Poster: 392

Preliminary Blinded Safety Data From R-ALLY, a Phase 3, Randomized, Double-Blind, Placebo-Controlled Study of Tenapanor in Pediatric Patients With IBS-C

Jonathan E. Markowitz,[1] Mihaela Ringheanu,[2] Karishma Raju,[3] Yang Yang,[3] and Susan Edelstein[3]

[1]Prisma Health Pediatric Gastroenterology, Greenville, SC, USA; [2]Texas Digestive Specialists, Harlingen, TX, USA; [3]Ardelyx, Inc., Waltham, MA, USA

Background

- Irritable bowel syndrome (IBS) is a common disorder of gut-brain interaction[1] that can affect children and adolescents.[2]
- Symptoms of IBS include abdominal pain associated with bowel changes like diarrhea and/or constipation.[3]
- IBS can negatively impact the quality of life of pediatric patients and their families, decrease school attendance, and contribute to significant healthcare costs.[2]
- The estimated prevalence of IBS ranges from 4% in children (aged 4-10 years) to 5.8% in adolescents (aged 11-18 years),[4,5] with IBS with constipation (IBS-C) as the predominant subtype.[6]
- Tenapanor is a novel, minimally absorbed, small-molecule inhibitor of intestinal sodium/hydrogen exchanger 3 (NHE3)[7,8] expressed on the luminal surface of the gastrointestinal tract, approved in the US and Canada for the treatment of IBS-C in adults.[9,10]
- Tenapanor improves symptoms of IBS-C by increasing sodium and water content in the intestinal lumen, which facilitates softer stool consistency and accelerates intestinal transit.[8,11]
- In nonclinical studies, tenapanor also modulated intestinal tight junctions, decreasing intestinal permeability to macromolecules[12] and reducing visceral hypersensitivity.[13]
- Tenapanor (50 mg twice a day [bid]) significantly improved complete spontaneous bowel movements (CSBMs) and abdominal pain in adults with IBS-C in two pivotal, phase 3, randomized, placebo-controlled studies (NCT02621892, NCT02686138).[14,15]
- Tenapanor (50 mg bid) demonstrated acceptable overall safety and tolerability for up to 55 consecutive weeks in adult patients in a phase 3, open-label extension study (NCT02727751), with diarrhea being the most common adverse effect.[16]
- A phase 3, randomized, double-blind, placebo-controlled study (R-ALLY; NCT05643534) is underway to assess the efficacy, safety, and tolerability of tenapanor (25 mg and 50 mg bid) in pediatric patients (aged ≥12 and <18 years) with IBS-C when administered for 12 consecutive weeks.
- We present preliminary blinded safety data from the R-ALLY study.

Study Design and Methods

- Enrollment is anticipated to be approximately 180 pediatric patients who meet all inclusion criteria, none of the exclusion criteria (**Table 1**), and demonstrate active IBS-C during a 2-week screening period at up to 60 US sites.

Table 1: Inclusion and Exclusion Criteria for the R-ALLY Study

Inclusion criteria	Exclusion criteria
Aged ≥12 and <18 years	Functional diarrhea as defined by Rome IV child/adolescent criteria
Patient weighs ≥18 kg at time of written consent	IBS with diarrhea, mixed IBS, or unsubtyped IBS, as defined by Rome IV child/adolescent criteria
Patient meets the Rome IV criteria for child/adolescent diagnosis of IBS-C	History of nonretentive fecal incontinence
Patient is willing to discontinue laxatives for the duration of the study (rescue medication is permitted after 72 hours without a bowel movement)	Patient has manual disimpaction any time prior to randomization (after consent)
Ability of both patient and parent/guardian/legally authorized representative to communicate with the Investigator and to comply with the requirements of the entire study, including an understanding of the assessments in the eDiary and how to use the eDiary app	Patient currently has both unexplained and clinically significant alarm symptoms (lower gastrointestinal bleeding, anemia, or weight loss) and systemic signs of infection or colitis, or any neoplastic process

IBS, irritable bowel syndrome; IBS-C, irritable bowel syndrome with constipation.

Primary Endpoint:

- 6 of 12-week abdominal pain and SBM (APS) +2 response, defined as achieving the weekly APS +2 response criteria (ie, achieving both weekly SBM +2 response and weekly abdominal pain response during the same week) for ≥6 out of the 12 weeks of the randomized treatment period.
 - The weekly SBM +2 response is defined as having an increase of ≥2 from baseline in the average weekly SBM frequency for a given week.
 - The weekly abdominal pain response is defined as having ≥30% reduction from baseline in the average weekly abdominal pain score for a given week.

Secondary Endpoints:

- 6 of 12-week SBM +2 response, defined as achieving the weekly SBM +2 response for ≥6 out of the 12 weeks of the randomized treatment period.
- 6 of 12-week abdominal pain response, defined as achieving the weekly abdominal pain response for ≥6 out of the 12 weeks of the randomized treatment period.
- Change from baseline in average weekly SBM frequency, stool consistency score, and abdominal pain score.
- Overall use of rescue medication.

- Eligible pediatric patients are randomized (1:1:1) to receive tenapanor 25 mg bid, 50 mg bid, or placebo bid (~60 patients/group) during the 12-week treatment period.
- No more than 2 uses of rescue medication (bisacodyl suppository, enema, or tablet) are allowed during the study, and none can be used during the 2 days prior to randomization.
- During the 12-week randomized treatment period, patients record their IBS-C symptoms with daily assessments using an eDiary (**Box**).
- Pediatric patients are assessed on site every 2 or 4 weeks (visits 3-6) for safety.
 - Assessments may include a physical exam, electrocardiogram, vital signs, height (at Screening and End of Study only) and clinical laboratory measurements.
 - Adverse events (AEs), concomitant medications, and compliance are evaluated at each visit.

Box. eDiary

The eDiary will collect information on daily bowel movement frequency, stool consistency, abdominal pain, and rescue medication usage during the screening period and the randomized treatment period.

Example questions include:

- *How would you rate your worst abdominal pain today? Please use the scale where 0 represents no abdominal pain and 10 represents very severe abdominal pain.*

None **0** 1 **2** 3 **4** 5 **6** 7 **8** 9 **10** Very Severe

- *How many bowel movements have you had today?*
 For each bowel movement:
 - *Please enter the time of the bowel movement using the 12-hour AM/PM format.*
 - *Refer to the Children's modified Bristol Stool Form Scale (mBSFS) given to you. Please enter the number that best describes the consistency of each bowel movement following the scale:*

1. for rabbit droppings: separate hard lumps, like nuts (hard to pass)
2. for bunch of grapes: sausage-shaped but lumpy
3. for corn on cob: like a sausage but with cracks on the surface
4. for sausage: like a sausage or snake, smooth and soft
5. for chicken nuggets: soft blobs with clear-cut edges
6. for porridge: fluffy pieces with ragged edges, a mushy stool (porridge)
7. for gravy: watery, no solids pieces, entirely liquid

- *Have you taken any rescue medication since yesterday? If yes:*
 - *Please enter the date you took the rescue medication, using the 8-digit format: 2 digits for the month, 2 digits for the day, and 4 digits for the year.*
 - *Please enter the time you took the rescue medication, using a 12-hour AM/PM format.*

Results

- As of May 26, 2023, 42 patients have been screened, 23 randomized, 23 treated, and 3 have completed the study. There were 12 screen failures and 2 early dropouts (**Figure 1**).
 - The 3 patients who completed the study entered the open-label extension study (NCT05905926).
- Baseline demographics are shown in **Table 2** for the 23 treated patients (ie, the safety analysis set).
- An interim summary of treatment-emergent adverse events (TEAEs) for the safety analysis set is shown in **Table 3**.
- No serious TEAEs were reported.
- All TEAEs were resolved and considered unrelated to study drug except diarrhea.

Table 2. Baseline Demographics (Safety Analysis Set)

Baseline demographics (N=23)	
Age, median (range), years	14.5 (12-17)
Female, n (%)	14 (60.9)
Race, n (%)	
White	20 (87.0)
Other	2 (8.7)
Multiple	1 (4.3)
Not Hispanic or Latino, n (%)	10 (43.5)
IBS-C duration at randomization (n=22), median (range), years	5.765 (0.05-17.26)

% = n/N*100. IBS-C, Irritable bowel syndrome with constipation.

Figure 1. Study Flow Chart (Consort Diagram)

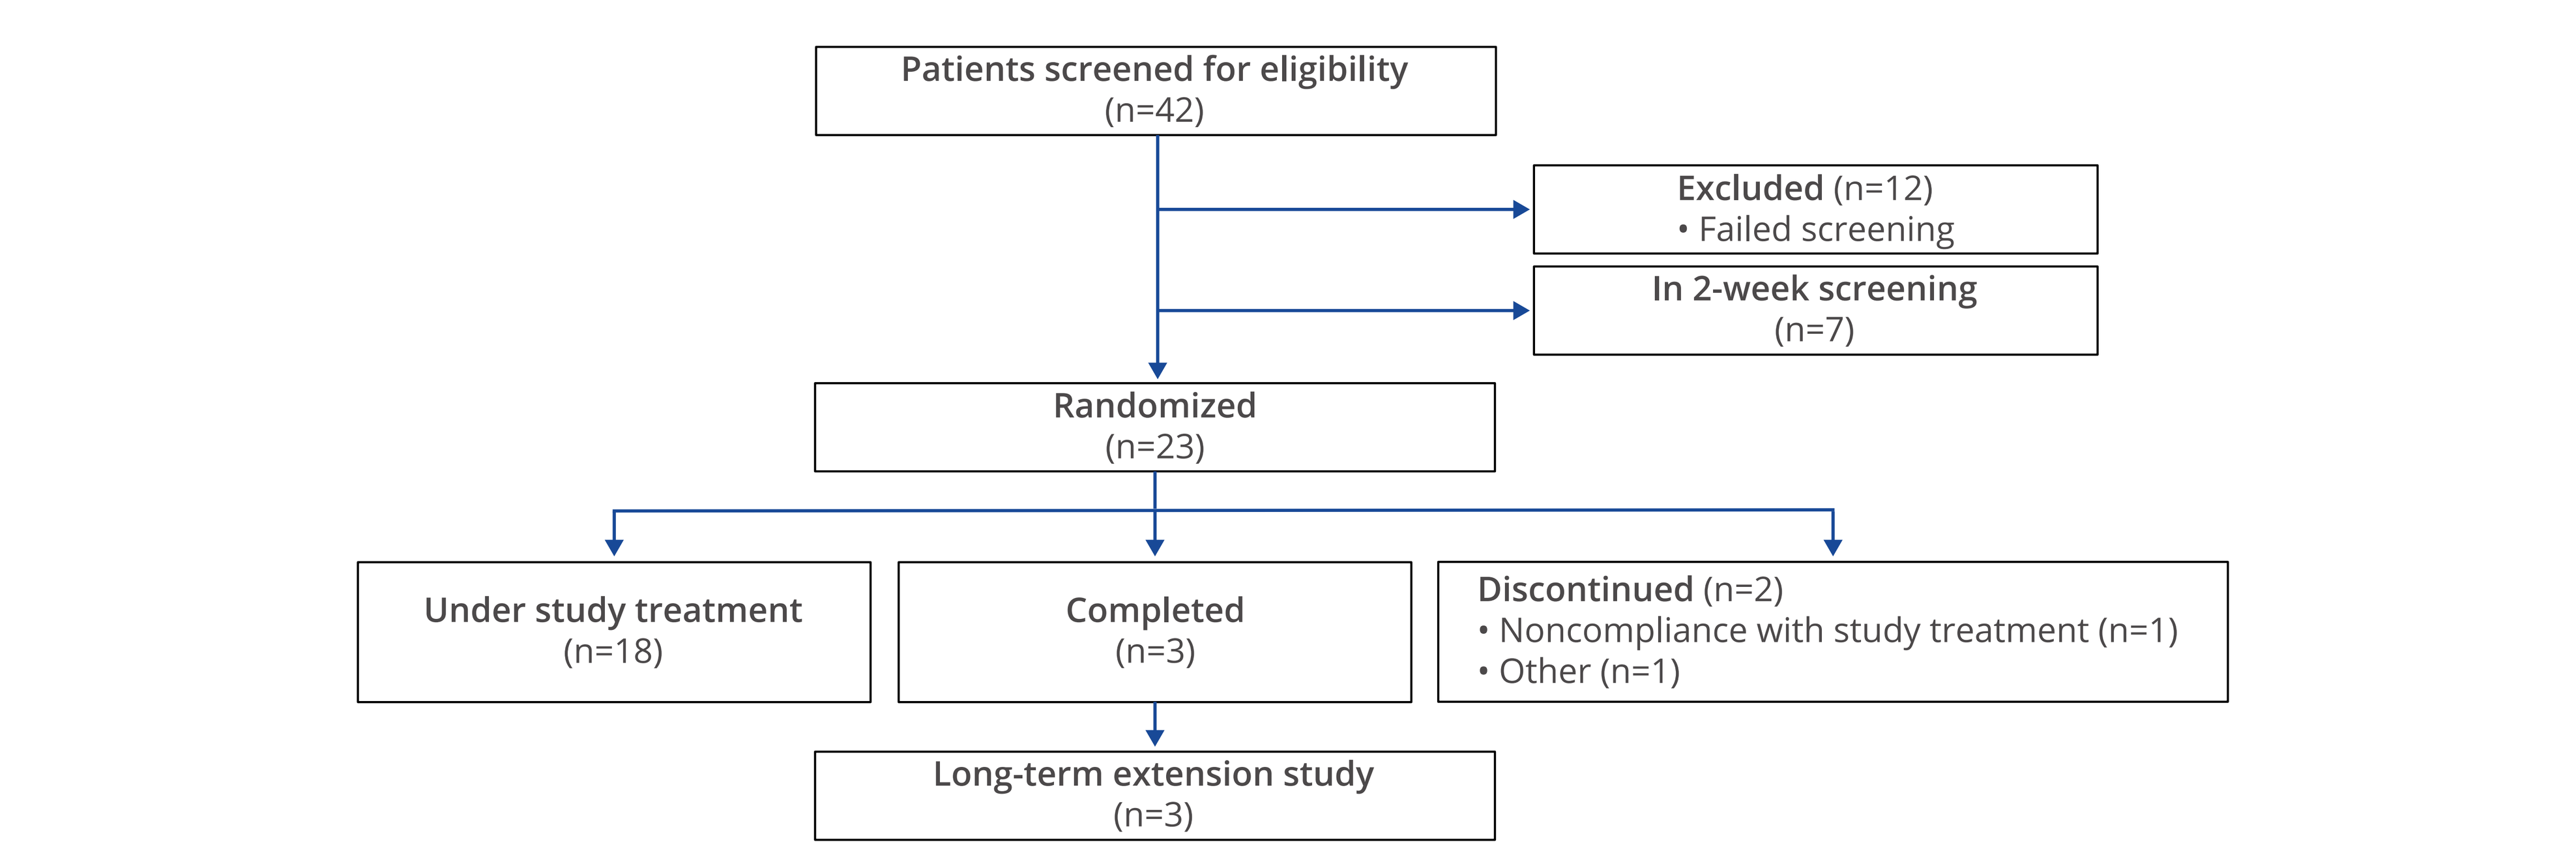
Patients screened for eligibility (n=42)
Excluded (n=12)
• Failed screening
In 2-week screening (n=7)
Randomized (n=23)
Under study treatment (n=18)
Completed (n=3)
Discontinued (n=2)
• Noncompliance with study treatment (n=1)
• Other (n=1)
Long-term extension study (n=3)

Table 3. Treatment-Emergent Adverse Events (Safety Analysis Set)

Treatment-emergent adverse events (N=23)	
Patients with any	**n (%)**
TEAE	6 (26.1)
Serious TEAE	0
Severe TEAE	2 (8.7)
Drug-related TEAE[a]	2 (8.7)
TEAE leading to study drug discontinuation	0
Reported term	
Diarrhea/loose stool	3 (13.0)
Abdominal pain	1 (4.3)
Anal irritation	1 (4.3)
Anal itching	1 (4.3)
Gastroenteritis	1 (4.3)
Headache	1 (4.3)
Runny nose	1 (4.3)

% = n/N*100. TEAE, treatment-emergent adverse event.
[a]All events that were considered probably or possibly related to study drug were mild in severity.

Conclusions

- Tenapanor is being investigated as a treatment for IBS-C in pediatric patients.
- The R-ALLY study is currently underway and enrolling pediatric patients (aged ≥12 and <18 years) with IBS-C to assess the safety and efficacy of tenapanor when administered bid for 12 weeks.
- The preliminary blinded safety results of this study are consistent with those from the previous studies of tenapanor in adult patients with IBS-C.
- To date, no unexpected safety concerns have been observed.

Disclosures

Jonathan Markowitz and Mihaela Ringheanu have no commercial interests or conflicts of interest to declare.

Karishma Raju, Yang Yang, and Susan Edelstein are employees of Ardelyx, Inc.

Acknowledgments

Medical writing support for the development of this poster, under the direction of the authors, was provided by Ashfield MedComms, an Inizio company, and funded by Ardelyx, Inc.

References

1. Lacy BE, et al. *Am J Gastroenterol.* 2021;116:17-44.
2. Devanarayana NM, et al. *World J Gastroenterol.* 2018;24:2211-35.
3. Enck P, et al. *Nat Rev Dis Primers.* 2016;2:16014.
4. Robin SG, et al. *J Pediatr.* 2018;195:134-39.
5. Scarpato E, et al. *Clin Gastroenterol Hepatol.* 2018;16:870-6.
6. Self MM, et al. *Clin Gastroenterol Hepatol.* 2014;12:1468-73.
7. Spencer AG, et al. *Sci Transl Med.* 2014;6:227ra236.
8. Rosenbaum DP, et al. *Clin Drug Investig.* 2018;38:341-51.
9. IBSRELA. Prescribing information. Ardelyx, Inc.;2022.
10. IBSRELA. Product monograph. Knight Therapeutics, Inc.;2023.
11. Johansson S, et al. *Clin Exp Nephrol.* 2017;21:407-16.
12. Wang J, et al. Poster presented at: Digestive Disease Week; June 2-5, 2018; Washington, DC.
13. Li Q, et al. Poster presented at: World Congress of Gastroenterology at The American College of Gastroenterology Annual Scientific Meeting; October 13–18, 2017; Orlando, FL, USA.
14. Chey WD, et al. *Am J Gastroenterol.* 2020;115:281-93.
15. Chey WD, et al. *Am J Gastroenterol.* 2021;116:1294-303.
16. Lembo A, et al. Poster presented at: ACG Annual Scientific Meeting October 5-10, 2018; Philadelphia, PA.

Dr. Markowitz can be contacted for further information on this study at jonathan.markowitz@prismahealth.org.

Copies of this poster obtained through the quick response (QR) code are for personal use only and may not be reproduced without permission from the authors.

North American Society For Pediatric Gastroenterology, Hepatology, and Nutrition (NASPGHAN 2023); October 4-7, 2023; San Diego, CA, USA