Poster P0641

Irritable Bowel Syndrome With Constipation Poses a Substantial Burden to Patient Overall Health Status and Quality of Life: Results From the IBS In America 2024 Real-World Survey

Eric Shah,[1] Johannah Ruddy,[2] Belinda Gist,[2] Elizabeth Stremke,[2] Laura Williams,[2] Baharak Moshiree[3]

[1]University of Michigan, Ann Arbor, MI, USA; [2]Ardelyx, Inc., Waltham, MA, USA; [3]Wake Forest Medical University, Charlotte, NC, USA

Background

- Irritable bowel syndrome (IBS) is a disorder of gut-brain interaction characterized by symptoms of recurring abdominal pain and disordered defecation.[1]
- IBS with constipation (IBS-C) accounts for approximately one-third of IBS cases and is characterized by hard and lumpy stools.[2,3]
- Patients with IBS-C also experience abdominal symptoms, such as bloating, discomfort, cramping, excessive gas, and fullness, that also contribute to the disease burden.[4,5]
- The IBS In America 2024 survey was conducted to gain insight into the experience of patients with IBS.
- Here, we report the overall health status and quality of life (QoL) reported by patients with IBS-C in the IBS In America 2024 survey.

Methods

- IBS In America 2024 was a 15-minute online survey fielded by Health Union from January 15 to April 14, 2024, in United States residents aged ≥18 years, covering demographics, comorbidities, QoL/impact, treatments, and health care provider (HCP) interactions.
- Patients who completed the survey and met the following criteria were invited to participate in an institutional review board–approved extension survey and are included in this analysis:
 - Diagnosed with IBS-C by an HCP.
 - Currently seeing an HCP to treat their IBS-C.
 - Had ever used an over-the-counter or prescription treatment for their IBS-C.

Results

- Respondents with IBS-C (N=284) had a mean age of 51.4 years and were predominantly female (92%) and White (87%; **Table 1**).
- More than one-third of the respondents were employed full-time or self-employed (36%), and most respondents had health insurance (99%; **Table 1**).

Table 1: Demographics and Baseline Characteristics of Patients With IBS-C Who Completed the Survey

	IBS-C respondents (N=284)
Age, mean (range), y	51.4 (18–86)
Female, n (%)	261 (92)
Race,[a] n (%)	
Asian or South Asian	3 (1)
Black or African American	28 (10)
Native Hawaiian or other Pacific Islander	1 (0.4)
White	247 (87)
Other[b]	11 (4)
Ethnicity	
Hispanic or Latino	11 (4)
Non-Hispanic or Latino	273 (96)
Employment status	
Employed full-time	91 (32)
Fully retired	74 (26)
On disability	54 (19)
Employed part-time	26 (9)
Unemployed	14 (5)
Homemaker/SAH parent	11 (4)
Self-employed	11 (4)
Student	6 (2)
Health insurance	
Group coverage	122 (43)
Medicare	97 (34)
Medicaid	28 (10)
Private	14 (5)
Health insurance exchange	11 (4)
Military, VA, Tricare	6 (2)
None	3 (1)

[a]Participants were instructed to select all that apply, permitting them to identify with multiple races/ethnicities. [b]Includes Native American or Alaska Native, "Prefer not to answer," and Other races/ethnicities. IBS-C, irritable bowel syndrome with constipation; SAH, stay-at-home; VA, Veterans Affairs.

Health Status

- More than one-third of patients described their QoL as "poor" or "fair" (39%), which they often attributed to having multiple health conditions (**Figure 1**).

Figure 1: Current Overall QoL

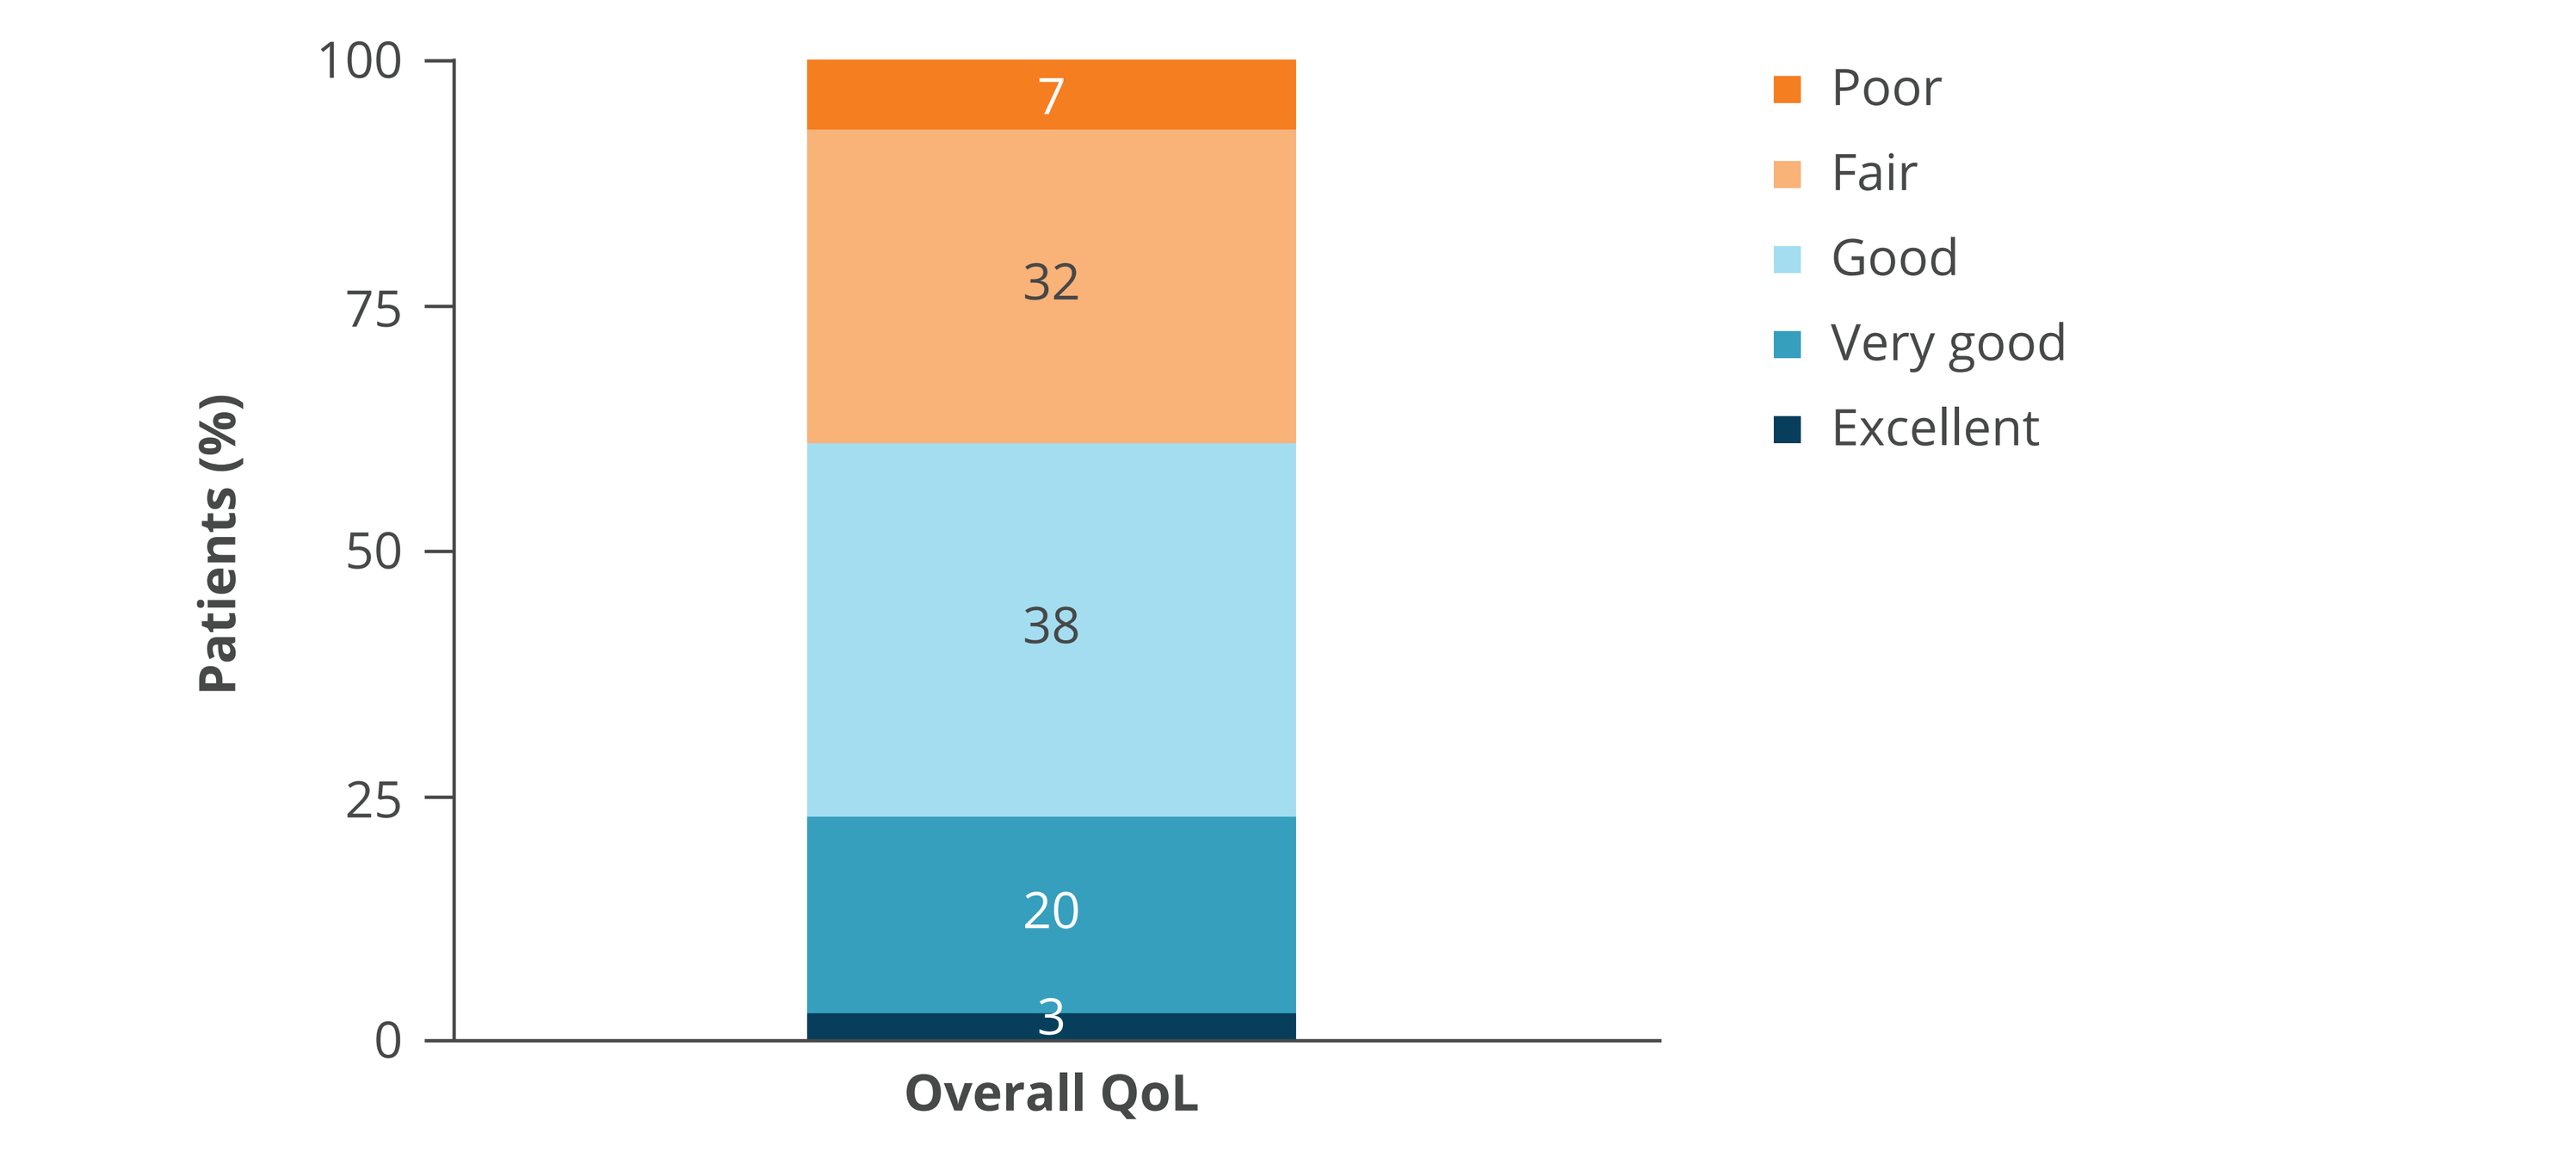

Patients (%)
100
75
50
25
0
7
32
38
20
3
Overall QoL
Poor
Fair
Good
Very good
Excellent

QoL, quality of life.

- Most comorbidities experienced by patients with IBS-C included allergies (58%), mental health conditions (anxiety, 57%; major depressive disorder/depression, 42%), chronic pain (42%), migraine (37%), chronic dry eye (34%), hyperlipidemia (32%), high blood pressure (29%), obesity (26%), and hypothyroidism (26%; **Figure 2**).
 - Patients reported that in the previous 30 days, their physical or mental health had been "not good" for a mean of 15 and 11 days, respectively.

Figure 2: Comorbidities Reported in at Least 10% of Patients With IBS-C

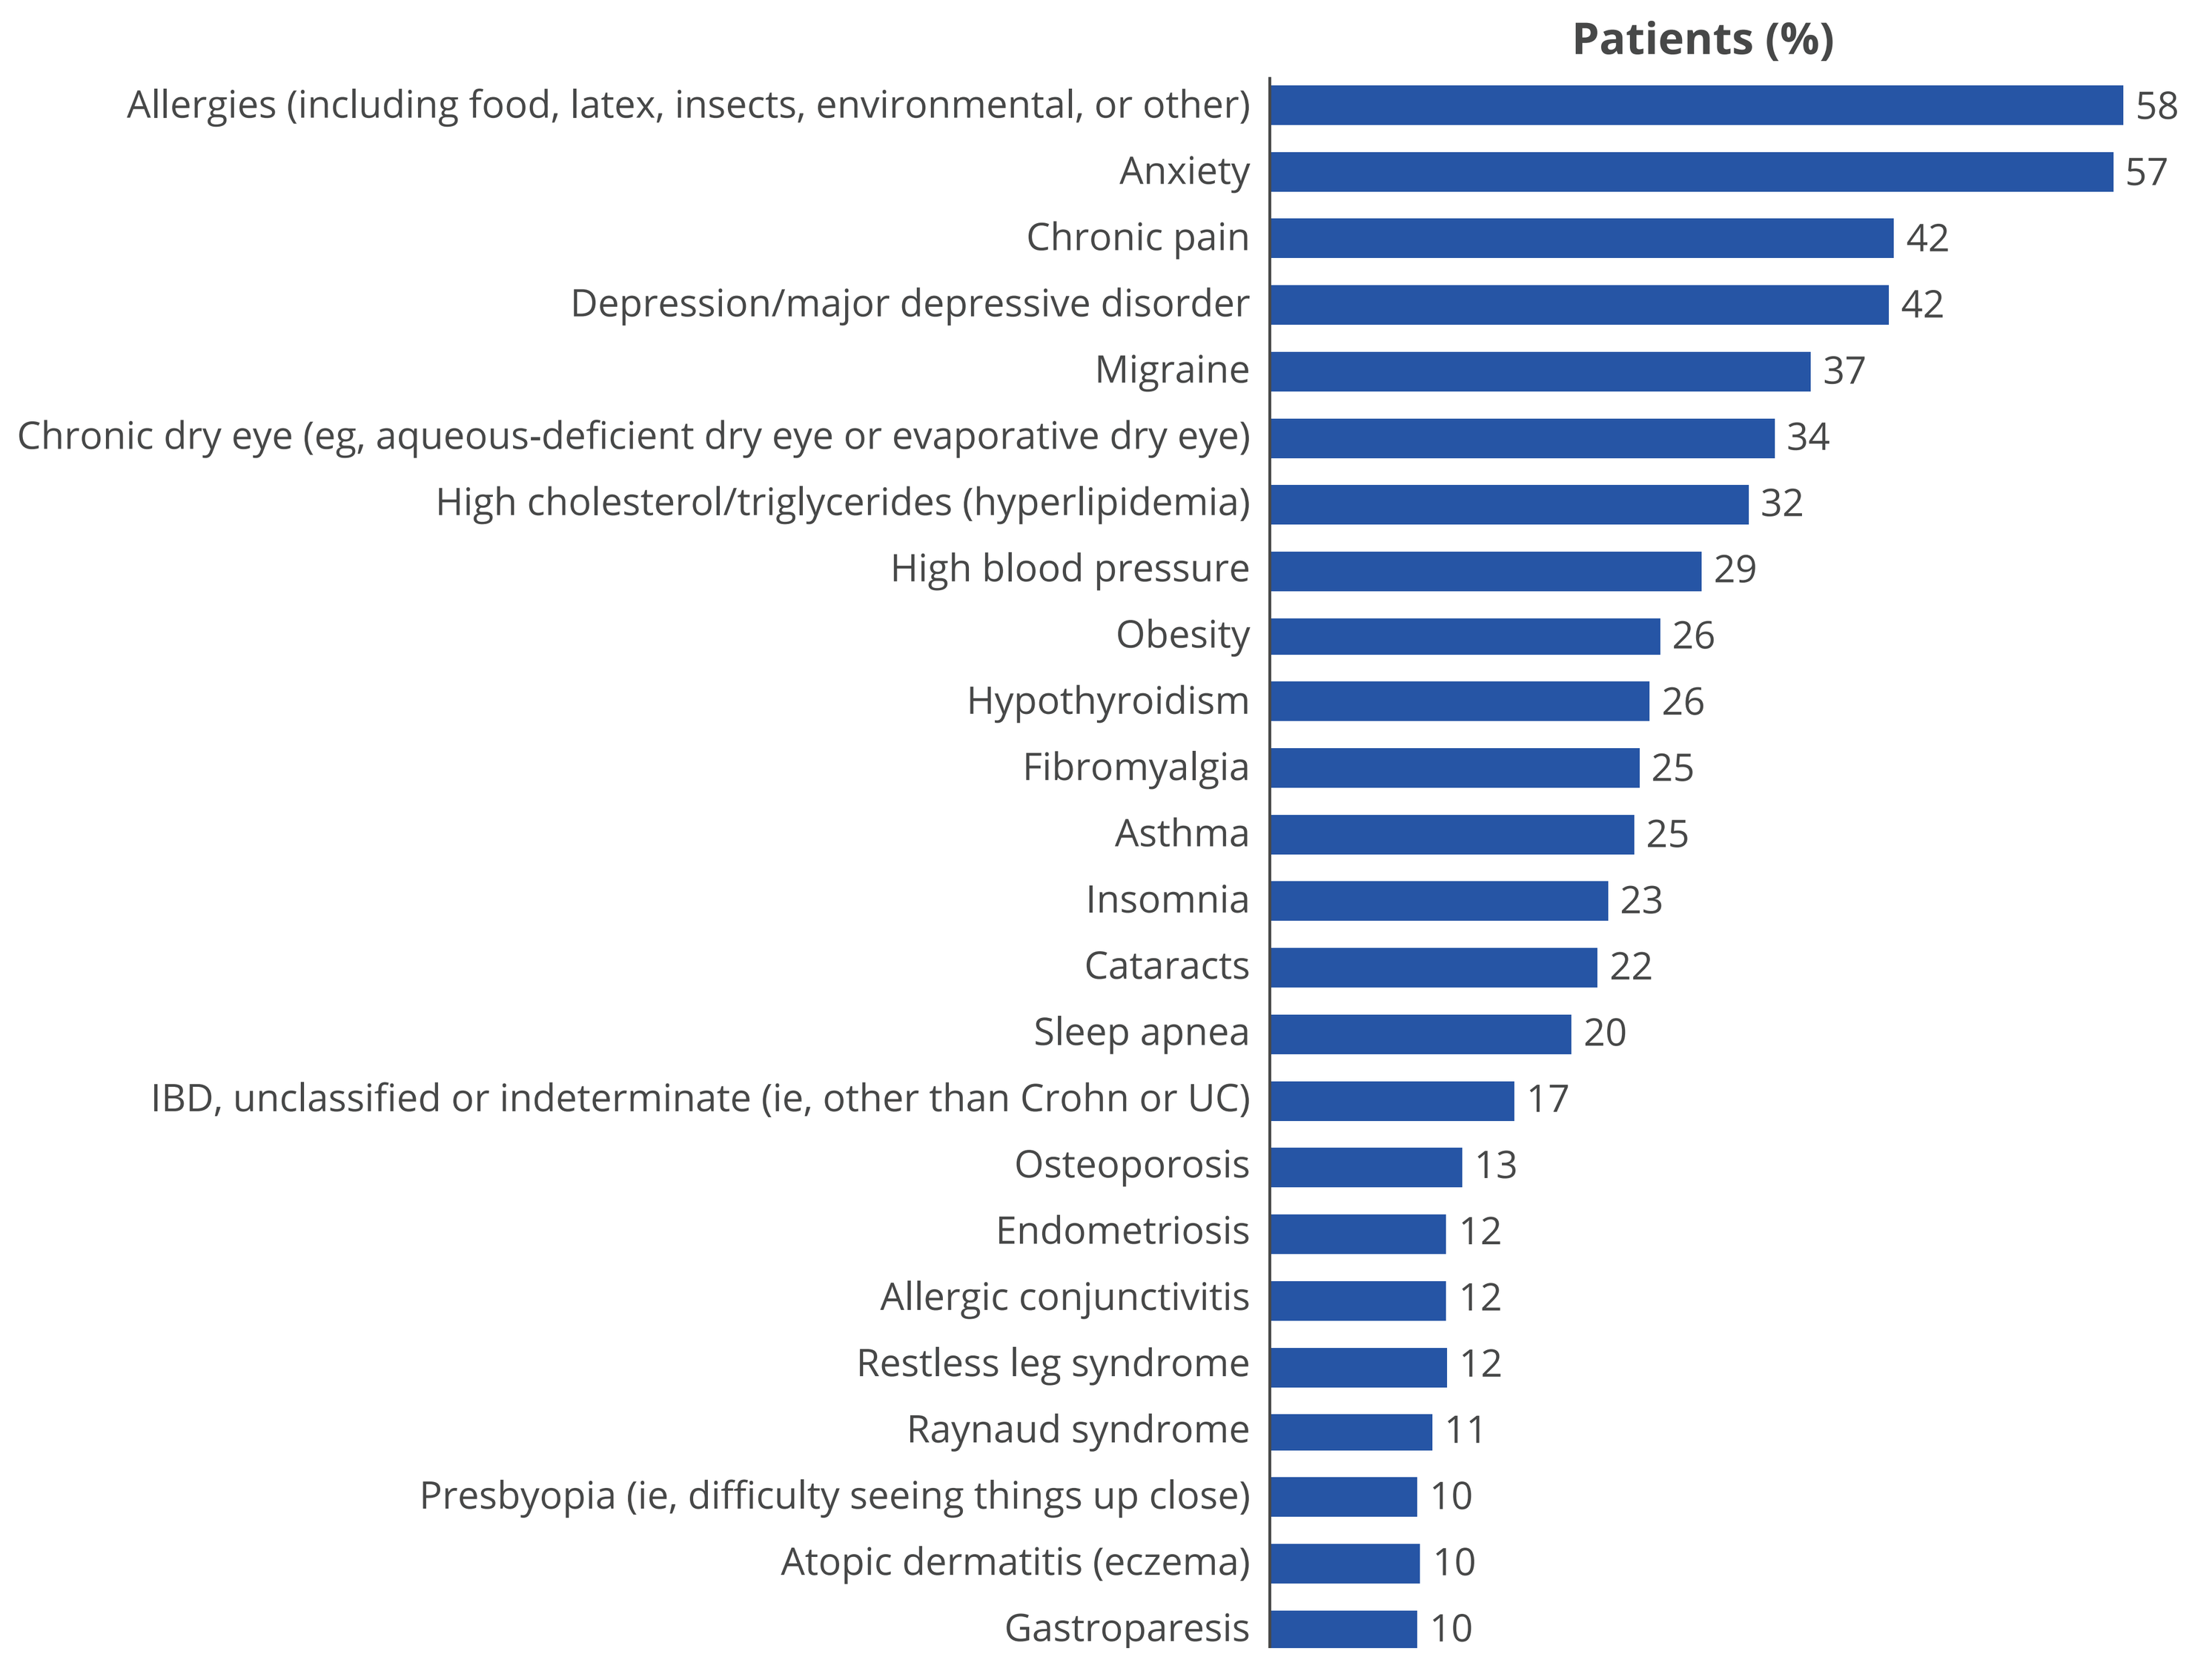

Patients (%)
Allergies (including food, latex, insects, environmental, or other) 58
Anxiety 57
Chronic pain 42
Depression/major depressive disorder 42
Migraine 37
Chronic dry eye (eg, aqueous-deficient dry eye or evaporative dry eye) 34
High cholesterol/triglycerides (hyperlipidemia) 32
High blood pressure 29
Obesity 26
Hypothyroidism 26
Fibromyalgia 25
Asthma 25
Insomnia 23
Cataracts 22
Sleep apnea 20
IBD, unclassified or indeterminate (ie, other than Crohn or UC) 17
Osteoporosis 13
Endometriosis 12
Allergic conjunctivitis 12
Restless leg syndrome 12
Raynaud syndrome 11
Presbyopia (ie, difficulty seeing things up close) 10
Atopic dermatitis (eczema) 10
Gastroparesis 10

IBD, inflammatory bowel disease; UC, ulcerative colitis.

Quality of Life

- Most patients (90%) had "some" or "significant" negative impact of IBS-C on their overall QoL, with 1 in 4 patients reporting a negative impact on their mental/emotional health (**Figure 3**).
- Most patients with IBS-C also reported "some" or "significant" negative impact on sexual health/intimacy ("some" 40%; "significant" 24%), a sense of independence (43%; 16%), and relationships with friends/family (42%; 14%; **Figure 3**).
- Almost half of patients (46%) reported satisfaction with social activities and relationships as "fair" or "poor," and 44% described their fatigue as "severe" or "very severe."

Figure 3: Impact of IBS-C on the QoL of Patients

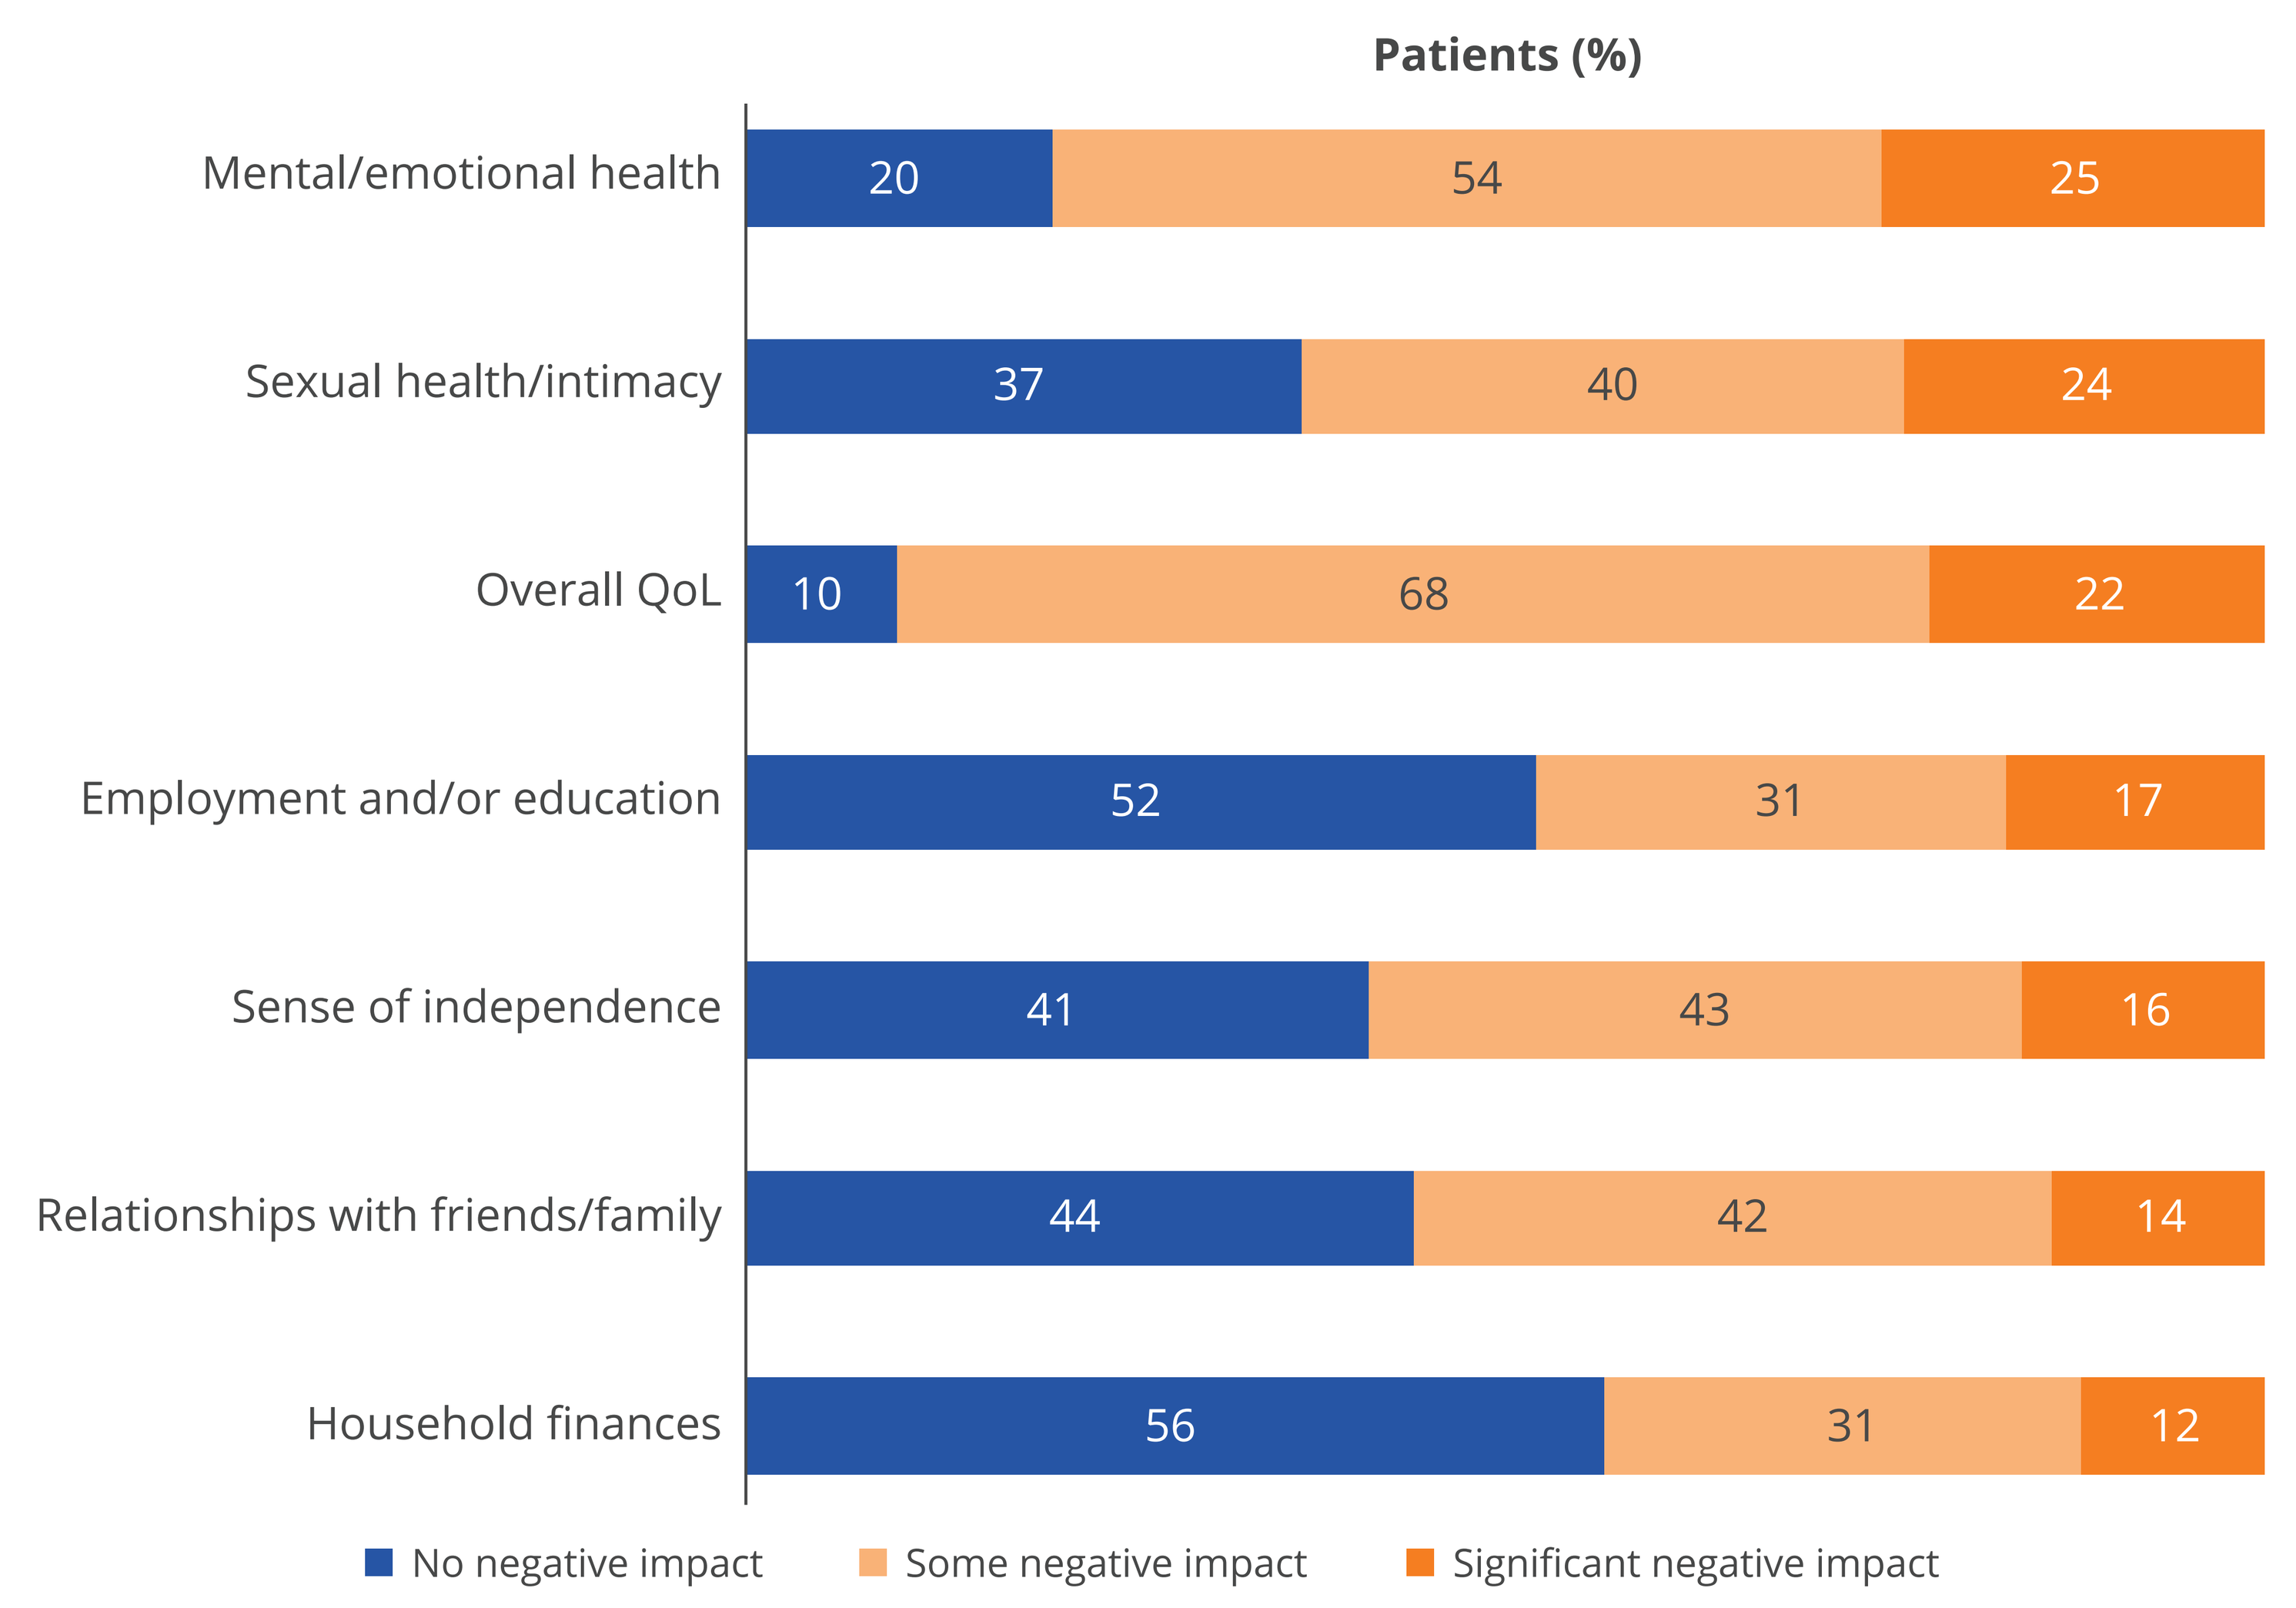

Patients (%)
Mental/emotional health 20 54 25
Sexual health/intimacy 37 40 24
Overall QoL 10 68 22
Employment and/or education 52 31 17
Sense of independence 41 43 16
Relationships with friends/family 44 42 14
Household finances 56 31 12
No negative impact
Some negative impact
Significant negative impact

QoL, quality of life.

Support and Understanding

- Twenty-six percent of patients strongly disagreed with the statement "IBS does not stop me from doing the things I enjoy," whereas only 23% strongly agreed, and most felt neutral (**Figure 4**).
- Less than one-third of patients (31%) reported that they had the support needed to manage their IBS-C, and even fewer patients (26%) reported that their loved ones understood what it was like to live with IBS-C (**Figure 4**).

Figure 4: Attitudes, Support, and Understanding Experienced by Patients With IBS-C

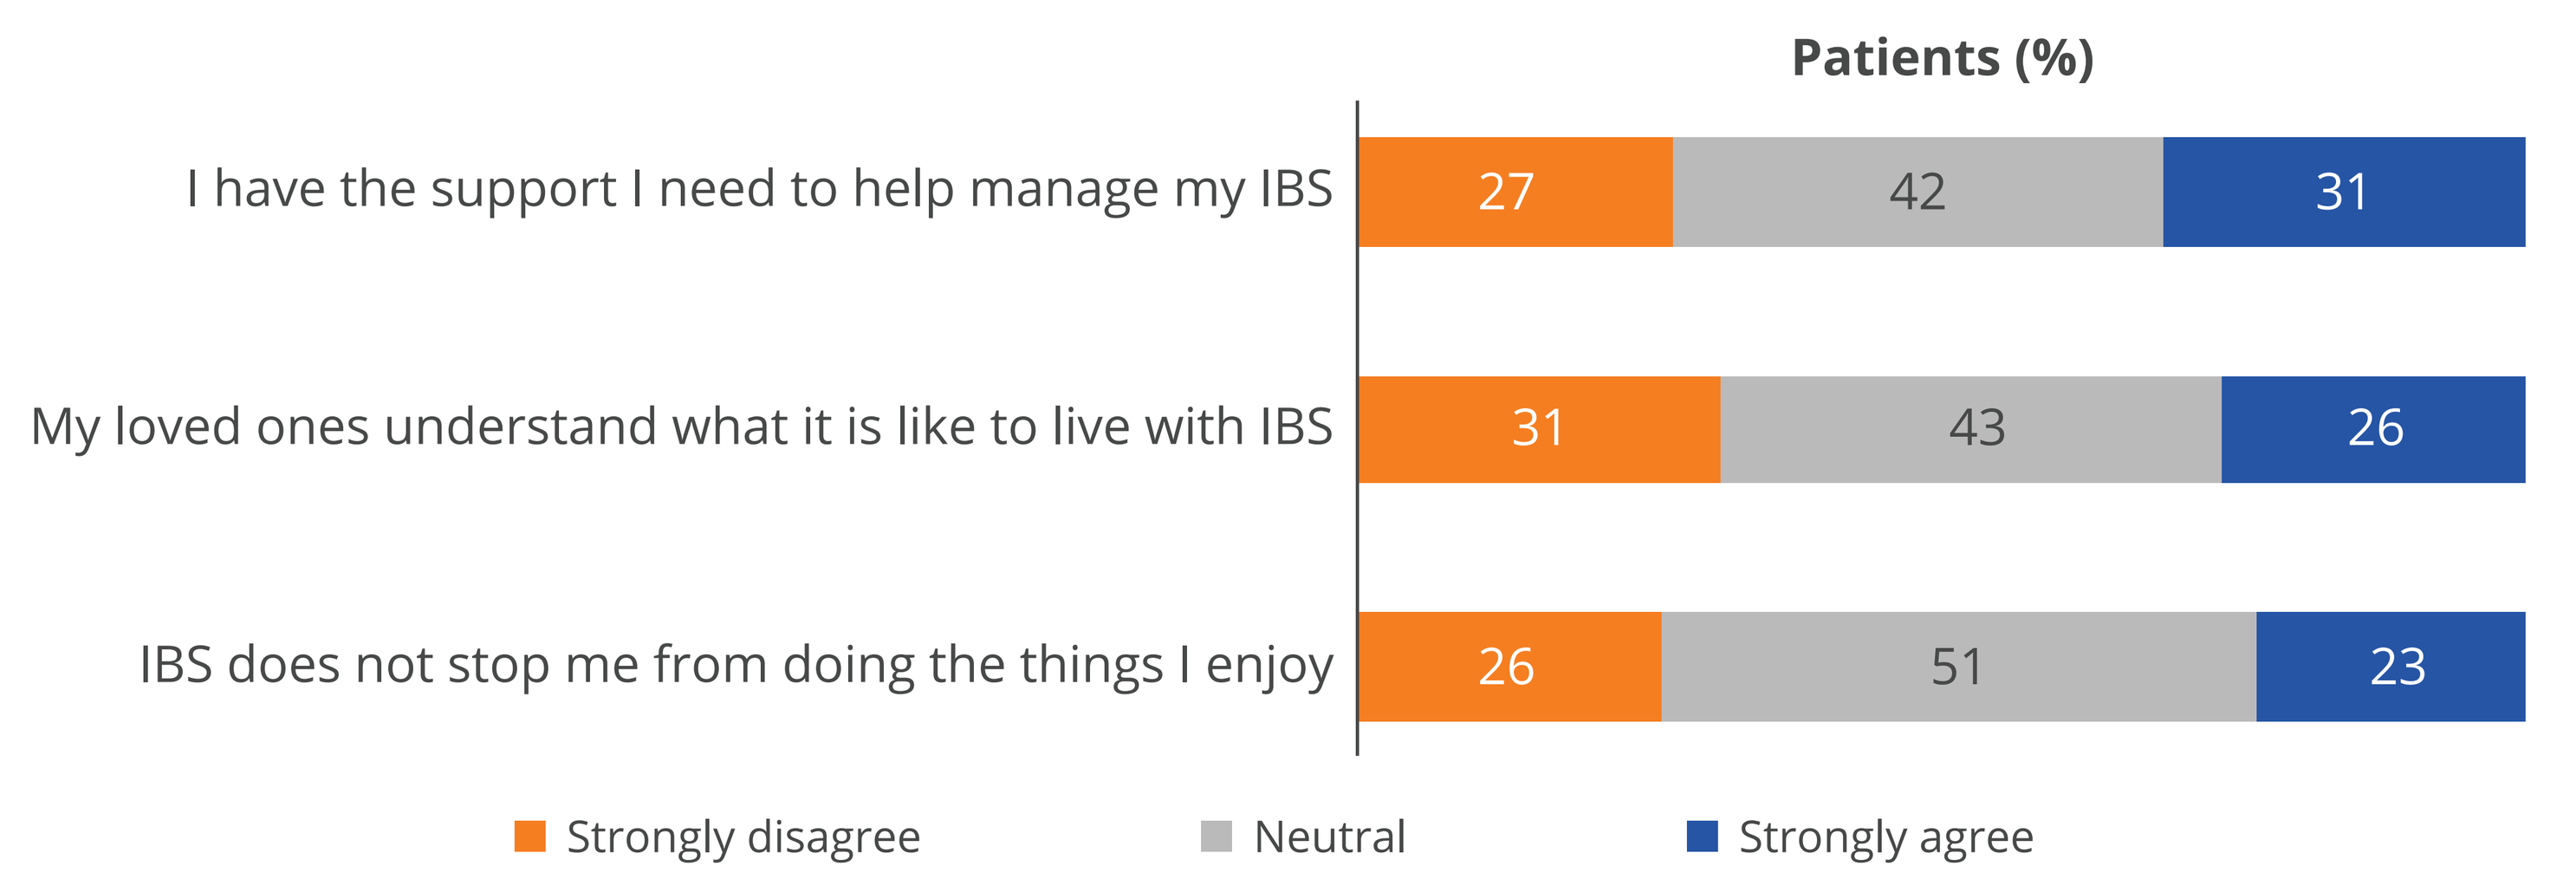

Patients (%)
I have the support I need to help manage my IBS 27 42 31
My loved ones understand what it is like to live with IBS 31 43 26
IBS does not stop me from doing the things I enjoy 26 51 23
Strongly disagree
Neutral
Strongly agree

IBS, irritable bowel syndrome; IBS-C, irritable bowel syndrome with constipation.

Financial Experiences

- Twenty-nine percent of patients with IBS-C reported that they are very frustrated that they cannot work or contribute as much as usual (**Figure 5**).
- IBS-C was a financial hardship to 47% of patients and their families (**Figure 5**).
- In total, 48% of patients with IBS-C reported that their out-of-pocket medical expenses were somewhat to very much more than anticipated (**Figure 5**).
- Forty-five percent of patients with IBS-C felt they were not in control of their financial situation.

Figure 5: Attitudes and Experiences of Patients With IBS-C Related to Finances

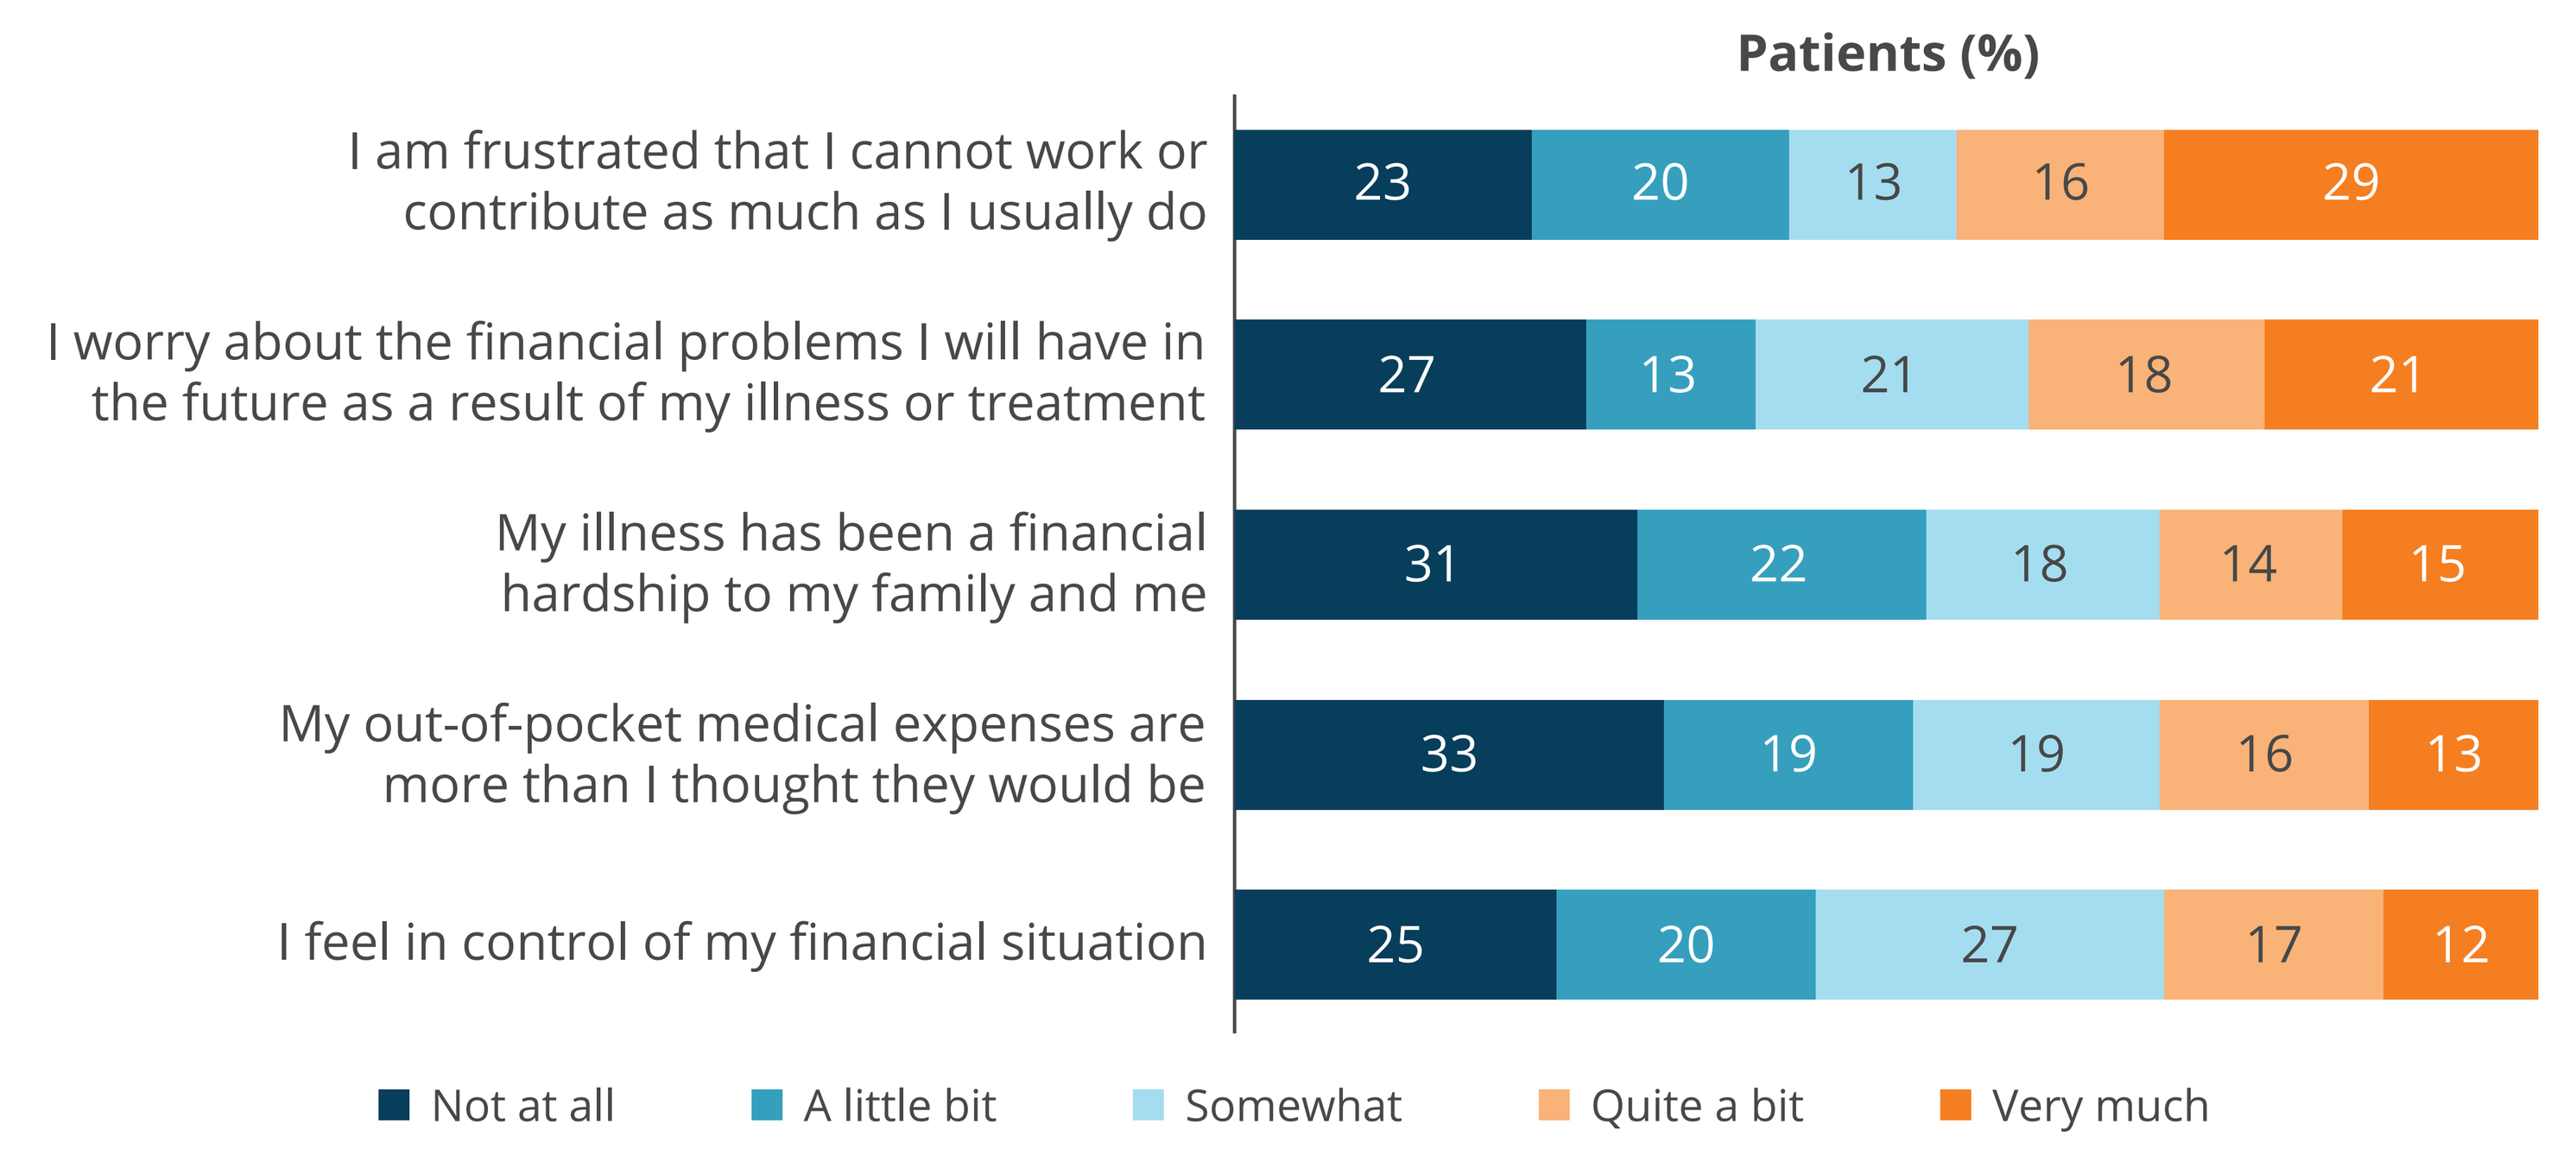

Patients (%)
I am frustrated that I cannot work or contribute as much as I usually do 23 20 13 16 29
I worry about the financial problems I will have in the future as a result of my illness or treatment 27 13 21 18 21
My illness has been a financial hardship to my family and me 31 22 18 14 15
My out-of-pocket medical expenses are more than I thought they would be 33 19 19 16 13
I feel in control of my financial situation 25 20 27 17 12
Not at all
A little bit
Somewhat
Quite a bit
Very much

IBS-C, irritable bowel syndrome with constipation.

Conclusions

IBS-C adversely affects many facets of patients' lives, imparting a substantial disease burden and decreased QoL in all areas, including sexual health; physical, mental, and emotional well-being; and overall relationships with others.

IBS-C also imposed a financial hardship on patients and their families due to reduced ability to work and higher-than-anticipated medical expenses.

Disclosures

Eric Shah has served as a consultant for Salix Pharmaceuticals. Johannah Ruddy, Belinda Gist, Elizabeth Stremke, and Laura Williams are employees of Ardelyx, Inc. Baharak Moshiree reports grant funding from Alfasigma, Atmo Biosciences, CinDOME, CinPhloro, the National Institutes of Health, and Restalsis; has served on advisory boards for Allergan, Alnylam, Ironwood Pharmaceuticals, Salix Pharmaceuticals, and Takeda; and has served on speakers' bureaus with Allergan, Bausch Pharmaceuticals, and QOL Medical.

References

1. Lacy BE et al. *Am J Gastroenterol*. 2021;116:17-44.
2. Lacy BE et al. *Gastroenterology*. 2016;150:1393-407.
3. Almario CV et al. *Gastroenterology*. 2023;165:1475-87.
4. Heidelbaugh JJ et al. *Am J Gastroenterol*. 2015;110:580-7.
5. Fehnel SE et al. *Value Health*. 2017;20:618-26.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

Dr Shah can be contacted for further information on this study at ERShah@umich.edu.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

American College of Gastroenterology 2024 Annual Scientific Meeting & Postgraduate Course; October 25–30, 2024; Philadelphia, PA, USA.