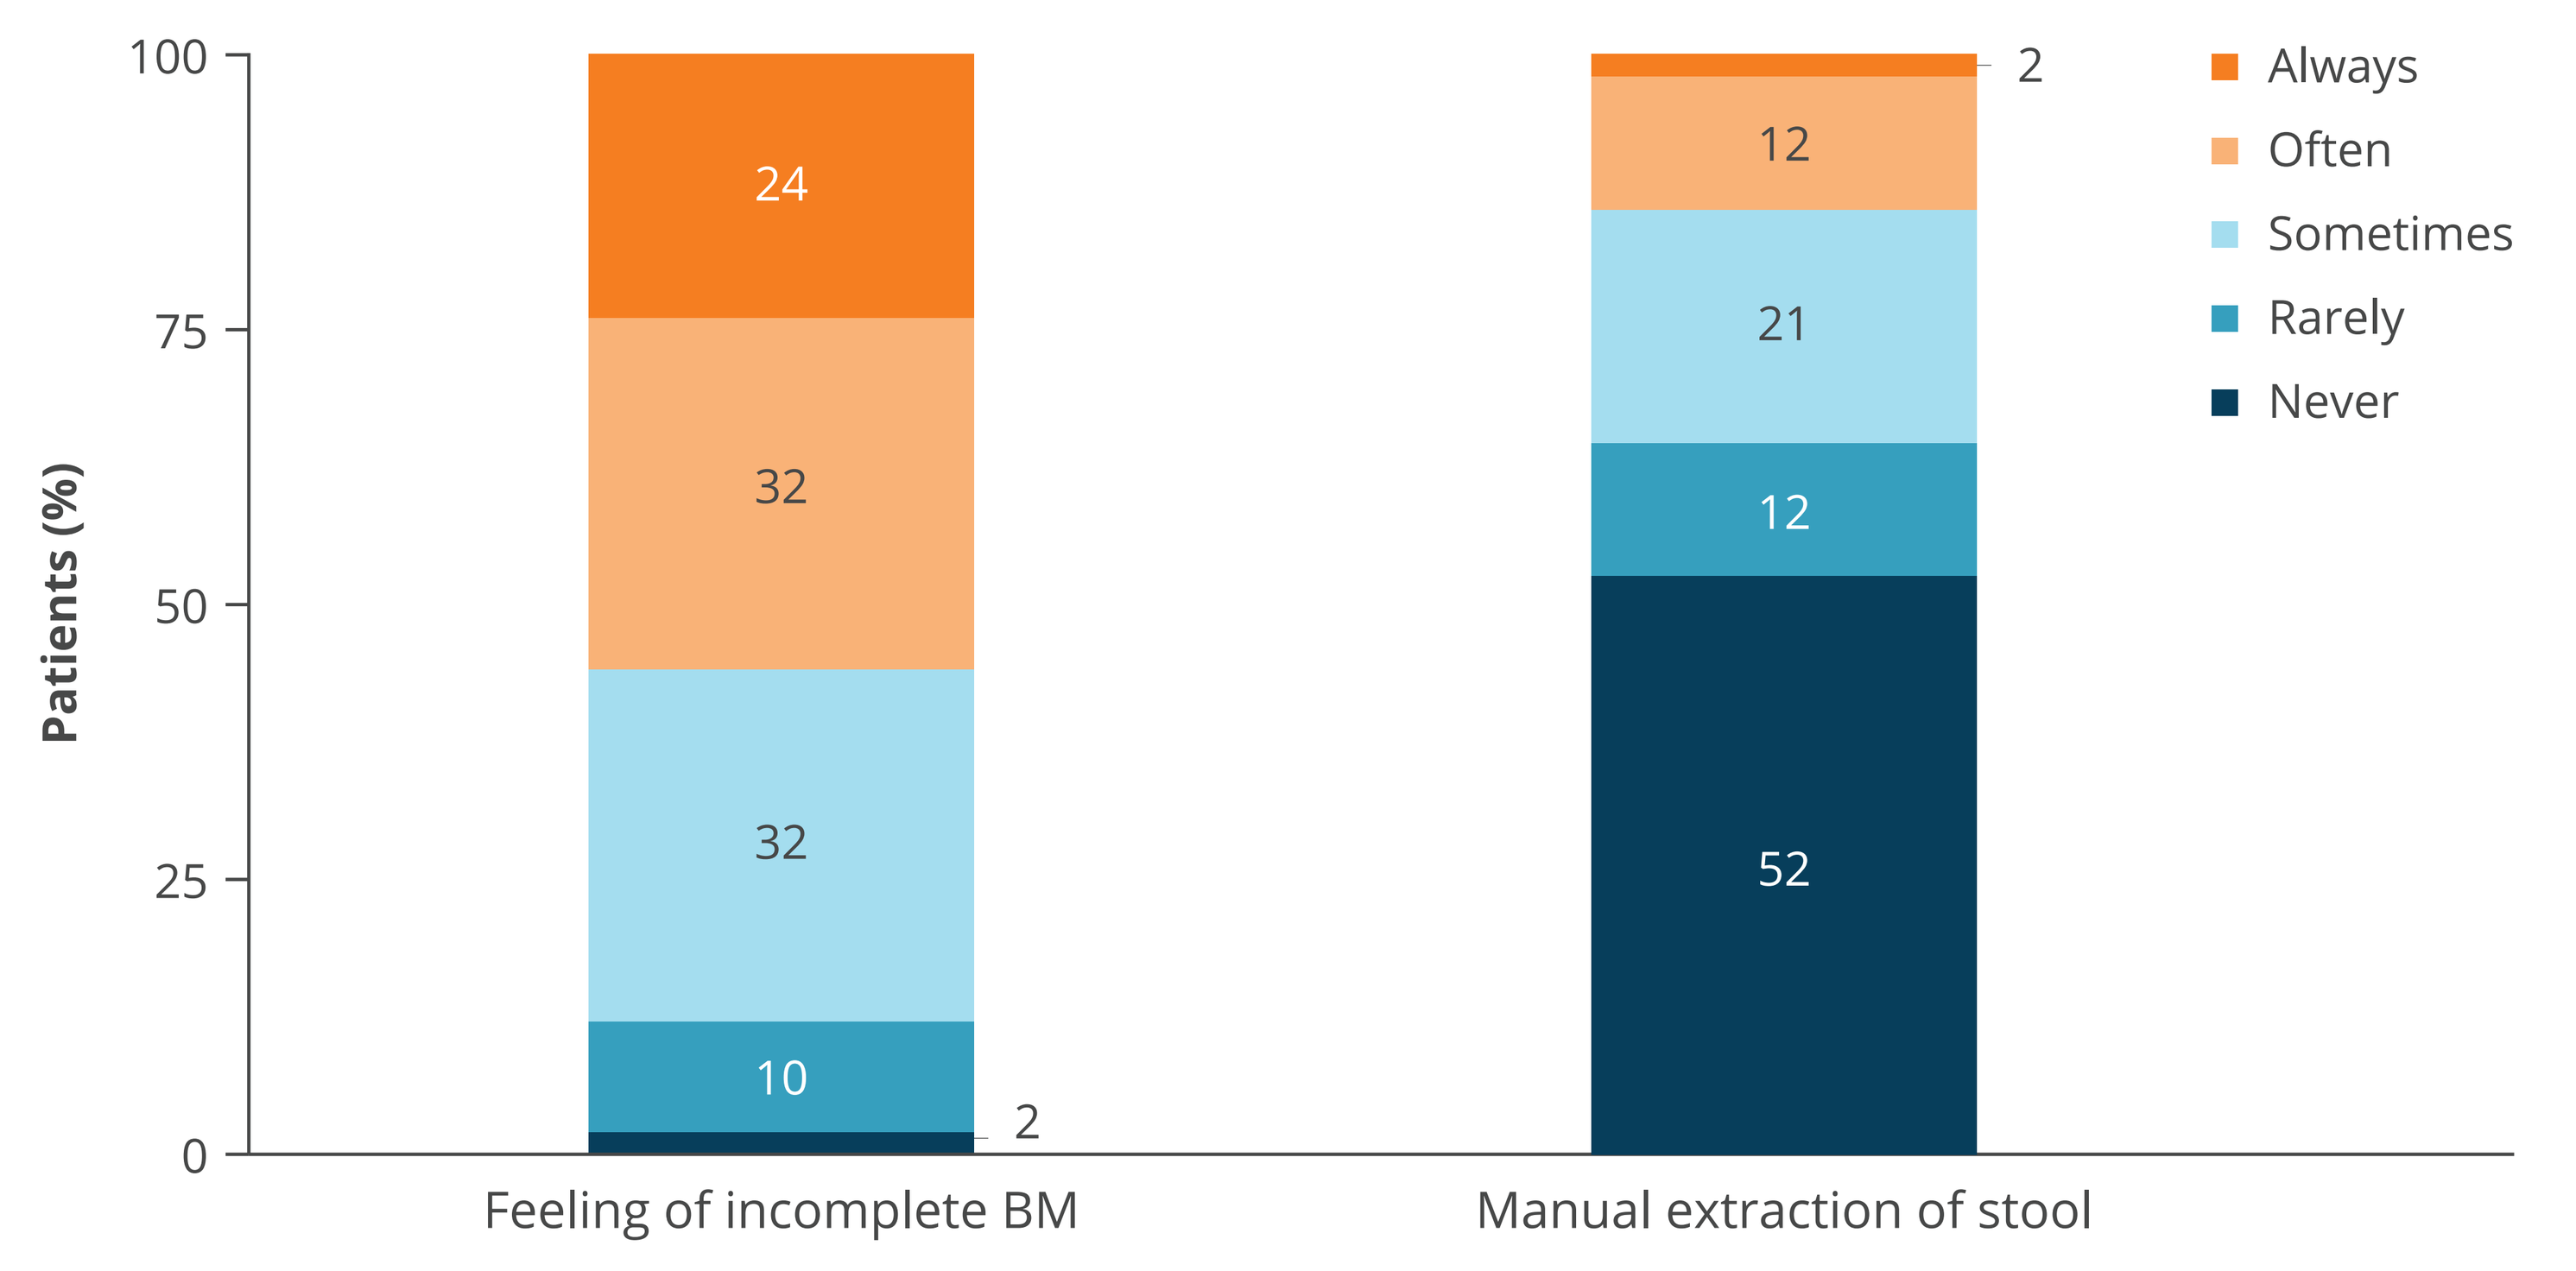

Patients (%)
100
75
50
25
0
24
32
32
10
2
Feeling of incomplete BM
2
12
21
12
52
Manual extraction of stool
Always
Often
Sometimes
Rarely
Never

BM, bowel movement; IBS-C, irritable bowel syndrome with constipation.

- In addition to constipation, ≥60% of patients with IBS-C also experienced bloating, abdominal cramps and pain, fullness in abdomen, excessive gas, and/or fatigue (**Figure 3**).
- Of the 95% of patients that experienced abdominal pain within the past 7 days, 33% described the pain as "quite bad" or "very bad," which interfered with their day-to-day activities "quite a bit" (20%) or "very much" (9%).

Figure 3: Symptoms Typically Experienced by Patients With IBS-C

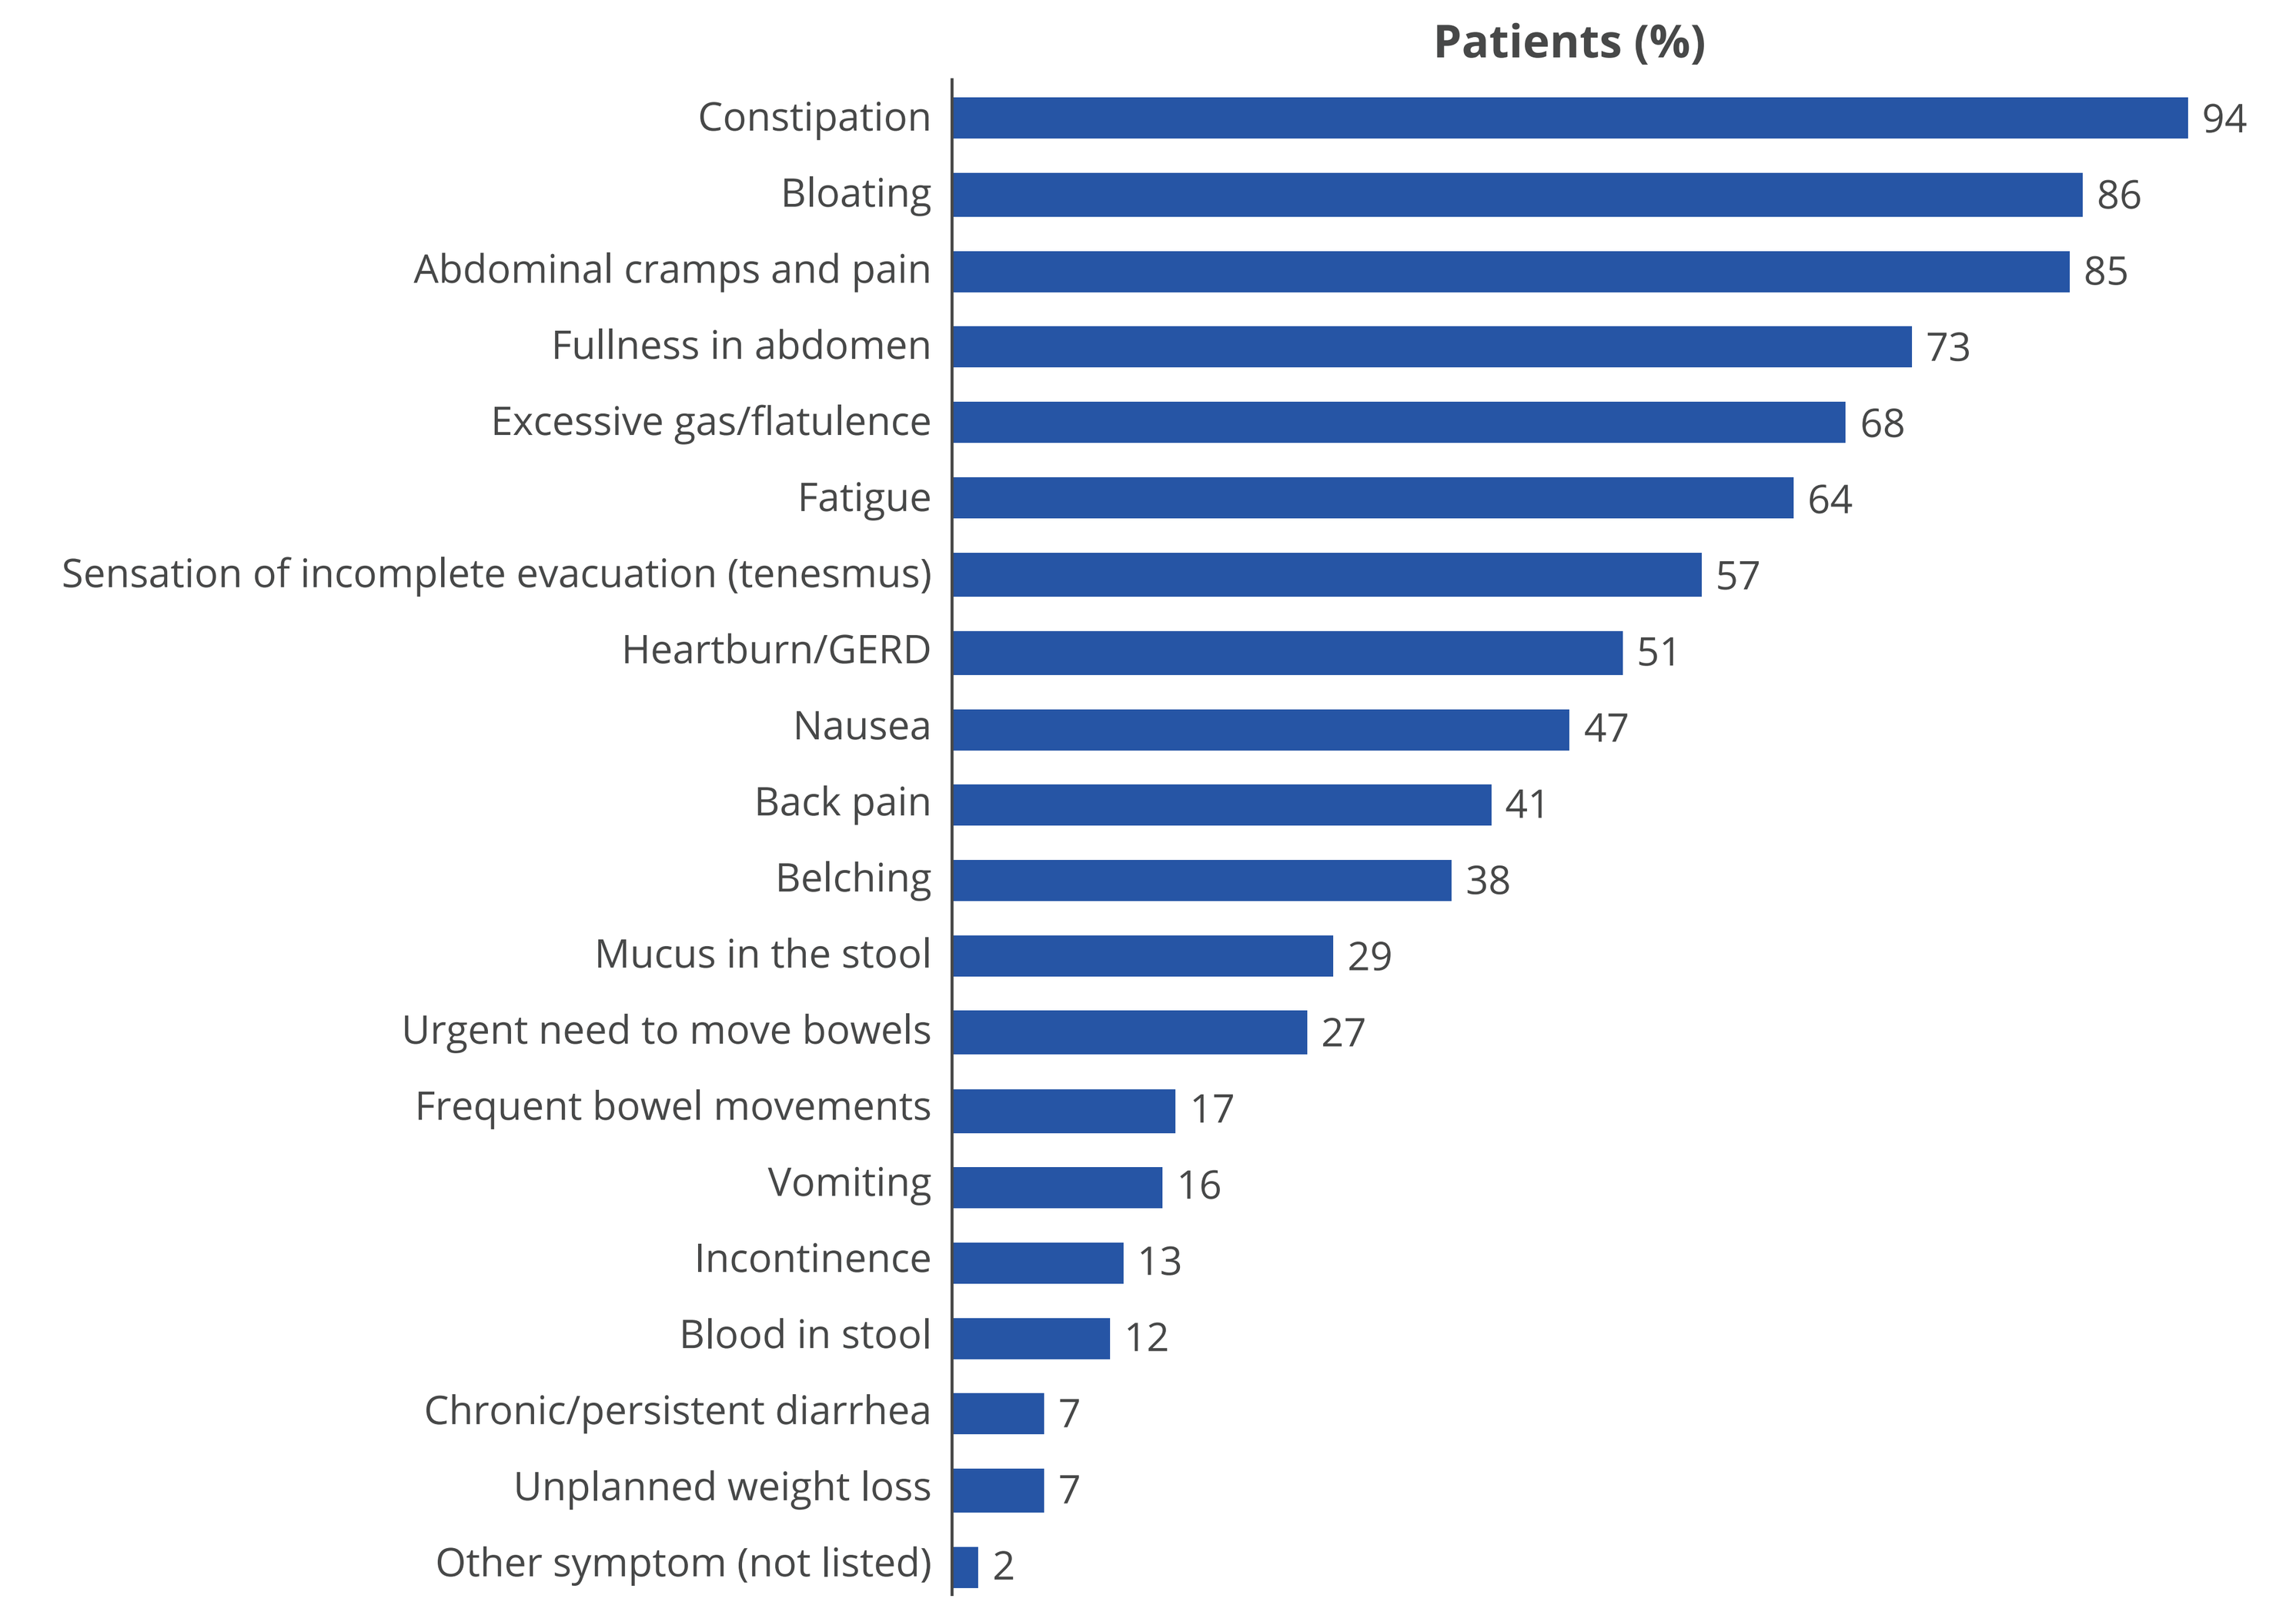

Patients (%)
Constipation 94
Bloating 86
Abdominal cramps and pain 85
Fullness in abdomen 73
Excessive gas/flatulence 68
Fatigue 64
Sensation of incomplete evacuation (tenesmus) 57
Heartburn/GERD 51
Nausea 47
Back pain 41
Belching 38
Mucus in the stool 29
Urgent need to move bowels 27
Frequent bowel movements 17
Vomiting 16
Incontinence 13
Blood in stool 12
Chronic/persistent diarrhea 7
Unplanned weight loss 7
Other symptom (not listed) 2

GERD, gastroesophageal reflux disease; IBS-C, irritable bowel syndrome with constipation.

- Of the 104 respondents who are currently perimenopausal or having menstrual cycles, 48% felt that menstruation made constipation worse and >80% felt it exacerbated abdominal pain and bloating symptoms (**Figure 4**).

Figure 4: Impact of Menstruation on IBS-C Symptoms Among Patients Who Currently Have Menstrual Cycles or Are Perimenopausal

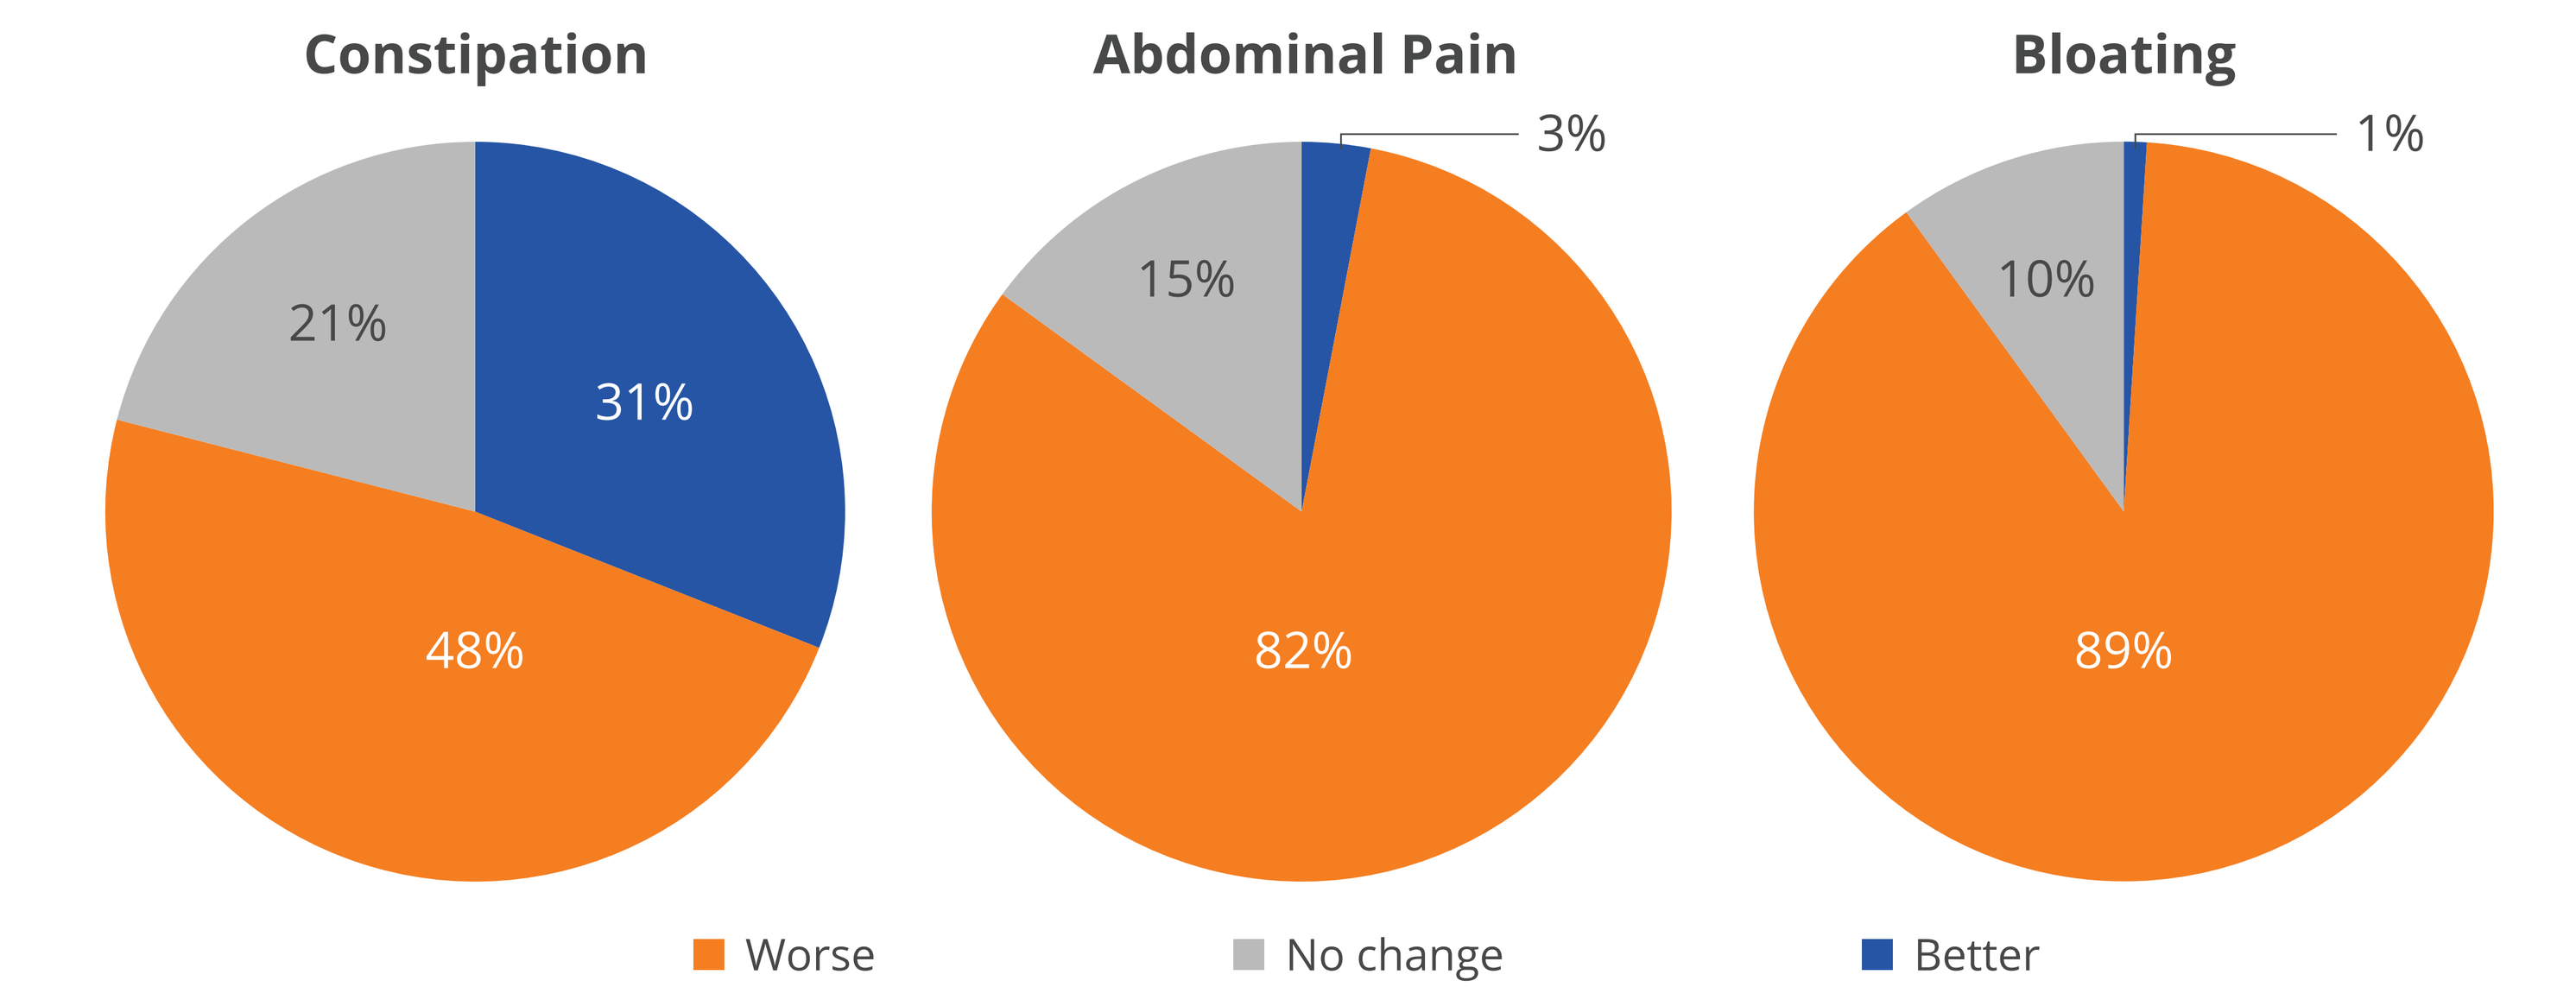

Constipation
21%
31%
48%
Abdominal Pain
3%
15%
82%
Bloating
1%
10%
89%
Worse
No change
Better

IBS-C, irritable bowel syndrome with constipation.

Conclusions

Patients with IBS-C reported many symptoms in addition to constipation, such as bloating, cramps, gas, fatigue, and sensations of incomplete stool evacuation, that negatively affected their lived experience.

The majority of patients also described a worsening of IBS-C symptoms around menstruation, suggesting an additional adverse effect on QoL that warrants further investigation.

Disclosures

Baharak Moshiree reports grant funding from Alfasigma, Atmo Biosciences, CinDOME, CinPhloro, the National Institutes of Health, and Restalsis; has served on advisory boards for Allergan, Alnylam, Ironwood, Salix Pharmaceuticals, and Takeda; has served on speakers' bureaus with Allergan, Bausch Pharmaceuticals, and QOL Medical. Johannah Ruddy, Belinda Gist, Elizabeth Stremke, and Laura Williams are employees of Ardelyx, Inc. Eric Shah has served as a consultant for Salix Pharmaceuticals.

References

1. Lacy BE et al. *Am J Gastroenterol*. 2021;116:17-44.
2. Lacy BE et al. *Gastroenterol*. 2016;150:1393-407.
3. Almario V et al. *Gastroenterol*. 2023;165:1475-87.
4. Heidelbaugh JJ et al. *Am J Gastroenterol*. 2015;110:580-7.
5. Fehnel SE et al. *Value Health*. 2017;20:618-26.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr Moshiree can be contacted for further information on this study at Baha.Moshiree@atriumhealth.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American College of Gastroenterology 2024 Annual Scientific Meeting & Postgraduate Course; October 25–30, 2024; Philadelphia, PA, USA.